Genome-wide CRISPRi screen identifies RPS27 as a novel synthetic lethal target for TP53 mutant cancers

Poster #2983

Stephen Paik, Yi Yu, Ashley H Choi, Samuel R Meier, Shangtao Liu, Binzhang Shen, Shanchuan Zhao, Xuewen Pan, Jannik N Andersen, and Teng Teng

INTRODUCTION

TP53 is a master tumor suppressor that regulates diverse cellular functions, including cell cycle progression, apoptosis, DNA damage repair and genome stability. Due to its critical role, TP53 is the most frequently mutated gene across human cancer and its loss often represents an early event in tumorigenesis. However, therapeutic strategies that specifically target TP53-mutant cancers remain limited.

To identify novel synthetic lethal targets for TP53 mutant cancers, we engineered and validated isogenic cell lines differing only in TP53 status and performed a genome wide CRISPR interference (CRISPRi) screen in paired A549 cells. The screen identified the small ribosome subunit protein RPS27 as the top dependency unique to TP53 knockout cells. Interestingly, this dependency was specific to the CRISPRi platform, as RPS27 did not score in a parallel CRISPR knockout (CRISPRko) screen. Further examination revealed that most CRISPRko gRNAs targeting RPS27 have multiple off-target sites with less than 3 mismatches. This likely causes general DNA-cutting-induced cytotoxicity that confounds true gene dependencies, which is avoided with the CRISPRi platform. Supporting this conclusion, re-analysis of the public DepMap/Achilles dataset showed that a single mismatch-free gRNA also demonstrate RPS27 dependency in TP53 mutant cell lines.

Mechanistically, TP53 drives the expression of RPS27L, a close paralog of RPS27. While TP53 wildtype cells express both RPS27L and RPS27, allowing for functional redundancy in ribosome assembly, TP53 mutant cells lack RPS27L expression and become largely dependent on RPS27 for ribosomal integrity and survival. Single gene validation confirmed that TP53 KO A549 cells are exquisitely sensitive to RPS27 knockdown and this dependency can be rescued by re-expression of either TP53, RPS27 or RPS27L. Together, these findings identify RPS27 as a novel synthetic lethal target for TP53 mutant cancers, revealing a previously unrecognized paralog-dependency within the ribosome and highlighting a potential therapeutic vulnerability.

RPS27 IS A TOP HIT IN TP53 ISOGENIC CRISPRi SCREEN

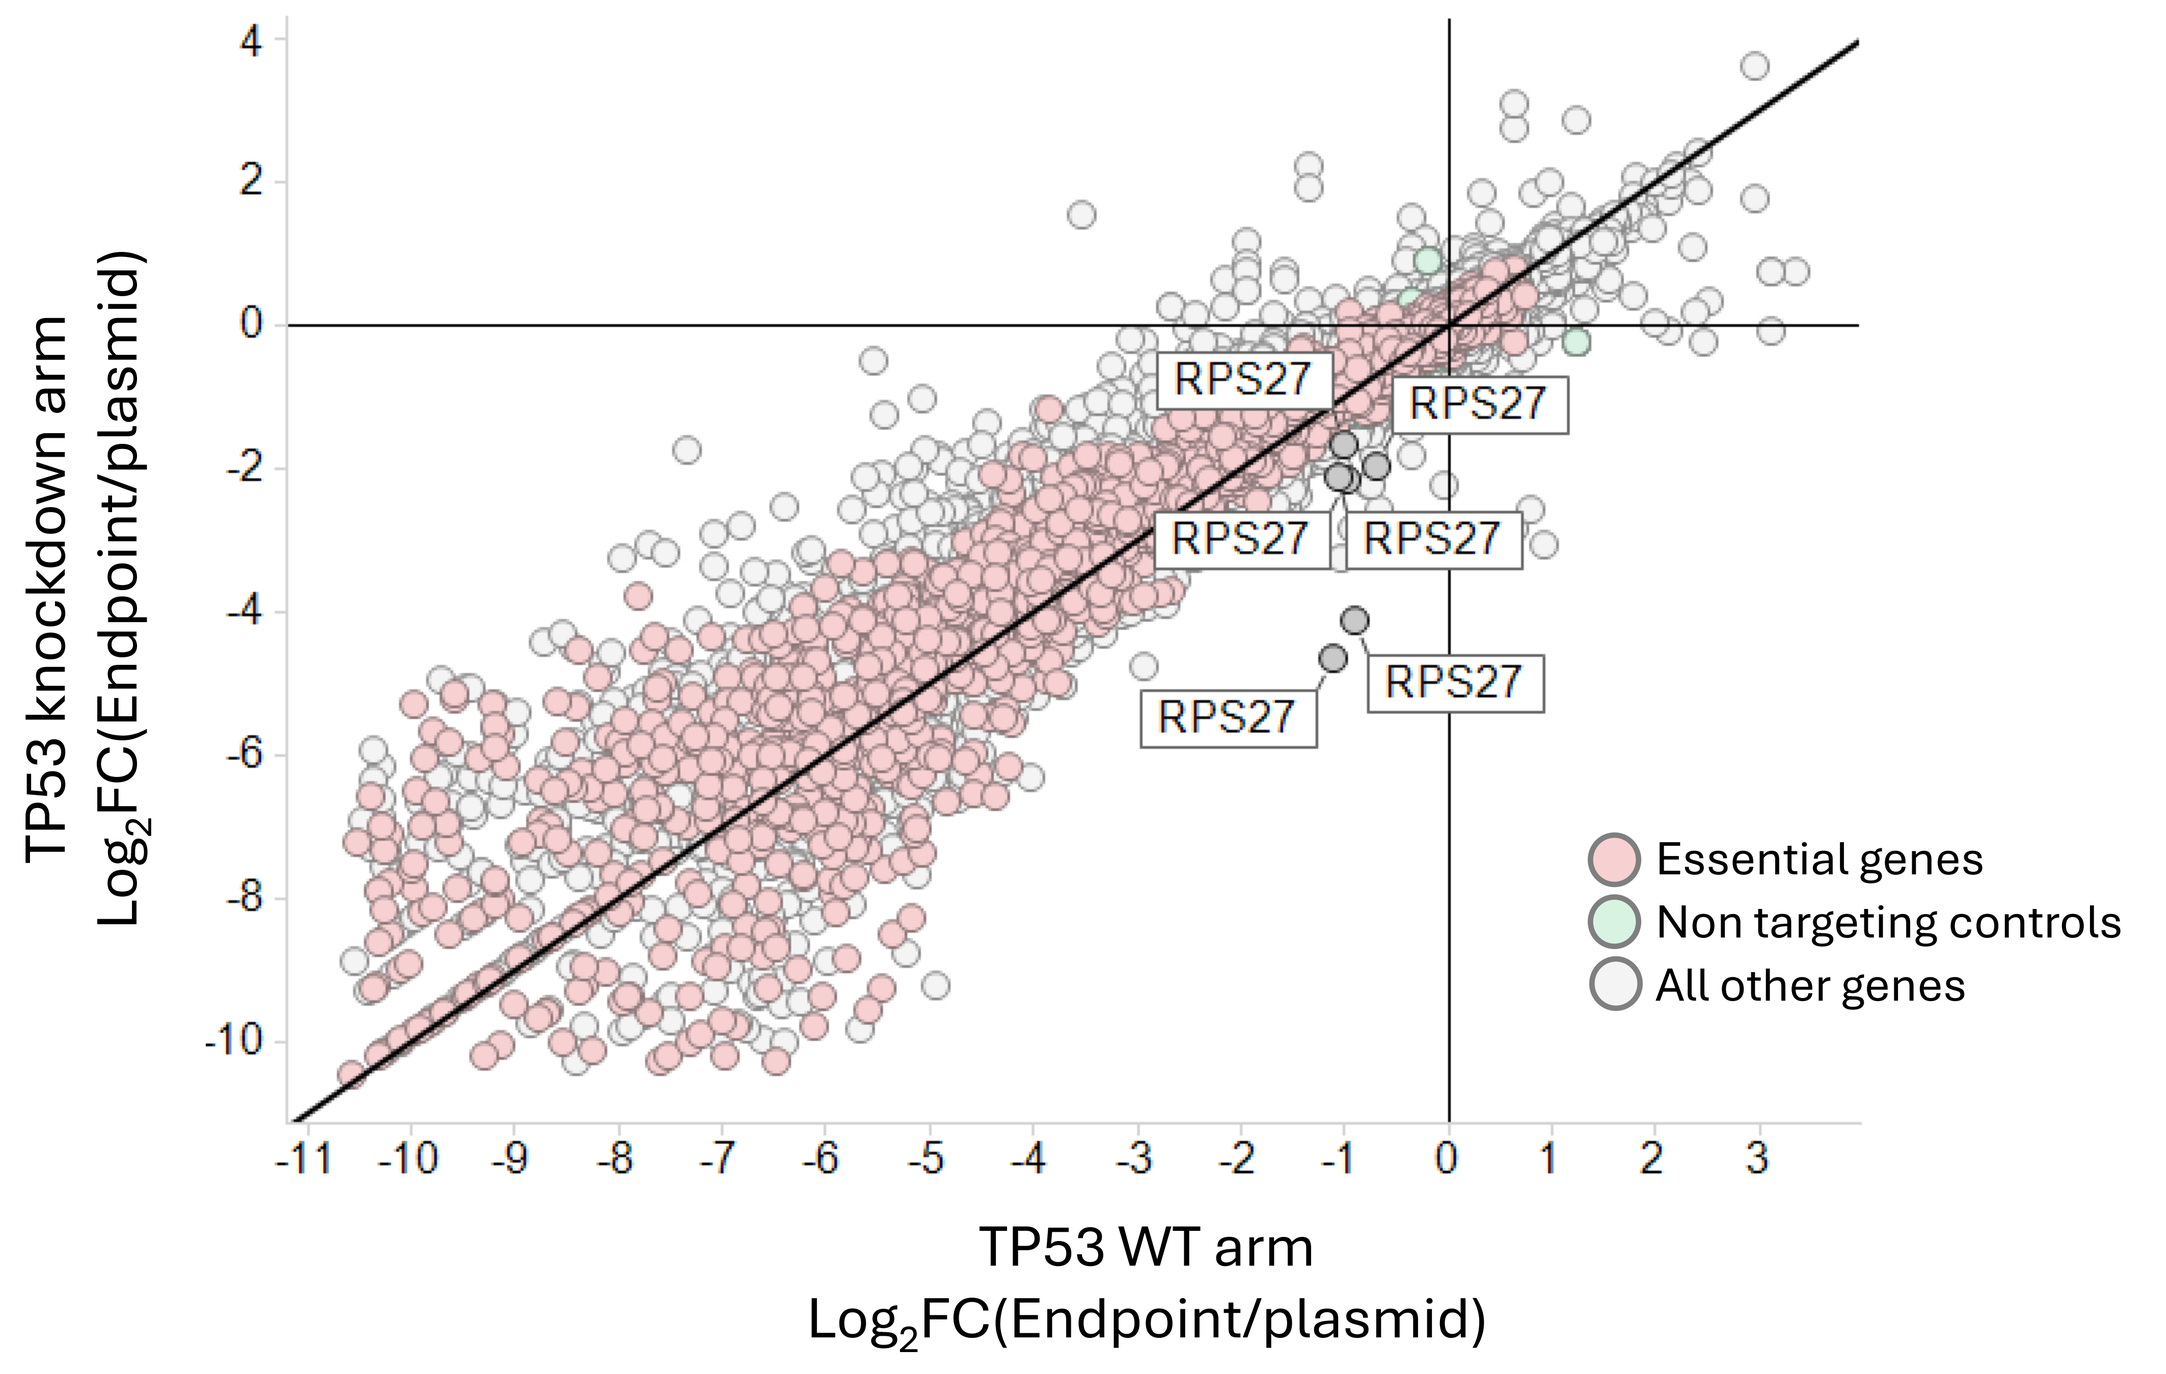
TP53 knockdown arm
Log_2FC(Endpoint/plasmid)
TP53 WT arm
Log_2FC(Endpoint/plasmid)
RPS27
Essential genes
Non targeting controls
All other genes

- Genome wide CRISPRi screen carried out in A549 TP53 WT and A549 TP53 knockdown isogenic pair cells
- RPS27 knockdown cells showed selective drop out only in TP53 knockdown arm

SINGLE GENE VALIDATION OF RPS27/TP53 SYNTHETIC LETHAL INTERACTION

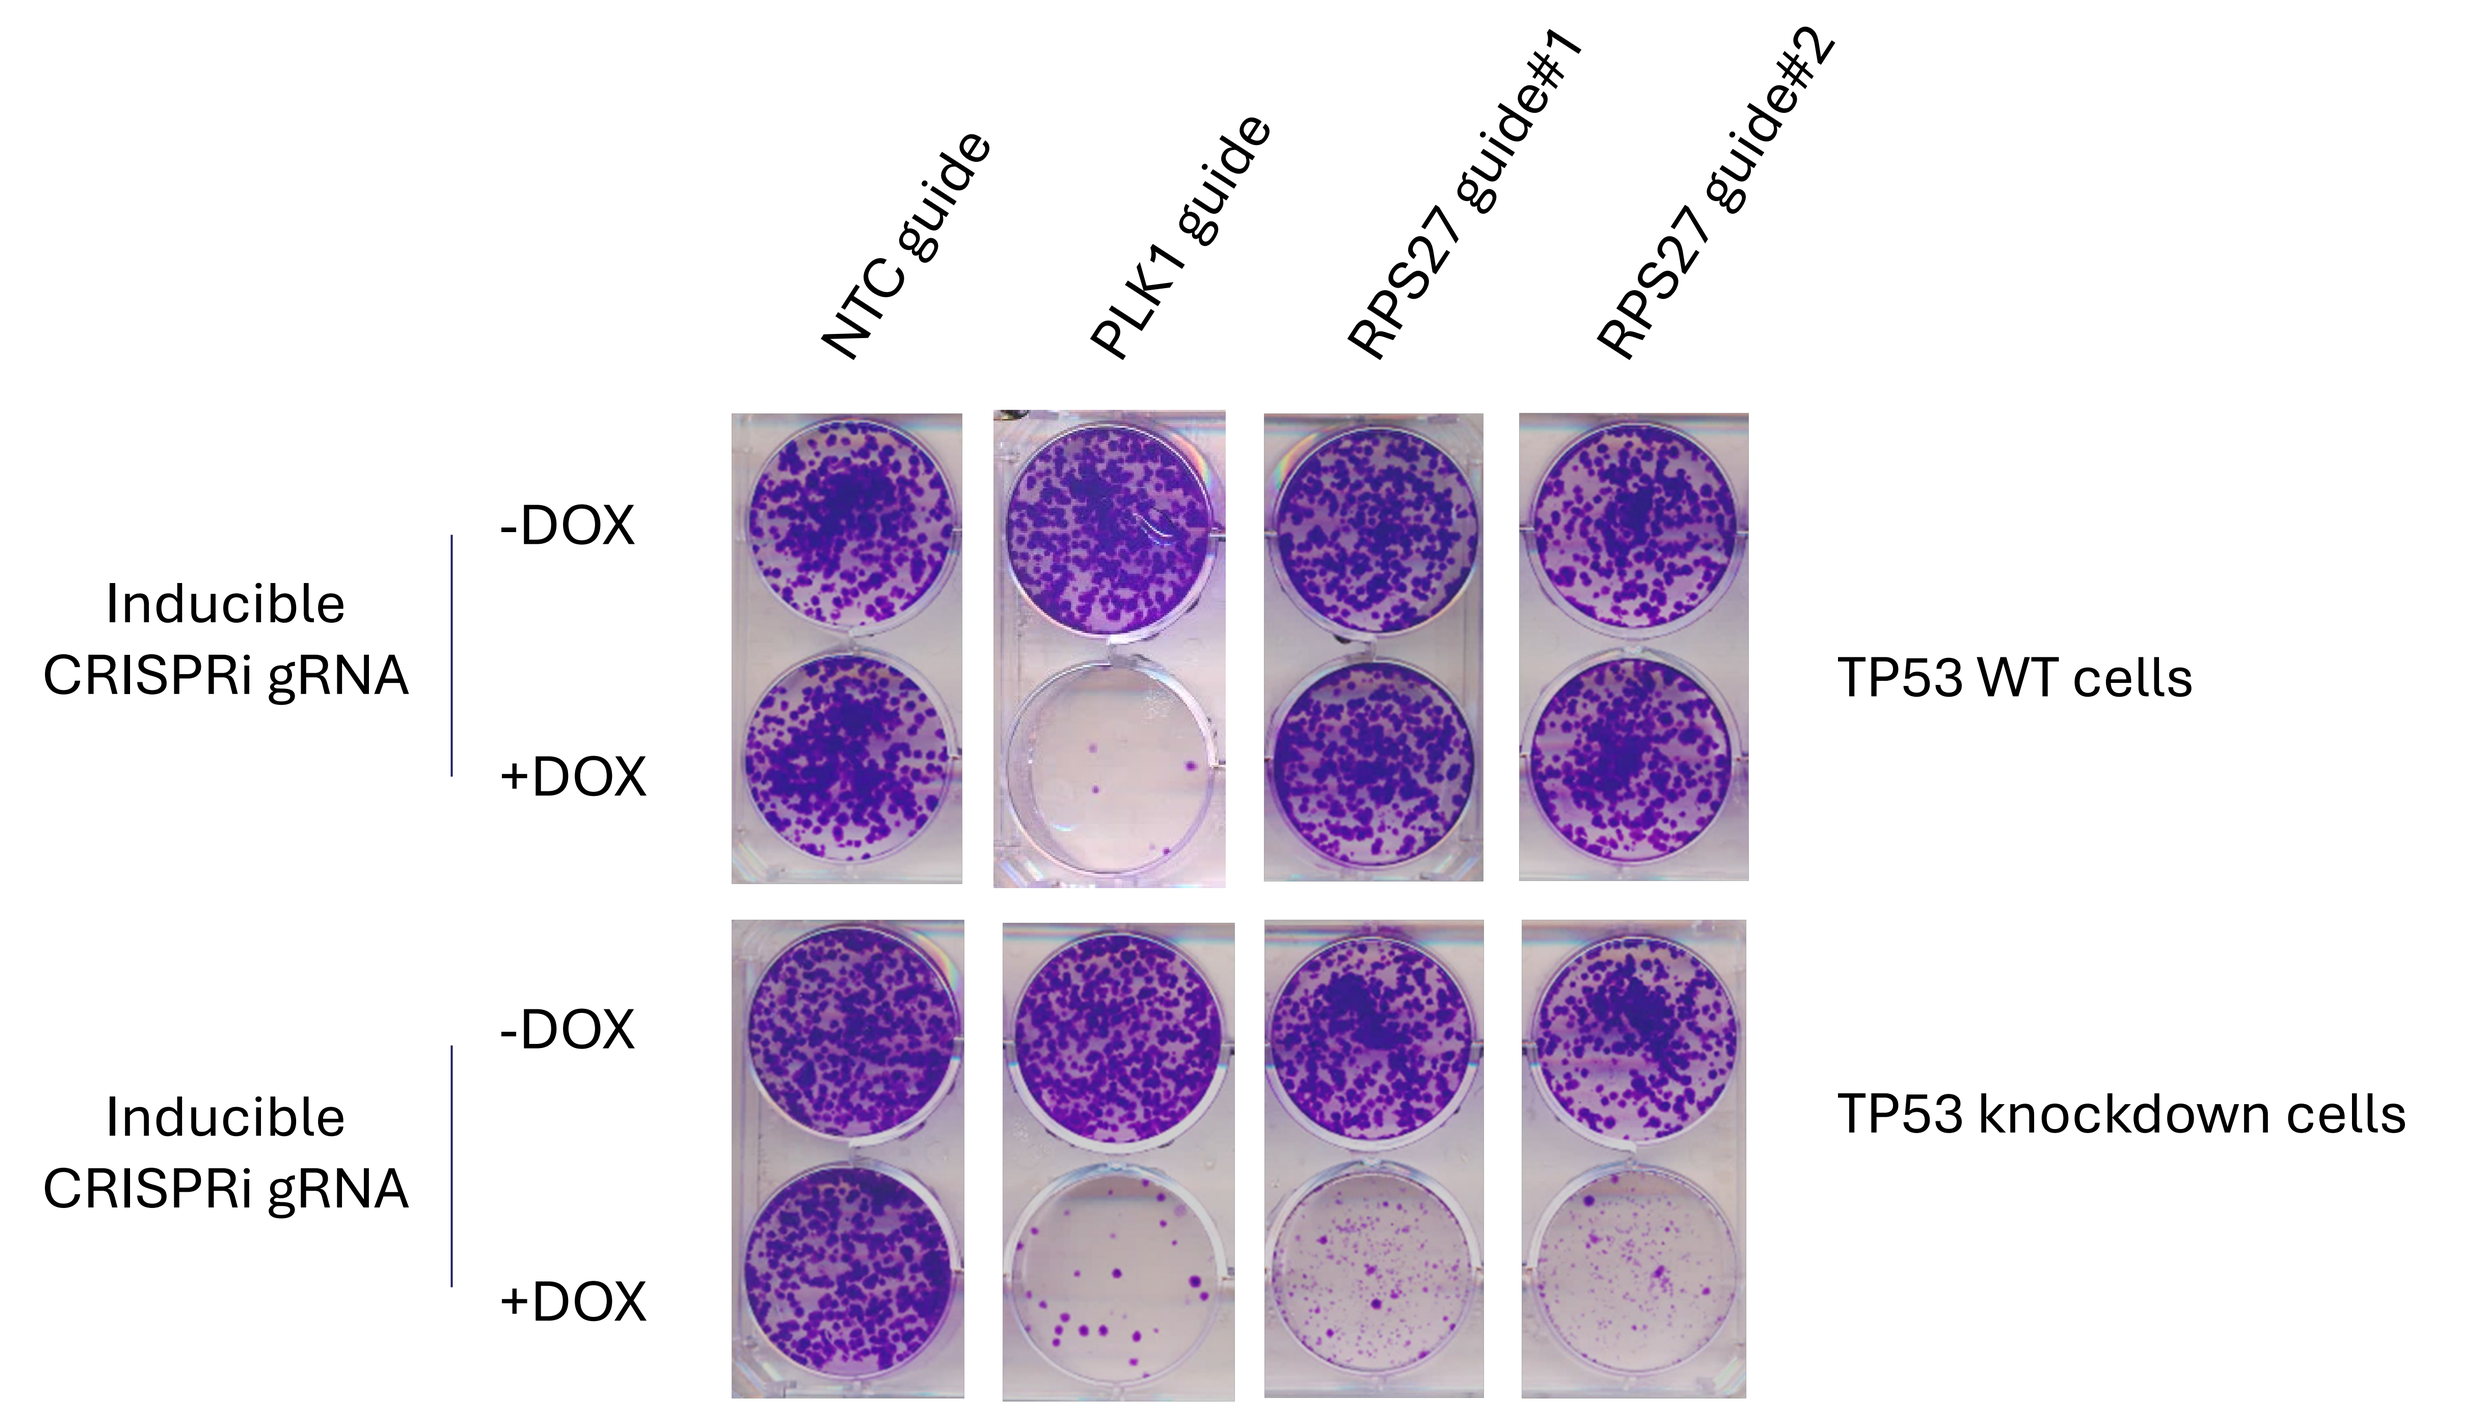
NTC guide
PLK1 guide
RPS27 guide#1
RPS27 guide#2
Inducible CRISPRi gRNA
-DOX
+DOX
TP53 WT cells
TP53 knockdown cells

- Colony formation assay validates RPS27 as a novel synthetic lethal target with TP53 loss using A549 isogenic cell line pair

TP53 RE-CONSTITUTION RESCUES RPS27 KNOCKDOWN INDUCED LETHALITY

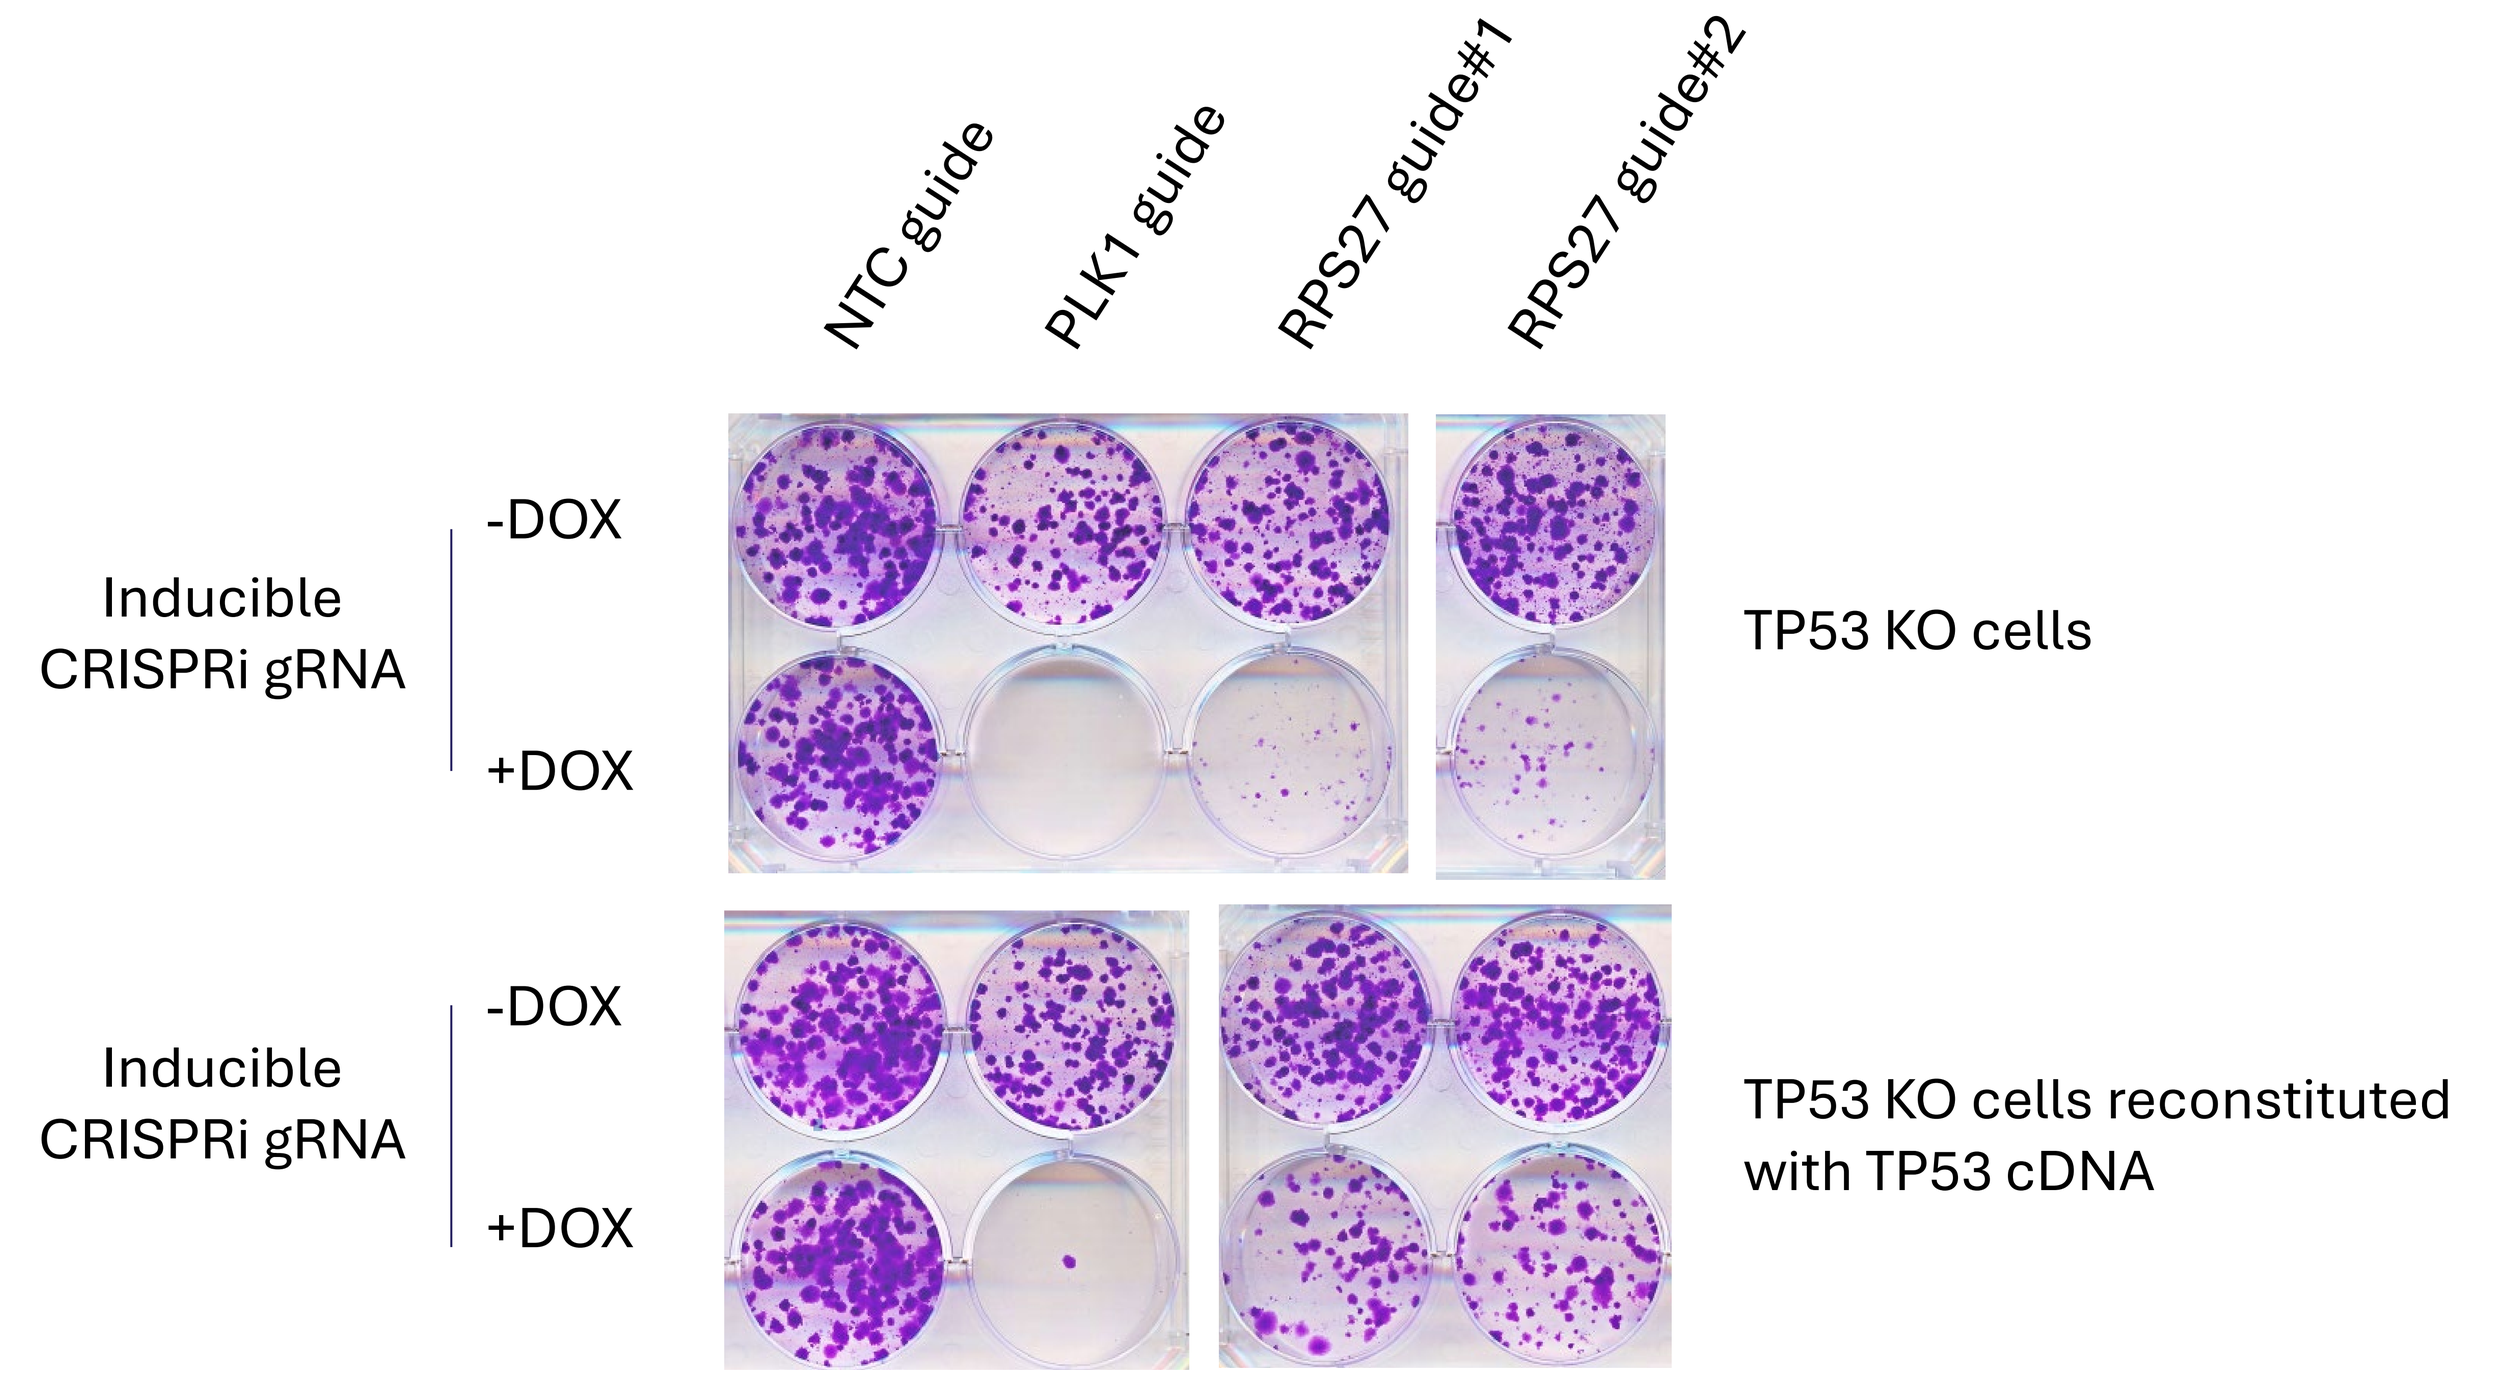
NTC guide
PLK1 guide
RPS27 guide#1
RPS27 guide#2
Inducible CRISPRi gRNA
-DOX
+DOX
TP53 KO cells
TP53 KO cells reconstituted with TP53 cDNA

- TP53 cDNA re-constitution rescues the RP27 dependent phenotype in A549 TP53 KO cells

TANGO FUNCTIONAL GENOMICS PLATFORM FOR NOVEL TARGET DISCOVERY

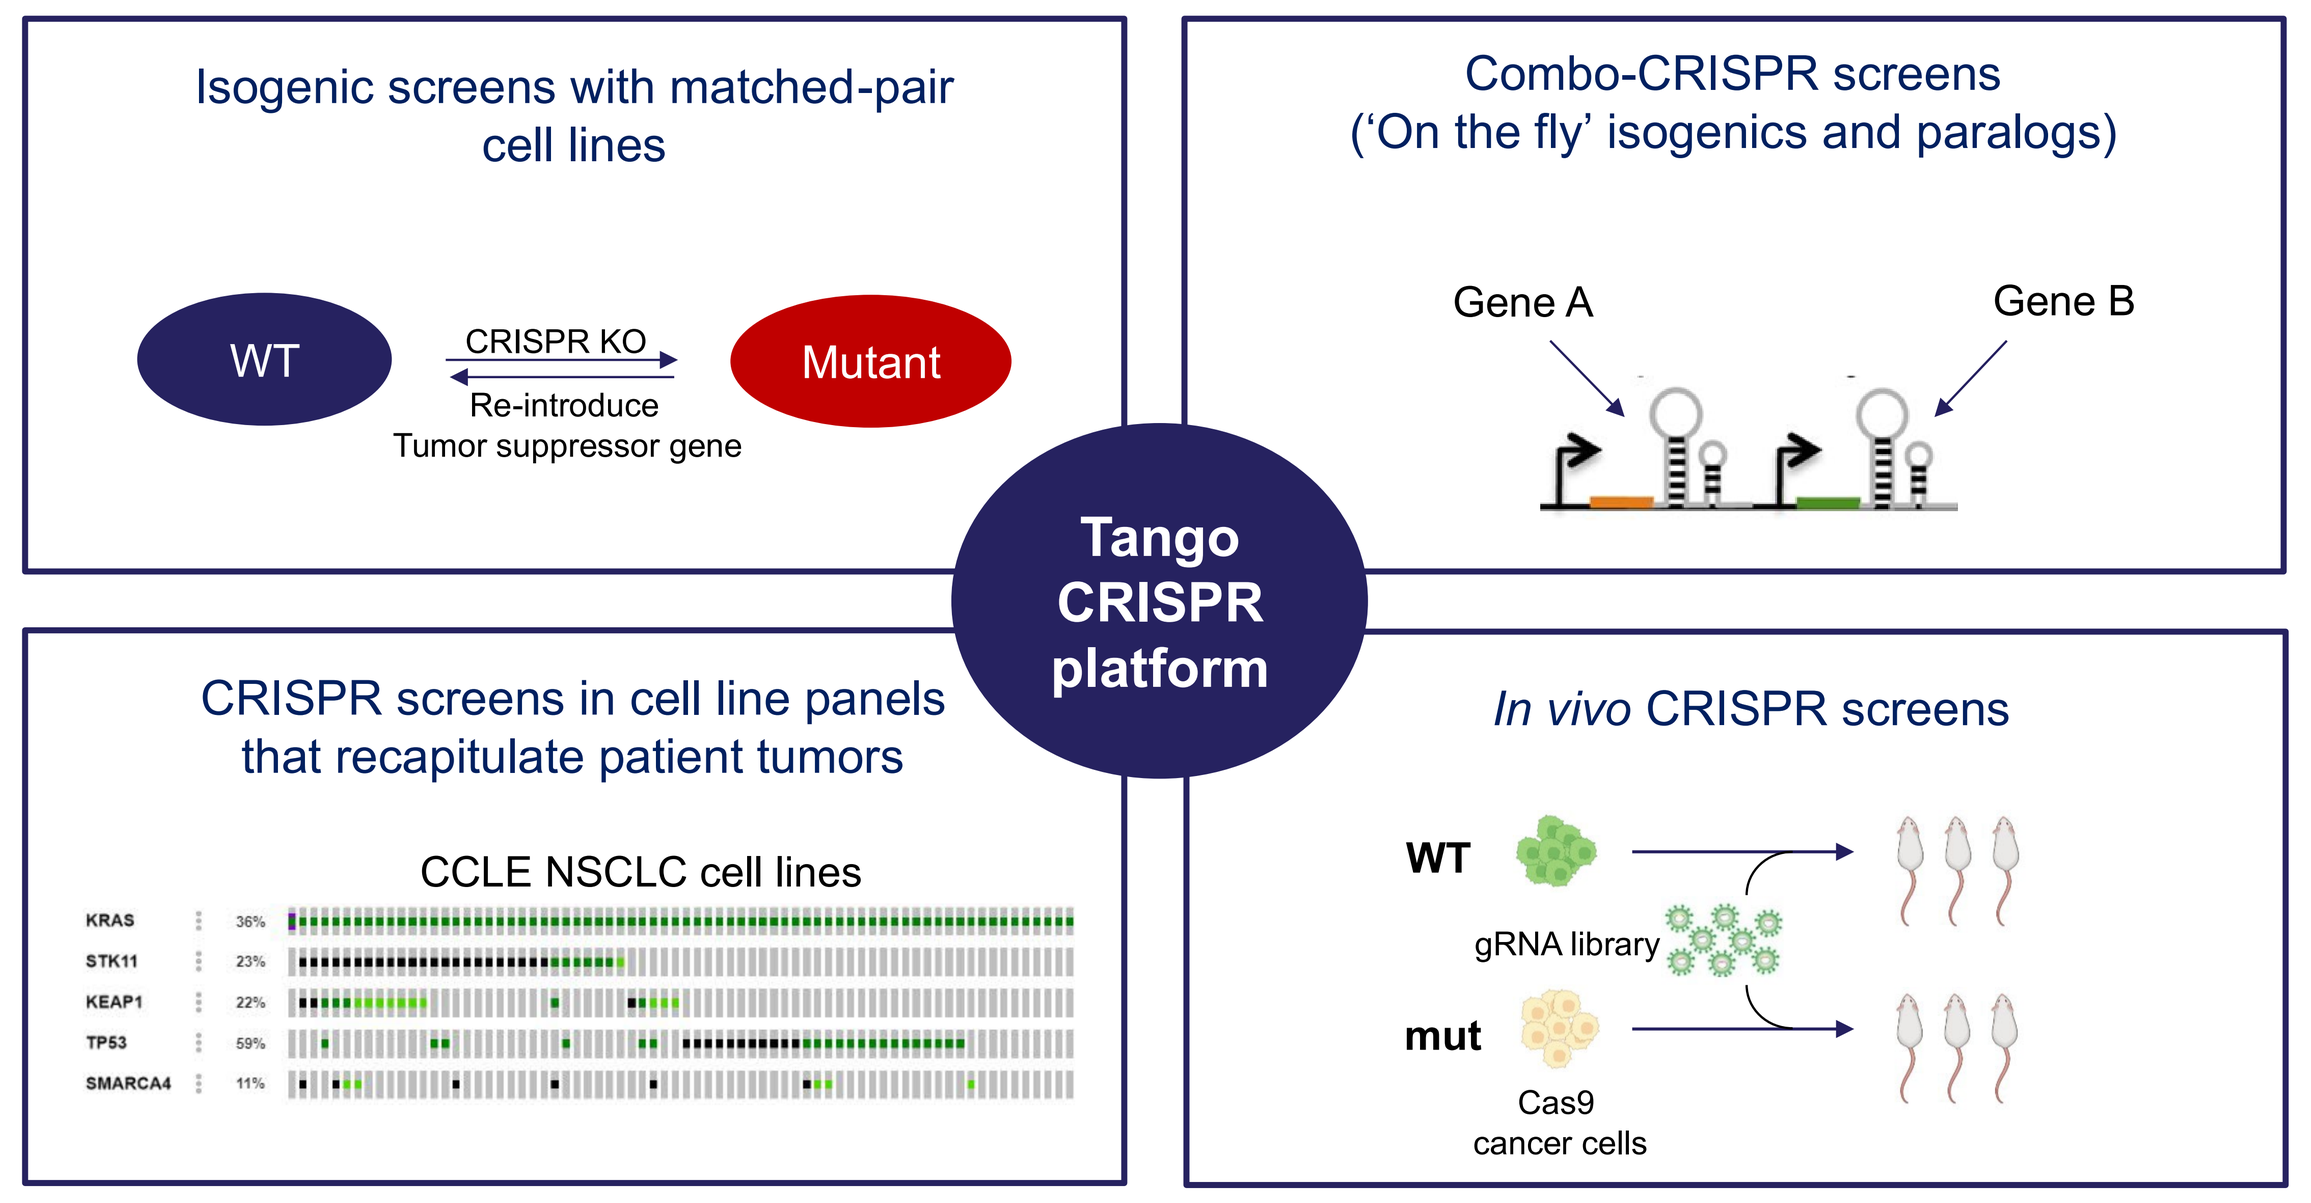
Isogenic screens with matched-pair cell lines
WT
CRISPR KO
Re-introduce
Tumor suppressor gene
Mutant
Combo-CRISPR screens
('On the fly' isogenics and paralogs)
Gene A
Gene B
Tango CRISPR platform
CRISPR screens in cell line panels that recapitulate patient tumors
CCLE NSCLC cell lines
KRAS
STK11
KEAP1
TP53
SMARCA4
In vivo CRISPR screens
WT
gRNA library
mut
Cas9 cancer cells

- Complementary platforms for novel target discovery and translational science

MECHANISM HYPOTHESIS OF THE RPS27/TP53 SYNTHETIC LETHAL INTERACTION

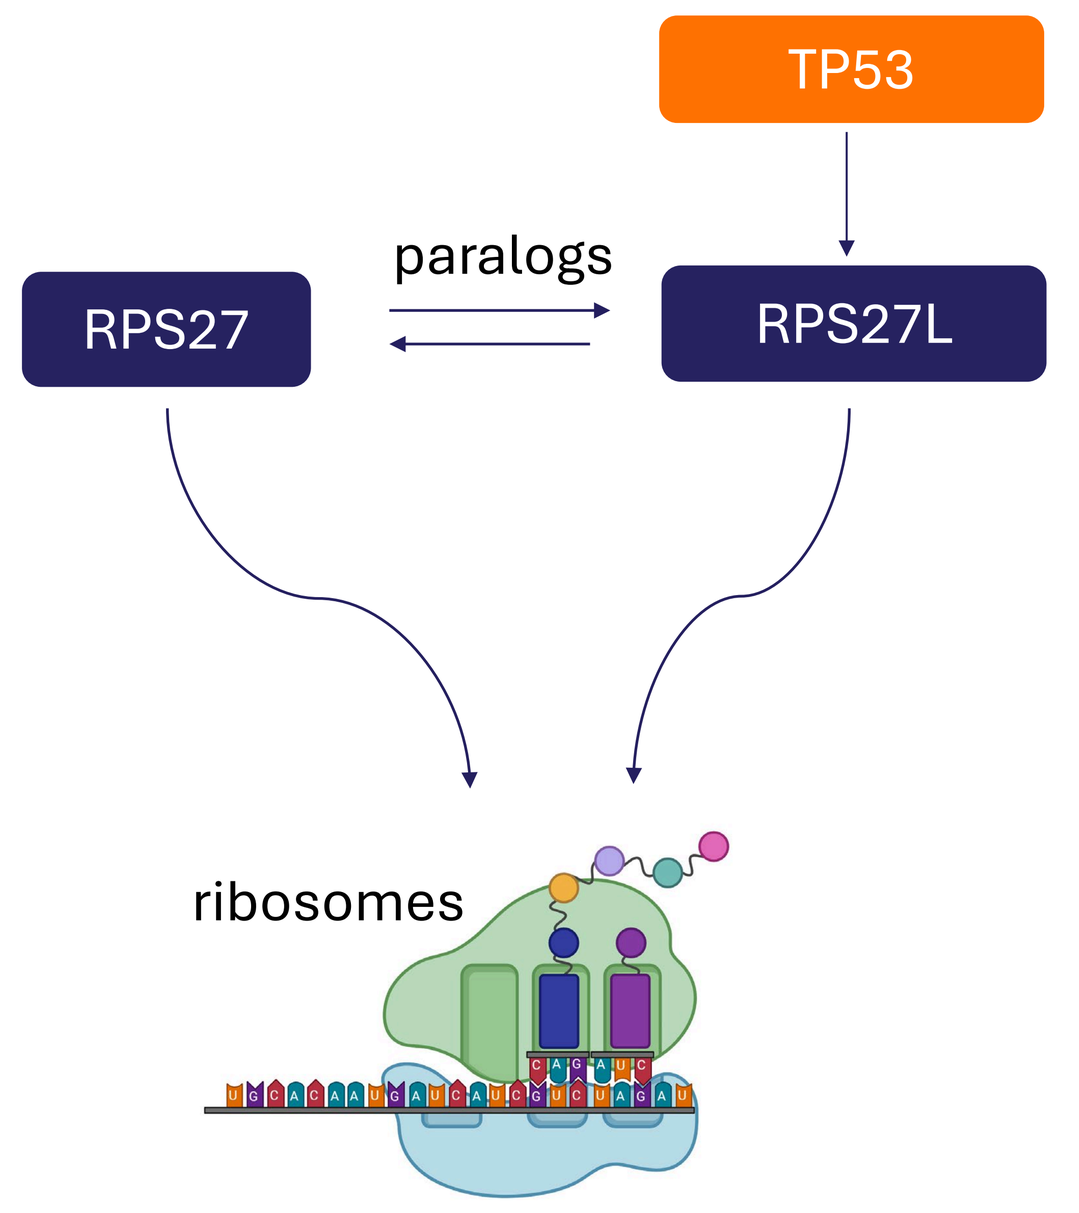
TP53
RPS27
paralogs
RPS27L
ribosomes

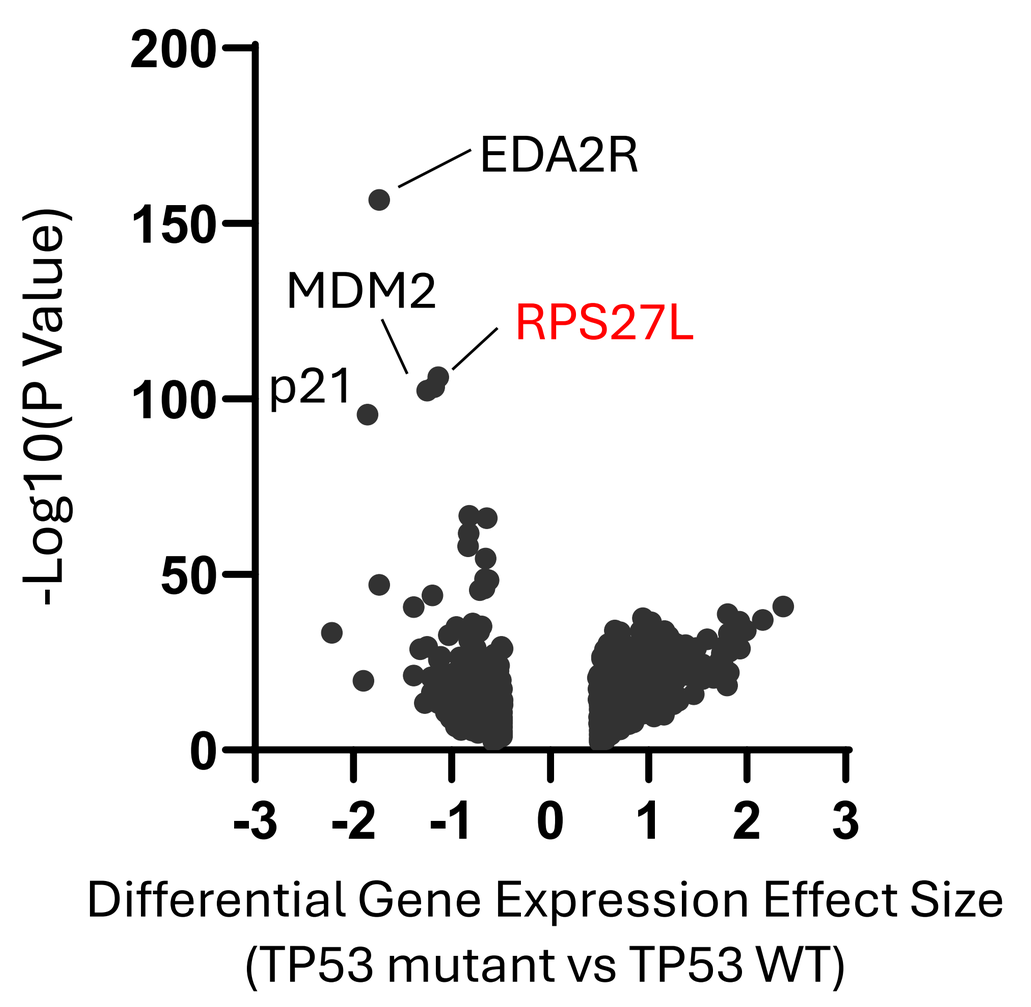
-Log10(P Value)
EDA2R
MDM2
RPS27L
p21
Differential Gene Expression Effect Size
(TP53 mutant vs TP53 WT)

- Therapeutic hypothesis of TP53 – RPS27 synthetic lethality
- RPS27L expression among the top correlated genes with TP53 mutation status

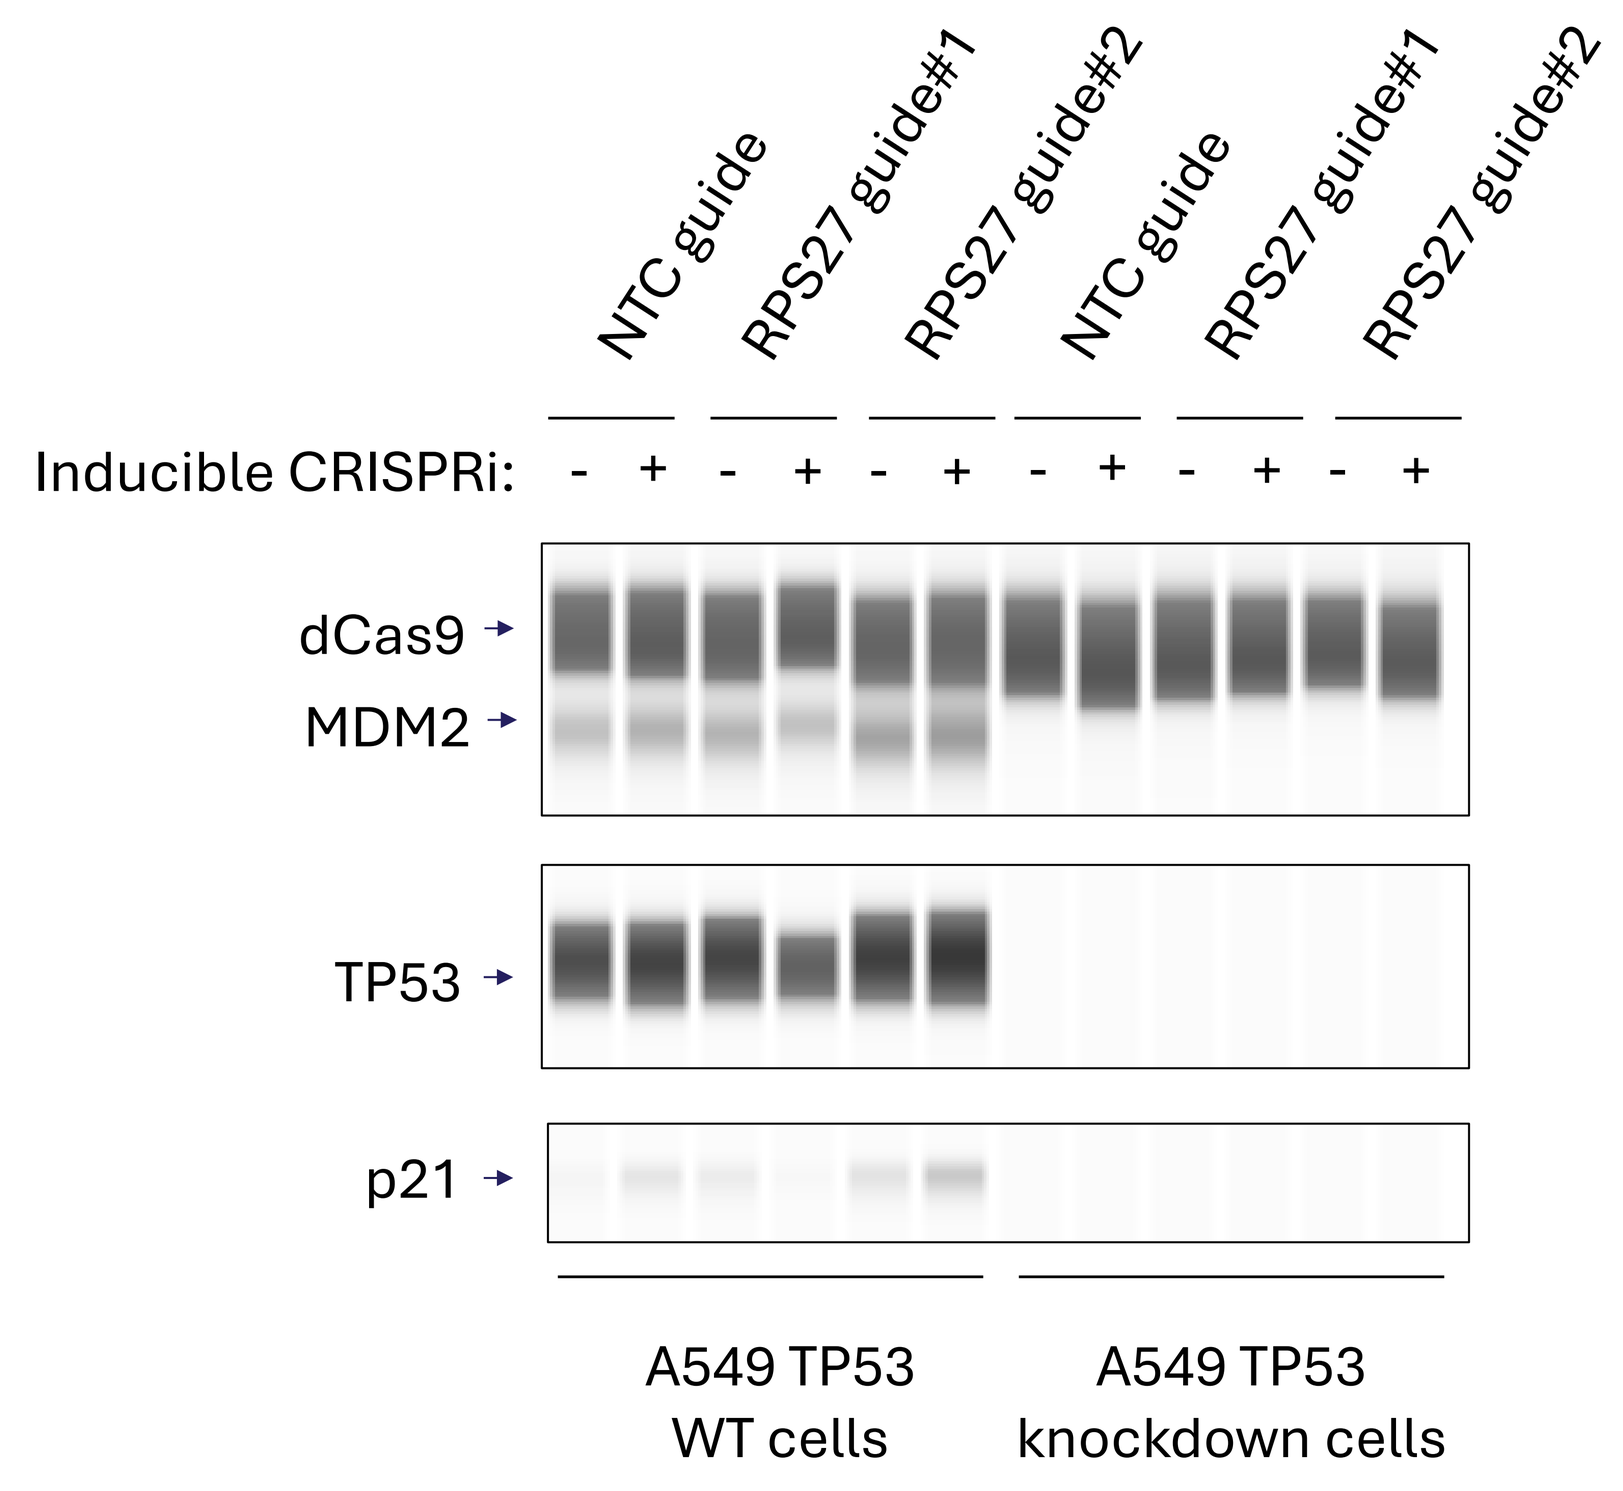
NTC guide
RPS27 guide#1
RPS27 guide#2
NTC guide
RPS27 guide#1
RPS27 guide#2
Inducible CRISPRi:
dCas9
MDM2
TP53
p21
A549 TP53 WT cells
A549 TP53 knockdown cells

- Parallel western blot analysis confirms TP53 status in the isogenic pair cell lines
- All samples treated with MDM2i to increase basal TP53, MDM2 and p21 expression

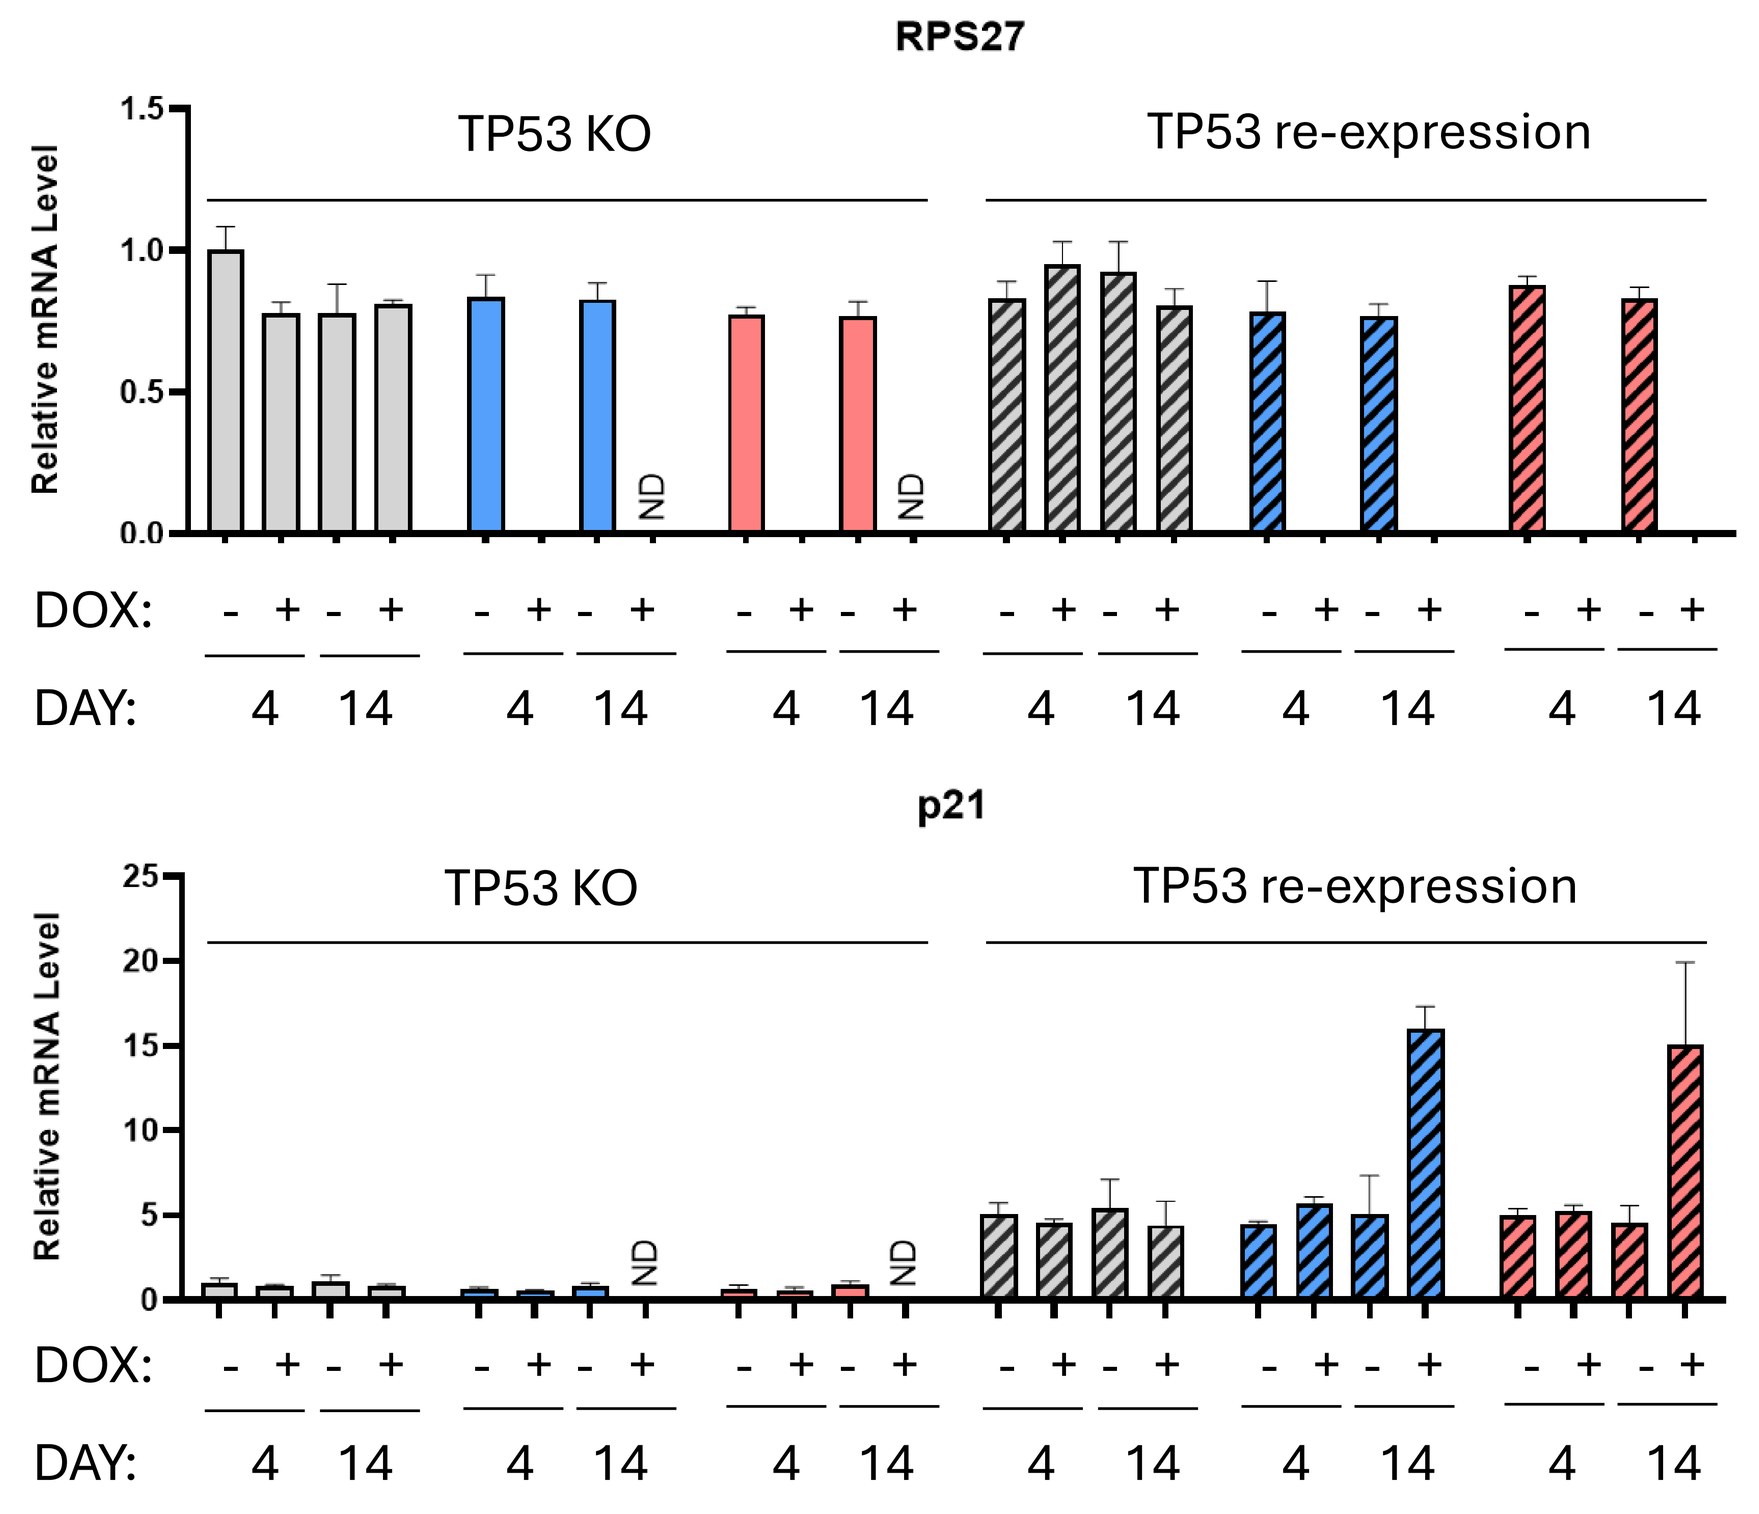
RPS27
Relative mRNA Level
TP53 KO
TP53 re-expression
ND
DOX:
DAY:
p21

- Parallel qRT-PCR analysis confirmed RPS27 knockdown and p21 induction

CRISPRko AND CRISPRi HAVE DIFFERENT ADVANTAGES

- Permanent mutagenesis
- More guides to choose from
- Cutting effect

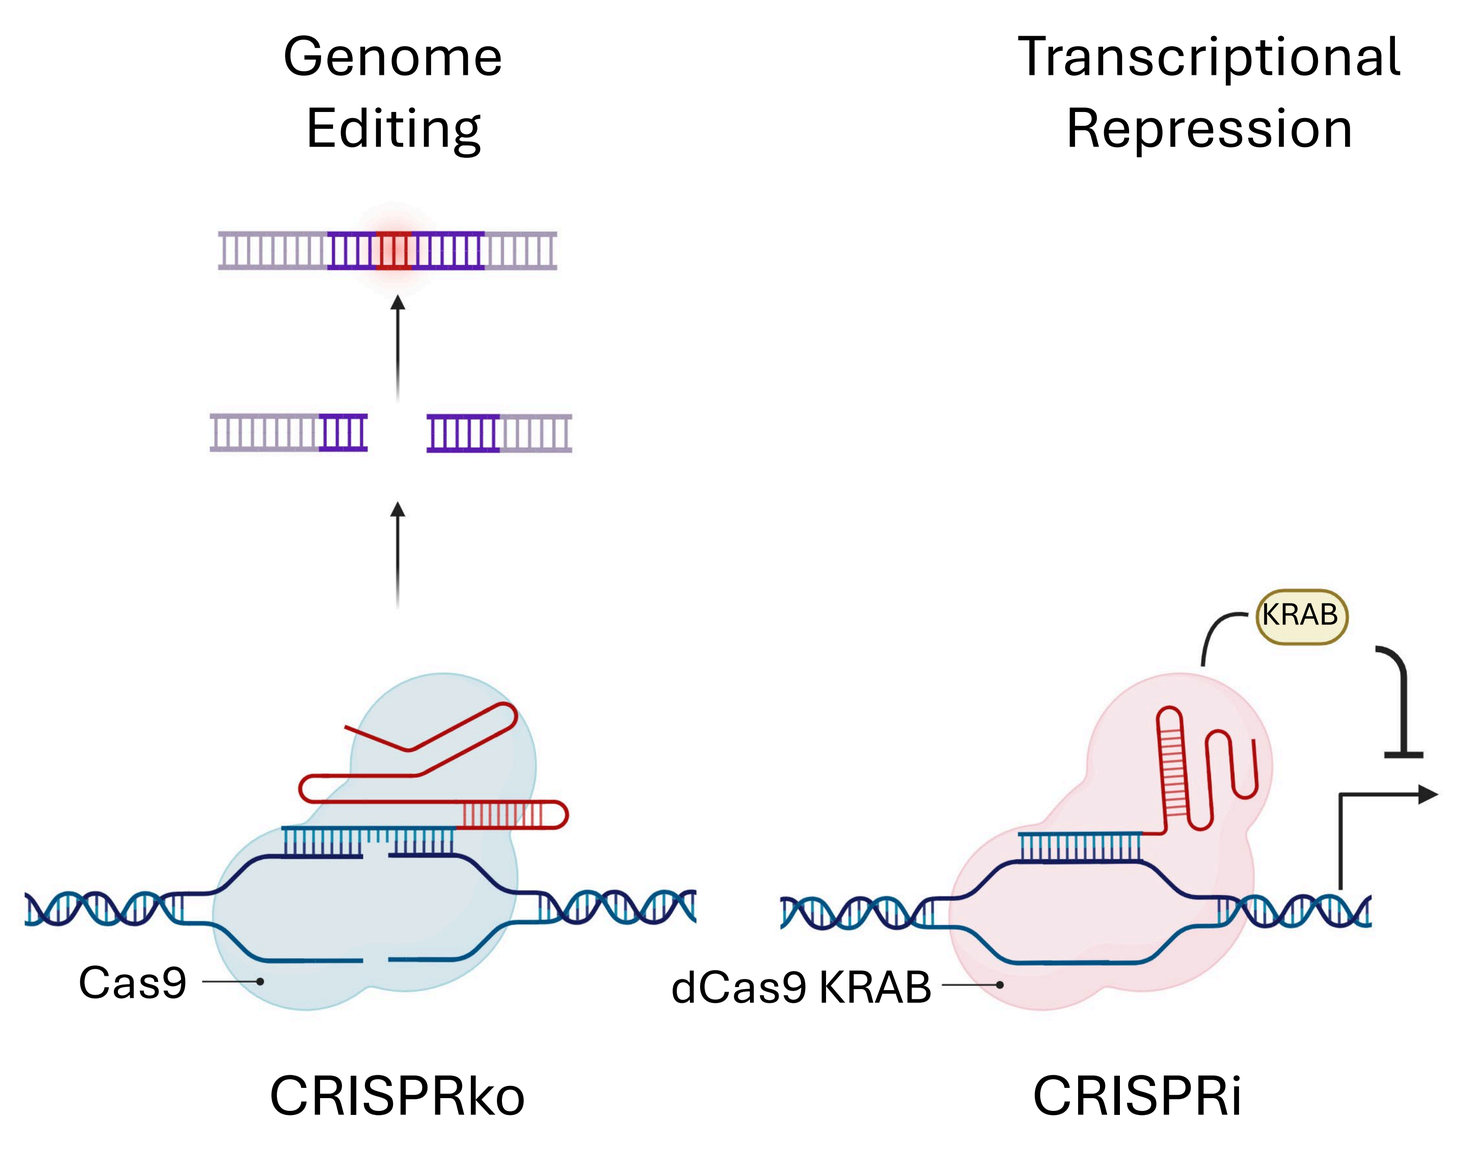
Genome Editing
Transcriptional Repression
KRAB
Cas9
dCas9 KRAB
CRISPRko
CRISPRi

- Need consistent repression
- Fewer guides to choose from
- Reversible
- No cutting effect

- Knock out vs. knock down offers different modes of genetic perturbation

COMMON CRISPRko gRNAS FOR RPS27 HAVE MULTIPLE OFF TARGET SITES

gRNA source	gRNA sequence	Off-target sites allowing 0 mismatch	Off-target sites allowing 1 mismatch	Off-target sites allowing 2 mismatch	Off-target sites allowing 3 mismatch
Brunello	ACAAAACTACCGTTTGTGCA	5	7	1	6
Brunello	GAAACACAAGAAGAAACGCC	8	7	3	21
Brunello	TTCATGGATGTGAAATGCCC	11	8	3	19
Brunello	TTGTGCATGGCTAAAGACCG	6	4	3	7
Avana	CCTTCATCCCTCTCCAGAAG	6	7	9	40
Avana	CTGTCCCTCTTAGCTCGCAA	0	0	0	4
Avana	GAAACACAAGAAGAAACGCC	8	7	3	21
Avana	GCATTTCACATCCATGAAGT	11	8	3	21

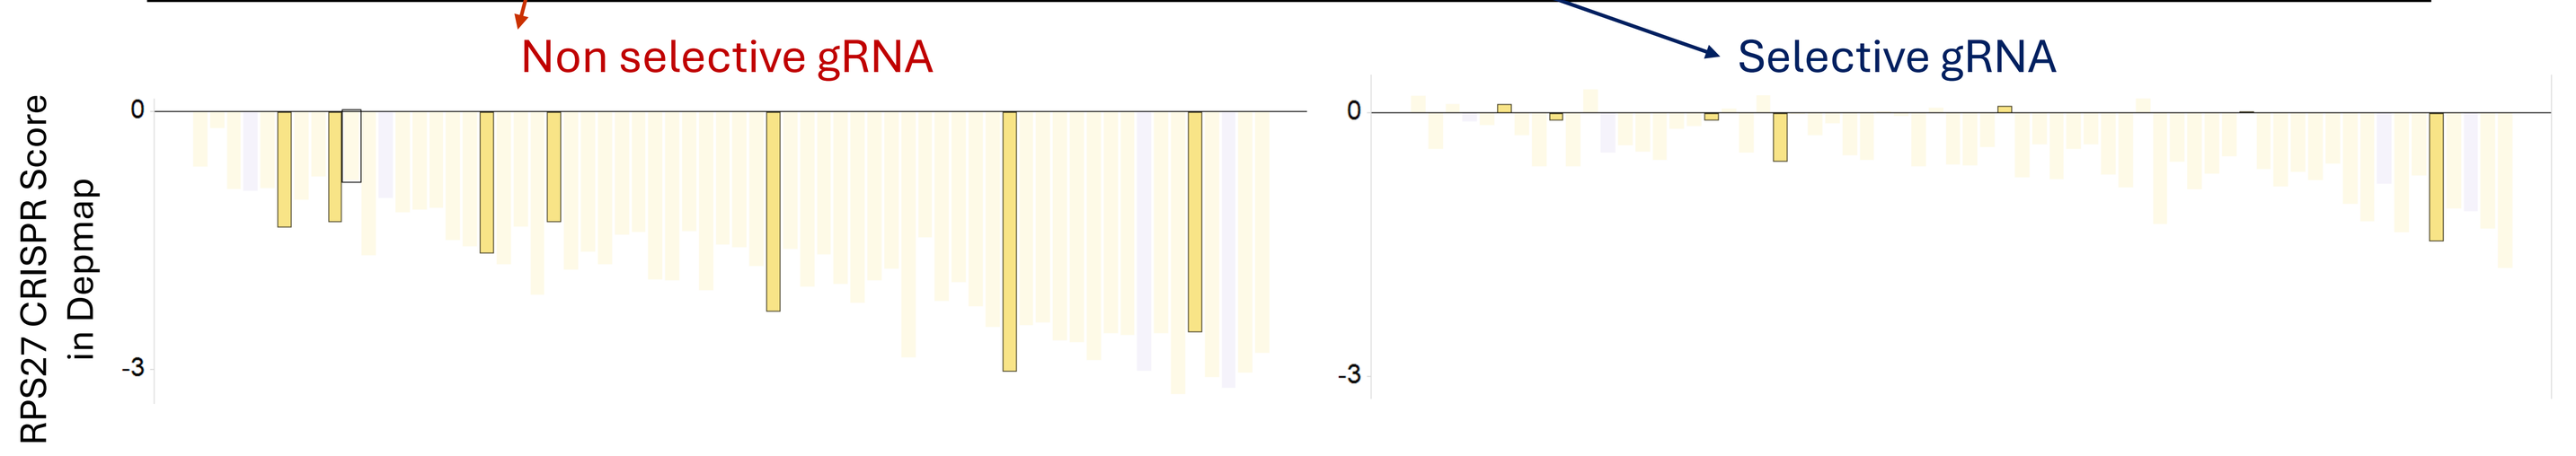
Non selective gRNA
Selective gRNA
RPS27 CRISPR Score in Depmap

Bold columns are TP53 WT lung cancer lines, selective RPS27 gRNA show context dependency for TP53 mutants whereas non-selective RPS27 gRNA shows pan-lethal phenotype

- CRISPRi platform overcomes the cutting effect limitation and noise from off-target sites

RPS27 OR RPS27L RE-EXPRESSION RESCUES RPS27 KNOCKDOWN INDUCED LETHALITY CELLS WITH TP53 LOSS

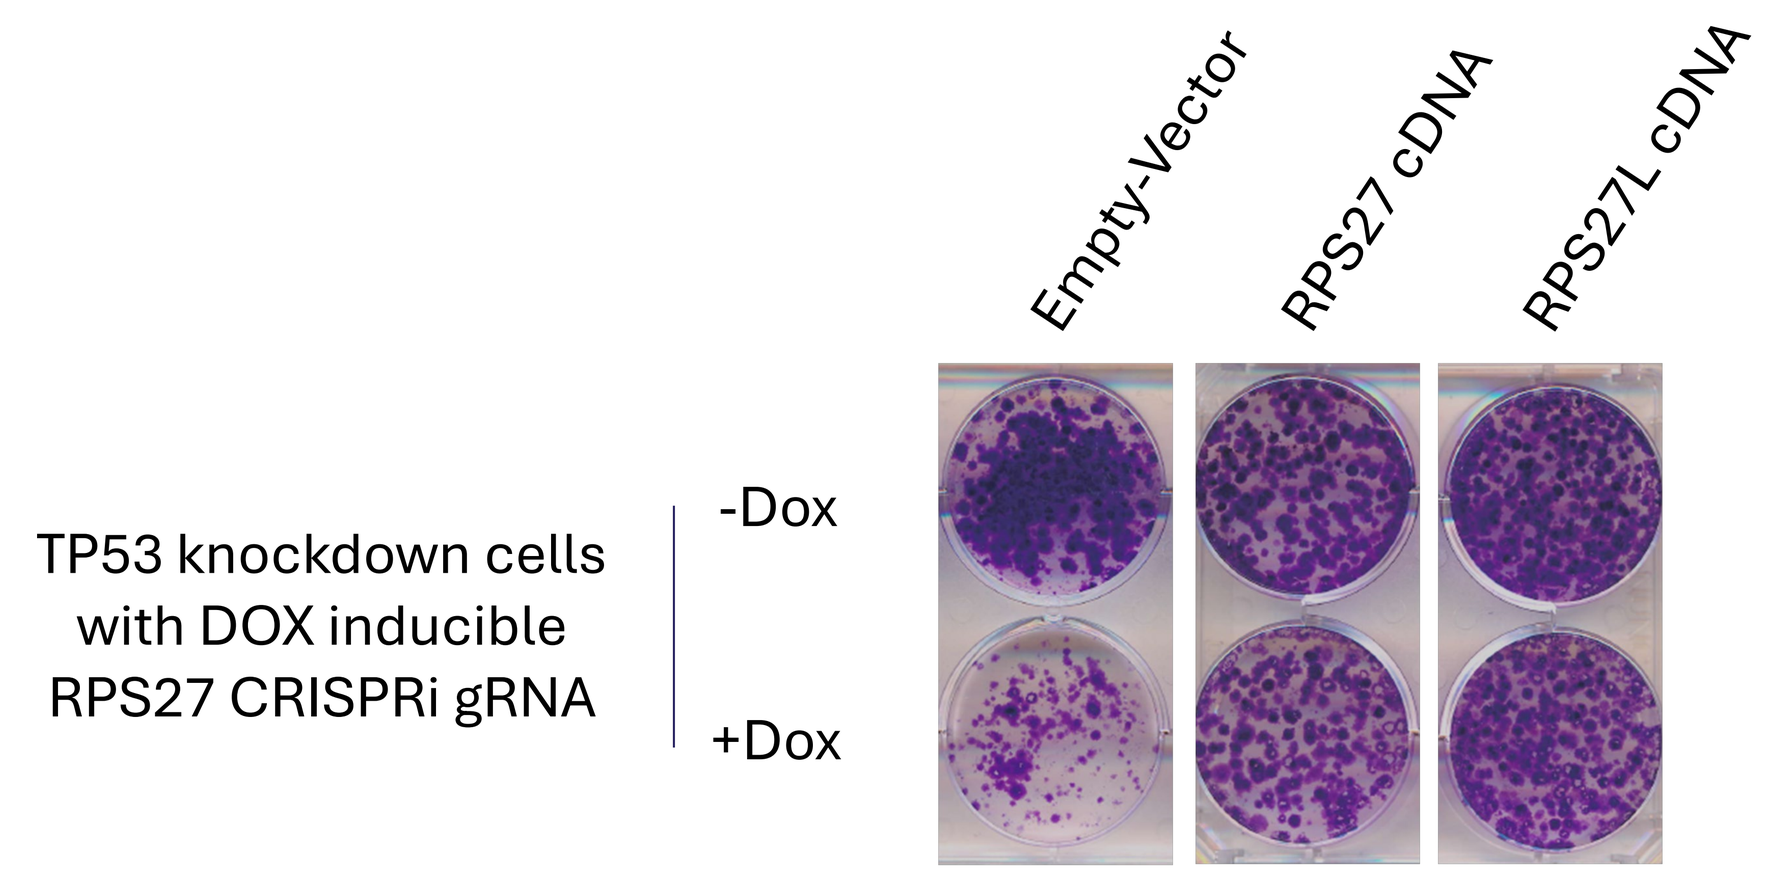
Empty-Vector
RPS27 cDNA
RPS27L cDNA
TP53 knockdown cells with DOX inducible RPS27 CRISPRi gRNA
-Dox
+Dox

- Re-expression of either RPS27 or RPS27L cDNA rescues the RP27 dependent phenotype in A549 TP53 knockdown cells

SUMMARY

- **Tango CRISPR platform enables precise identification of novel synthetic lethal targets**
- **CRISPRi platform offers distinct advantage and complements CRISPRko screen**
- **Whole genome TP53 isogenic CRISPRi screen identified RPS27 as a novel synthetic target**
- **CRISPRko RPS27 gRNAs have multiple off target sites, which likely caused cutting effect and buried RPS27 dependency in CRISPRko screens**
- **Single gene validation demonstrates strong selective effect from RPS27 knockdown in TP53 loss cells (due to low RPS27L expression)**
- **TP53, RPS27 or RPS27L re-expression can rescue the lethality induced by RPS27 knockdown, confirming the therapeutic hypothesis**
- **TP53 loss results in low RPS27L expression which leads to dependency on the paralog RPS27**

References

- Graphic Illustration created with BioRender.com
- DepMap, Broad (2025). DepMap Public 25Q3. Dataset. depmap.org
- Arafeh, R., et al. The present and future of the Cancer Dependency Map. Nat Rev Cancer 25, 59-73 (2025)
- Xiong, X., et al. Ribosomal protein S27-like and S27 interplay with p53-MDM2 axis as a target, a substrate and a regulator. Oncogene, 30(15), 1798–1811 (2011)