TNG348 is a potent and selective inhibitor of USP1 for the treatment of BRCA1/2mut and HRD+ cancers

Abstract #4968

Antoine Simoneau, Hsin-Jung Wu, Madhavi Bandi, Katherine Lazarides, Sining Sun, Shangtao Liu, Samuel Meier, Ashley Choi, Hongxiang Zhang, Binzhang Shen, Douglas Whittington, Sirimas Sudsakorn
Wenhai Zhang, Yi Yu, Yong Liu, Colin Liang, Michael Palmieri, Yingnan Chen, Brian Haines, Alice Tsai, Minjie Zhang, Alan Huang, Jannik Andersen, Tianshu Feng, Scott Throner, John Maxwell

INTRODUCTION

TNG348 is a selective and potent inhibitor of the deubiquitinating enzyme USP1 designed to target BRCA1/2mut vulnerabilities in breast and ovarian tumors. Here we present the biochemical, mechanistic, and in vitro and in vivo characterization of TNG348, an oral, allosteric and highly potent inhibitor of USP1. Upon treatment, TNG348 causes loss of viability in breast and ovarian cell lines with BRCA1/2 mutations. Cell line panel analysis shows TNG348 activity extends beyond BRCA1/2mut models with homologous recombination deficiency (HRD) as an additional feature correlating with USP1 inhibition sensitivity. We show that TNG348 induces cell death through a pathway that is distinct from PARPi and TNG348 demonstrates robust synergy when combined with first- or second-generation PARPi. Moreover, TNG348 exhibits strong tumor growth inhibition in combination with PARPi in BRCA1/2mut and HRD+ xenograft models as well as models of acquired PARPi resistance. We plan to evaluate TNG348 as single agent and in combination with PARP1i in patients with BRCA1/2 mutant or HRD+ tumors that are naïve to PARPi and with prior PARPi treatment history.

USP1 was identified as a synthetic lethal target in BRCA1/2 mutant cell lines

2 BRCAmut vs 11 BRCAwt cell lines

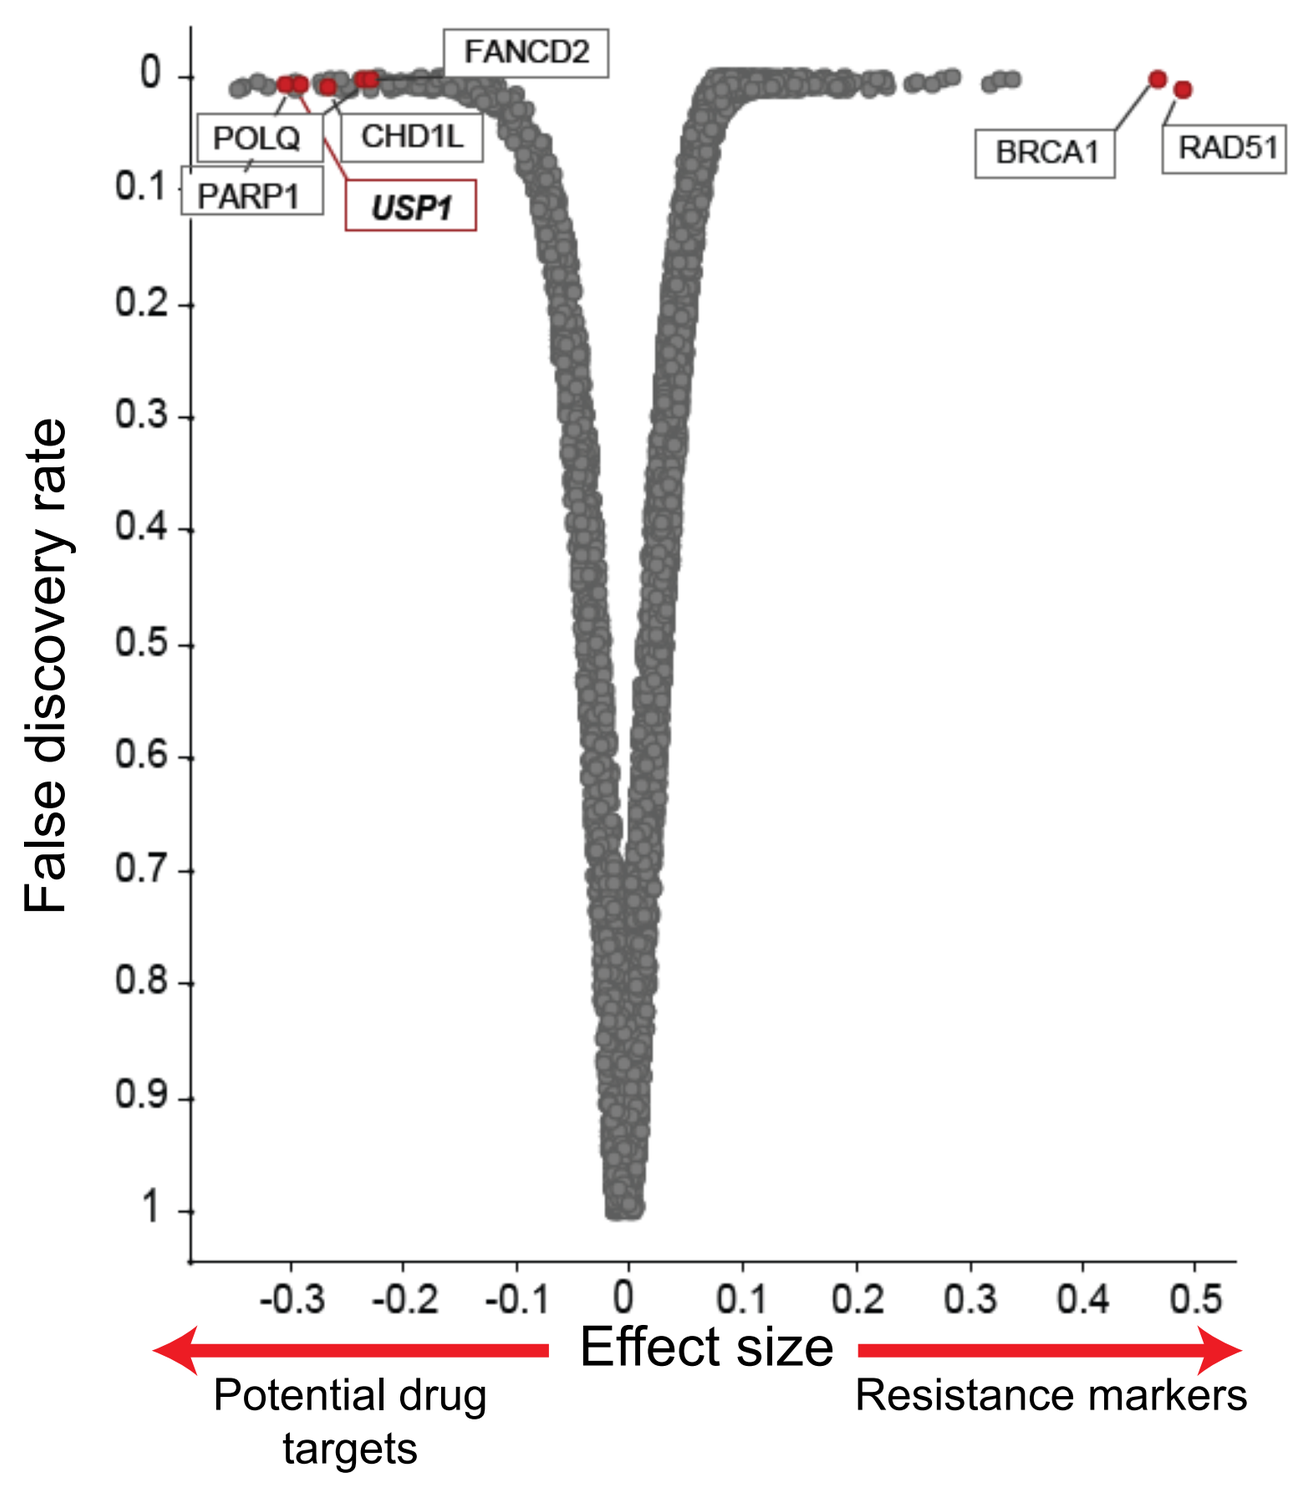
FANCD2
POLQ
CHD1L
PARP1
USP1
BRCA1
RAD51
False discovery rate
Effect size
Potential drug targets
Resistance markers

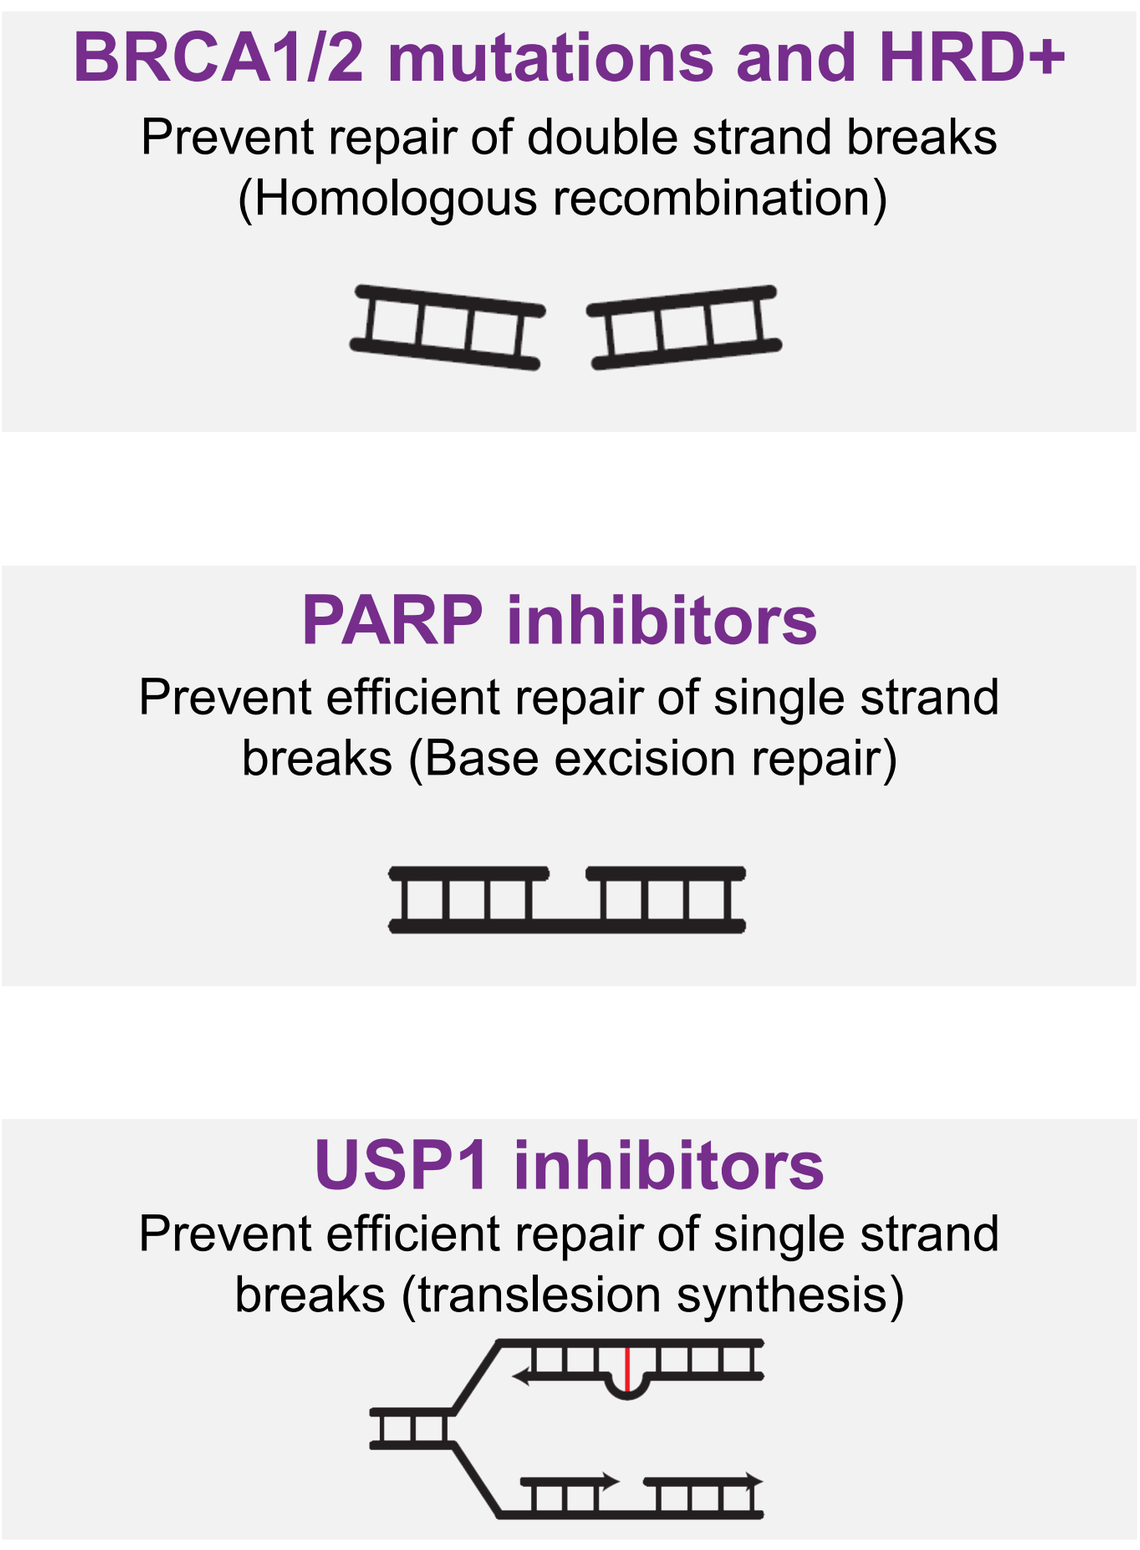
BRCA1/2 mutations and HRD+
Prevent repair of double strand breaks (Homologous recombination)
PARP inhibitors
Prevent efficient repair of single strand breaks (Base excision repair)
USP1 inhibitors
Prevent efficient repair of single strand breaks (translesion synthesis)

TNG348 acts through a ub-PCNA-dependent pathway that is distinct from PARP inhibitors

USP1i anchor screen

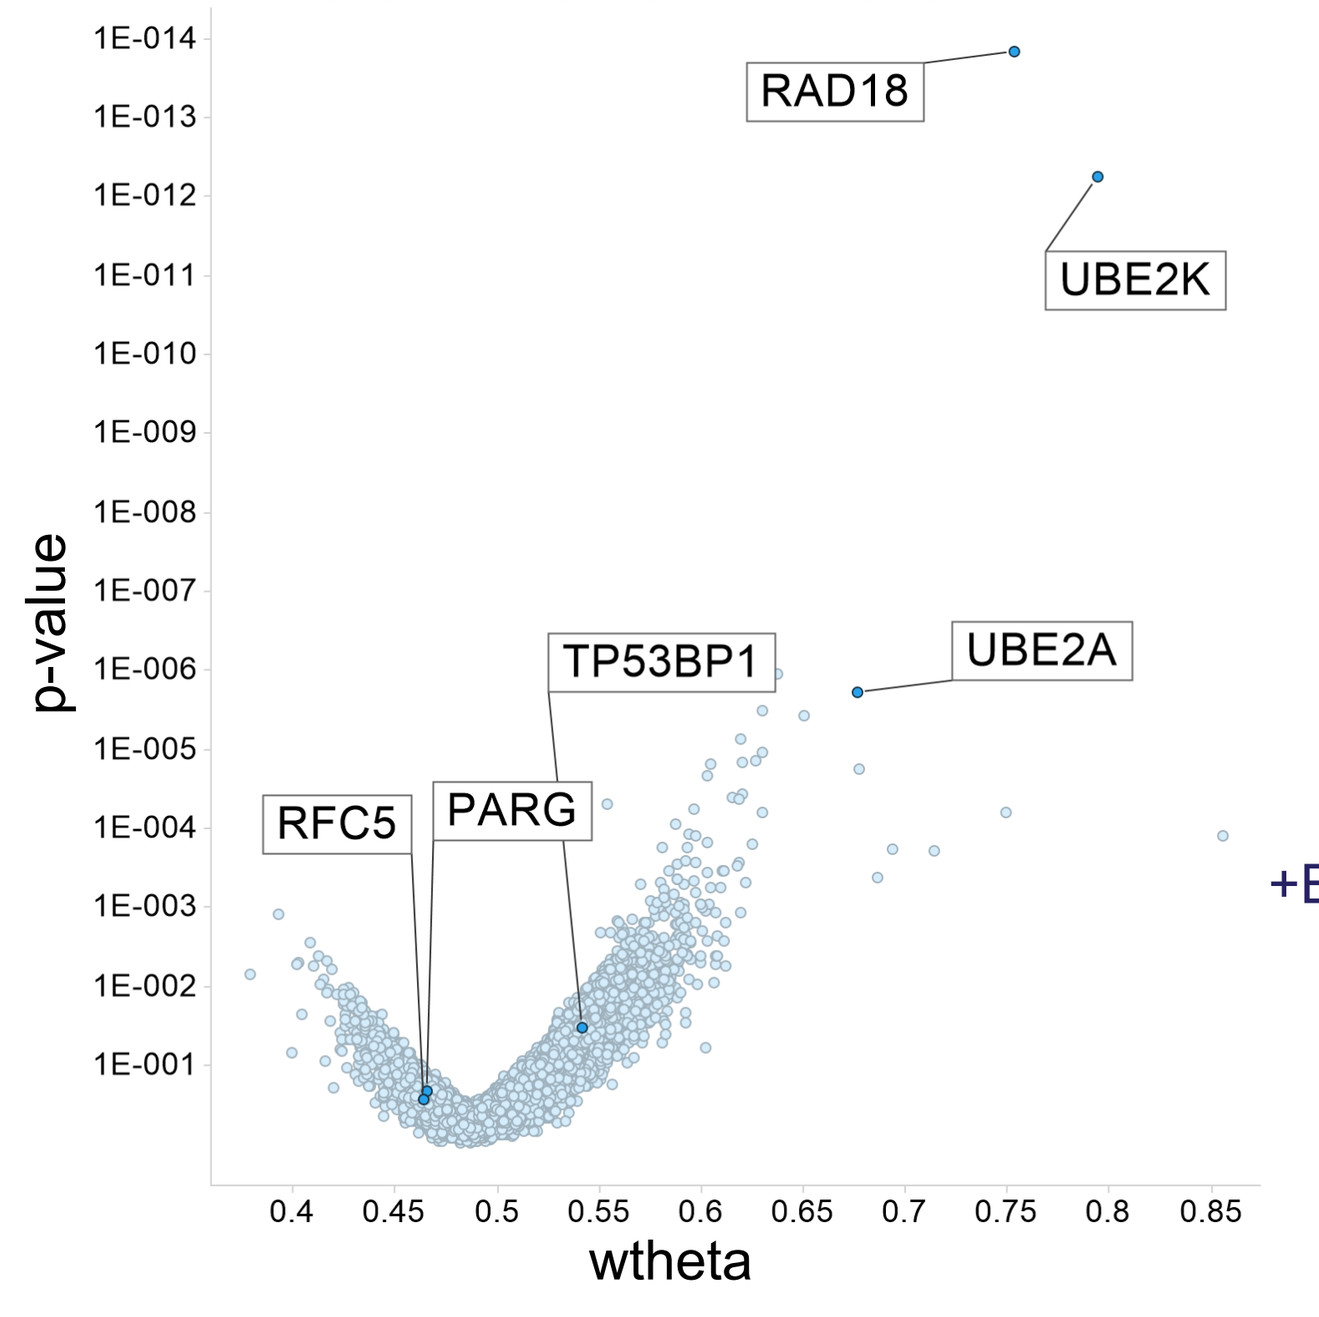
RAD18
UBE2K
TP53BP1
UBE2A
RFC5
PARG
p-value
wtheta

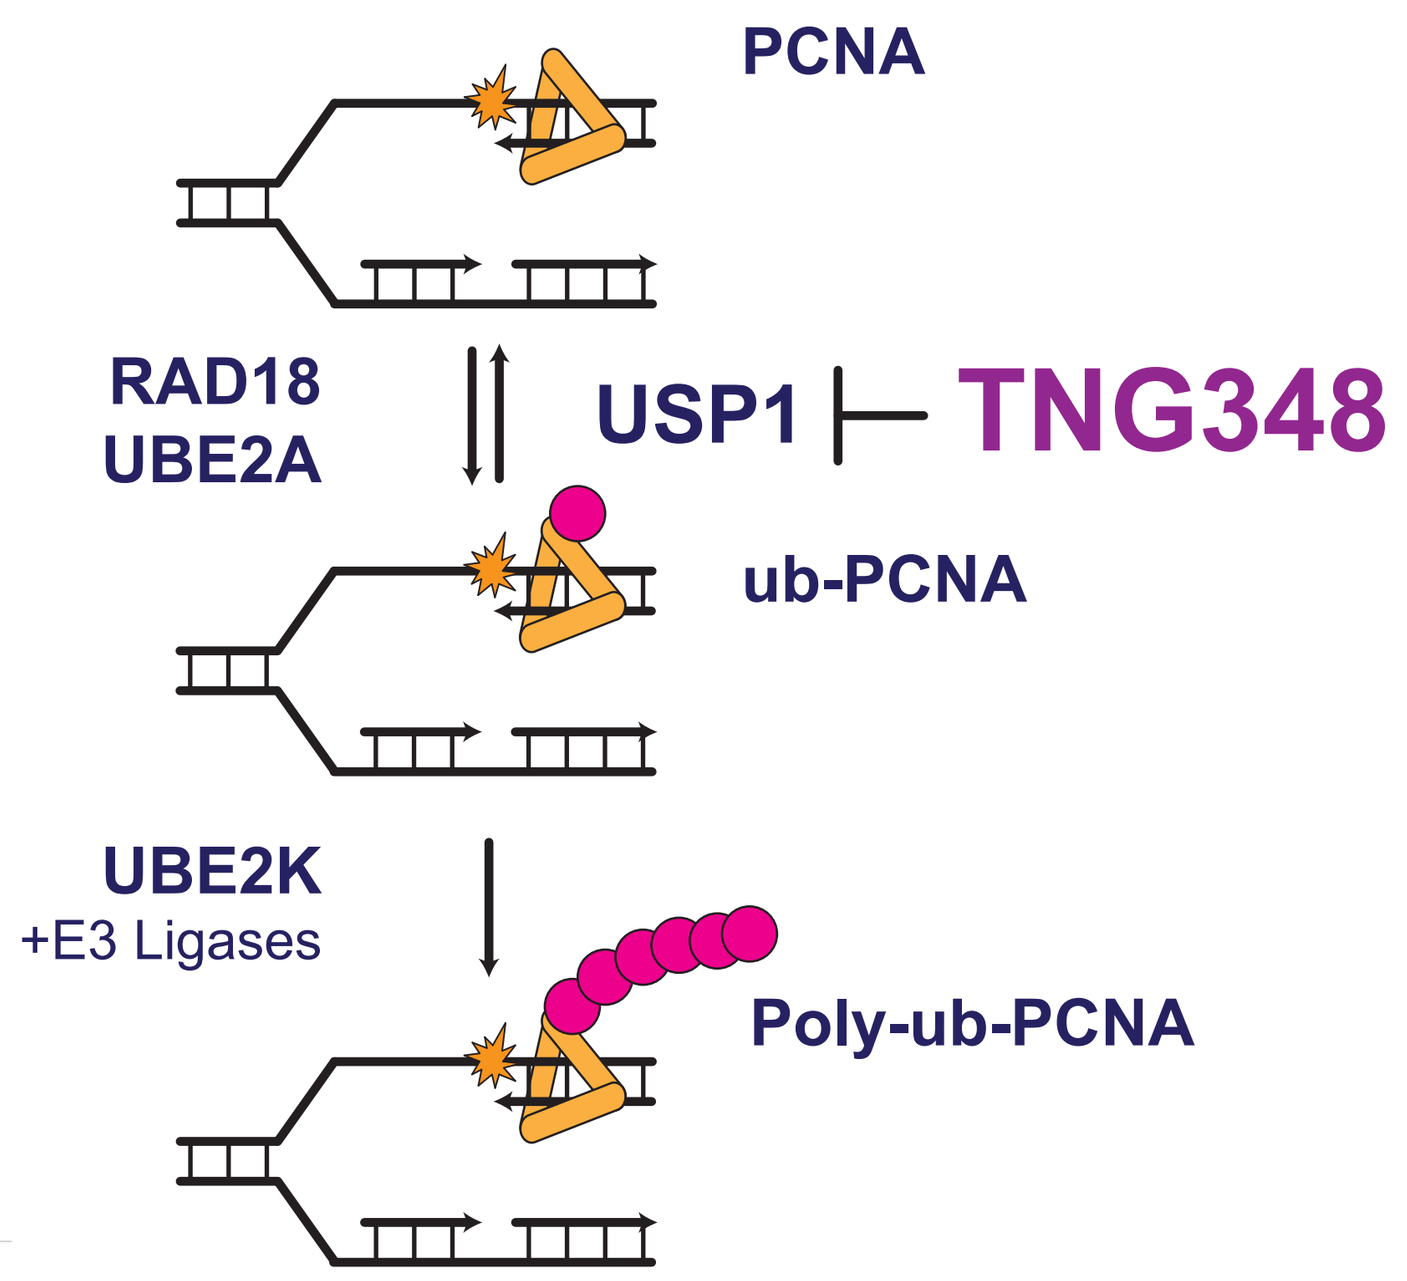
PCNA
RAD18
UBE2A
USP1 ⊢ TNG348
ub-PCNA
UBE2K
+E3 Ligases
Poly-ub-PCNA

TNG348 is a potent inhibitor of USP1

Biochemical assay

ub-Rho110 assay

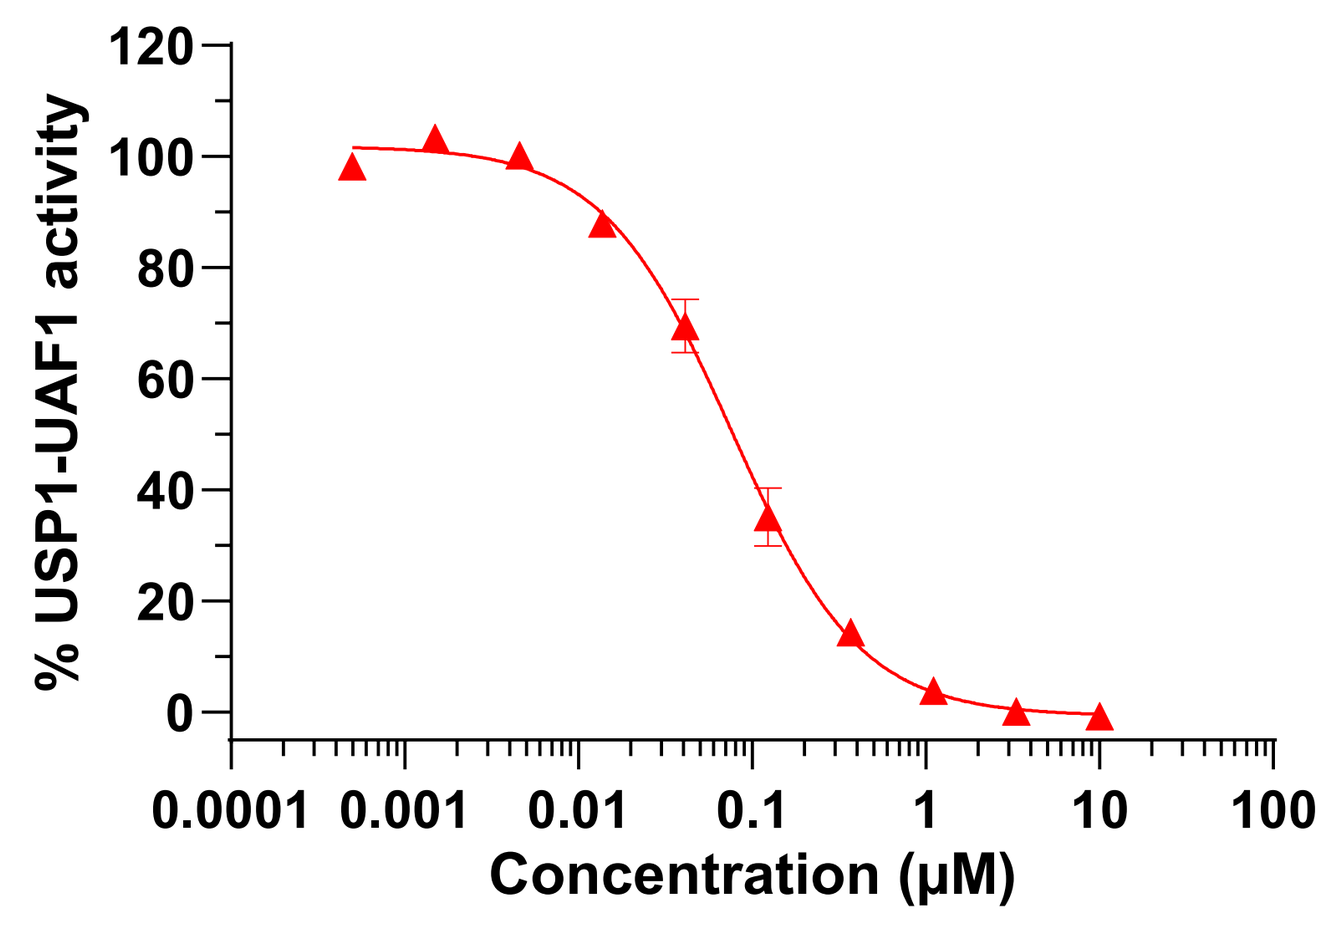
% USP1-UAF1 activity
Concentration (µM)

IC50: 82 nM

Cellular PD

MDA-MB-436 (BRCA1mut)

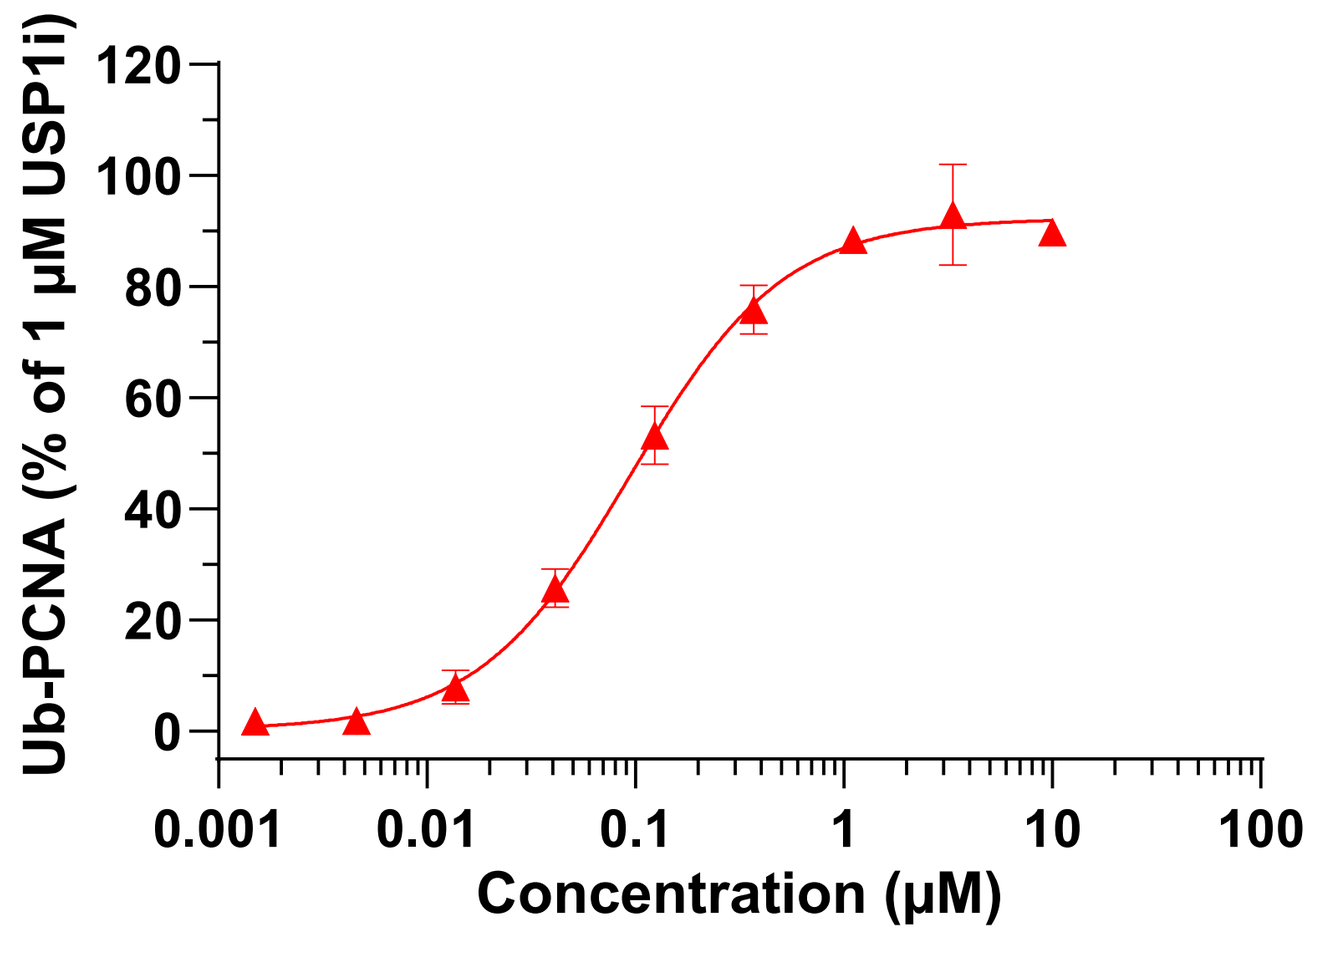
Ub-PCNA (% of 1 µM USP1i)
Concentration (µM)

IC50: 95 nM

TNG348 is selective for BRCA1/2mut and HRD+

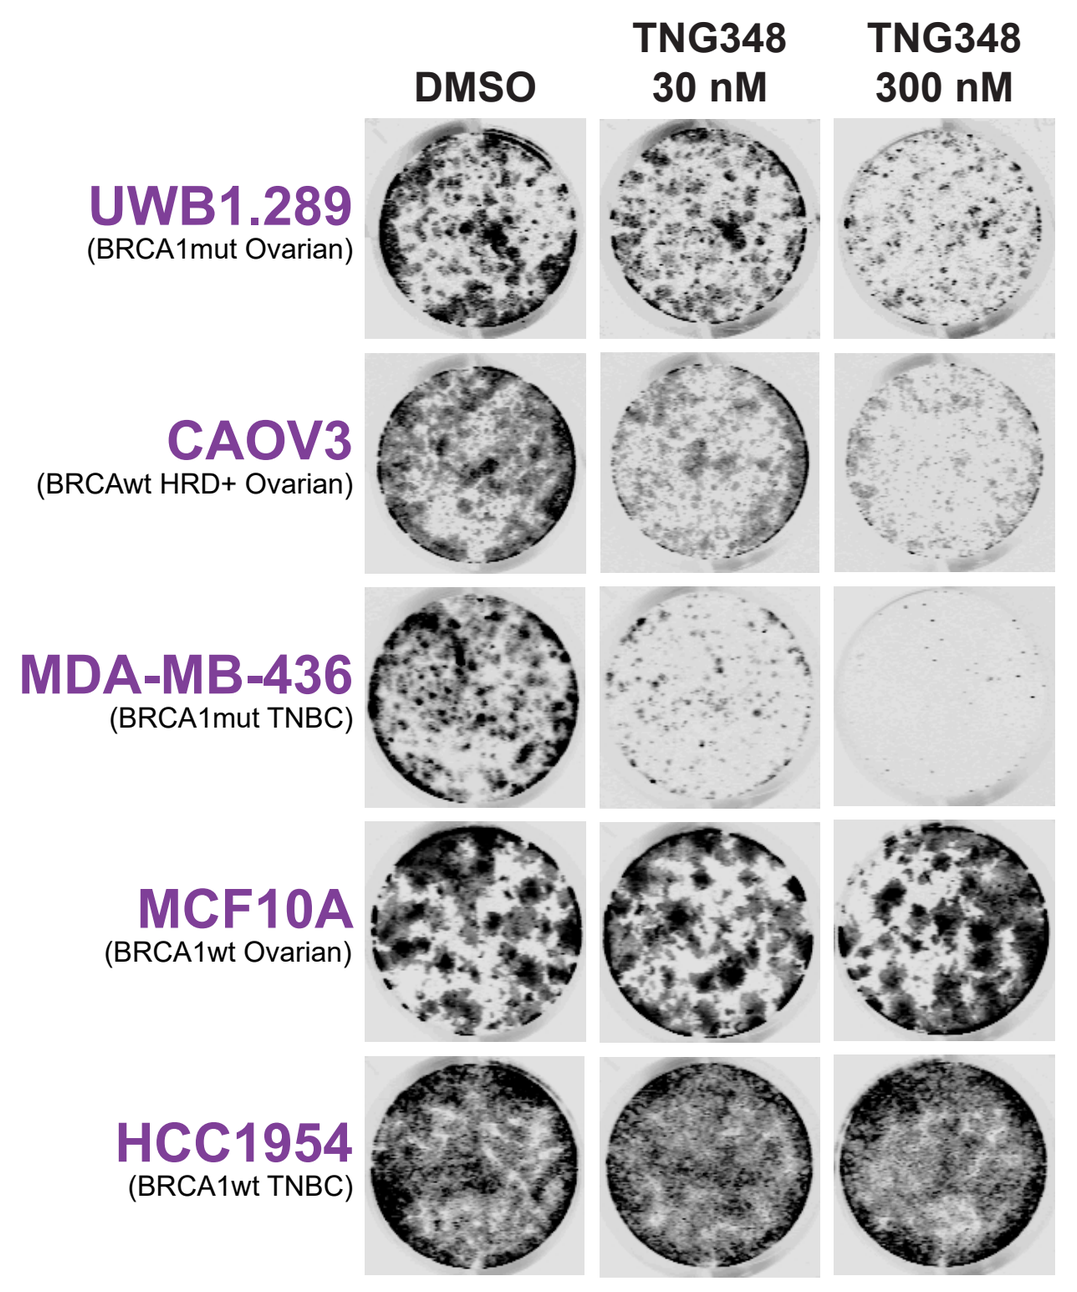
DMSO
TNG348 30 nM
TNG348 300 nM
UWB1.289
(BRCA1mut Ovarian)
CAOV3
(BRCAwt HRD+ Ovarian)
MDA-MB-436
(BRCA1mut TNBC)
MCF10A
(BRCA1wt Ovarian)
HCC1954
(BRCA1wt TNBC)

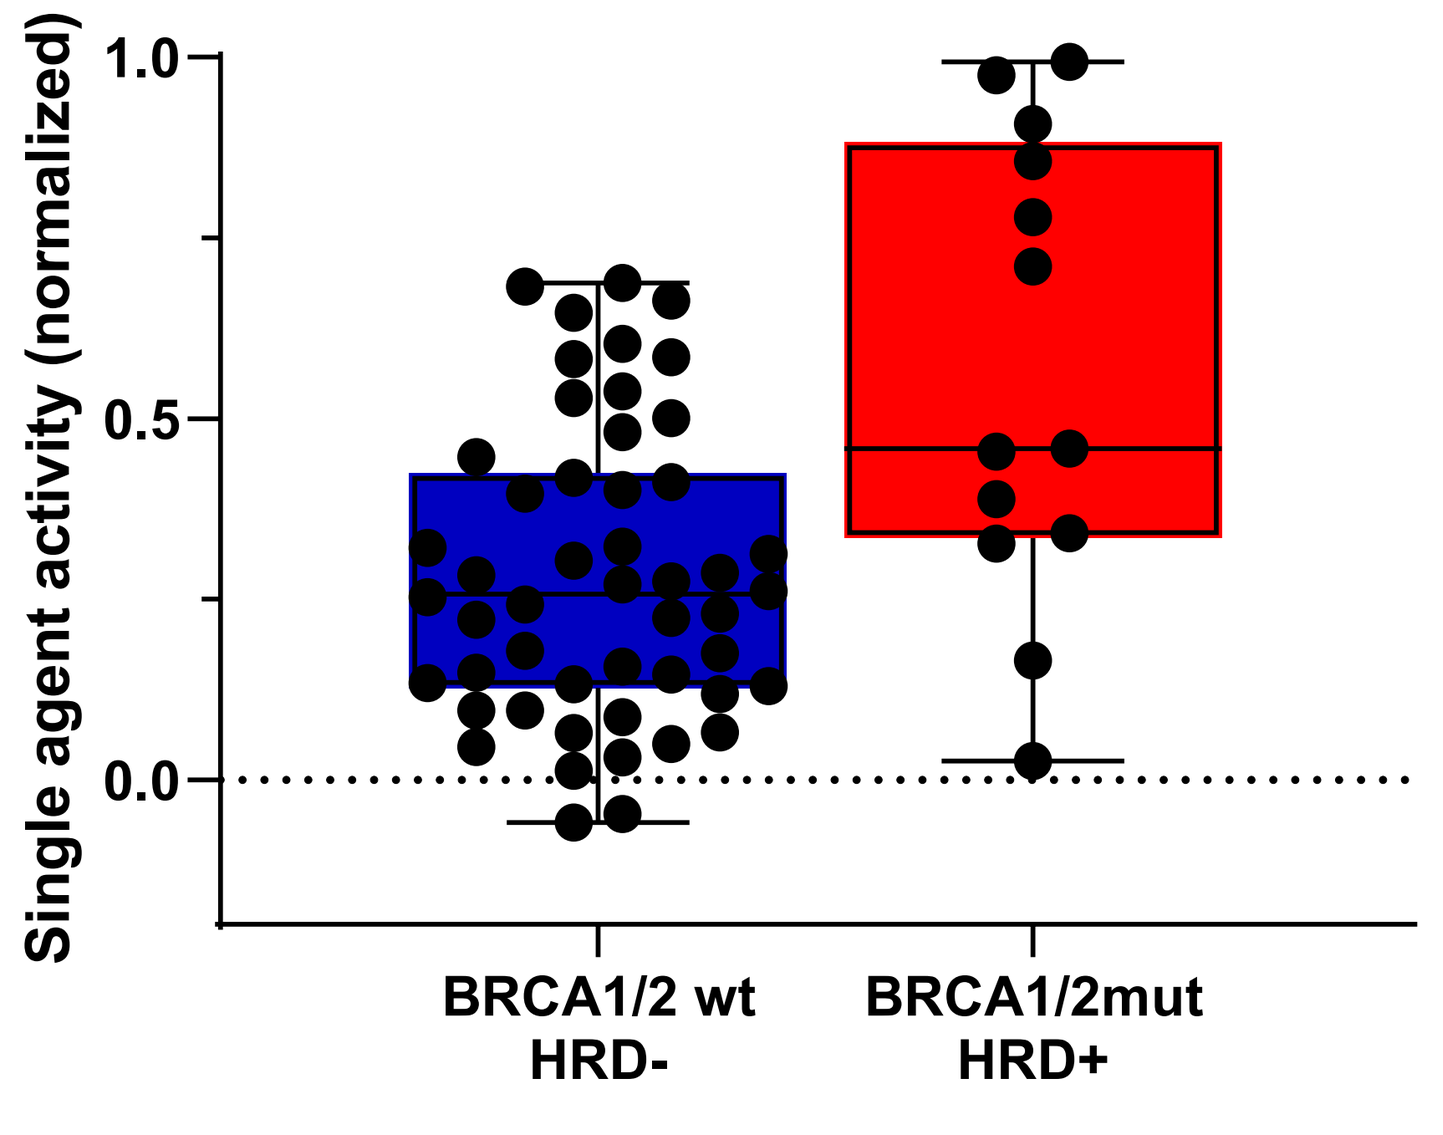
Single agent activity (normalized)
BRCA1/2 wt HRD-
BRCA1/2mut HRD+

TNG348 synergizes with PARP inhibitors in vitro

TNG348-olaparib

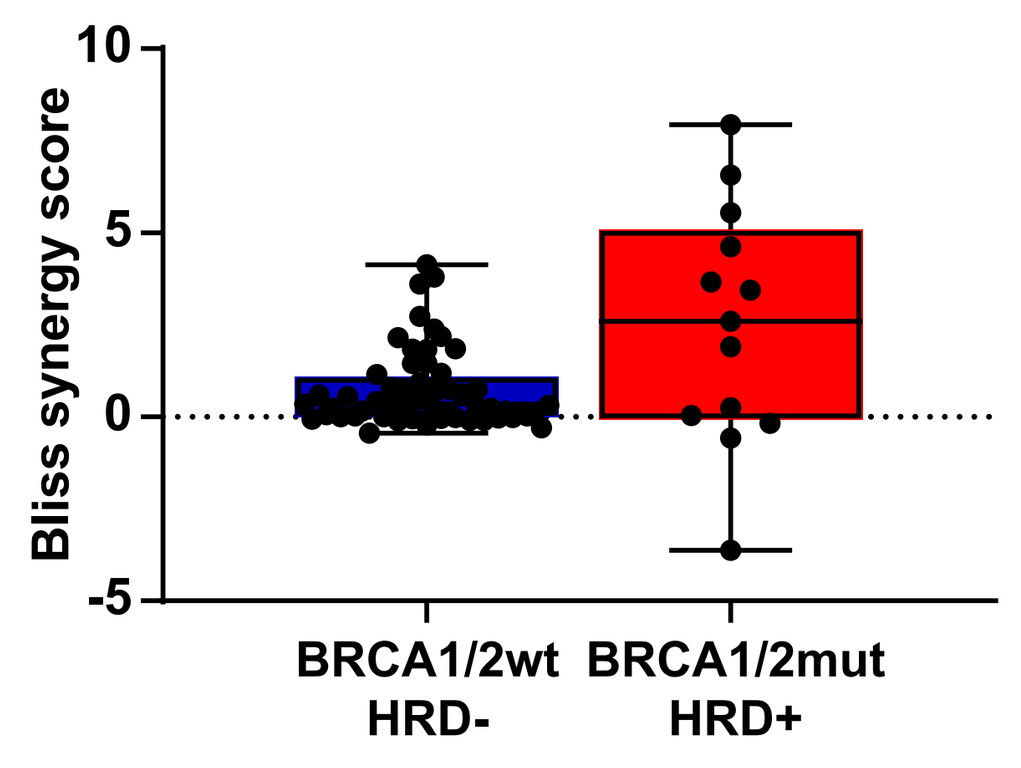
Bliss synergy score
BRCA1/2wt HRD-
BRCA1/2mut HRD+

TNG348-AZD5305

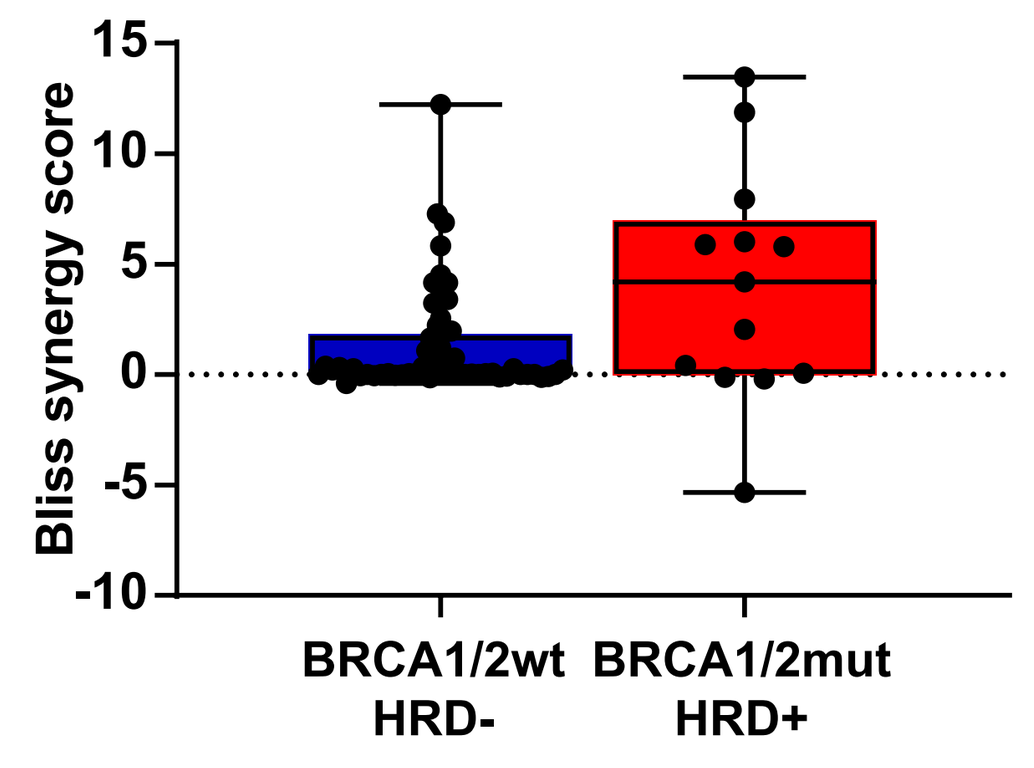
Bliss synergy score
BRCA1/2wt HRD-
BRCA1/2mut HRD+

Increasing doses of olaparib with or without TNG348

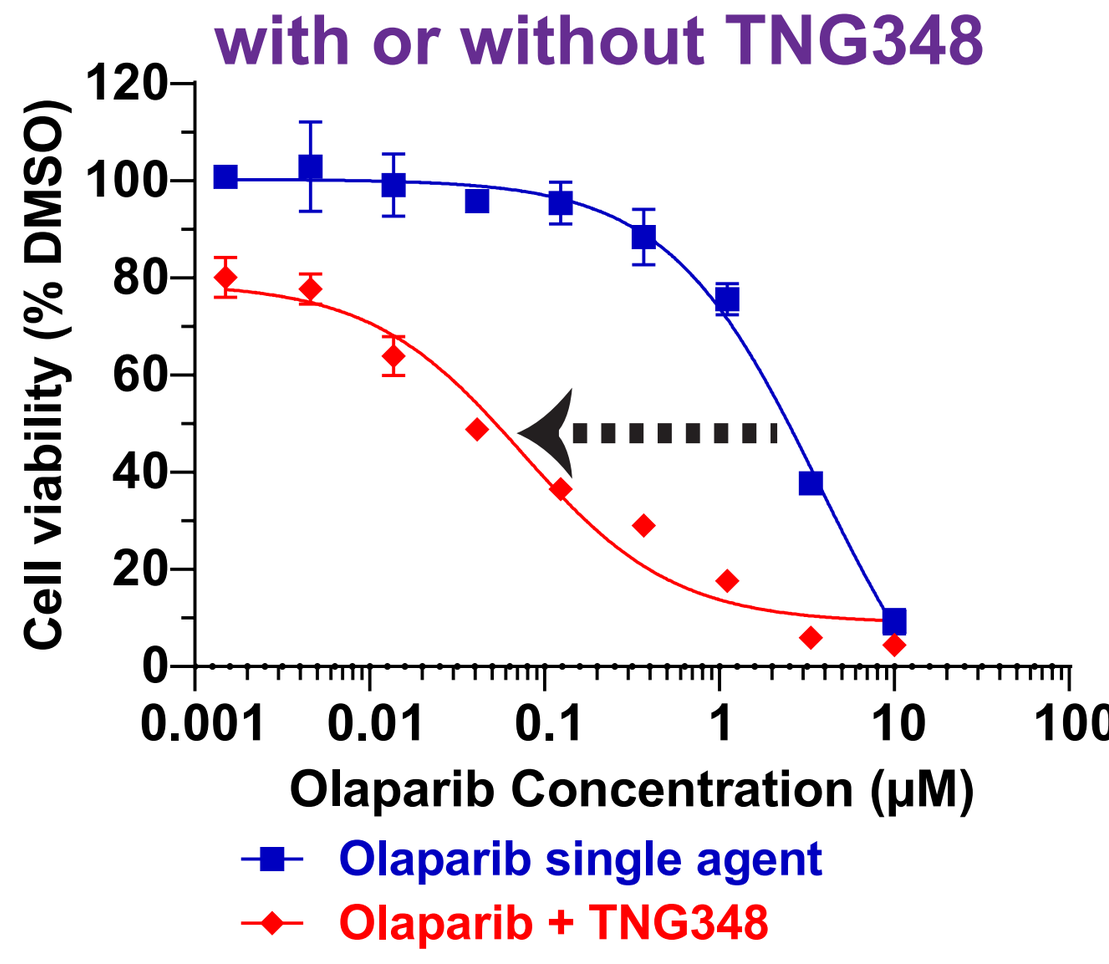
Cell viability (% DMSO)
Olaparib Concentration (µM)
Olaparib single agent
Olaparib + TNG348

Increasing doses of TNG348 with or without olaparib

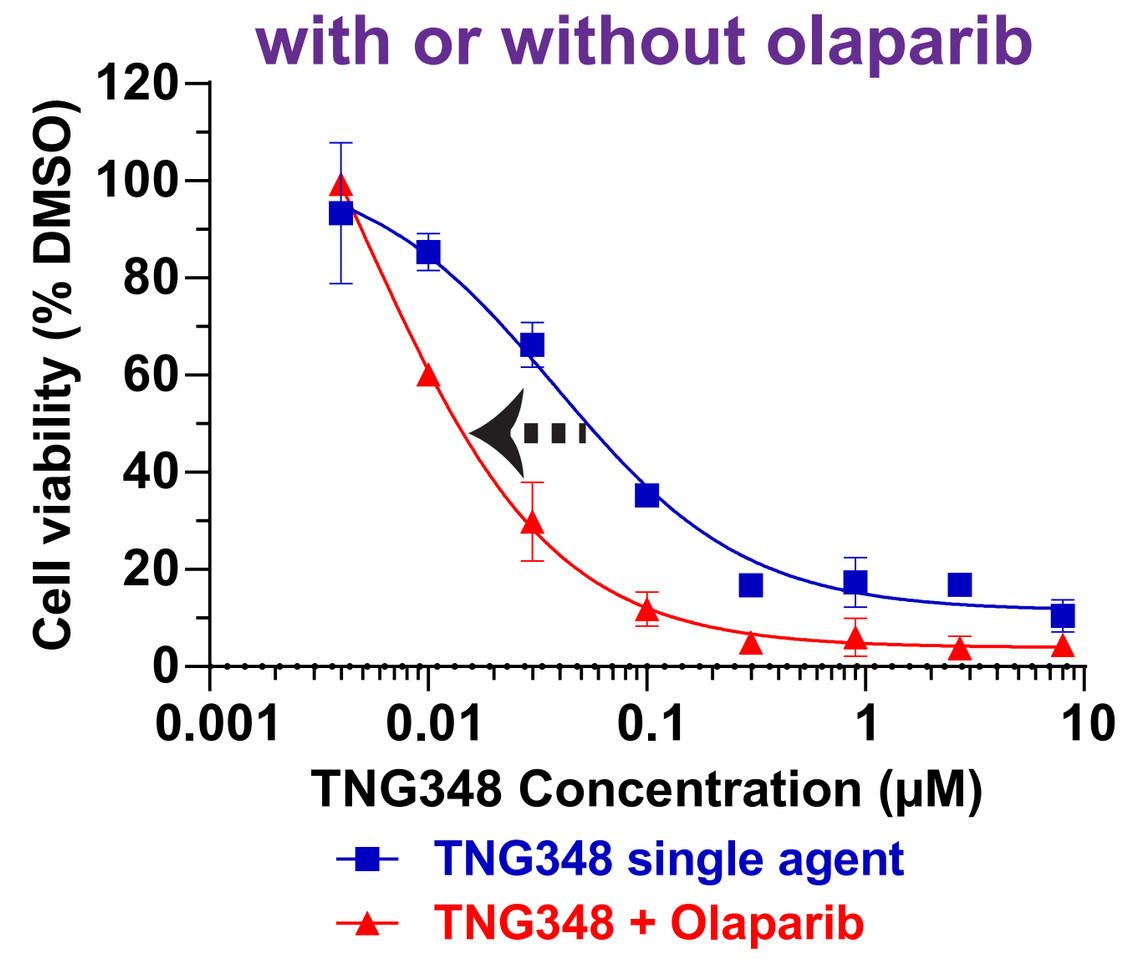
Cell viability (% DMSO)
TNG348 Concentration (µM)
TNG348 single agent
TNG348 + Olaparib

TNG348 synergizes in vivo with PARPi

BRCA1mut

TNBC PDX

BRCA2mut

Ovarian PDX

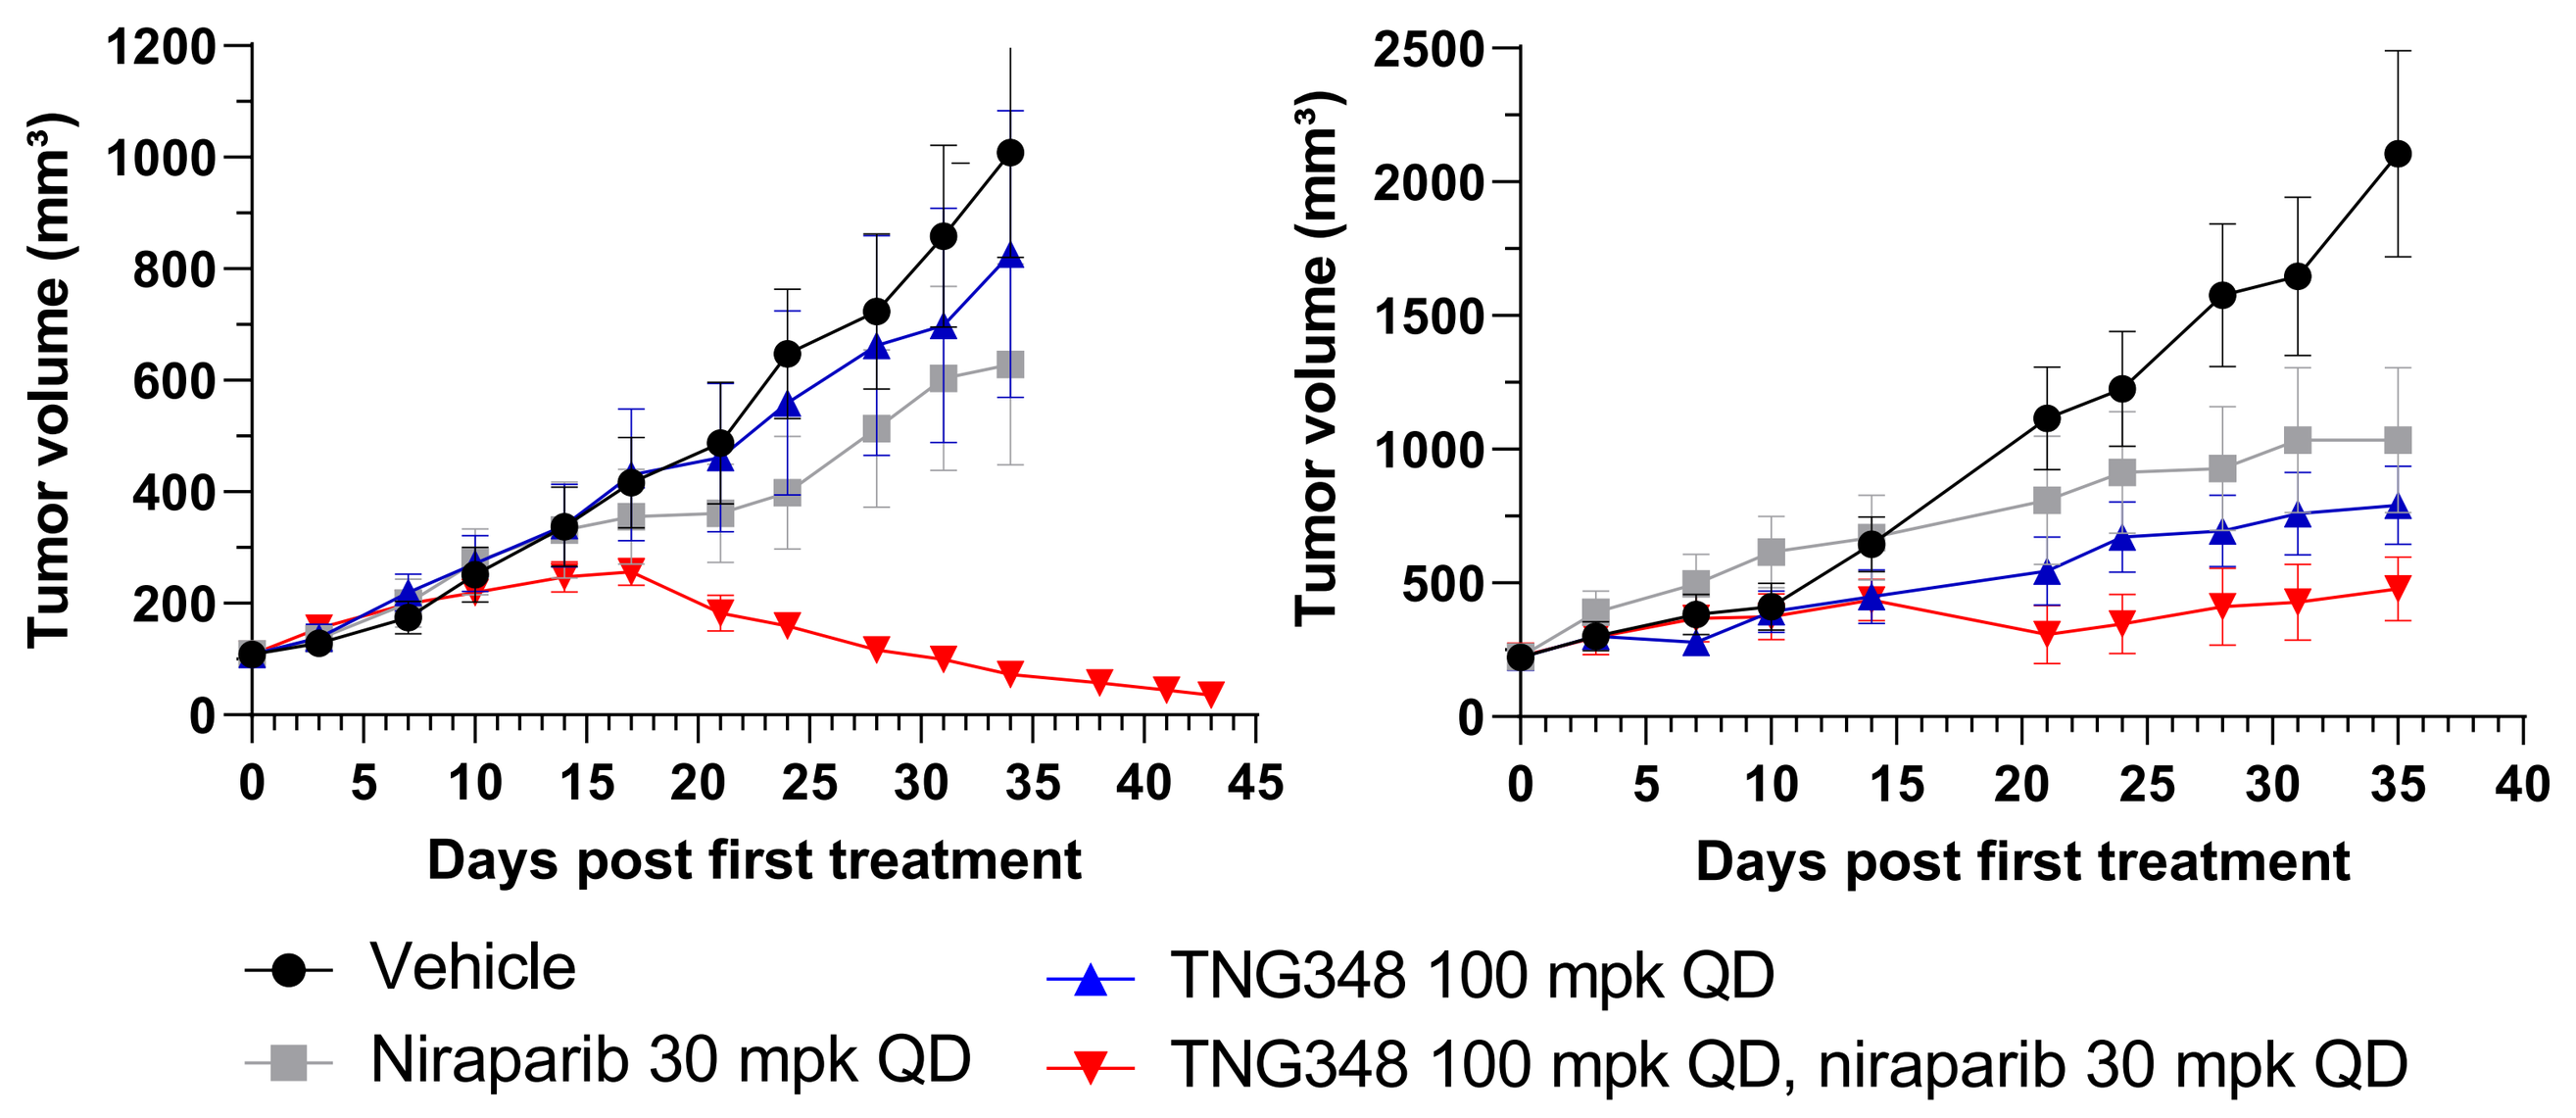
Tumor volume (mm³)
Days post first treatment
Vehicle
Niraparib 30 mpk QD
TNG348 100 mpk QD
TNG348 100 mpk QD, niraparib 30 mpk QD

BRCAwt HRD+

TNBC PDX

BRCAwt HRD+

Ovarian PDX

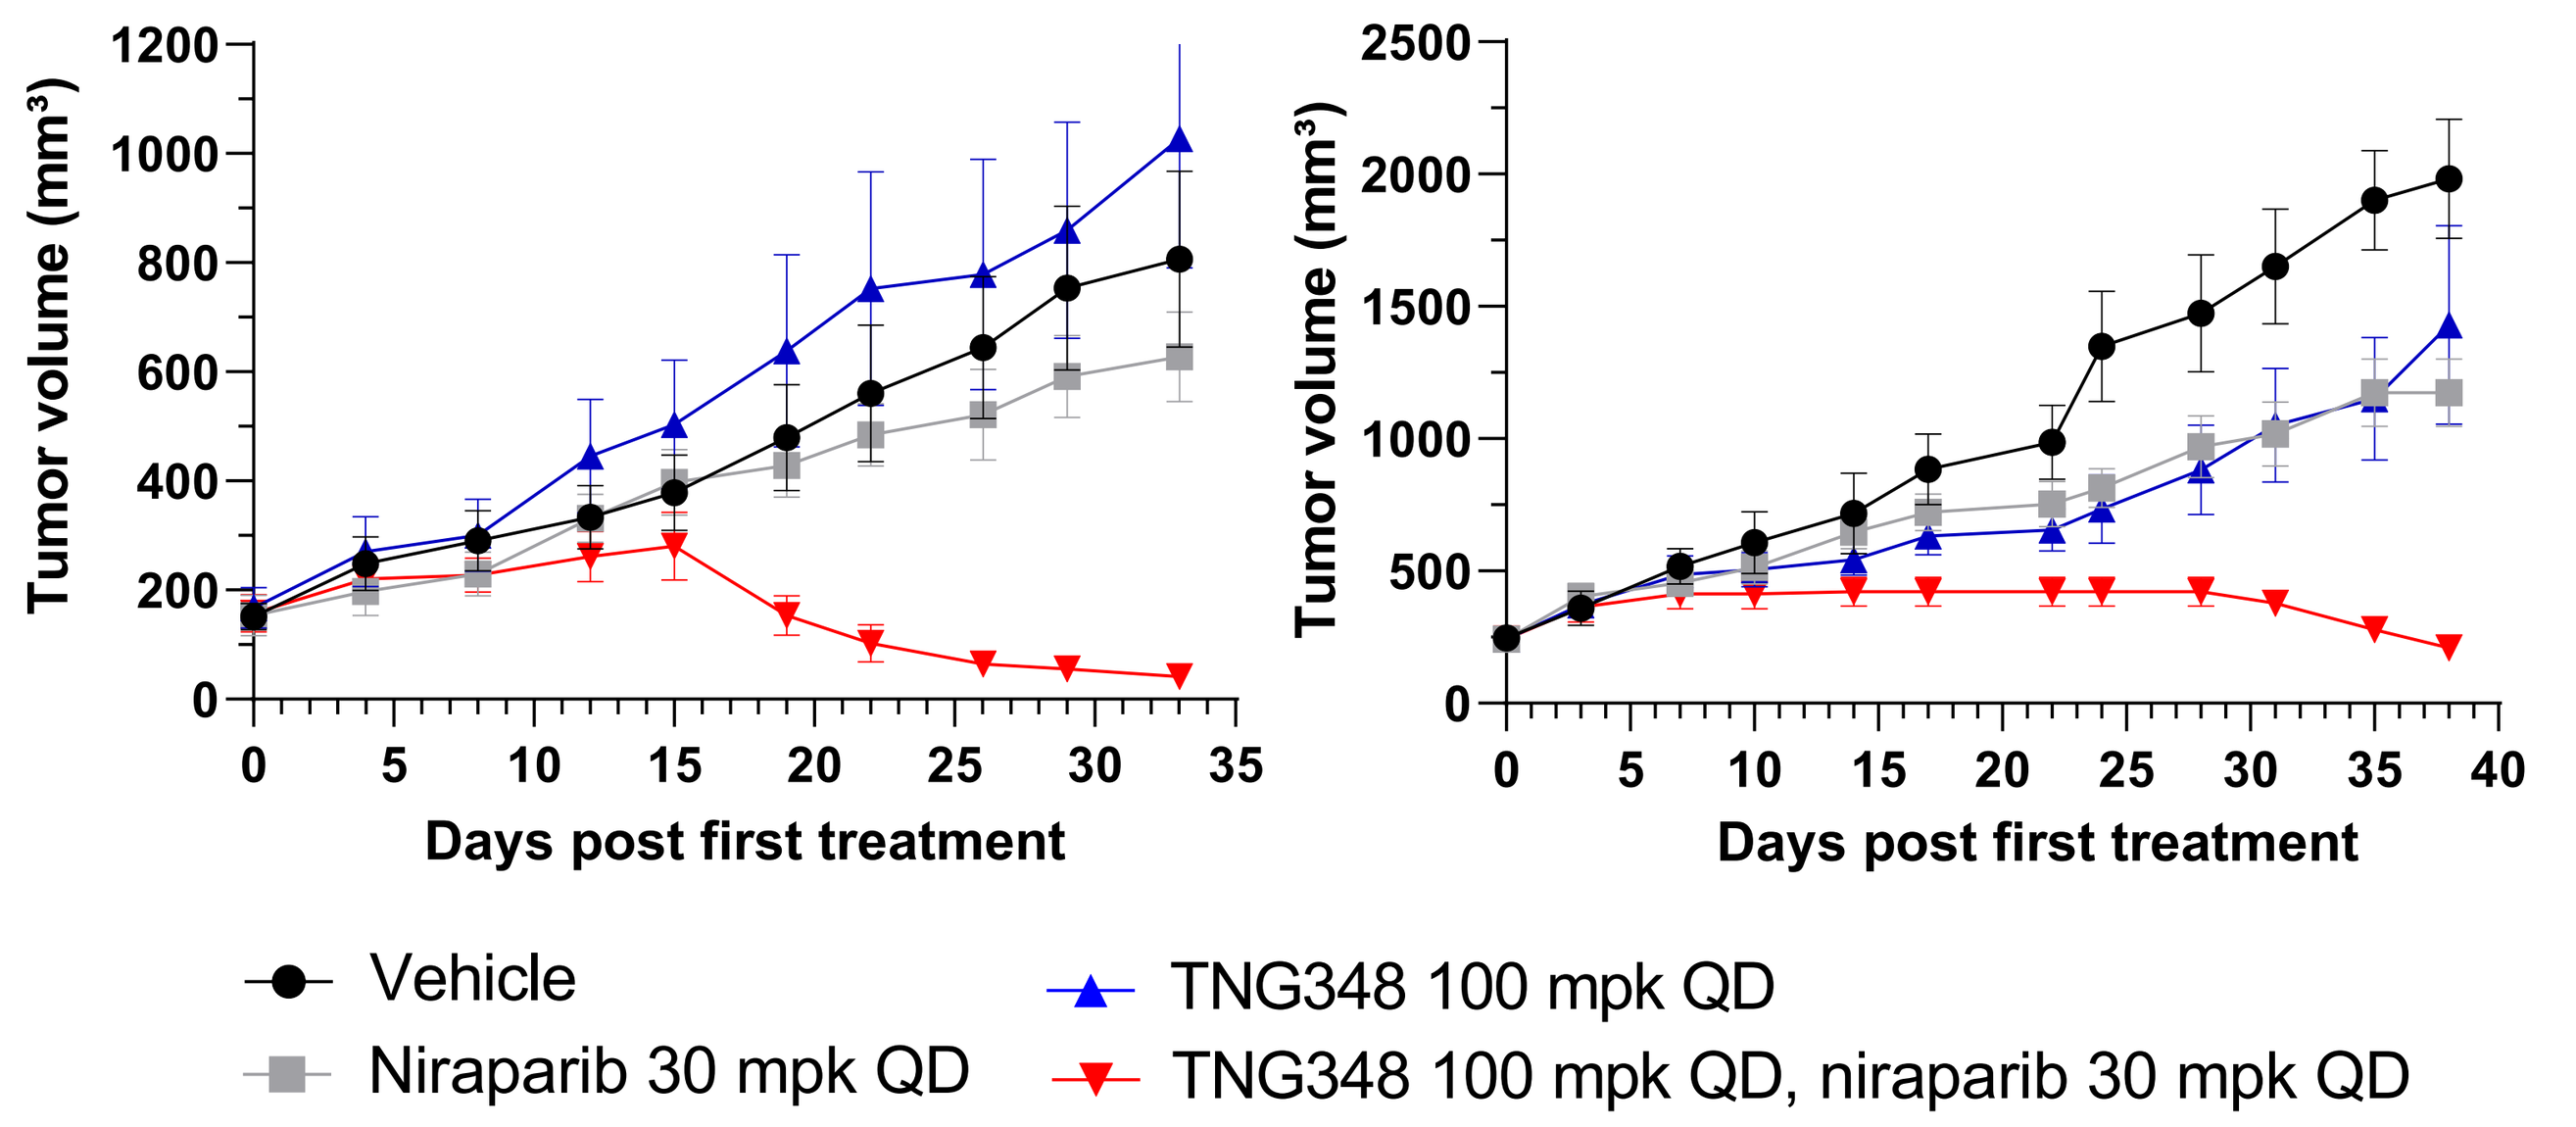
Tumor volume (mm³)
Days post first treatment
Vehicle
Niraparib 30 mpk QD
TNG348 100 mpk QD
TNG348 100 mpk QD, niraparib 30 mpk QD

TNG348 can overcome acquired PARPi resistance

BRCA1mut

Parental PARPi-sensitive TNBC PDX

BRCA1mut

Acquired PARPi resistance TNBC PDX

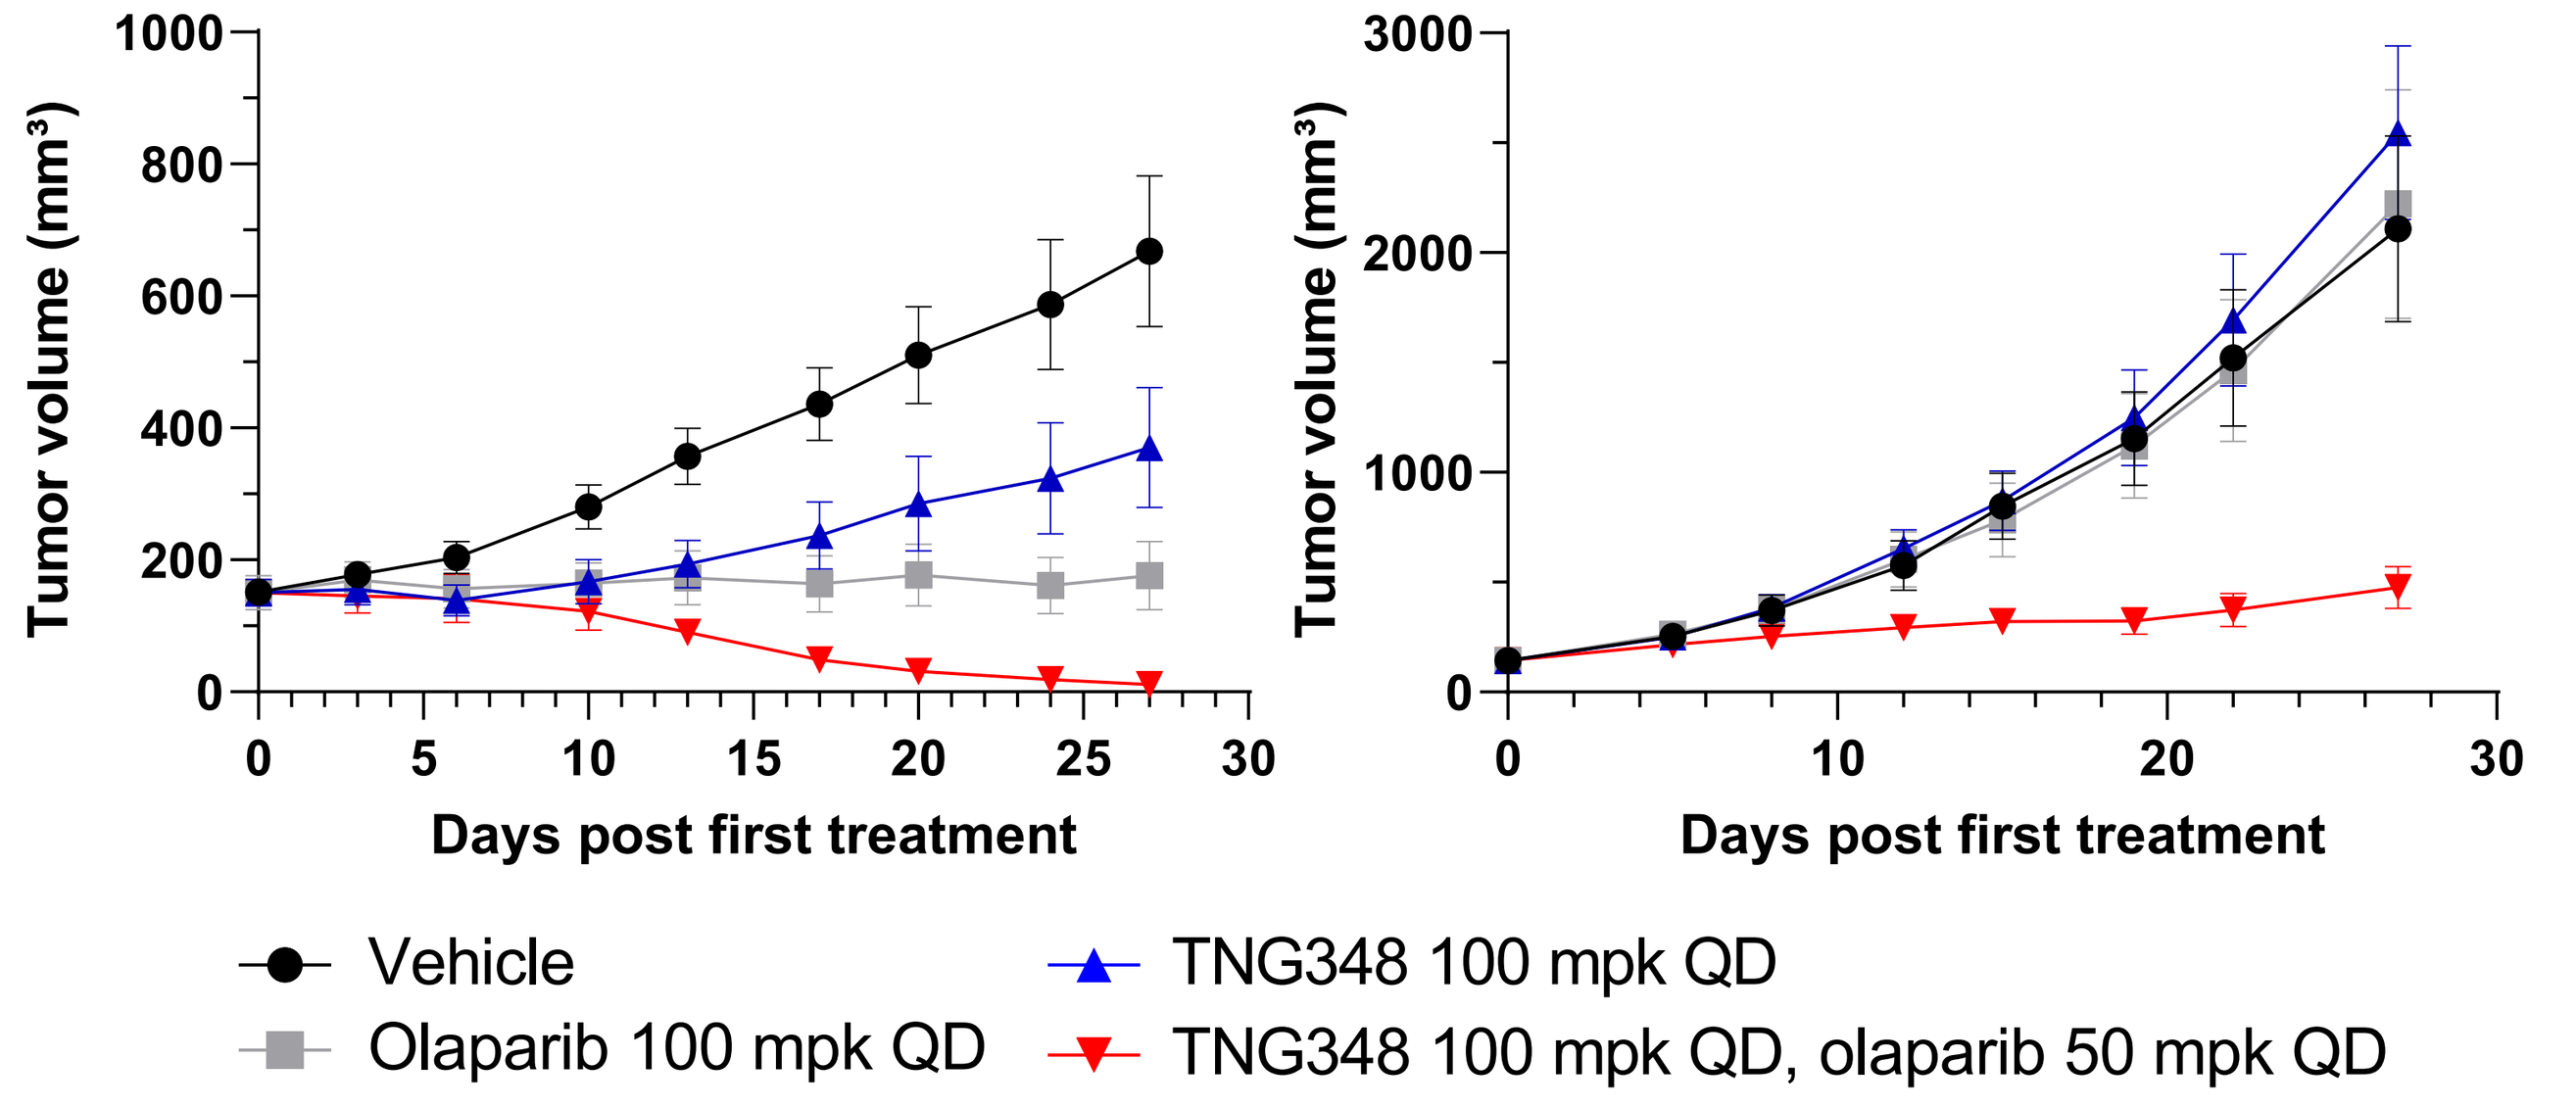
Tumor volume (mm³)
Days post first treatment
Vehicle
Olaparib 100 mpk QD
TNG348 100 mpk QD
TNG348 100 mpk QD, olaparib 50 mpk QD

- **PARPi resistant model was generated by consecutive passages in mice with constant olaparib exposure**
- **TNG348 is well tolerated, no body weight loss was observed in in vivo studies**

TNG348 is a selective allosteric inhibitor of USP1

USP1/UAF1 Initial Velocity

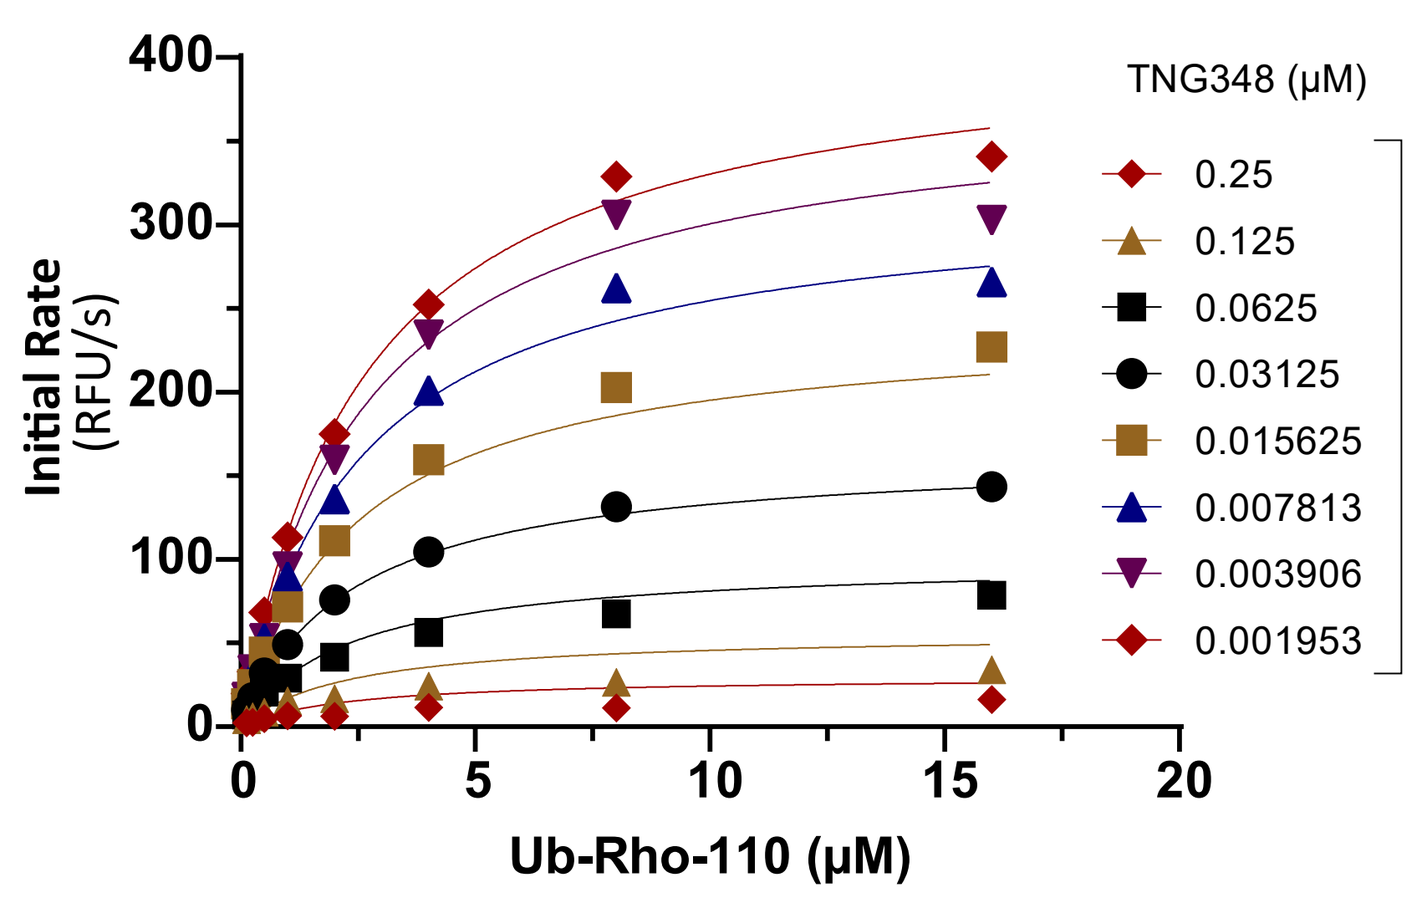
Initial Rate (RFU/s)
Ub-Rho-110 (µM)
TNG348 (µM)
0.25
0.125
0.0625
0.03125
0.015625
0.007813
0.003906
0.001953

Cryo-EM structure

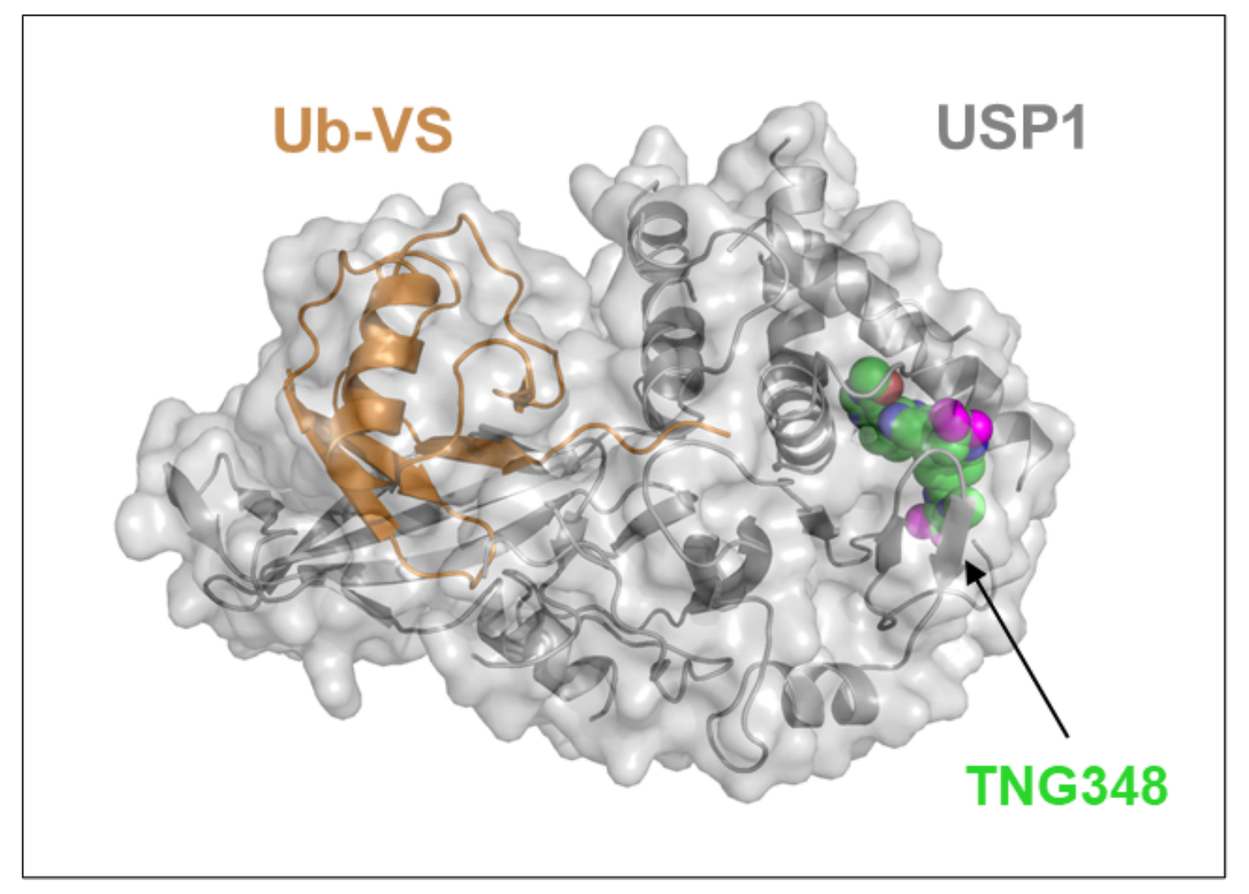
Ub-VS
USP1
TNG348

Selectivity across DUBs

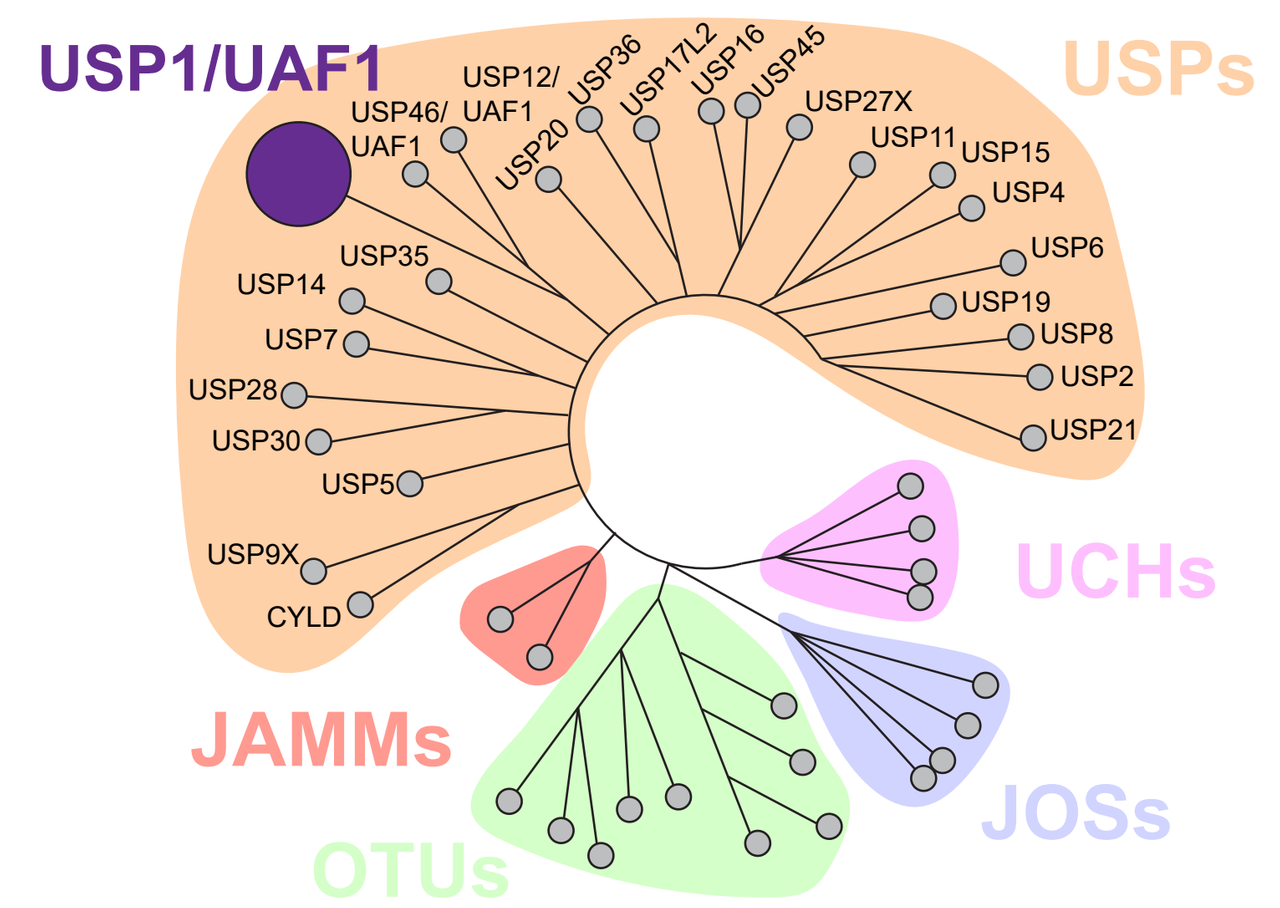
USP1/UAF1
USPs
UCHs
JAMMs
OTUs
JOSs

Catalytic activity

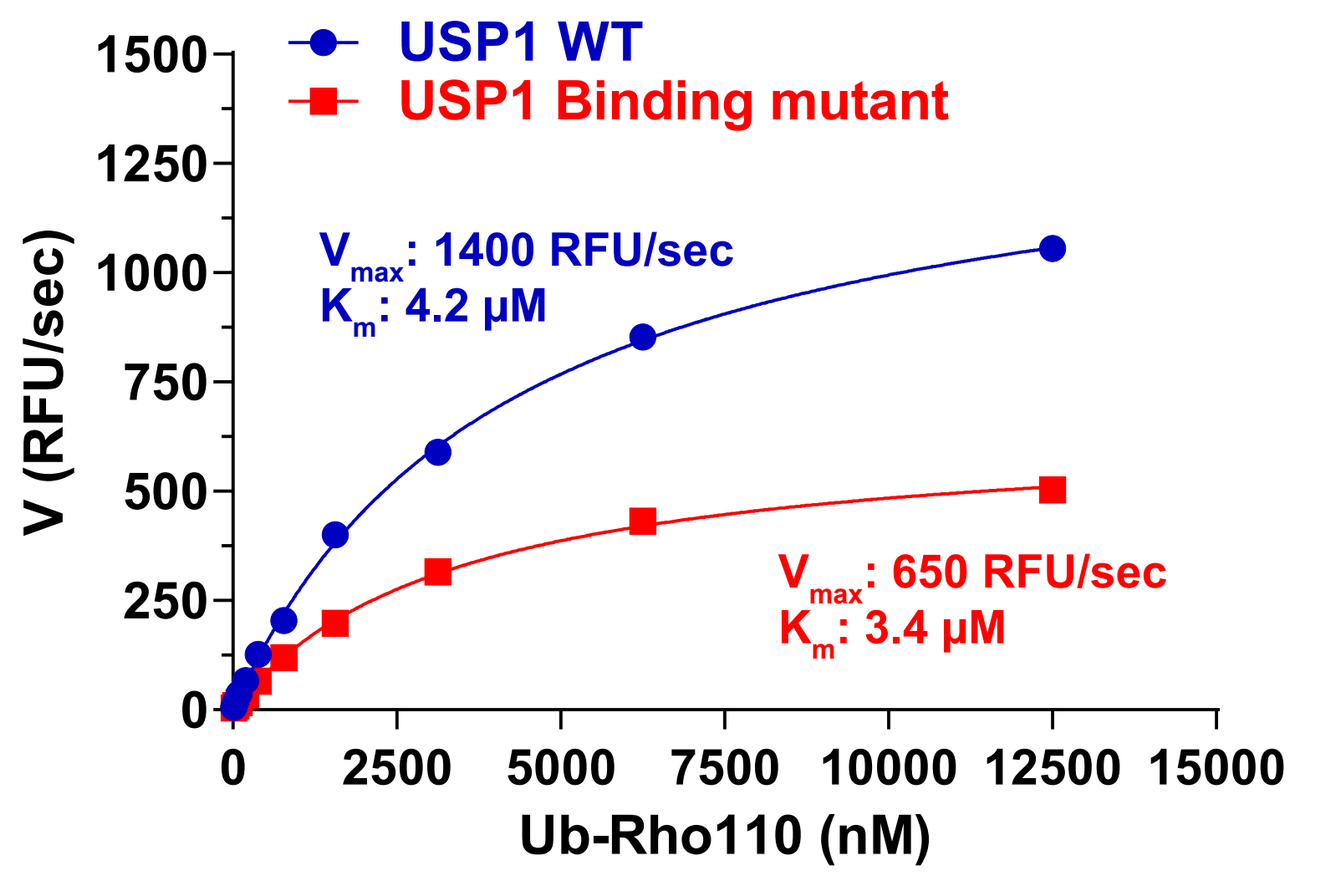
USP1 WT
USP1 Binding mutant
Vmax: 1400 RFU/sec
Km: 4.2 µM
Vmax: 650 RFU/sec
Km: 3.4 µM
V (RFU/sec)
Ub-Rho110 (nM)

TNG348 potency

ub-Rho110 assay

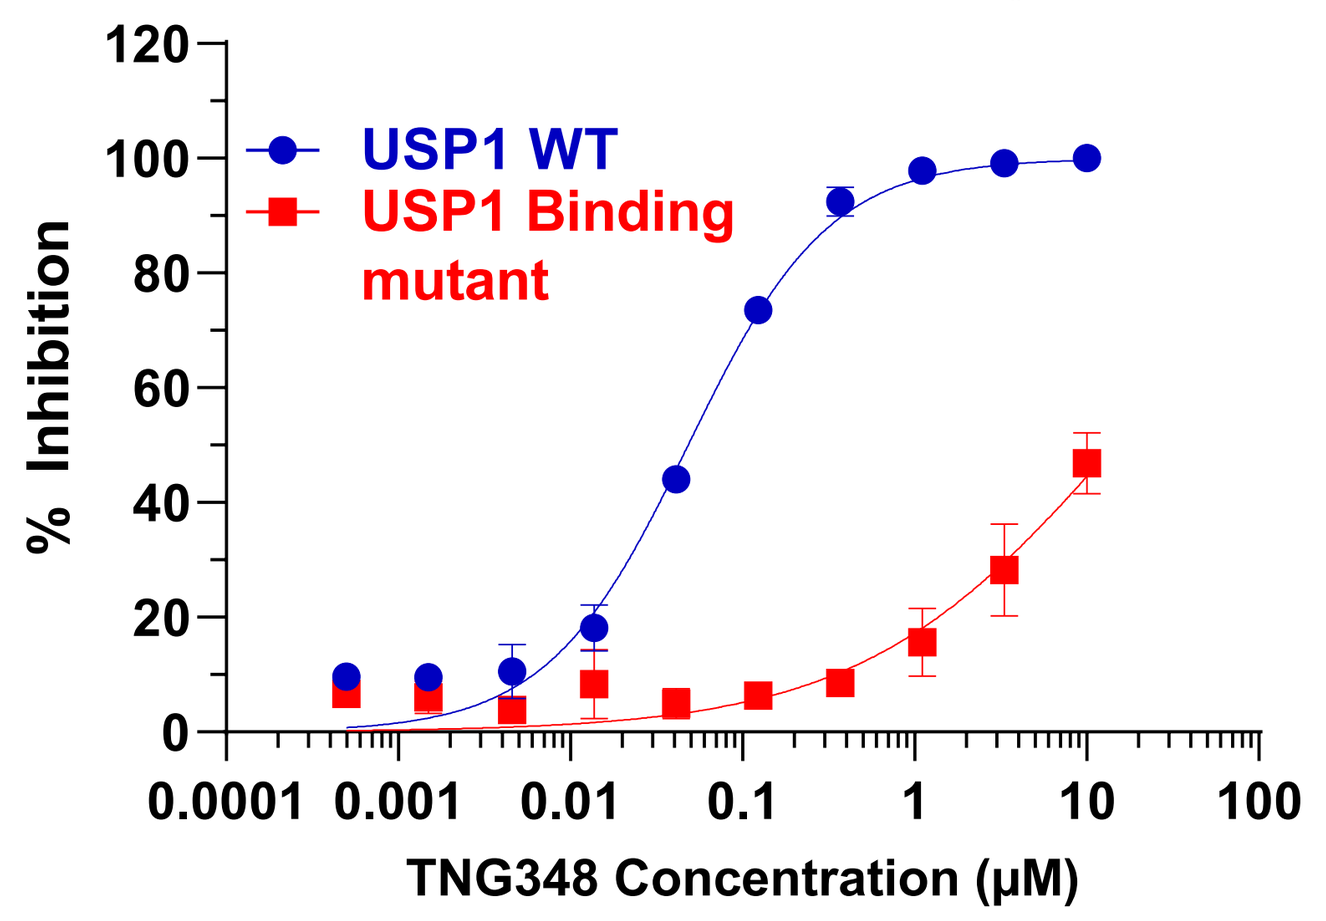
% Inhibition
USP1 WT
USP1 Binding mutant
TNG348 Concentration (µM)

Cell viability

MDA-MB-436

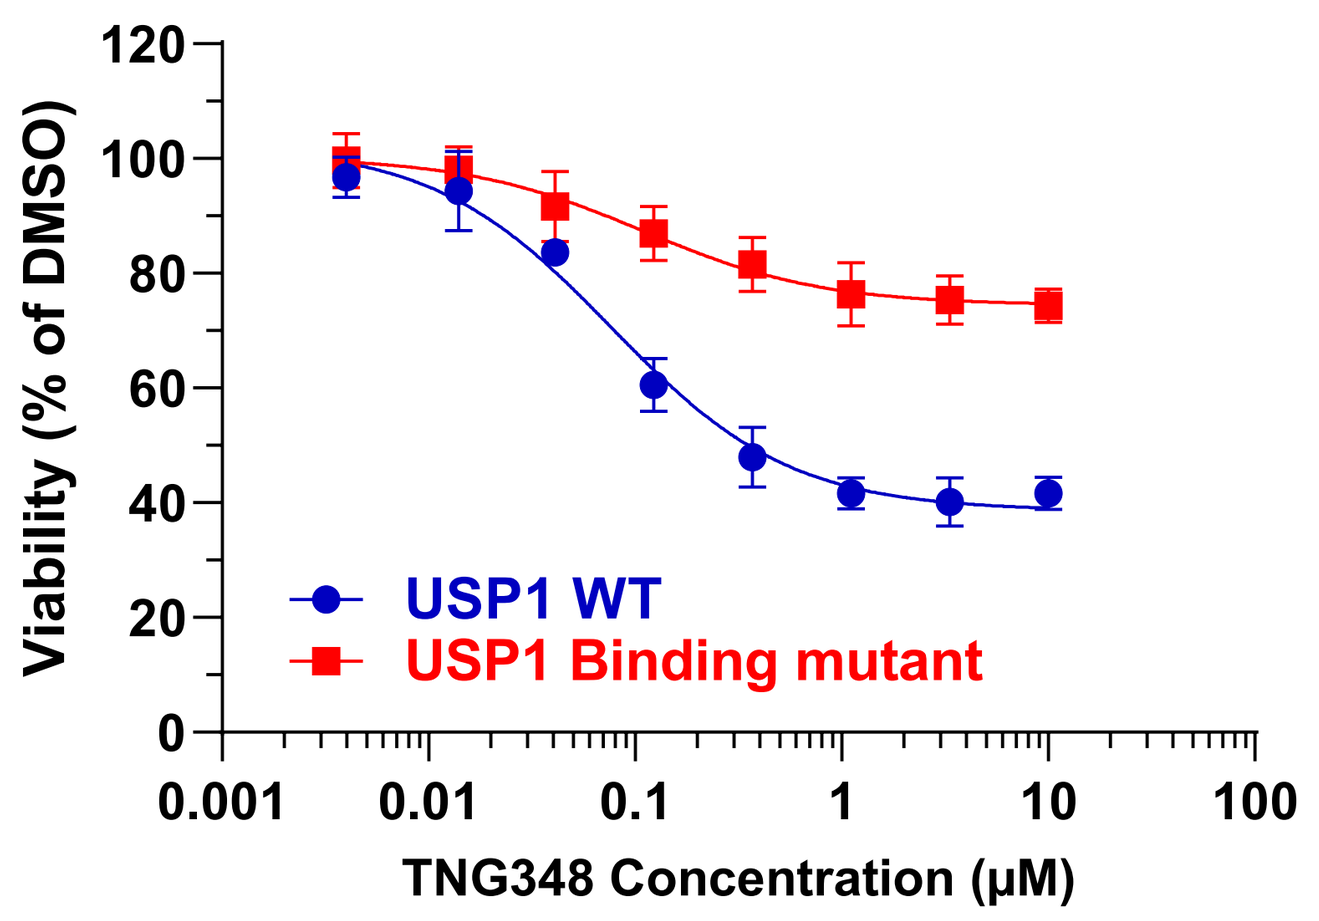
Viability (% of DMSO)
USP1 WT
USP1 Binding mutant
TNG348 Concentration (µM)

SUMMARY

- USP1 inhibition is synthetic lethal with BRCA1/2 mutations through a disctinct mechanism of action from PARPi
- Highly selective for USP1 in a panel of DUBs, consistent with its allosteric binding mode
- Single agent activity and strong PARPi synergy in breast and ovarian models with BRCA1/2 mutation or that are BRCA1/2wt but HRD+
- HRD+ cancers, including BRCA1/2 mutations, represent up to 50% of ovarian cancers, 25% of breast cancers, 10% of prostate cancers and 5% of pancreatic cancers
- Synergy in both PARPi-sensitive and -resistant models provides a clinical path in a large patient population
- TNG348 is clinical development candidate and we plan to file an IND in mid-2023

ACKNOWLEDGEMENTS

Chempartner, WuXi AppTec, Pharmaron, Xentech, XenoStart, Biortus
Champions Oncology, Crown Bioscience, Enamine