Experimental 'loss-of function' annotation of STK11 mutations with prognostic and therapeutic implications

Leanne Ahronian, Preksha Shahagadkar, Lauren Flynn, Lauren Grove, Shangtao Liu, Sam Meier, Binzhang Shen, Hannah Stowe, Hsin-Jung Wu, Yi Yu, Andre Mignault, Iga Sienczylo, Heather DiBenedetto, Silvia Fenoglio, Teng Teng

Abstract #5584

Introduction

Loss of function (LOF) mutations in Serine Threonine Kinase 11 (STK11) occur in 15% of lung adenocarcinoma and have been shown to drive resistance to immune checkpoint blockade clinically, as well as in preclinical models. Though STK11 is commonly inactivated in human cancer with strong implications for treatment outcomes, few STK11 mutations identified from tumor samples have been functionally characterized.

TNG260 is an inhibitor of CoREST that is currently being investigated in combination with pembrolizumab for the treatment of STK11-mutant cancer (NCT05887492). Patients are eligible for enrollment in the TNG260 phase 1/2 trial if their tumor contains a deleterious STK11 mutation. To begin classifying non-annotated variants, over 2,000 distinct mutations in the STK11 gene were identified from STK11 literature or public repositories of tumor sequencing data such as AACR Project GENIE and ClinVar. Where possible, loss-of-function annotations were captured from literature or predictive tools such as PolyPhen-2. However, many STK11 variants, particularly missense mutations, have never been functionally characterized.

We developed a functional screening approach to characterize STK11 alterations using the lung adenocarcinoma cell line A549. A549 cells contain homozygous loss of STK11 via a truncation mutation at Q37, and re-expression of wild-type STK11 in these cells strongly impairs their growth in vitro and in vivo. We created a library of STK11 variant cDNAs, each containing a unique barcode. This library was expressed in A549, and cells were maintained in vitro or in vivo to allow for positive selection of STK11 loss-of-function variants and depletion of variants that behave like wild-type STK11. At the end of the screen, variants were quantified by NGS using each mutant cDNA's unique barcode and compared to well-annotated controls.

These data were assembled to generate TNG260mutationfinder.com -- the first website to curate STK11 variants with functional annotations.

STK11 deficiency causes resistance to immune checkpoint blockade

In vivo CRISPR screen identifies STK11 as driver of α-PD1 resistance

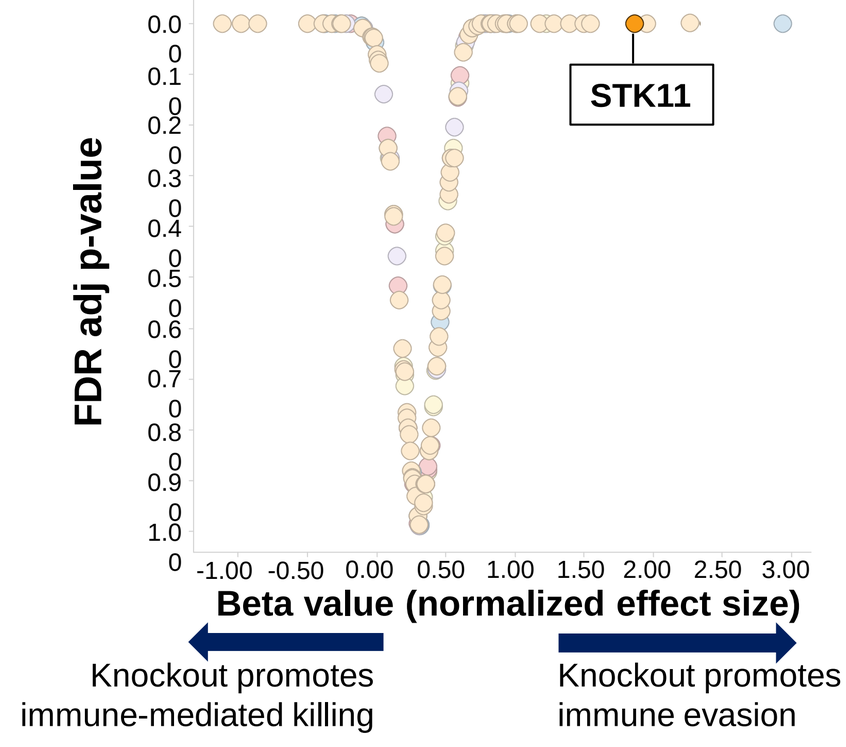
STK11
FDR adj p-value
Beta value (normalized effect size)
Knockout promotes immune-mediated killing
Knockout promotes immune evasion

MC38 - Control

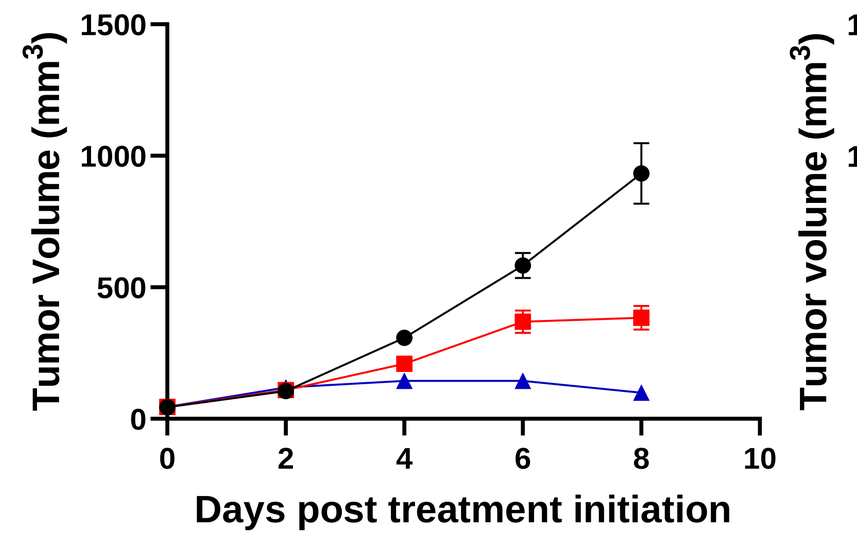
Tumor Volume (mm³)
Days post treatment initiation

MC38 - STK11 Deficient

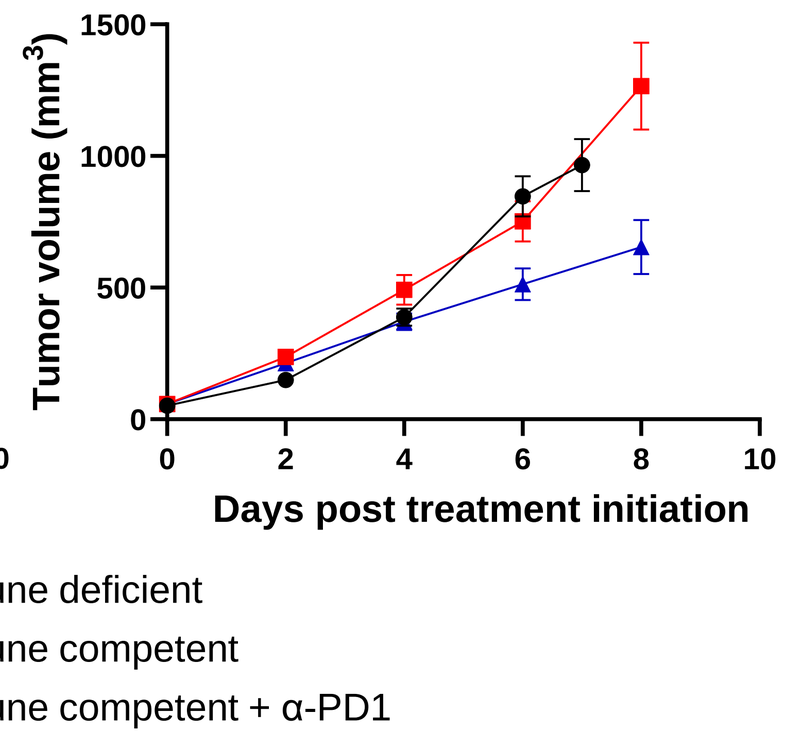
Tumor volume (mm³)
Days post treatment initiation
Immune deficient
Immune competent
Immune competent + α-PD1

STK11-mutated lung adenocarcinoma patients do not respond as well to immune checkpoint blockade as STK11 wild-type

TNG260 is a CoREST complex inhibitor that reverses α-PD1 resistance caused by loss of STK11

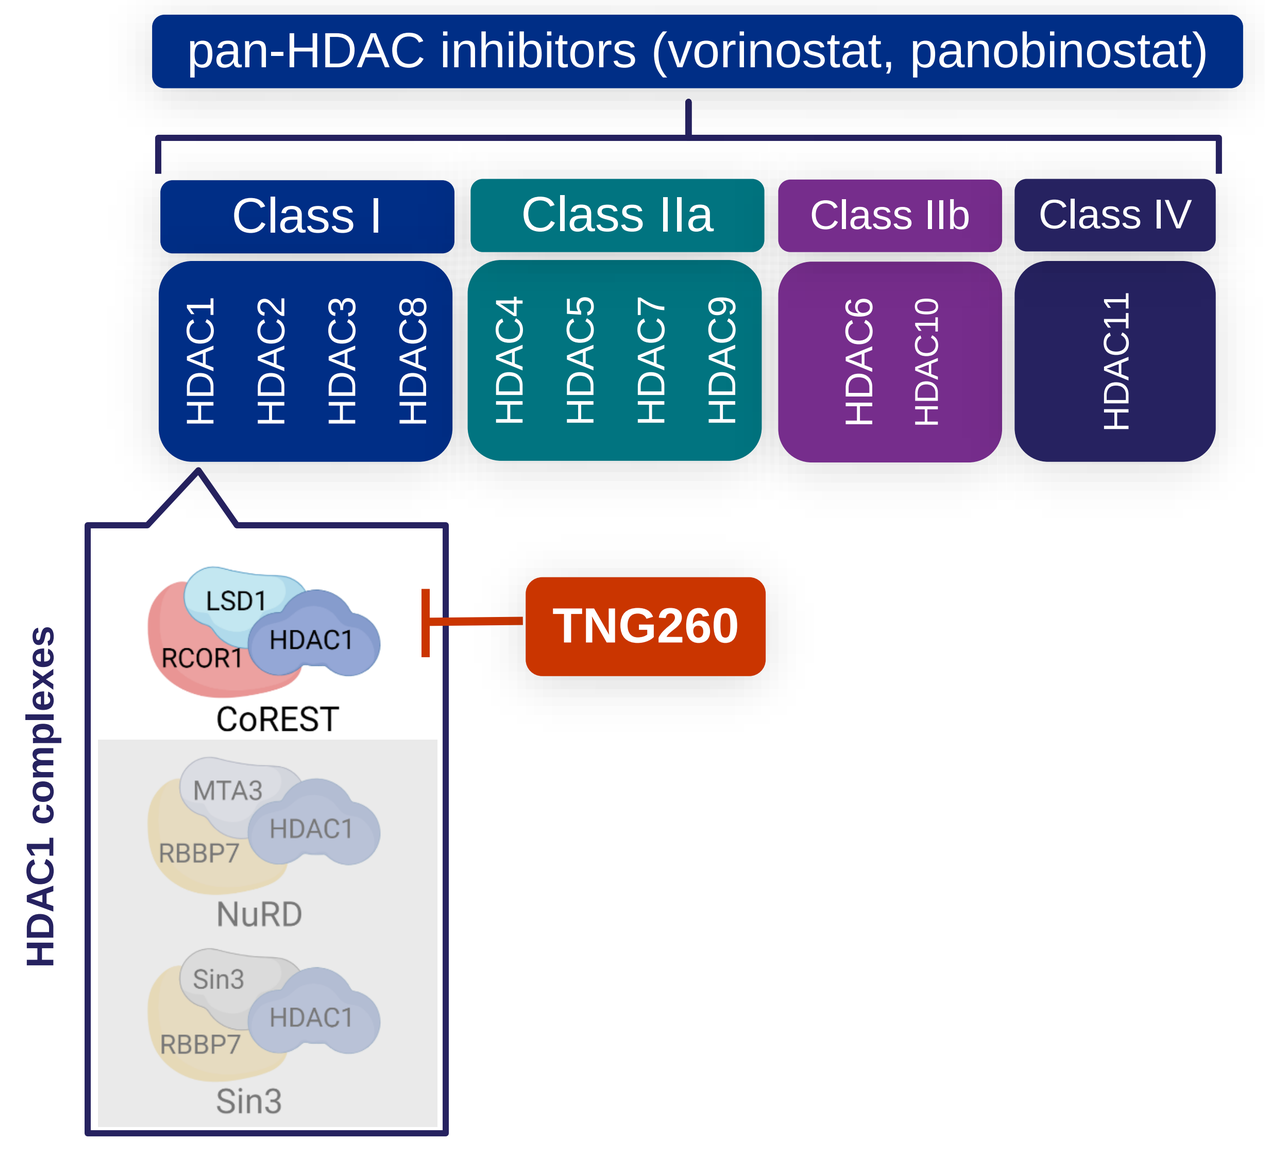
pan-HDAC inhibitors (vorinostat, panobinostat)
Class I
Class IIa
Class IIb
Class IV
HDAC1 HDAC2 HDAC3 HDAC8
HDAC4 HDAC5 HDAC7 HDAC9
HDAC6 HDAC10
HDAC11
HDAC1 complexes
LSD1 RCOR1 HDAC1
CoREST
MTA3 RBBP7 HDAC1
NuRD
Sin3 RBBP7 HDAC1
Sin3
TNG260

TNG260 is CoREST-selective

		CoREST	NCoR	NuRD	Sin3
Pan	Vorinostat	0.06	0.53	0.13	0.16
Class I	Tucidinostat	0.12	0.20	1.40	>100
	Domatinostat	0.09	0.19	30.0	>100
CoREST	TNG260	0.17	>100	>100	>100

STK11-null MC38 tumor model

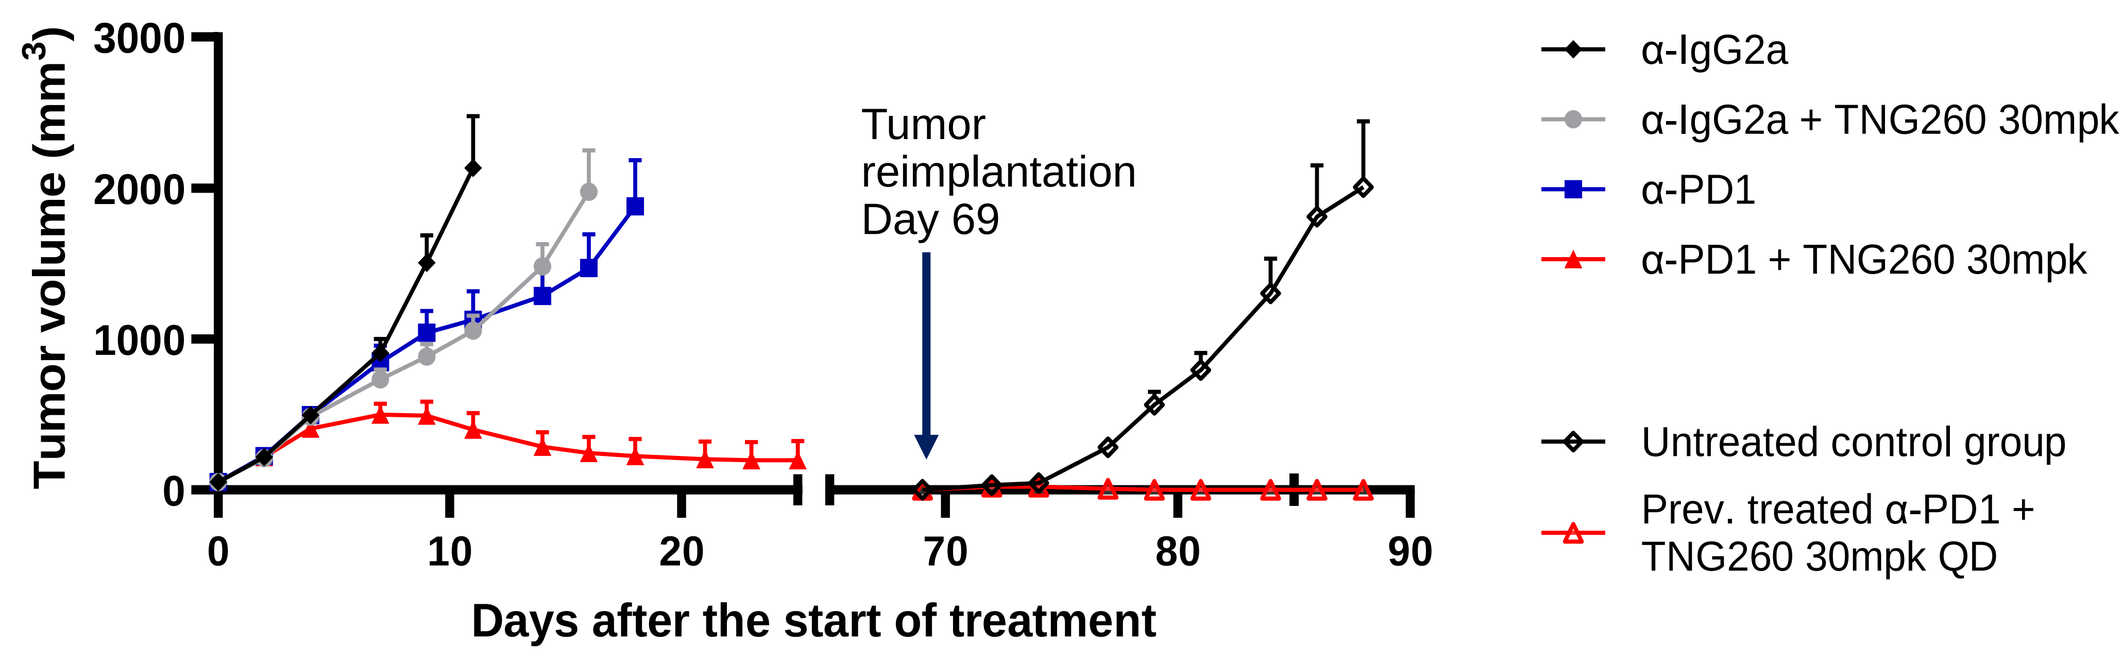
Tumor volume (mm³)
Tumor reimplantation Day 69
Days after the start of treatment
α-IgG2a
α-IgG2a + TNG260 30mpk
α-PD1
α-PD1 + TNG260 30mpk
Untreated control group
Prev. treated α-PD1 + TNG260 30mpk QD

5/8 mice had complete tumor regression by day 34

5/5 mice with complete regression rejected tumor reimplantation

Verifying STK11 loss-of-function status is critical for determining the preferred approach for patient treatment

% Frequency of STK11 mutations

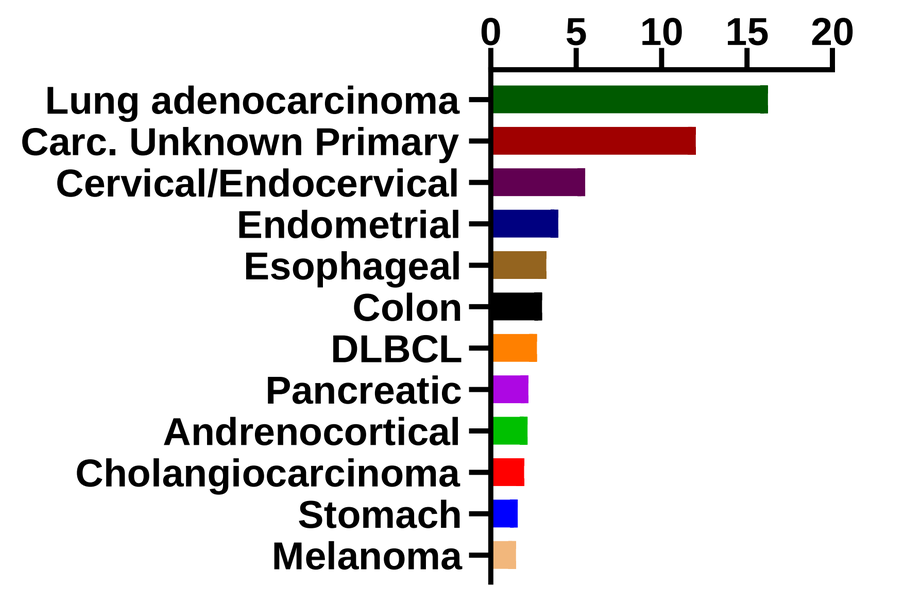
Lung adenocarcinoma
Carc. Unknown Primary
Cervical/Endocervical
Endometrial
Esophageal
Colon
DLBCL
Pancreatic
Andrenocortical
Cholangiocarcinoma
Stomach
Melanoma

STK11 variants in lung adenocarcinoma

Variant Type	Frequency
Missense	22%
Nonsense	20.8%
Frameshift (indel)	31.1%
Splicing	7.7%
Deletion	17%
Rearrangement	1.4%

STK11 missense mutations in lung adenocarcinoma

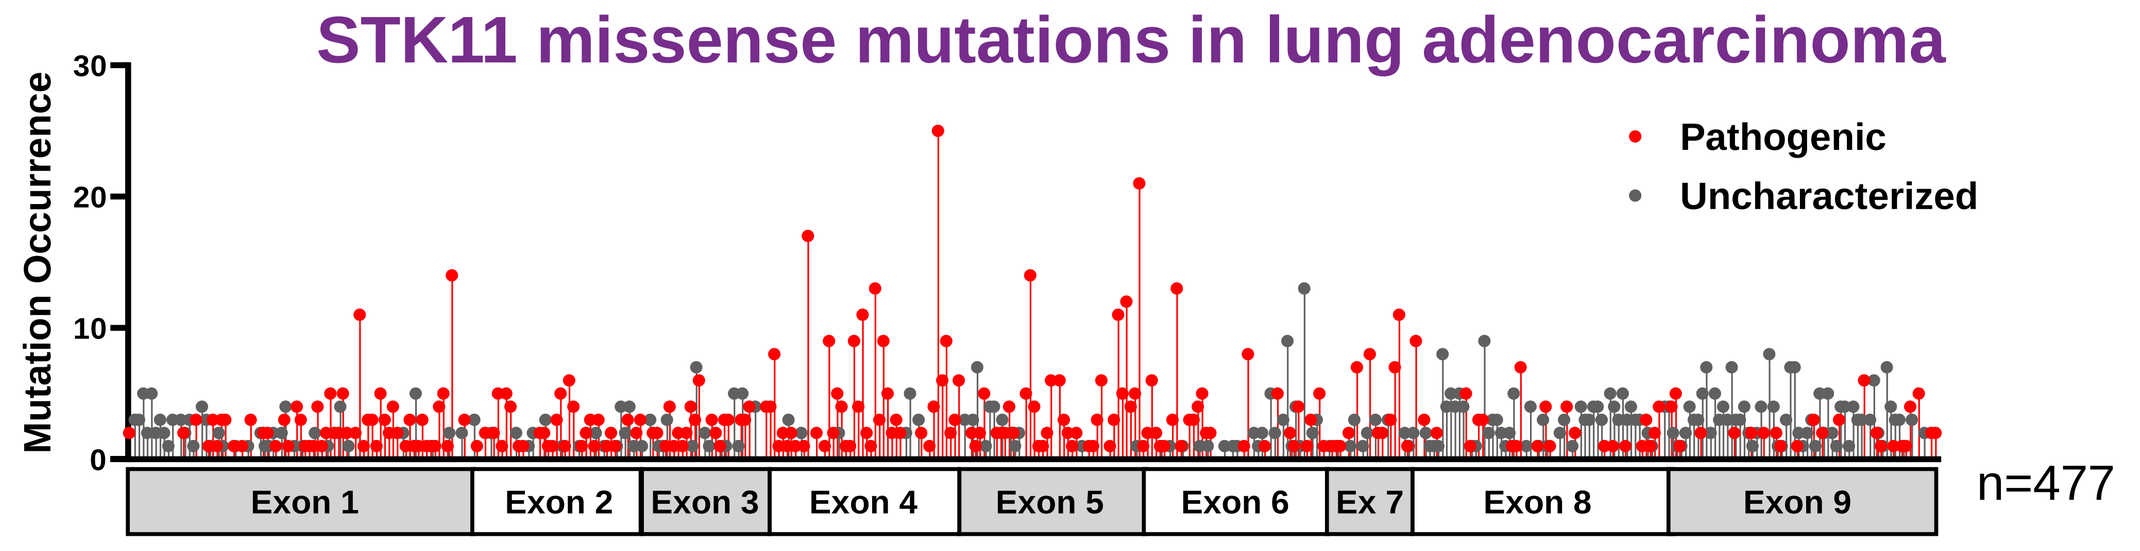
Mutation Occurrence
Pathogenic
Uncharacterized
Exon 1
Exon 2
Exon 3
Exon 4
Exon 5
Exon 6
Ex 7
Exon 8
Exon 9
n=477

Existing computational algorithms do not agree on pathogenicity of STK11 missense mutations identified from the literature

Alpha Missense Pathogenicity score v. Ensemble score

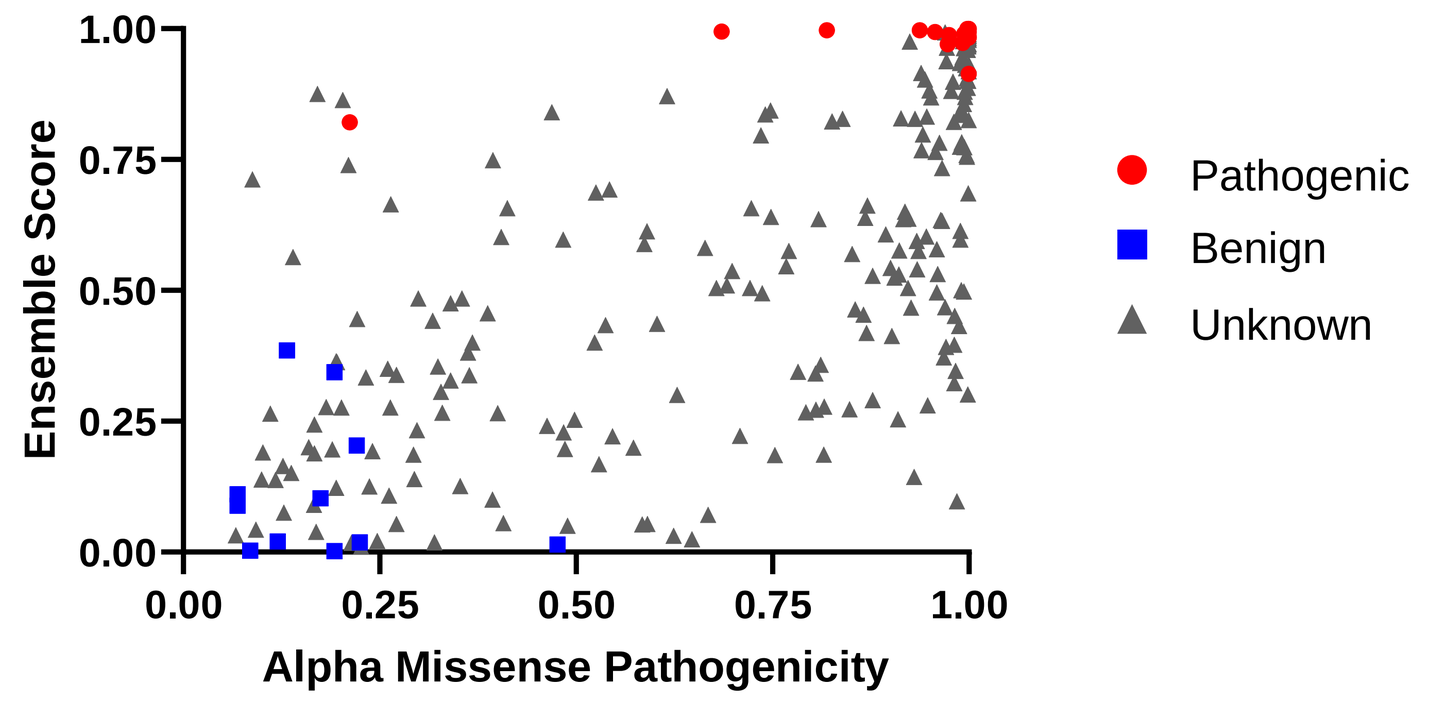
Ensemble Score
Alpha Missense Pathogenicity
Pathogenic
Benign
Unknown

Design of a functional enrichment screen to annotate patient-derived STK11 mutations for TNG260 clinical trial

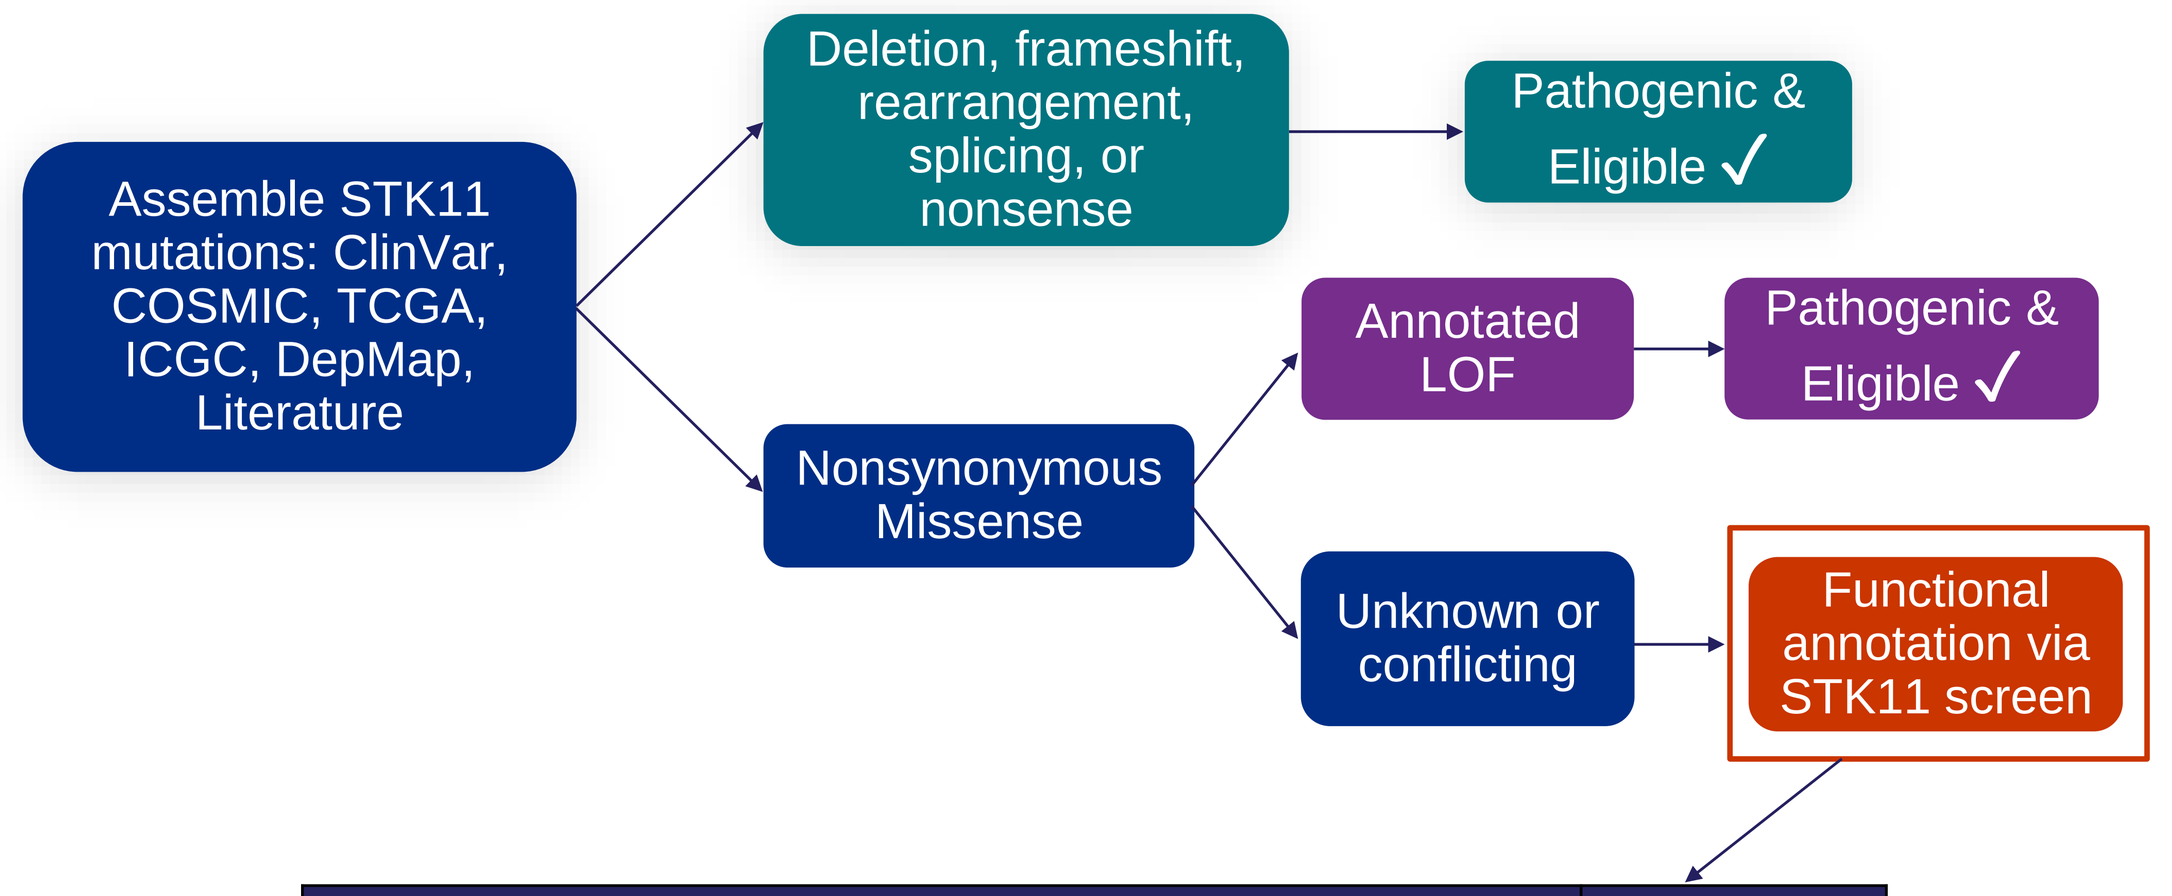
Assemble STK11 mutations: ClinVar, COSMIC, TCGA, ICGC, DepMap, Literature
Deletion, frameshift, rearrangement, splicing, or nonsense
Pathogenic & Eligible ✓
Nonsynonymous Missense
Annotated LOF
Pathogenic & Eligible ✓
Unknown or conflicting
Functional annotation via STK11 screen

Category	# of cDNAs
EGFP	3
STK11 wild-type	3
Benign mutants	3
Annotated pathogenic mutants	20
Mutants with conflicting data	15
Truncations near end of sequence	3
Pathogenic computational score, but no supporting data	252

A549 3D Culture

Cells expressing STK11 LOF mutations will enrich

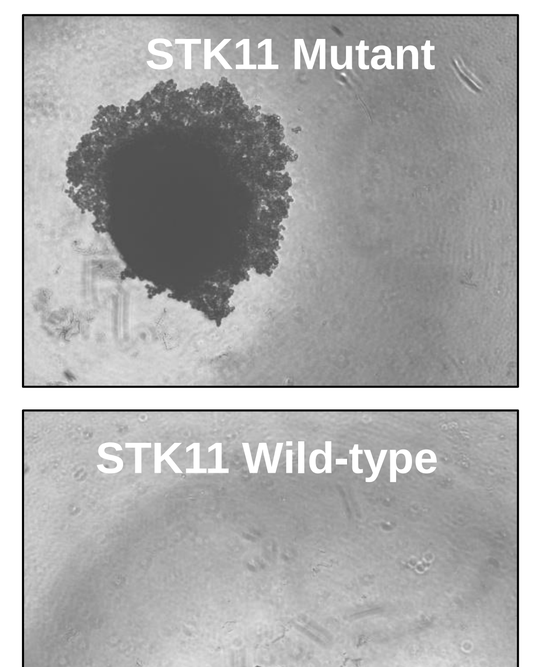
STK11 Mutant
STK11 Wild-type

A549 Xenografts

Cells expressing STK11 LOF mutations will enrich

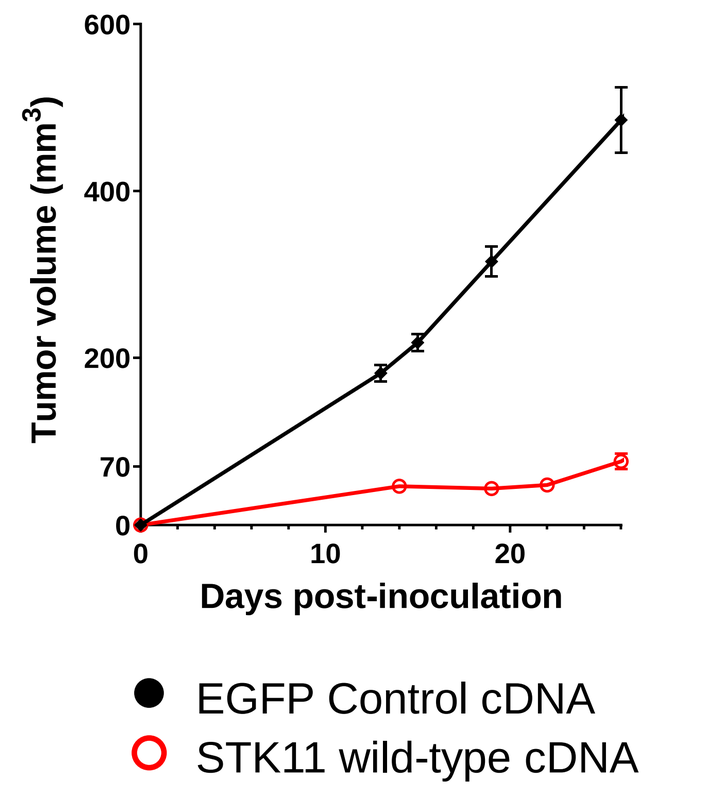
Tumor volume (mm³)
Days post-inoculation
EGFP Control cDNA
STK11 wild-type cDNA

STK11 Enrichment Screen

Cells with LOF variants will outcompete and enrich compared to benign variants

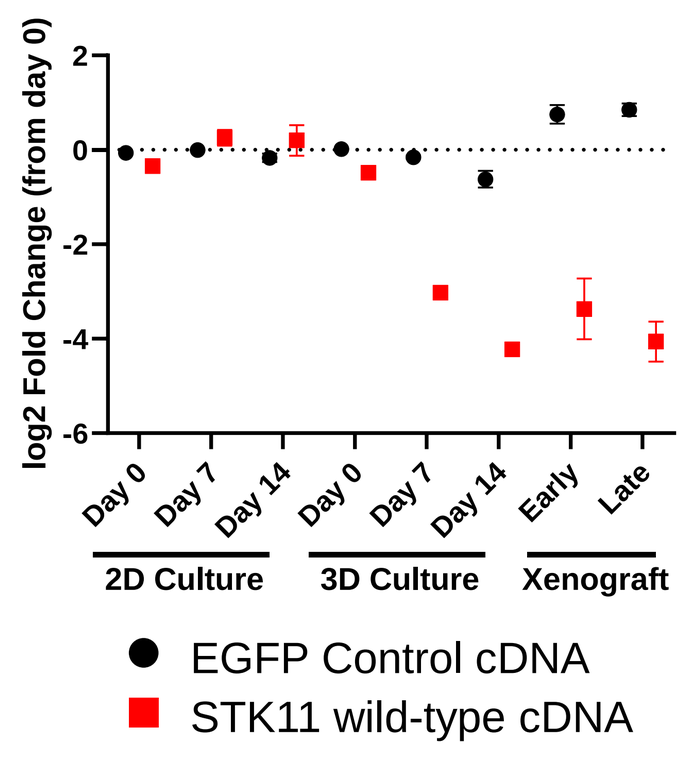
log2 Fold Change (from day 0)
Day 0
Day 7
Day 14
Early
Late
2D Culture
3D Culture
Xenograft
EGFP Control cDNA
STK11 wild-type cDNA

STK11 functional screen performed in 3D culture and xenografts

Xenograft Tumor Volume

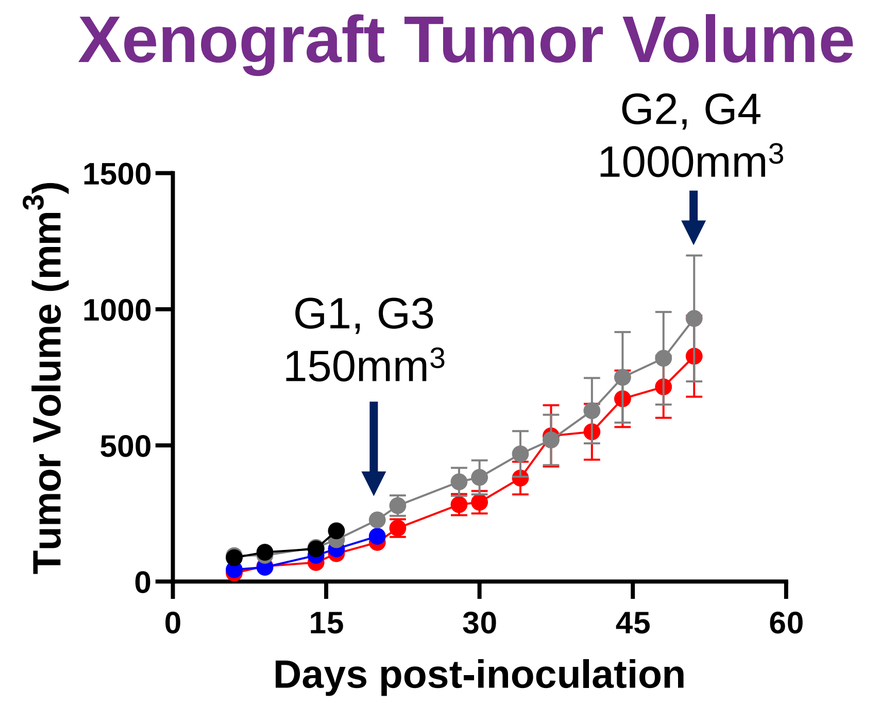
G2, G4 1000mm³
G1, G3 150mm³
Tumor Volume (mm³)
Days post-inoculation

Xenograft Tumor Weight

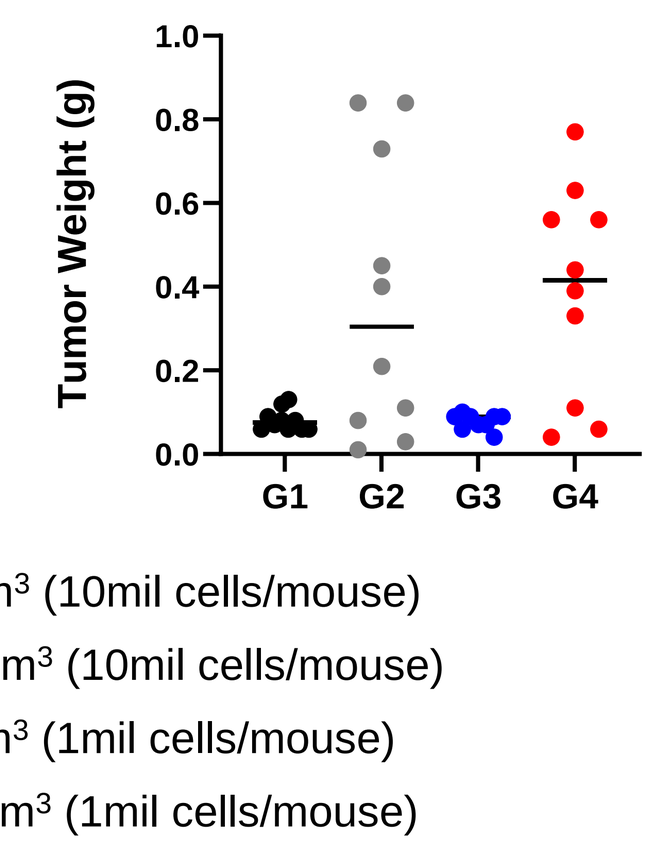
Tumor Weight (g)
G1
G2
G3
G4
G1 - 150mm³ (10mil cells/mouse)
G2 - 1000mm³ (10mil cells/mouse)
G3 - 150mm³ (1mil cells/mouse)
G4 - 1000mm³ (1mil cells/mouse)

3D in vitro samples were collected at day 0, day 7 and day 14 of growth in 3D culture. In vivo xenograft samples were collected at 150mm³ and 1000mm³, representing "early" and "late" timepoints, respectively. Samples were prepared for NGS and sequenced for a unique barcode representing each STK11 variant.

Functional screen enriches true loss-of-function STK11 mutations over benign variants

Functional screen enriches positive control loss of function variants compared to benign variants or wild-type STK11

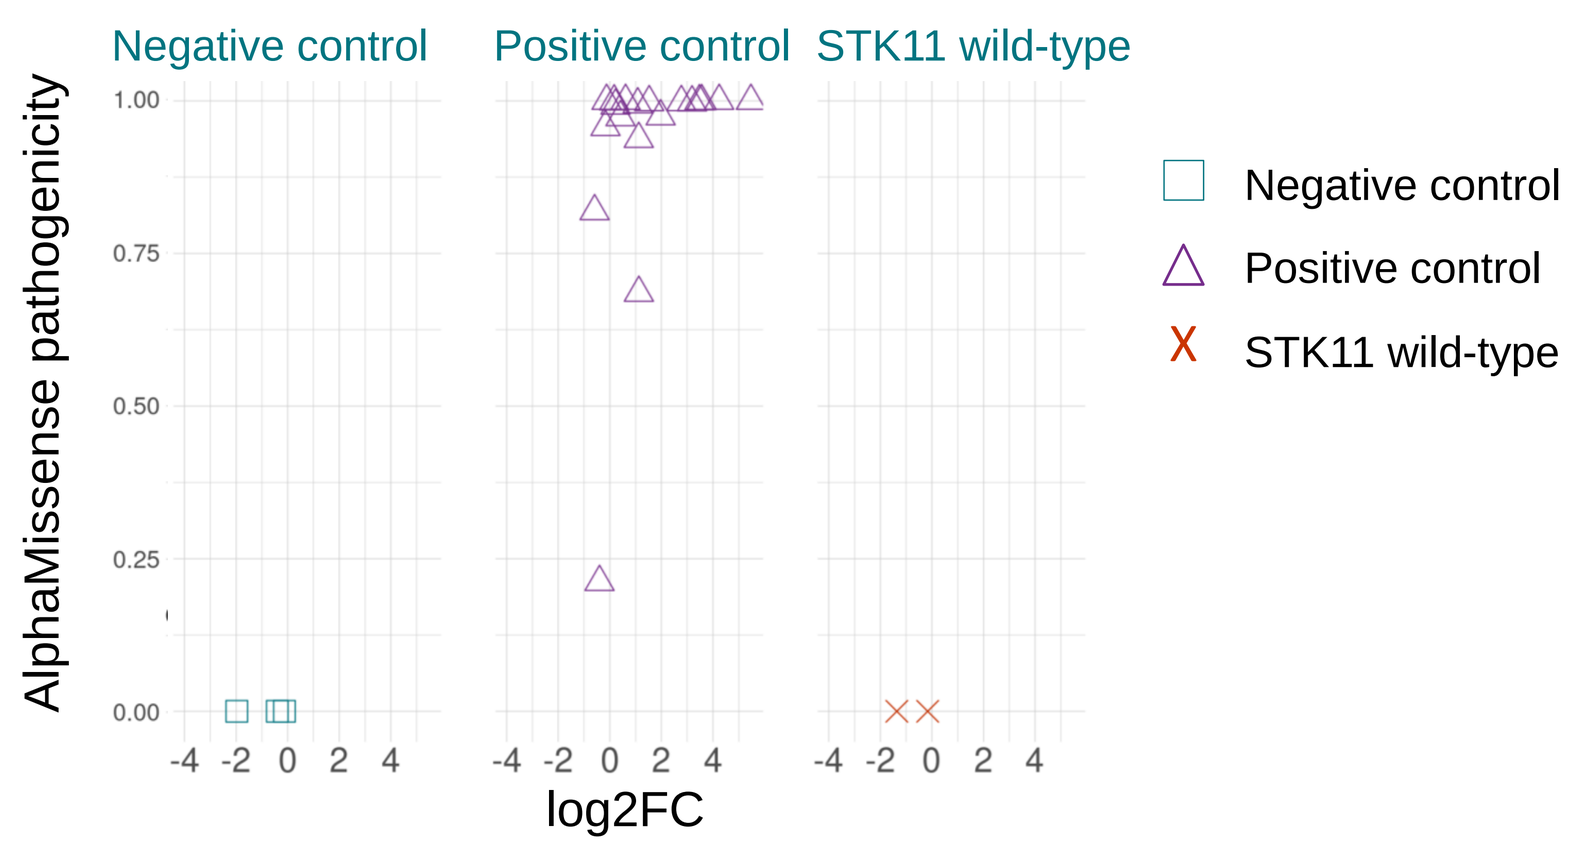
Negative control
Positive control
STK11 wild-type
AlphaMissense pathogenicity
log2FC
Negative control
Positive control
STK11 wild-type

Each STK11 variant was compared to day 0 to determine the log2 fold change (log2FC) of each variant. Log2FC values that exceed the WT and benign controls indicate pathogenic variants. Pathogenicity scores from each missense mutation were determined by AlphaMissense and plotted against log2FC.

Screen enriches for true loss of function STK11 variants and not benign variants

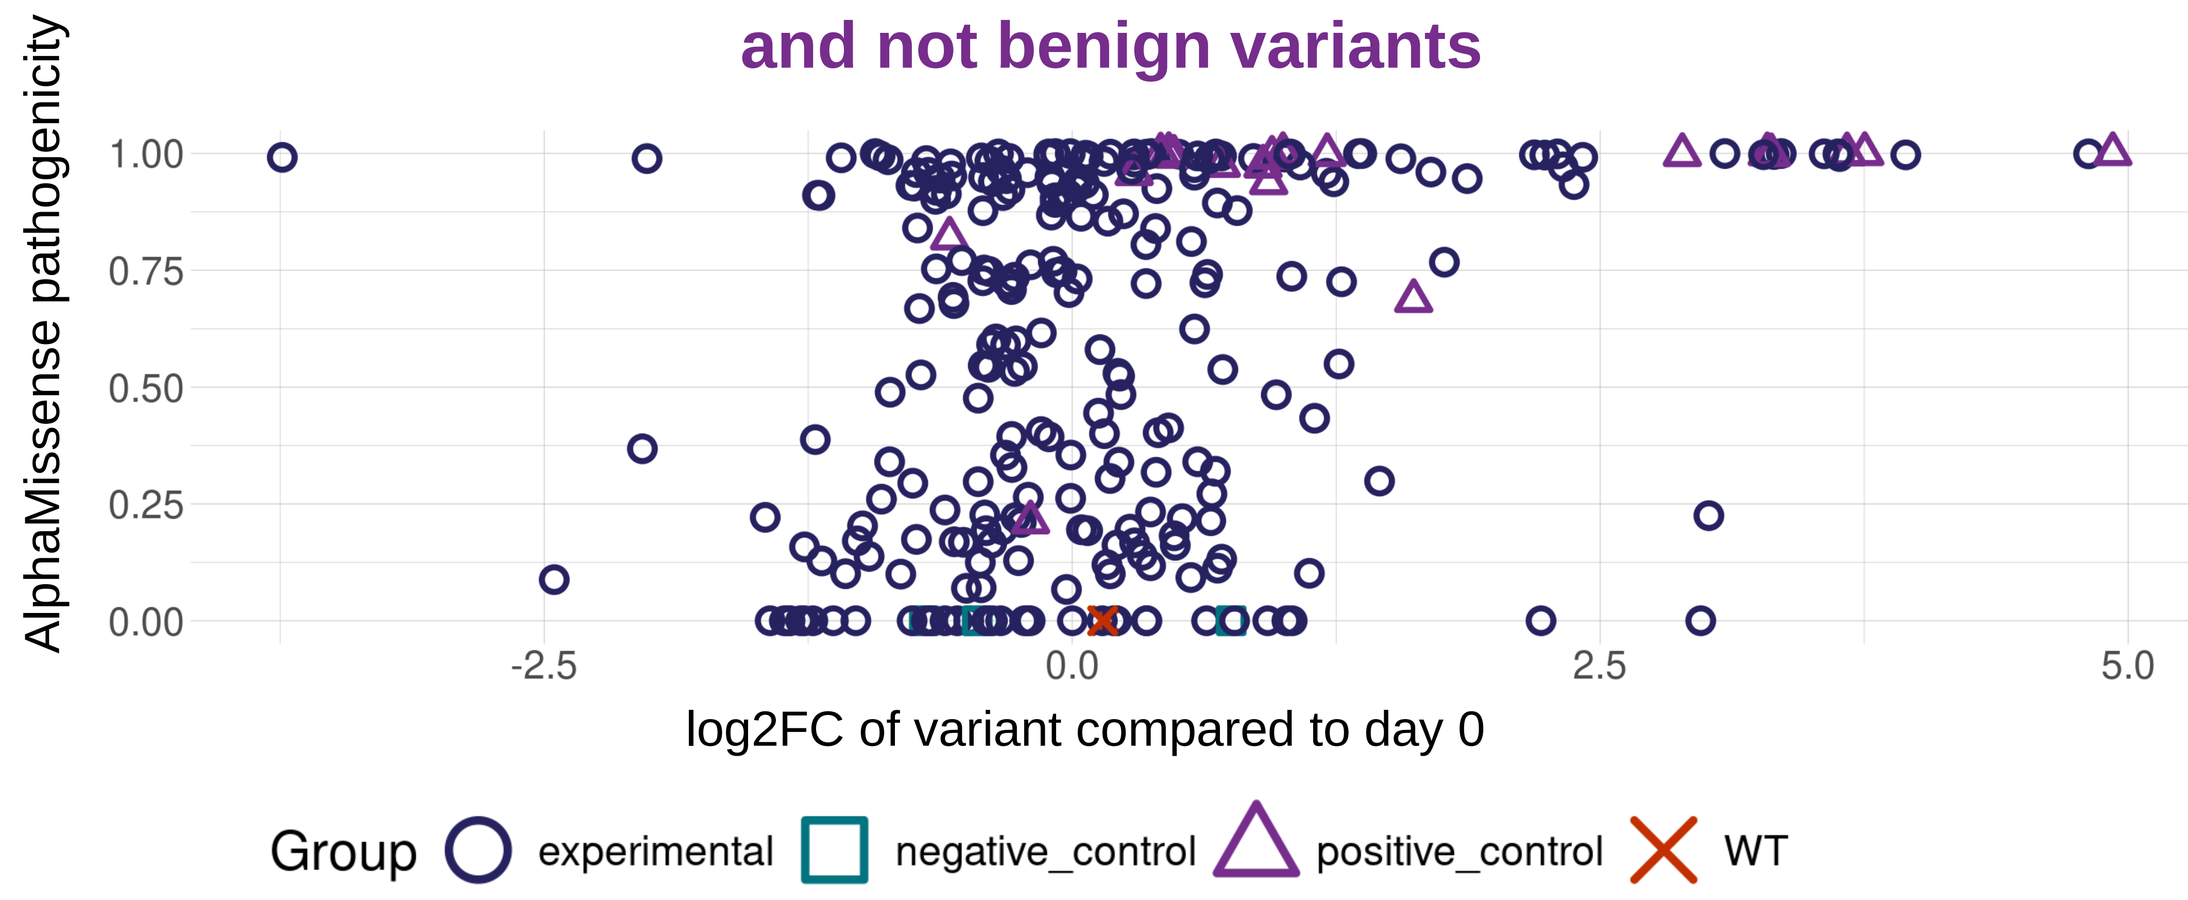
AlphaMissense pathogenicity
log2FC of variant compared to day 0
Group
experimental
negative_control
positive_control
WT

Each STK11 variant was compared to day 0 to determine the log2 fold change (log2FC) of each variant. Pathogenic variants are those with log2FC values that exceed wild-type STK11 and negative controls. Pathogenicity scores from each missense mutation were determined by AlphaMissense and plotted against log2FC.

Change in STK11 mutation frequency over time

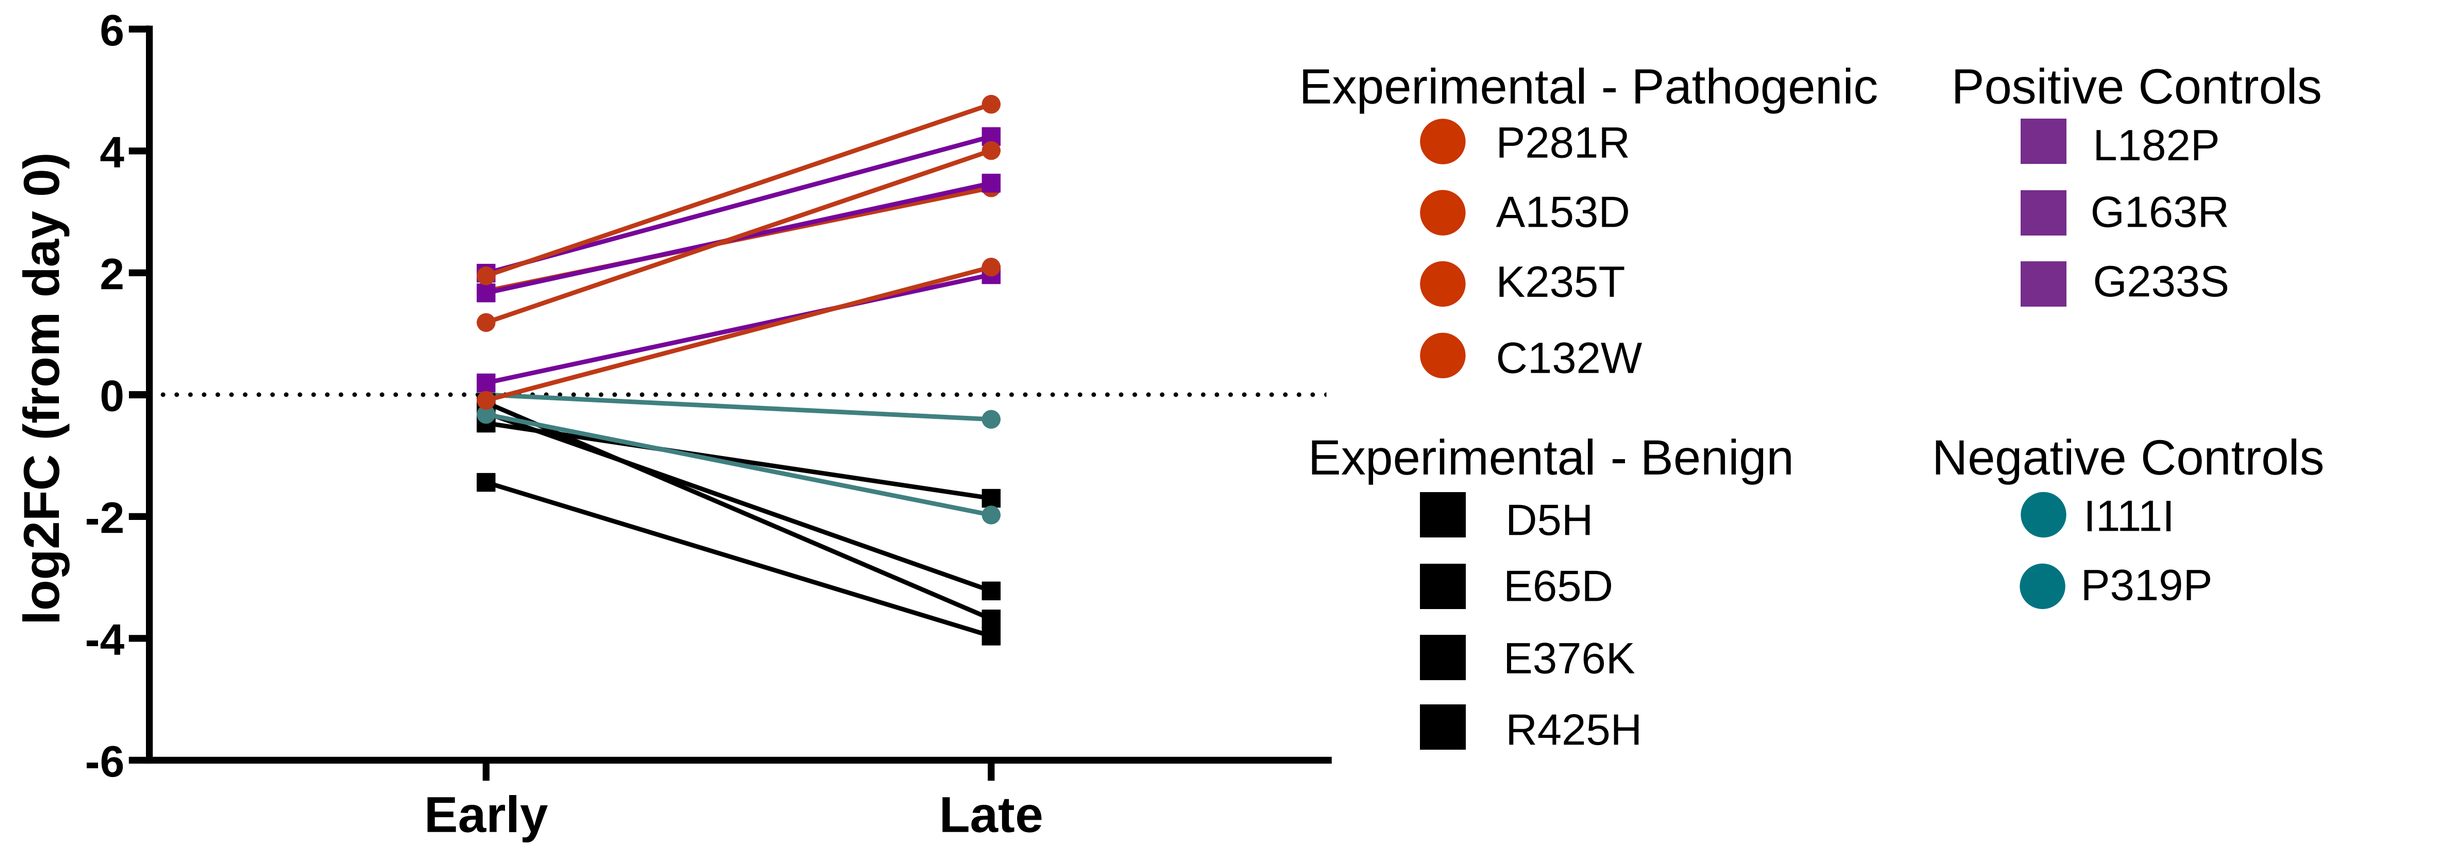
log2FC (from day 0)
Early
Late
Experimental - Pathogenic
P281R
A153D
K235T
C132W
Positive Controls
L182P
G163R
G233S
Experimental - Benign
D5H
E65D
E376K
R425H
Negative Controls
I111I
P319P

Log2 fold change in representative STK11 variants between "early" and "late" timepoints. Early tumors were collected at 150mm³, and late timepoint tumors were collected at 1000mm³. Positive controls enriched during the screen, while negative controls were static or depleted. Experimental mutations were pathogenic if log2FC values increased over time, while experimental mutations were benign if they stayed static or depleted over time.

Annotation of novel pathogenic missense mutations in STK11

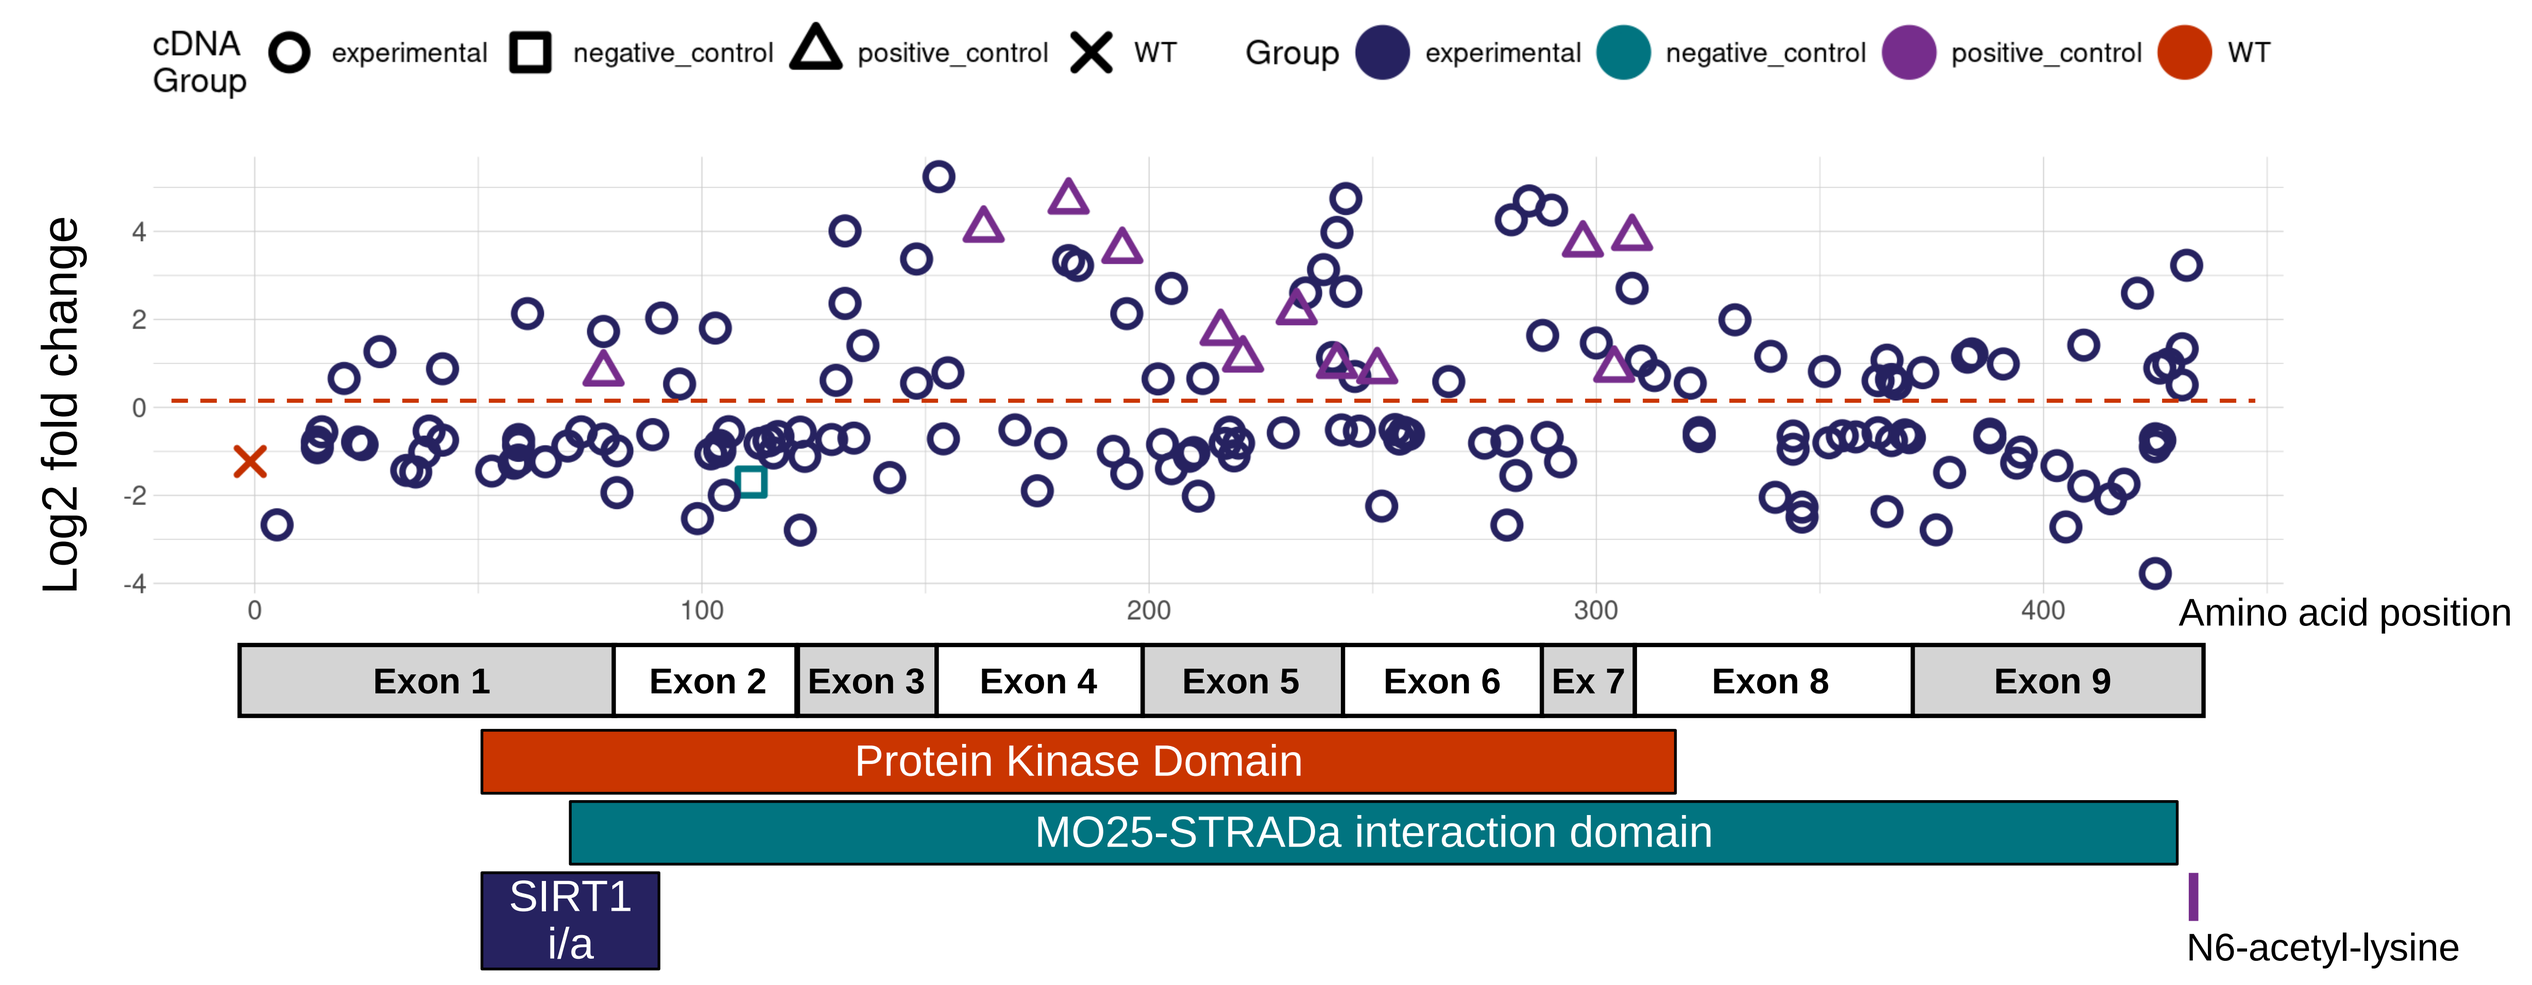
cDNA Group
experimental
negative_control
positive_control
WT
Group
Log2 fold change
Amino acid position
Exon 1
Exon 2
Exon 3
Exon 4
Exon 5
Exon 6
Ex 7
Exon 8
Exon 9
Protein Kinase Domain
MO25-STRADa interaction domain
SIRT1 i/a
N6-acetyl-lysine

Each STK11 variant was compared to day 0 to determine its log2 fold change (log2FC). Each variant's log2FC was plotted by amino acid sequence of STK11, shown with corresponding domains. Variants that fall above the red dotted line are pathogenic.

Benign Mutations		Pathogenic Mutations	
R405W	E376K	L290P	C132R
K81N	R409P	T244P	I300N
K122R	K175R	L285P	F148S
R415L	V34F	C132W	K235T
I99T	A205P	A153D	L182R
L252M	L105S	G242E	G61S
Y340H	G346S	P281R	
D5H	E65D	L184P	
P280L	C418Y	T244N	
D53N	G346V	W308R	
R425H		G288V	

TNG260mutationfinder.com, a searchable database of 2600 STK11 loss-of-function variants identified from patient tumors

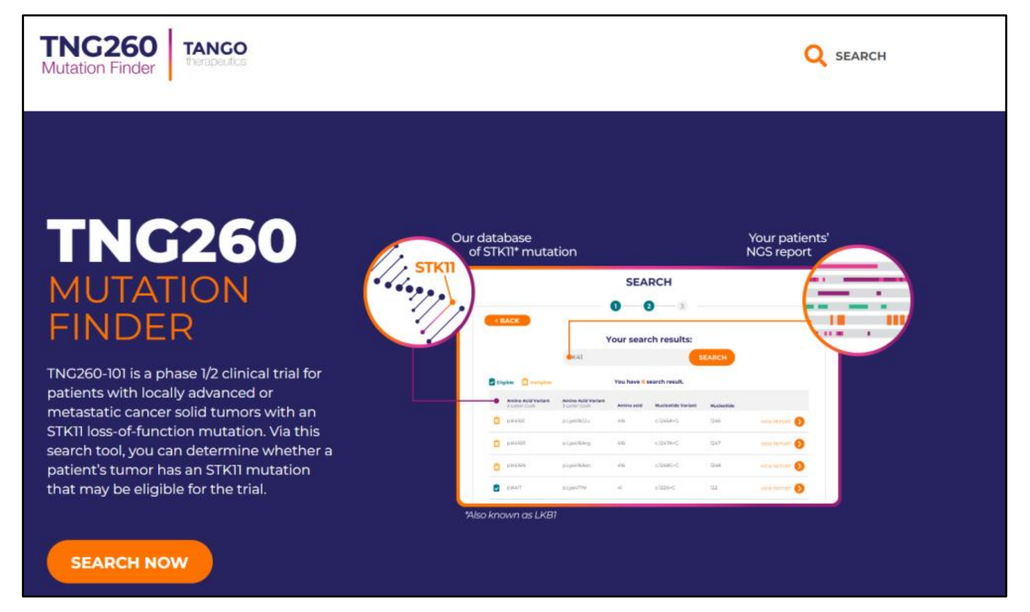
TNG260 MUTATION FINDER
SEARCH NOW

TNG260 Mutation Finder

Summary

- **Tumors with STK11 loss of function are resistant to immune checkpoint blockade**
- **STK11 is altered in 15% of NSCLC, but STK11 missense mutations are poorly annotated**
- **The functional screen identified LOF mutations that had low pathogenicity scores, and vice versa – indicating that functional validation of in silico analyses are necessary for STK11 missense variants**
- **Data was assembled to generate TNG260mutationfinder.com, a repository of patient-derived STK11 alterations with pathogenic or benign calls**
- **Annotation of STK11 is critical for identifying patients with true STK11 LOF mutations which can help inform treatment options**
- **STK11 mutation is one criterion used to inform patient eligibility for NCT05887492, a clinical trial studying TNG260 + pembrolizumab in patients with STK11-mutated solid tumors**

ACKNOWLEDGEMENTS

We acknowledge the hard work and contribution of Tiziani Whitmyre, Inc., for the construction and design of TNG260mutationfinder.com. We also thank the Charles River Accelerator and Development lab for their vivarium services.

REFERENCES

Alexander, B., Sokol, E. S., Danziger, N. A., Pavlick, D. C., Elvin, J., Killian, J. K., et al. *Annals of Oncology* 2020;31, S283–S284.

Ricciuti B, Arbour KC, Lin JJ, Vajdi A, Vokes N, Hong L, et al. *J Thorac Oncol*. 2022;17:399–410.

Skoulidis F, Goldberg ME, Greenawalt DM, Hellmann MD, Awad MM, Gainor JF, et al. *Cancer Discov*. 2018;8:CD-18-0099.

TCGA Research Network: www.cancer.gov/tcga