TNG260, a CoREST-selective deacetylase inhibitor, reverses anti-PD1 resistance driven by loss of STK11

Leanne G. Ahronian, Minjie Zhang, Chengyin Min, Alice W. Tsai, Jacques Ermolieff, Patrick McCarren, Margaret Wyman, David Guerin, Ye Wang, Alborz Bejnood, Kenjie Amemiya, Brian McMillan, Nikitha Das, Brian Doyon, Andre Mignault, Colin Liang, Vassil Elitzin, Samuel R. Meier, Ashley Choi, Yi Yu, John P. Maxwell, Alan Huang, Xinyuan Wu

ABSTRACT #
444

ABSTRACT

Background: Histone deacetylase 1 (HDAC1) was identified from a novel in vivo CRISPR screening platform as a target gene whose inhibition reverses α-PD1 resistance driven by loss of STK11. Histone deacetylases are a well-studied class of oncology drug targets, but existing non-isoform-selective HDAC inhibitors have few approved clinical applications due to toxicities that limit sufficient exposure in solid tumors. Our data suggest that HDAC3, an essential gene, is a primary driver of bone marrow toxicity caused by HDAC inhibitors that target multiple isoforms.

Methods: We discovered and developed TNG260, a small molecule which inhibits HDAC1 with 10-fold selectivity over HDAC3 in cells, and 500-fold selectivity for the CoREST complex over the other HDAC1-containing complexes, NuRD and Sin3.

Results: Treatment of an α-PD1 resistant STK11-mutant MC38 syngeneic tumor model with TNG260 re-sensitizes this model to treatment with α-PD1. The combination of TNG260 and α-PD1 led to durable complete tumor regressions in the majority of treated animals. All mice with complete responses remained tumor-free until tumor rechallenge (21 days) and rejected engraftment of tumor cells. Unlike previously developed HDAC inhibitors designed for tumor cell cytotoxicity, TNG260 has no anti-tumor efficacy in immunocompromised mice, indicating the tumor cell killing with TNG260 is immune-mediated and not due to direct cell killing. Immune profiling of tumors following treatment with TNG260 and α-PD1 showed a decoupling of $T_{effector}$ and $T_{regulatory}$ cell recruitment caused by α-PD1 monotherapy, leading to a more active immune microenvironment. TNG260 also decreased intratumoral infiltration of neutrophils, an immune suppressive cell type associated with STK11-mutant NSCLC. Toxicity profiling of TNG260 shows it has less viability impact on erythroid and myeloid cells in vitro than other HDAC inhibitors, and in vivo toxicity studies showed bone marrow suppression only at TNG260 doses that are no longer selective for HDAC1/2.

Conclusions: TNG260 is a potent, highly selective small molecule CoREST inhibitor with good drug-like properties. It reverses the immune evasion phenotype caused by loss of STK11 and induces tumor regressions in an STK11-mutant model in combination with α-PD1. The TNG260 clinical development plan will be among the first to combine the power of genetic patient selection and immunotherapy, evaluating patients with STK11 mutant cancers in a trial combining TNG260 and a checkpoint inhibitor.

HDAC1 inhibition re-sensitizes STK11-deleted tumors to immune checkpoint blockade

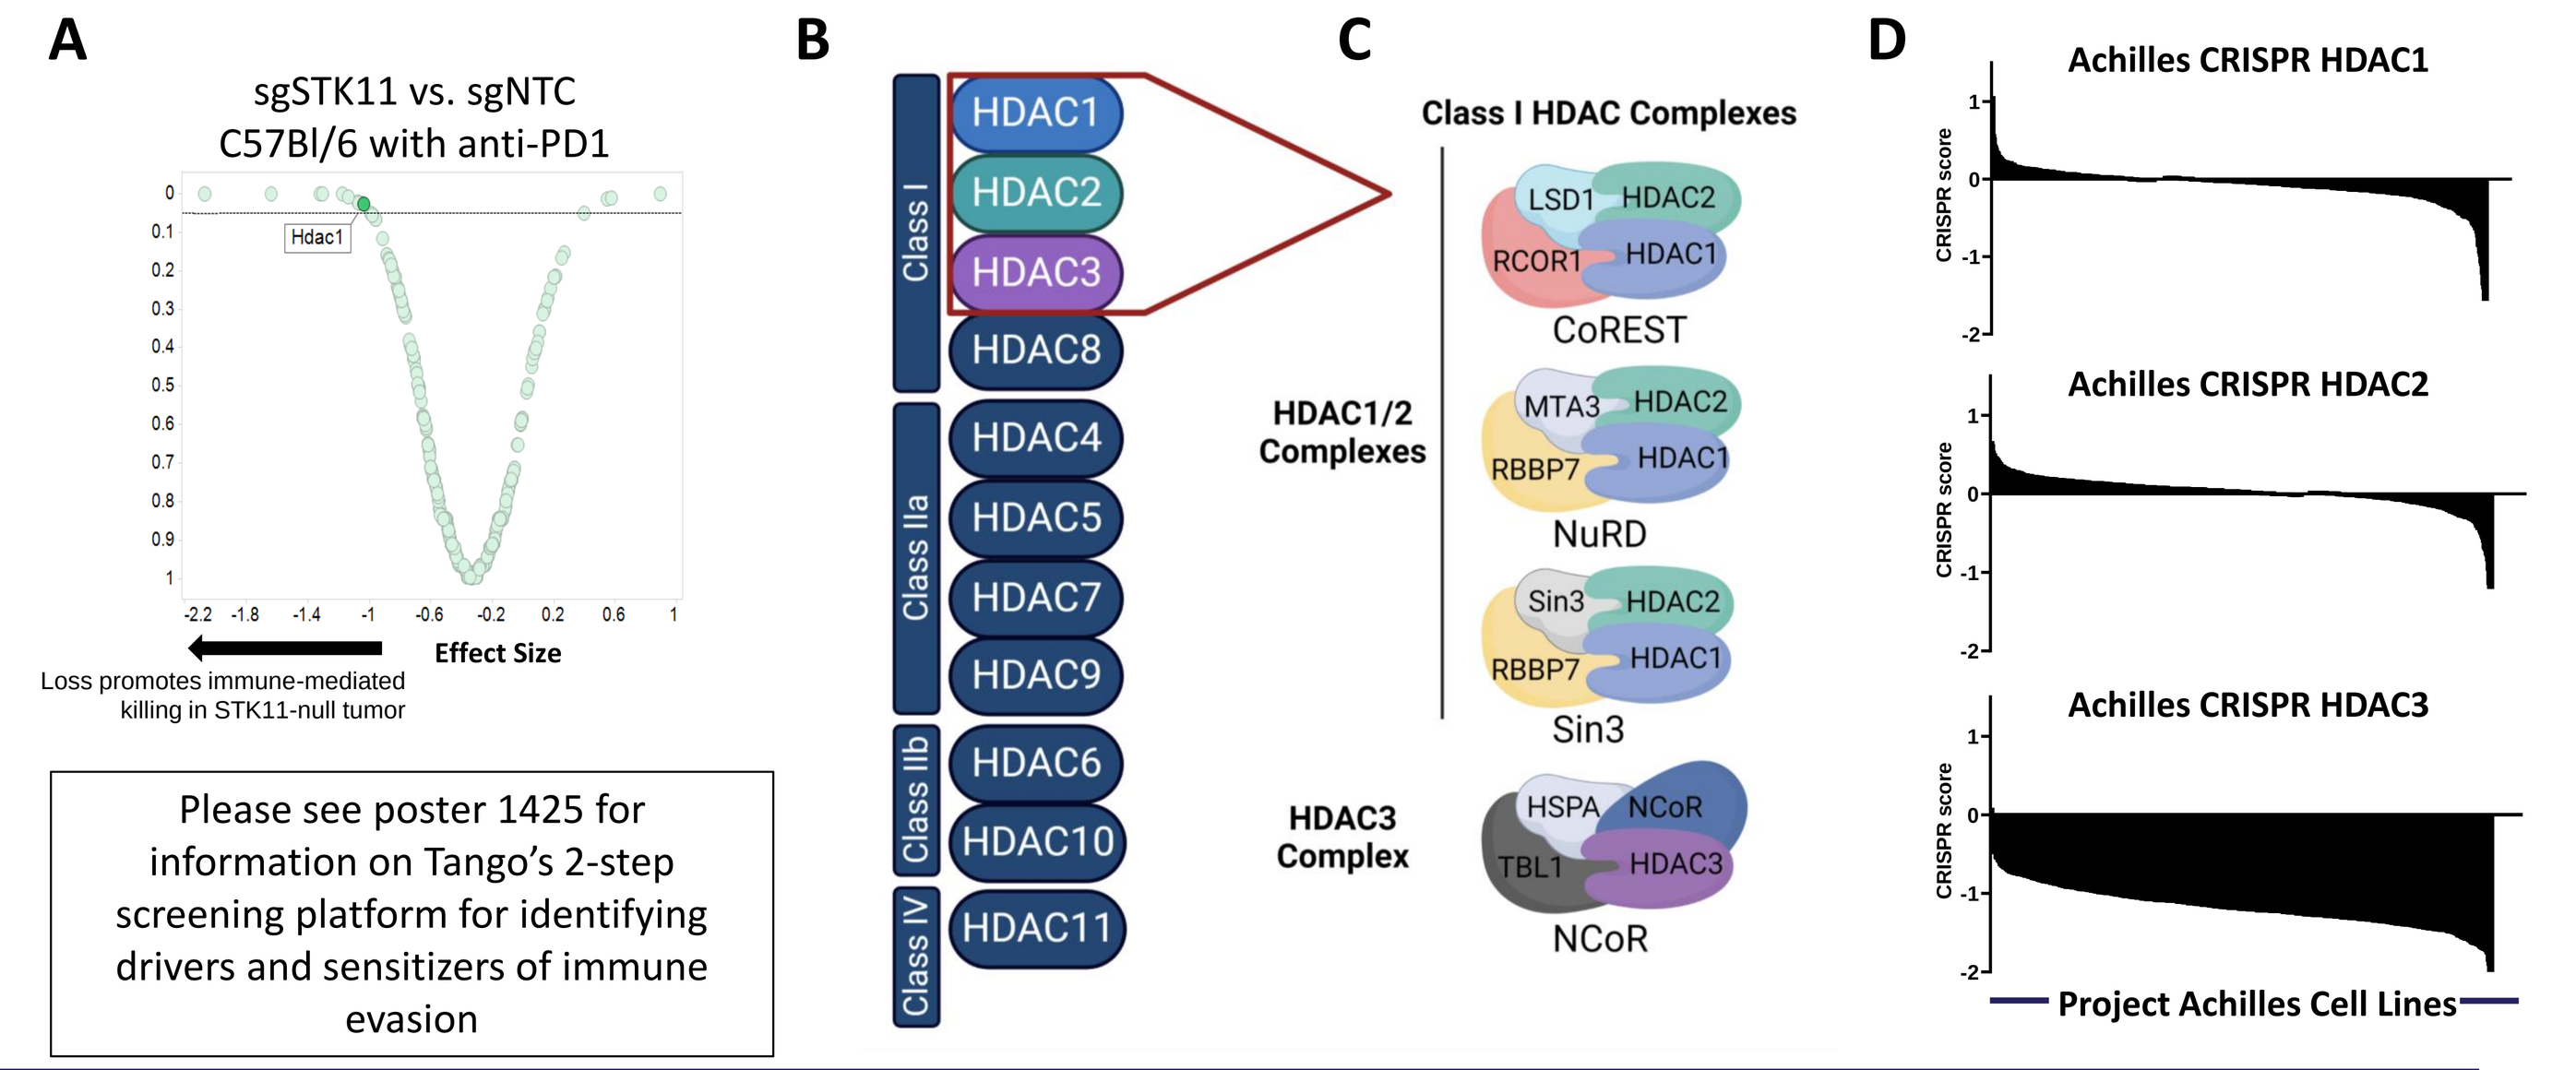
A
sgSTK11 vs. sgNTC
C57Bl/6 with anti-PD1
Hdac1
Effect Size
Loss promotes immune-mediated killing in STK11-null tumor
Please see poster 1425 for information on Tango's 2-step screening platform for identifying drivers and sensitizers of immune evasion
B
Class I
Class IIa
Class IIb
Class IV
HDAC1
HDAC2
HDAC3
HDAC8
HDAC4
HDAC5
HDAC7
HDAC9
HDAC6
HDAC10
HDAC11
C
Class I HDAC Complexes
HDAC1/2 Complexes
HDAC3 Complex
CoREST
NuRD
Sin3
NCoR
D
Achilles CRISPR HDAC1
Achilles CRISPR HDAC2
Achilles CRISPR HDAC3
CRISPR score
Project Achilles Cell Lines

Figure 1: CoREST was identified as a sensitizer to anti-PD1 in an in vivo screen of STK11-deleted cancer. (A) Volcano plot of an unbiased in vivo CRISPR screen identifying HDAC1 knockout as a sensitizer to anti-PD1 in STK11-deleted MC38 tumors. (B) There are 11 Zn-binding HDAC enzymes divided into 4 classes. (C) Class I HDAC enzymes function through 4 main complexes. HDAC1/2 are in the CoREST, NuRD, and Sin3 complexes, while HDAC3 is in the NCoR complex. (D) Waterfall plots of the Project Achilles CRISPR scores for HDAC1, HDAC2, and HDAC3 in a panel of cell lines. Negative scores indicate depletion of cells with knockout of the indicated gene.

TNG260 is a CoREST-selective deacetylase inhibitor

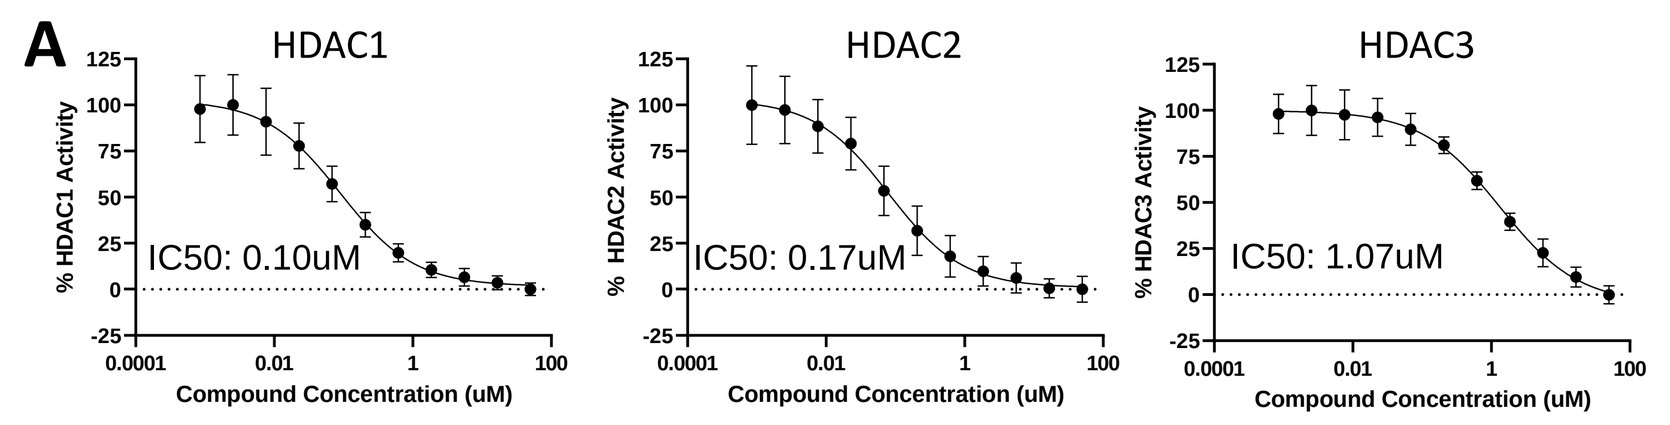
A
HDAC1
HDAC2
HDAC3
IC50: 0.10uM
IC50: 0.17uM
IC50: 1.07uM
Compound Concentration (uM)

B

Compound	HDAC1	HDAC2	HDAC3	HDAC6	HDAC10
TNG260	0.10	0.17	1.07	>30	>30
TCA	0.01	0.02	0.08	5.09	0.20
Entinostat	1.45	0.38	1.12	>30	>30
Chidamide	0.84	0.25	0.41	>30	>30

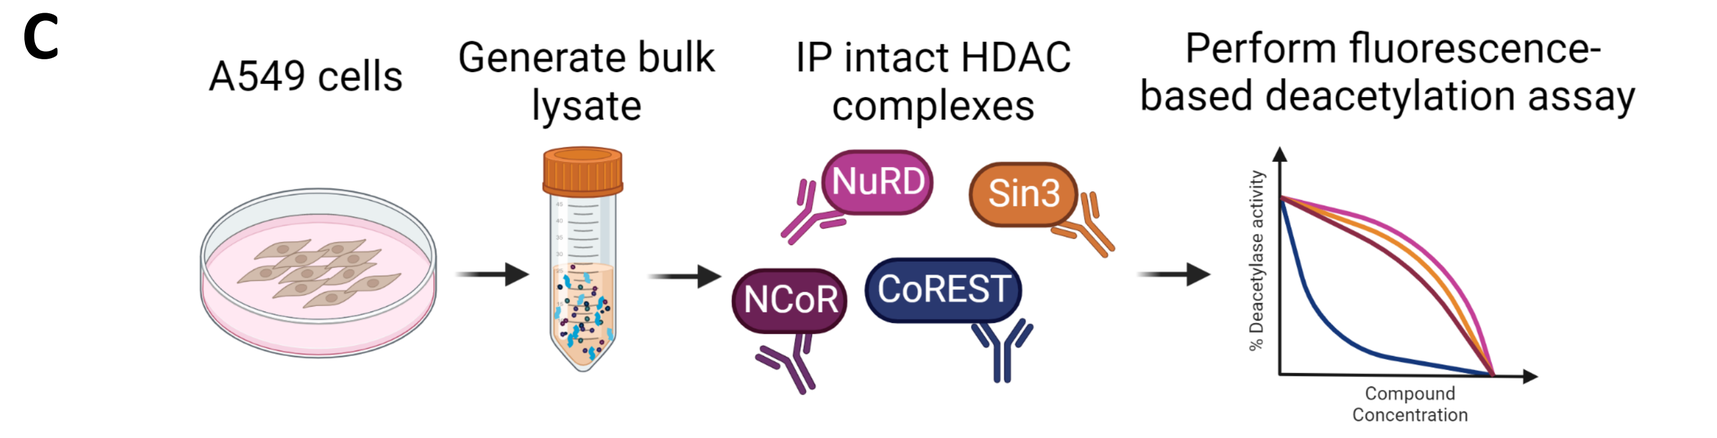
C
A549 cells
Generate bulk lysate
IP intact HDAC complexes
Perform fluorescence-based deacetylation assay
NuRD
Sin3
NCoR
CoREST
% Deacetylase activity
Compound Concentration

D

	CoREST	NCoR	NuRD	Sin3
Vorinostat	0.061	0.525	0.133	0.160
Tucidinostat	0.120	0.200	1.400	>100
Domatinostat	0.087	0.193	30.000	>100
TNG260	0.170	>100	>100	>100

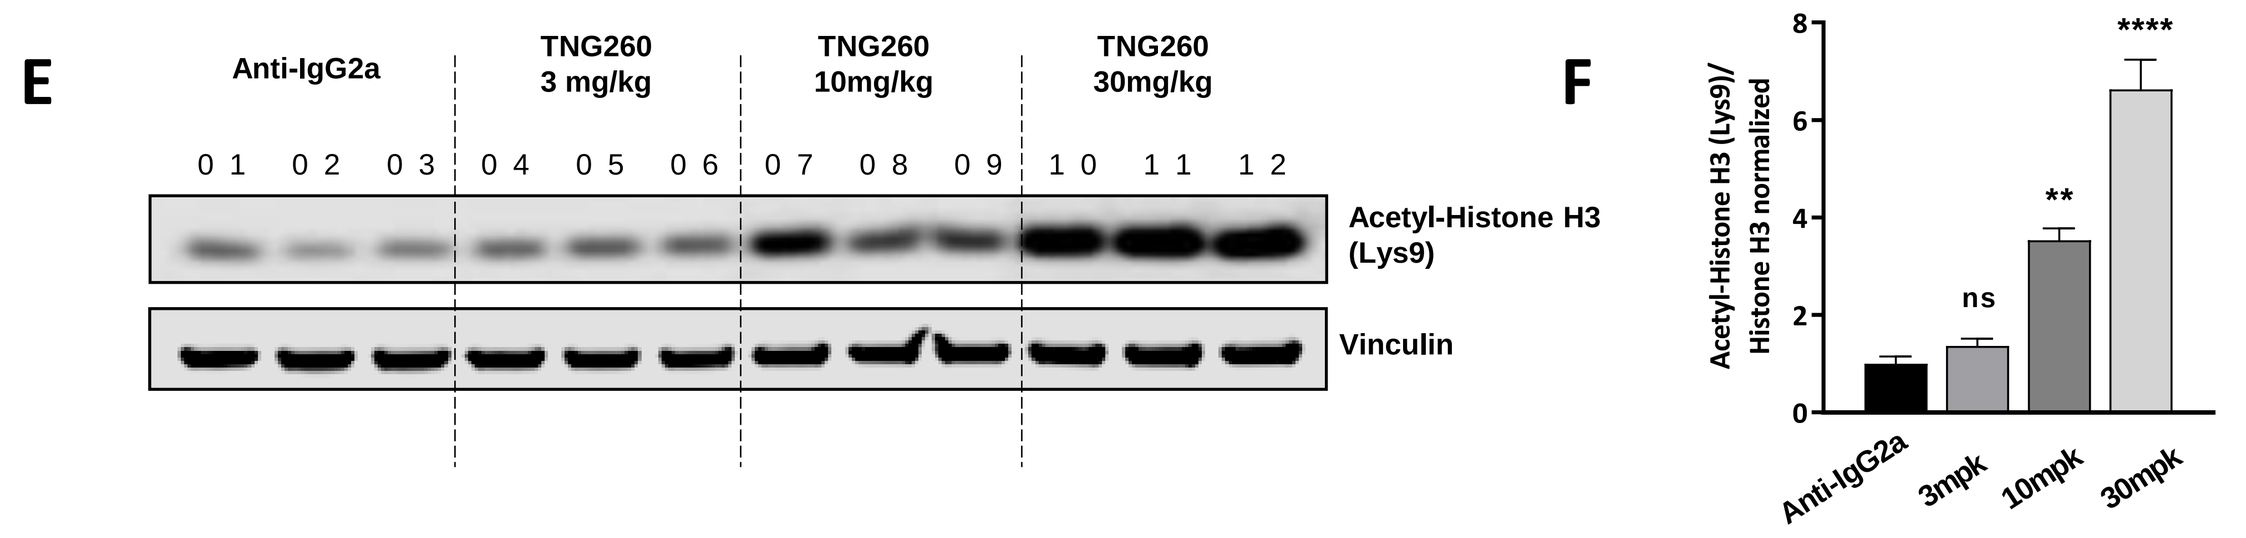
E
Anti-IgG2a
TNG260 3 mg/kg
TNG260 10mg/kg
TNG260 30mg/kg
Acetyl-Histone H3 (Lys9)
Vinculin
F
Acetyl-Histone H3 (Lys9)/ Histone H3 normalized
ns
**

Anti-IgG2a
3mpk
10mpk
30mpk

Figure 2: TNG260 is an HDAC1/2 inhibitor that is selective for the CoREST complex. (A) Dose-dependent binding of TNG260 to HDAC1, HDAC2, and HDAC3 by cellular NanoBRET target engagement assay. (B) IC50s of TNG260 and less-selective HDAC inhibitors evaluated in a cellular NanoBRET assay for HDAC 1, 2, 3, 6 and 10. (C) HDAC complexes CoREST, NCoR, NuRD and Sin3 were co-immunoprecipitated from A549 cells and profiled in a fluorescence-based deacetylase assay. (D) Heat map of IC50s of the indicated compounds in a deacetylase assay using purified CoREST, NCoR, NuRD, and Sin3. (E) Western blot of acetylated histone 3 lysine 9 (H3K9Ac) from mouse MC38 tumor tissue after treatment with TNG260 for 7 days at the indicated dose. Tissue was collected 8 hours post last dose. (F) Quantification of the H3K9Ac western blot in (E) and normalized to total histone H3.

TNG260 reverses resistance to immune checkpoint blockade driven by loss of STK11

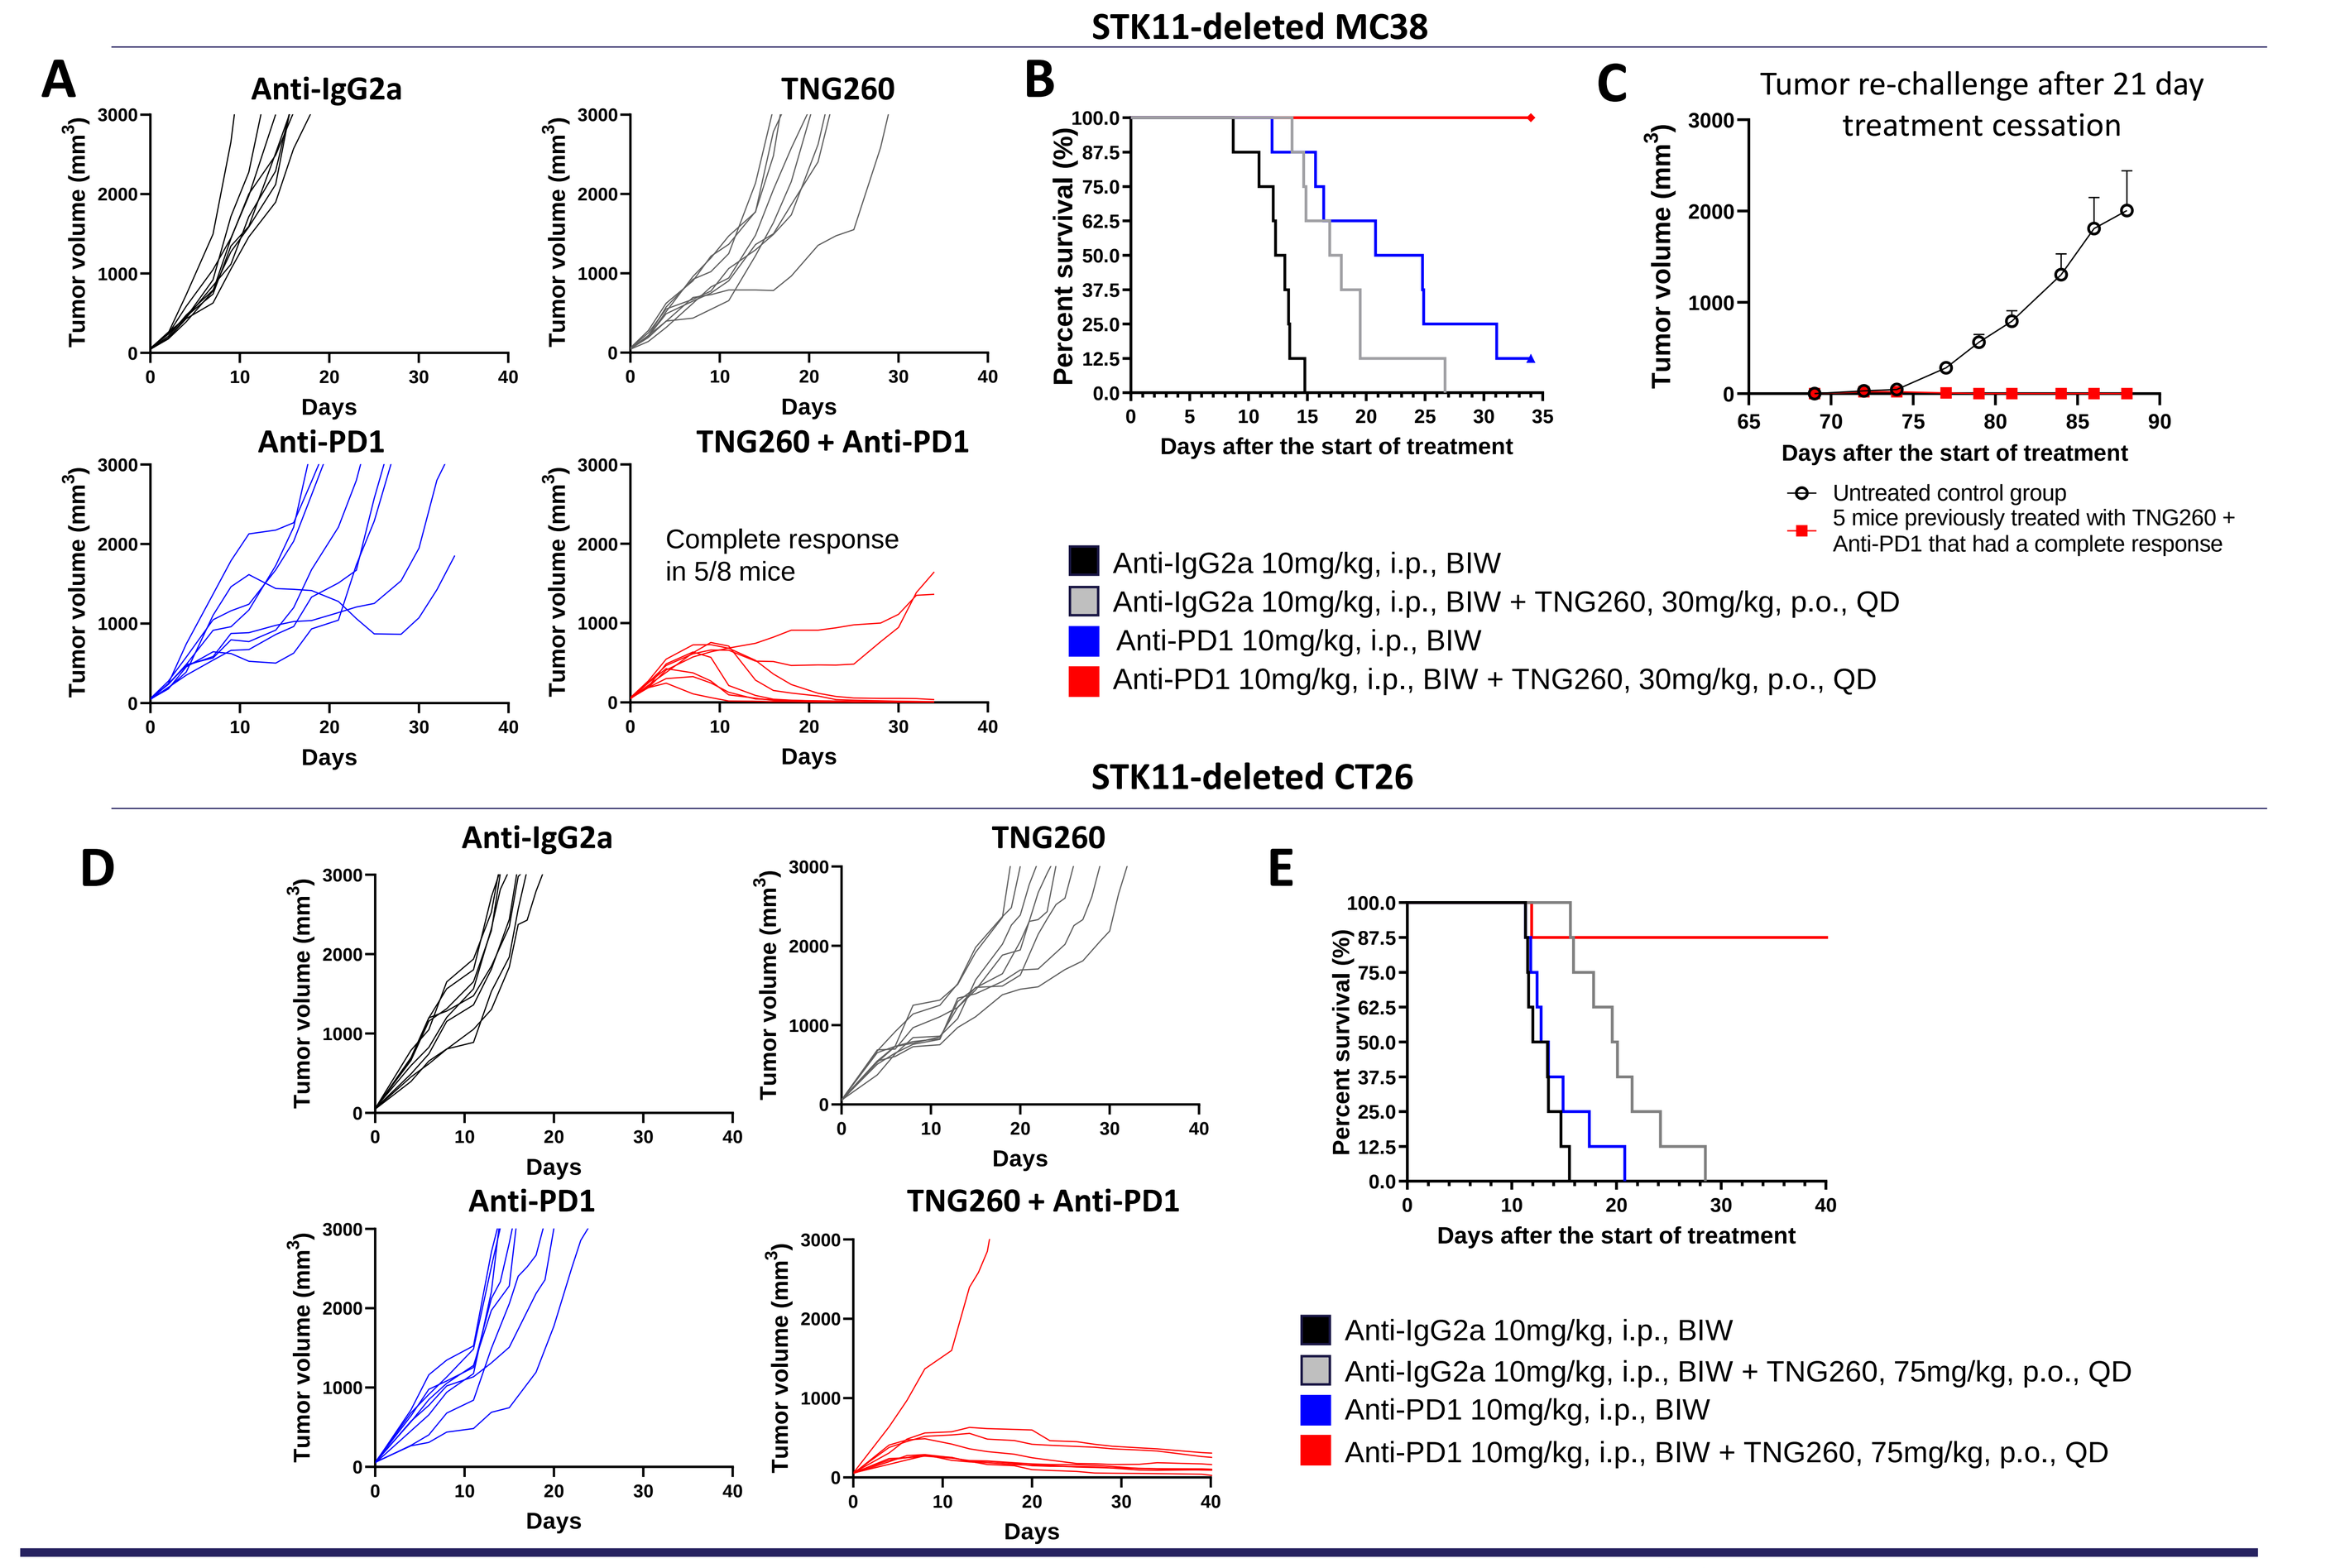
STK11-deleted MC38
A
Anti-IgG2a
TNG260
Anti-PD1
TNG260 + Anti-PD1
Complete response in 5/8 mice
Tumor volume (mm³)
Days
B
Percent survival (%)
Days after the start of treatment
C
Tumor re-challenge after 21 day treatment cessation
Untreated control group
5 mice previously treated with TNG260 + Anti-PD1 that had a complete response
Anti-IgG2a 10mg/kg, i.p., BIW
Anti-IgG2a 10mg/kg, i.p., BIW + TNG260, 30mg/kg, p.o., QD
Anti-PD1 10mg/kg, i.p., BIW
Anti-PD1 10mg/kg, i.p., BIW + TNG260, 30mg/kg, p.o., QD
STK11-deleted CT26
D
E
Anti-IgG2a 10mg/kg, i.p., BIW
Anti-IgG2a 10mg/kg, i.p., BIW + TNG260, 75mg/kg, p.o., QD
Anti-PD1 10mg/kg, i.p., BIW
Anti-PD1 10mg/kg, i.p., BIW + TNG260, 75mg/kg, p.o., QD

Figure 3: TNG260 with anti-PD1 is efficacious in STK11-deleted syngeneic mouse models. (A) The MC38 mouse model is made resistant to murine anti-PD1 by knockout of STK11. Mice were treated with TNG260 orally once daily and with anti-PD1 twice per week. Tumor volume was monitored over the course of treatment and plotted by individual animal. (B) Survival plot of mice with STK11-deleted MC38 tumors. (C) 5 mice that exhibited a complete regression in the TNG260 + anti-PD1 combination arm were monitored for 3 weeks after treatment cessation and no tumor regrowth was observed. These 5 animals were re-challenged with STK11-deleted MC38 tumors in parallel with a control group of previously untreated mice. All animals remained off-treatment, and tumor size was plotted over time after re-challenge. (D) STK11 knockout renders CT26 tumors resistant to anti-PD1 treatment. Mice were treated with TNG260 orally once daily and with anti-PD1 twice per week. Tumor volume was monitored over the course of treatment and plotted by individual animal. (E) Survival plot of mice with STK11-deleted CT26 tumors.

Efficacy of TNG260 + Anti-PD1 is T cell-mediated

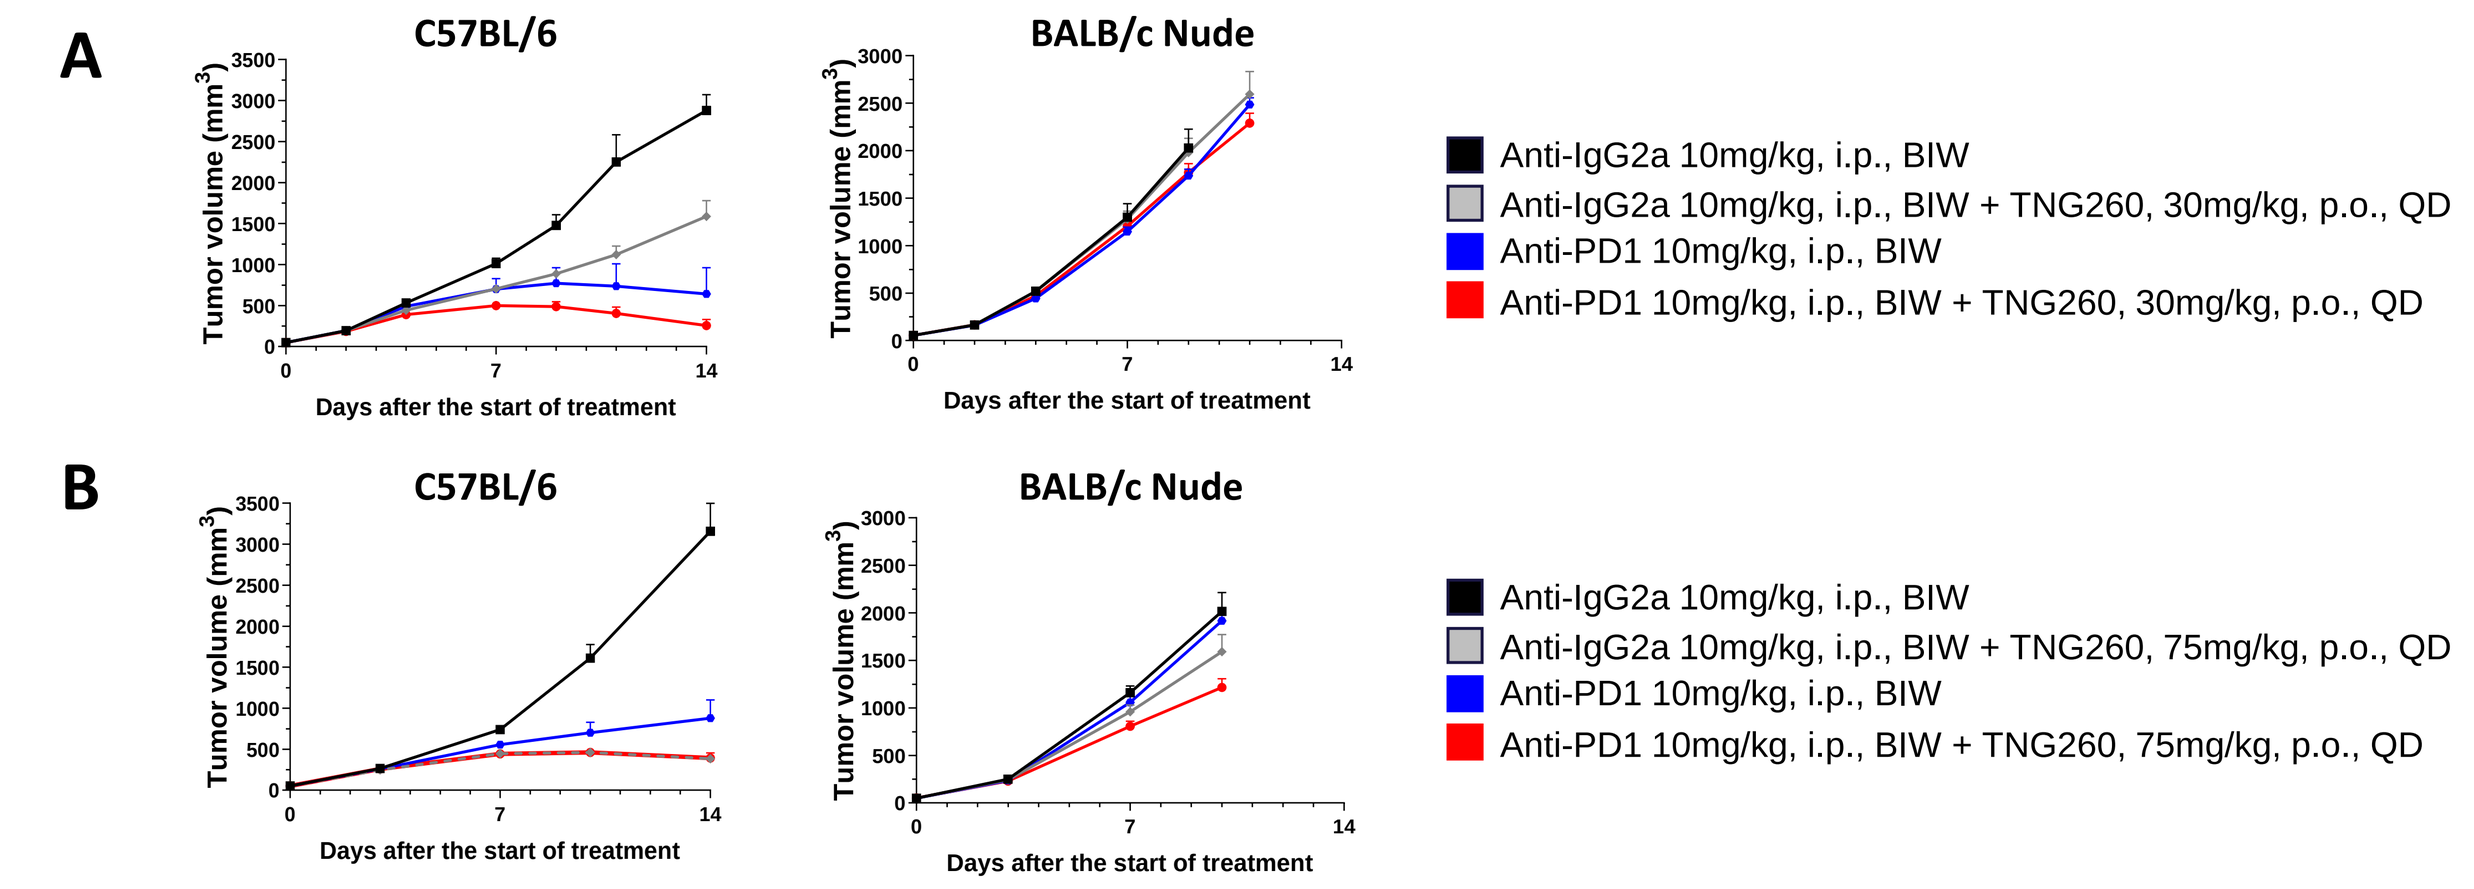
A
C57BL/6
BALB/c Nude
Anti-IgG2a 10mg/kg, i.p., BIW
Anti-IgG2a 10mg/kg, i.p., BIW + TNG260, 30mg/kg, p.o., QD
Anti-PD1 10mg/kg, i.p., BIW
Anti-PD1 10mg/kg, i.p., BIW + TNG260, 30mg/kg, p.o., QD
B
C57BL/6
BALB/c Nude
Anti-IgG2a 10mg/kg, i.p., BIW
Anti-IgG2a 10mg/kg, i.p., BIW + TNG260, 75mg/kg, p.o., QD
Anti-PD1 10mg/kg, i.p., BIW
Anti-PD1 10mg/kg, i.p., BIW + TNG260, 75mg/kg, p.o., QD
Tumor volume (mm³)
Days after the start of treatment

Figure 4: Efficacy of TNG260 with anti-PD1 requires an intact T cell compartment. (A) A head-to-head comparison of efficacy in STK11-deficient MC38 tumor cells in C57BL/6 animals and athymic BALB/c Nude mice with TNG260 at 30mg/kg alone or in combination with anti-PD1. (B) A head-to-head comparison of efficacy in STK11-deficient MC38 tumor cells in C57BL/6 animals and athymic BALB/c Nude mice with TNG260 at 75mg/kg alone or in combination with anti-PD1.

TNG260 treatment drives expression of cytokines that promote anti-tumor activity

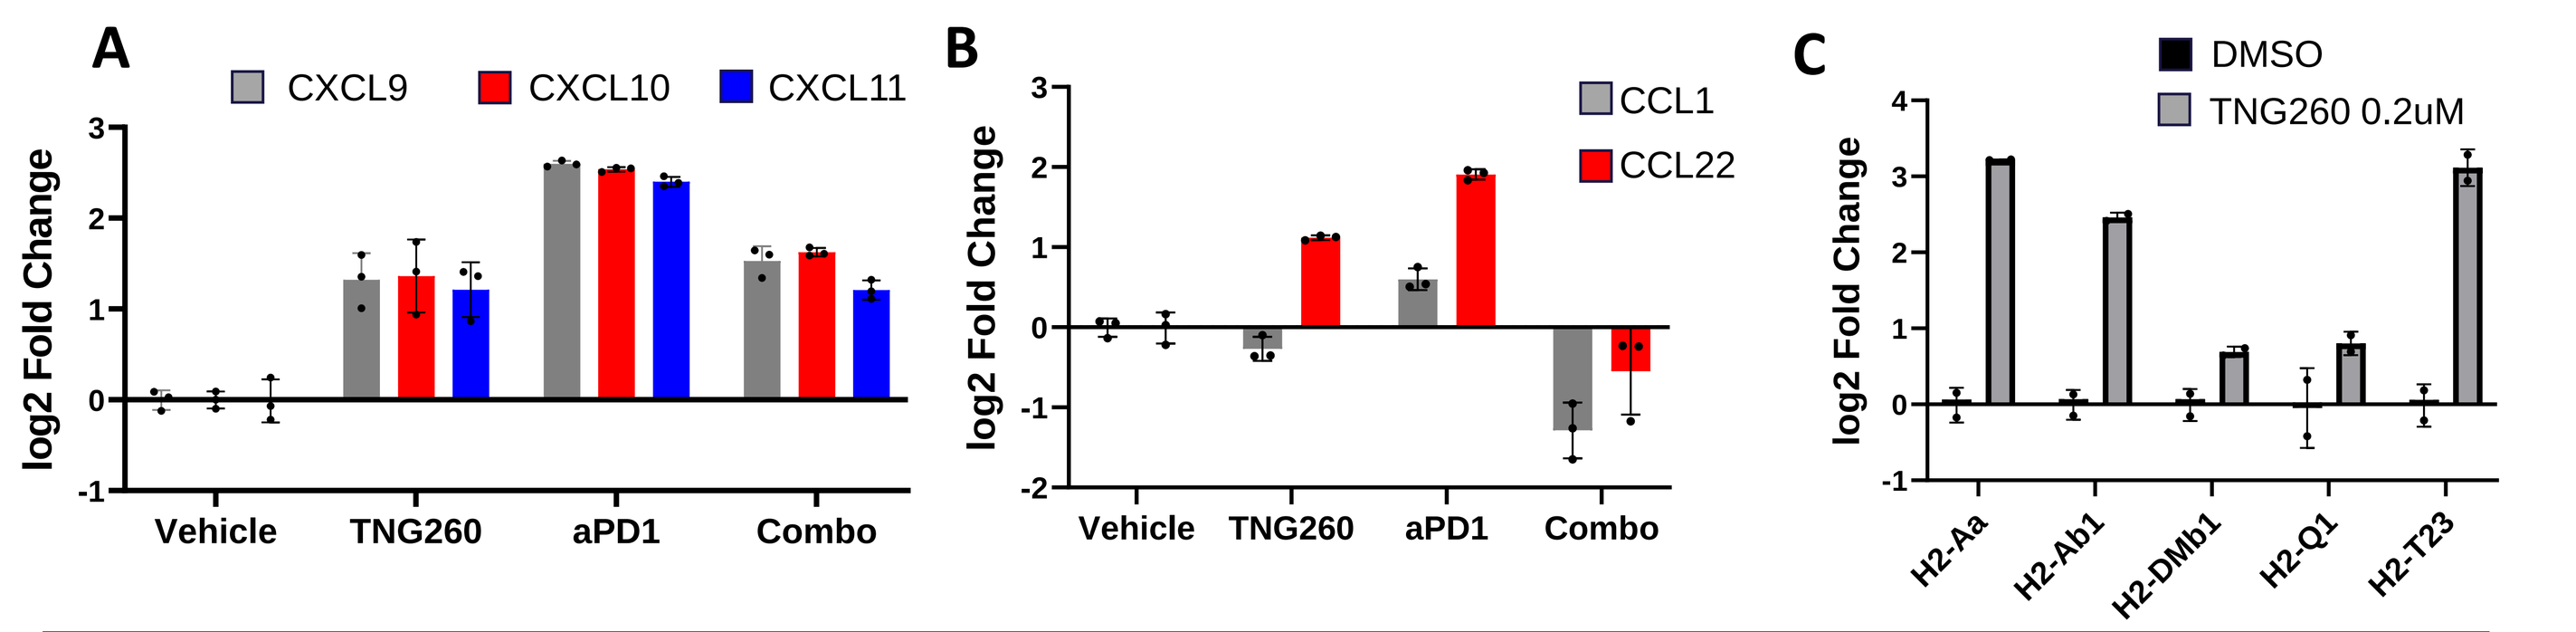
A
CXCL9
CXCL10
CXCL11
log2 Fold Change
Vehicle
TNG260
aPD1
Combo
B
CCL1
CCL22
C
DMSO
TNG260 0.2uM
H2-Aa
H2-Ab1
H2-DMb1
H2-Q1
H2-T23

Figure 5: TNG260 reverses immune evasion on tumor cells by changing expression of cytokines and antigen presentation genes. (A) Gene expression profiling of CXCL9, 10, and 11 by Nanostring PanCancer IO 360 on STK11-/- MC38 tumors treated for 7 days with 30mg/kg of TNG260 or anti-PD1 alone or in combination. Tumors were collected 8 hours post last dose. (B) Gene expression profiling of Treg-recruiting chemokines CCL1 and CCL22 by Nanostring PanCancer IO 360 on STK11-/- MC38 tumors treated for 7 days with 30mg/kg of TNG260 or anti-PD1 alone or in combination. Tumors were collected 8 hours post last dose. (C). STK11-deficient MC38 cells were profiled for HLA gene expression by Nanostring IO360 panel in vitro after 4 days of treatment with TNG260 at 0.2uM compared to solvent control.

TNG260 + anti-PD1 depletes intratumoral Tregs and increases T cell activity

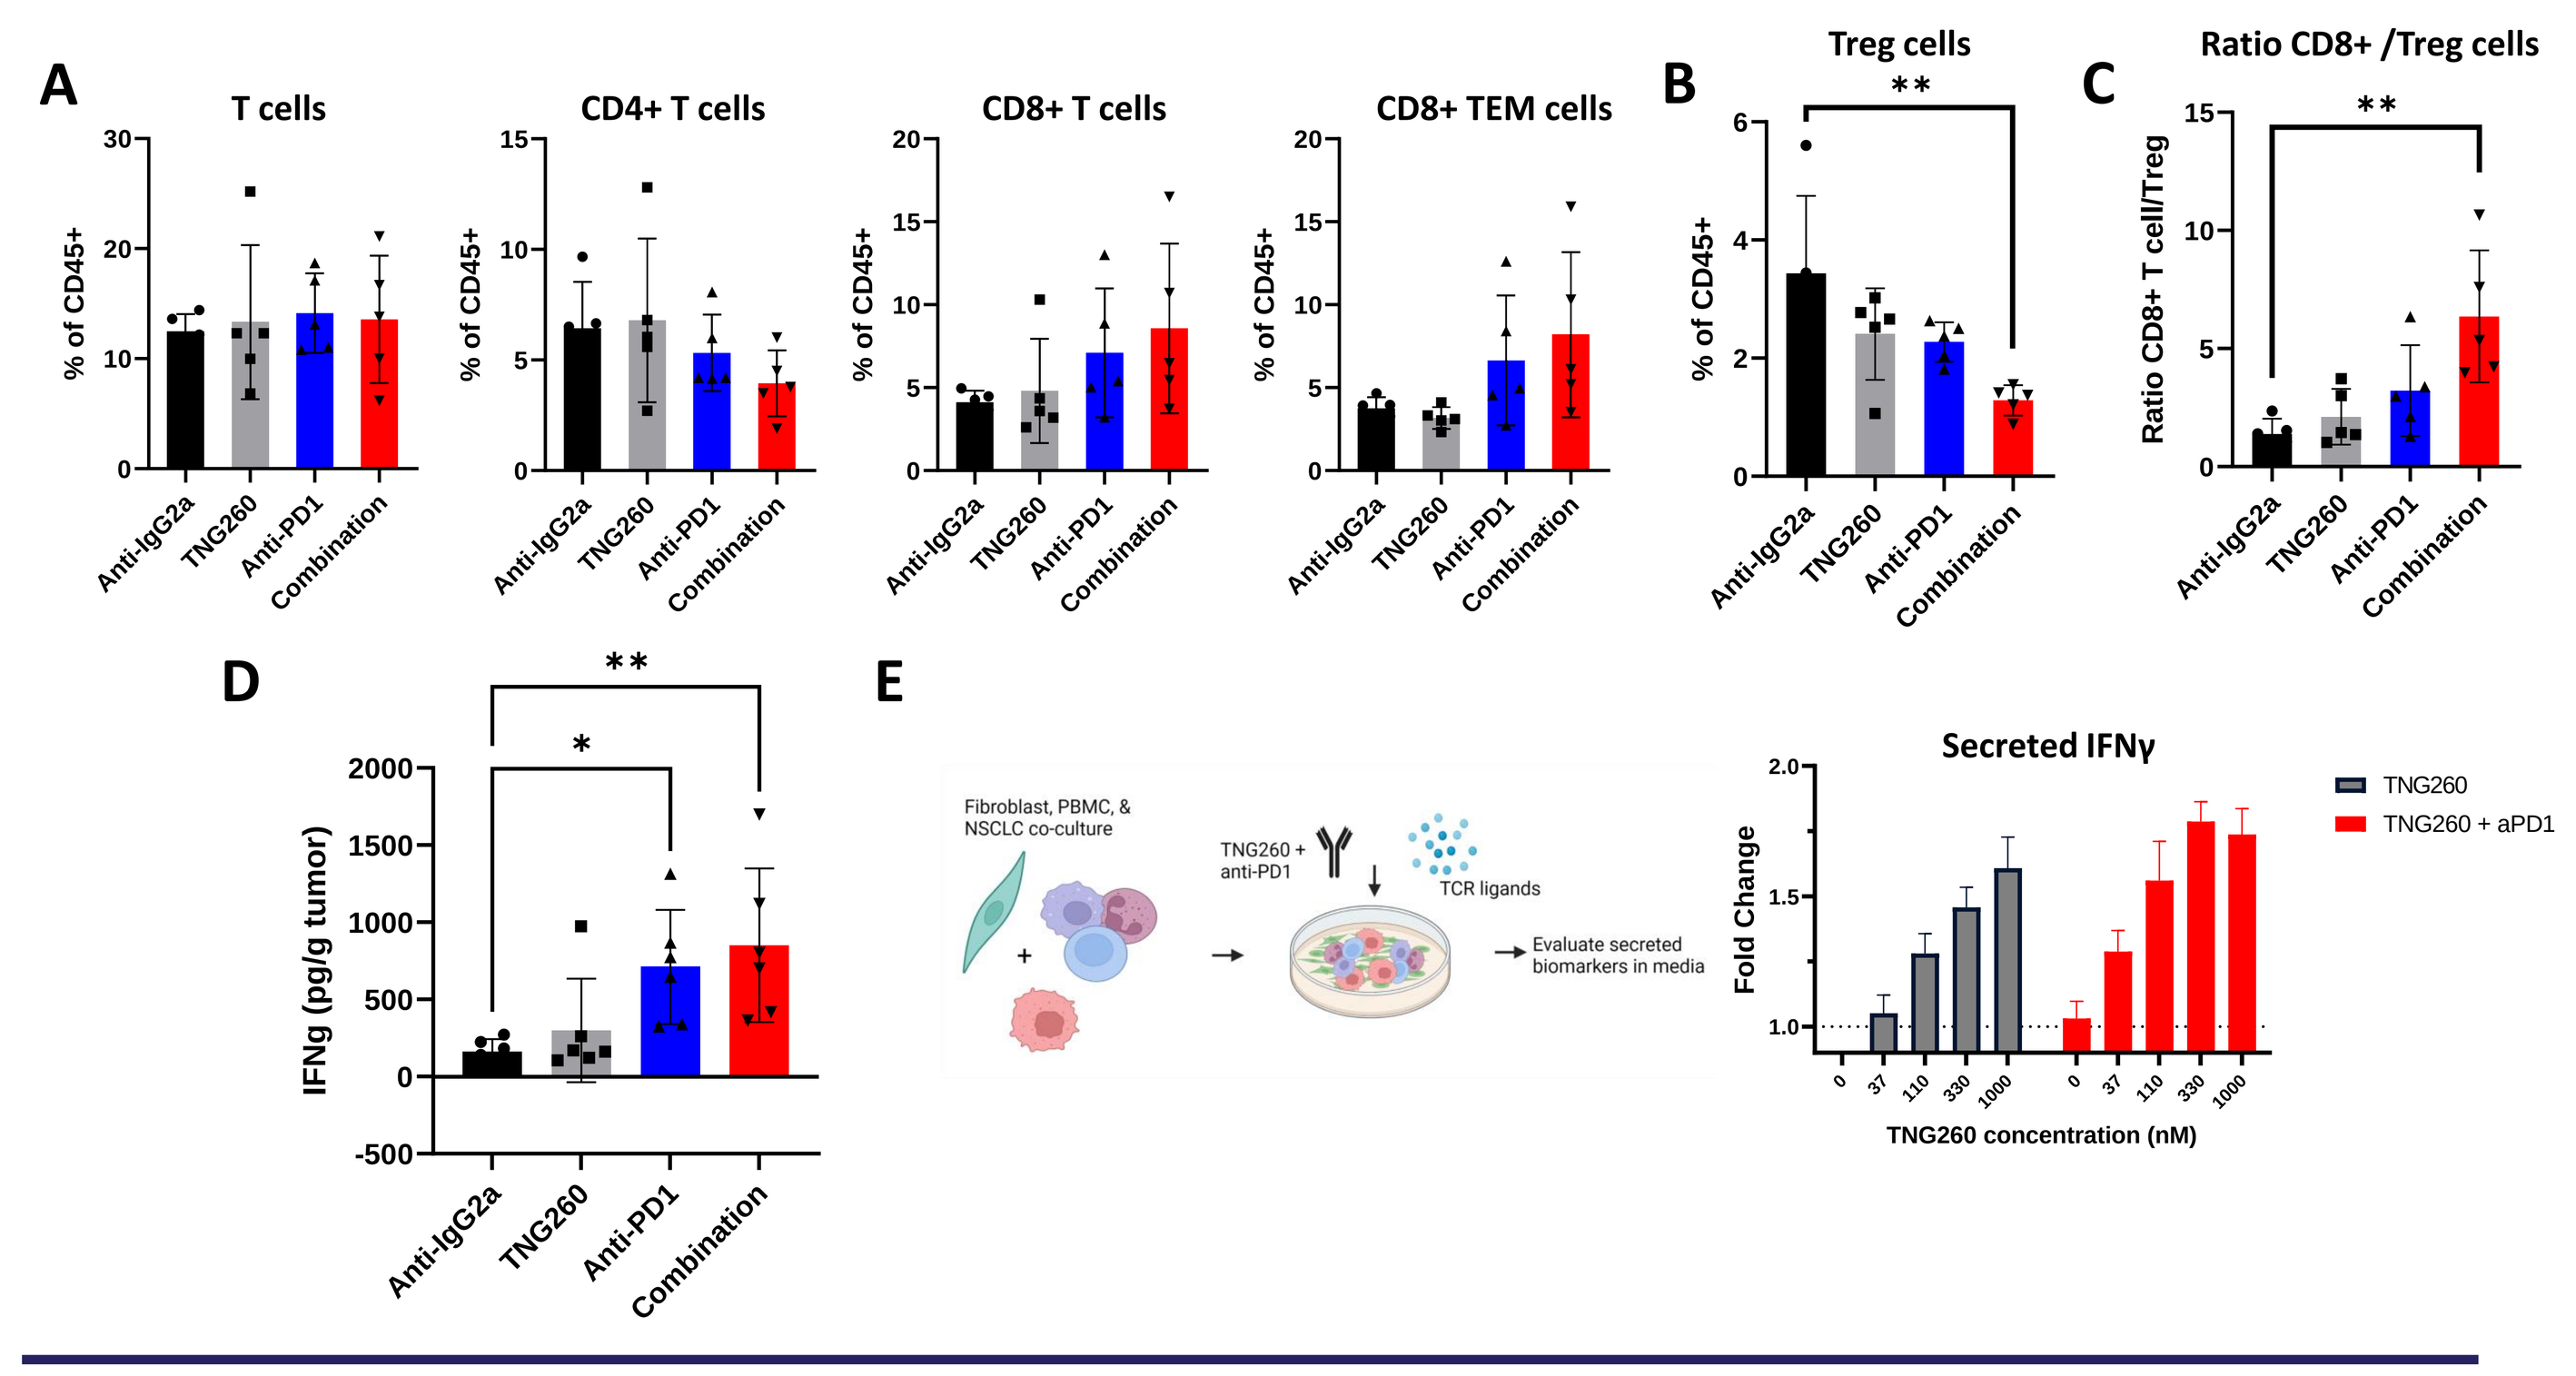
A
T cells
CD4+ T cells
CD8+ T cells
CD8+ TEM cells
% of CD45+
B
Treg cells
C
Ratio CD8+ /Treg cells
Ratio CD8+ T cell/Treg
Anti-IgG2a
TNG260
Anti-PD1
Combination
D
IFNg (pg/g tumor)
E
Fibroblast, PBMC, & NSCLC co-culture
TNG260 + anti-PD1
TCR ligands
Evaluate secreted biomarkers in media
Secreted IFNγ
TNG260
TNG260 + aPD1
Fold Change
TNG260 concentration (nM)

Figure 6: The combination of TNG260 and anti-PD1 increases T cell activity in the tumor microenvironment. (A) TIL profiling by flow cytometry of STK11-deleted MC38 tumors treated for 7 days with 10mg/kg of TNG260 alone or in combination with anti-PD1. Tumor tissue was collected 8 hours post last dose. (B) Flow cytometry of Tregulatory cells showing a significant decrease in frequency of Treg cells in the combination arm. (C) The ratio of CD8+ T effector cells to T regulatory cells in each of the treatment groups showing a significantly increased ratio in the combination arm. (D) IFNγ levels were evaluated by Luminex in tumors treated with 30mg/kg of TNG260 alone or in combination with anti-PD1. (E) A co-culture of human NSCLC cells with PBMCs and fibroblasts was treated with a dose response of TNG260 alone or in combination with a fixed dose of anti-PD1 for 72hours. IFNγ levels were quantified from tissue culture supernatant by ELISA.

TNG260 regulates immune function more selectively than other HDAC inhibitors

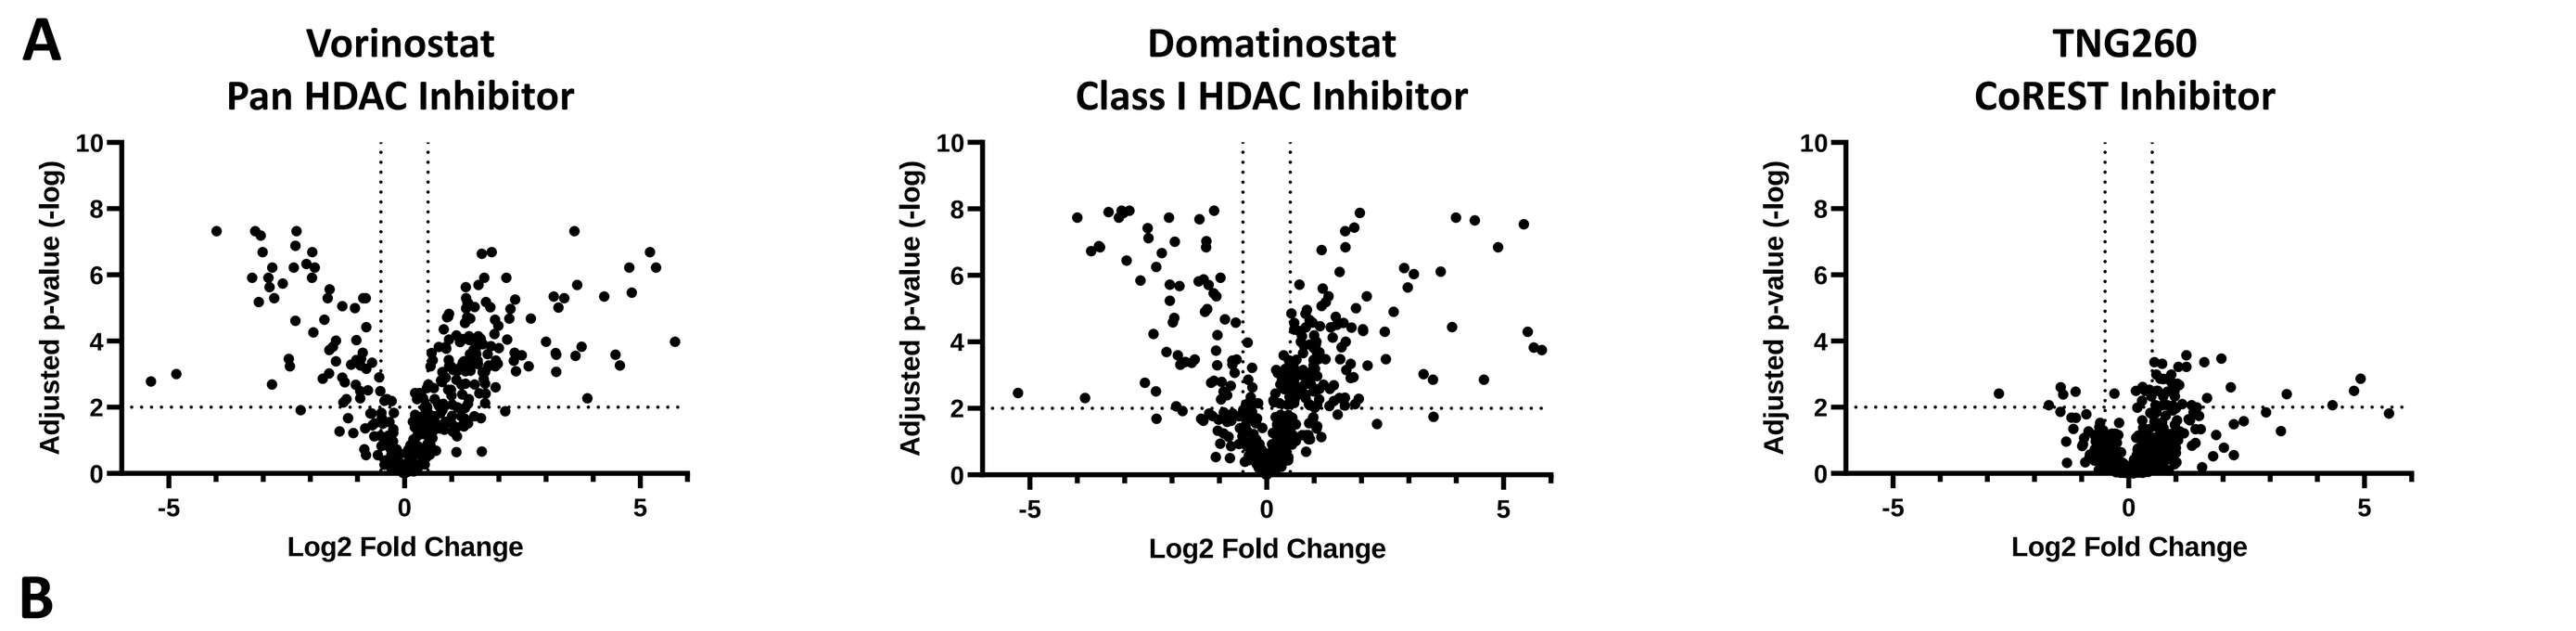
A
Vorinostat
Pan HDAC Inhibitor
Domatinostat
Class I HDAC Inhibitor
TNG260
CoREST Inhibitor
Adjusted p-value (-log)
Log2 Fold Change

B

Vorinostat

Term	Rank
Cell Proliferation	1
DNA Damage Repair	2
Wnt Signaling	3

Domatinostat

Term	Rank
Cell Proliferation	1
DNA Damage Repair	2
Epigenetic Regulation	3

TNG260

Term	Rank
Immune Cell Adhesion and Migration	1
Matrix Remodeling and Metastasis	2
Antigen Presentation	3

Figure 7: Comparison of gene expression changes between TNG260 and other HDAC inhibitors. (A) Nanostring profiling of vorinostat, domatinostat and TNG260 using the PanCancer IO360 panel in A549 cells treated for 96 hours. To select comparable doses of each compound, an H3K9Ac AlphaLISA was performed, and the dose that increased H3K9Ac two-fold was chosen. (B) The top three ranked gene ontology groups for each compound as determined from the Nanostring data in (A) using the nSolver software.

TNG260 is less cytotoxic and has an improved therapeutic index over less-selective HDAC inhibitors

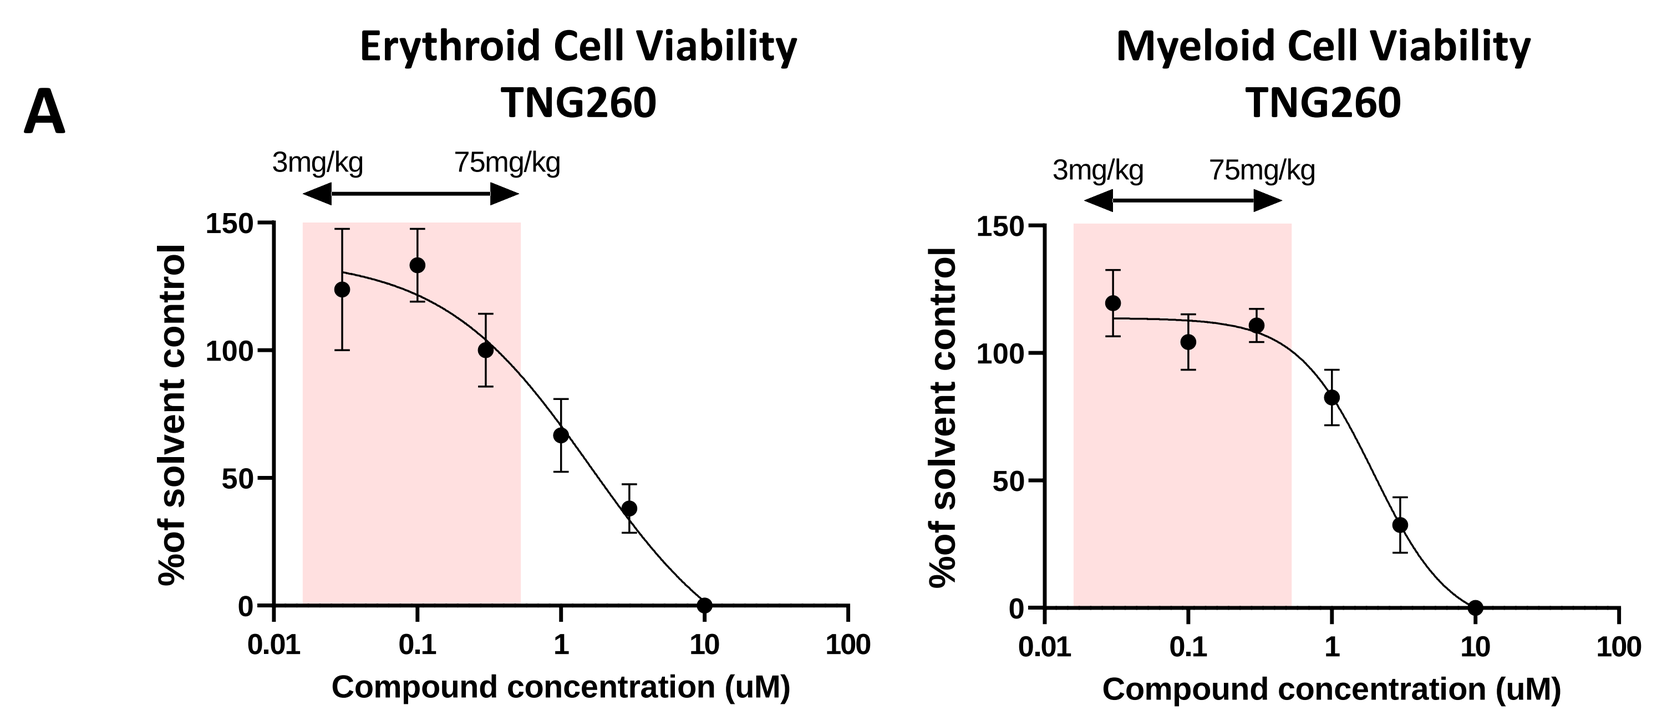
A
Erythroid Cell Viability
TNG260
Myeloid Cell Viability
TNG260
3mg/kg
75mg/kg
%of solvent control
Compound concentration (uM)

B

Relative IC50s of HDAC inhibitors on erythroid and myeloid cell viability

		Erythroid/ HDAC1 IC50	Myeloid/ HDAC1 IC50
Class I-selective	Mocetinostat	0.483	1.741
	Entinostat	0.634	0.210
	Tucidinostat	3.421	1.109
	TNG-0770701	13.673	15.282
	TNG260	15.932	18.136

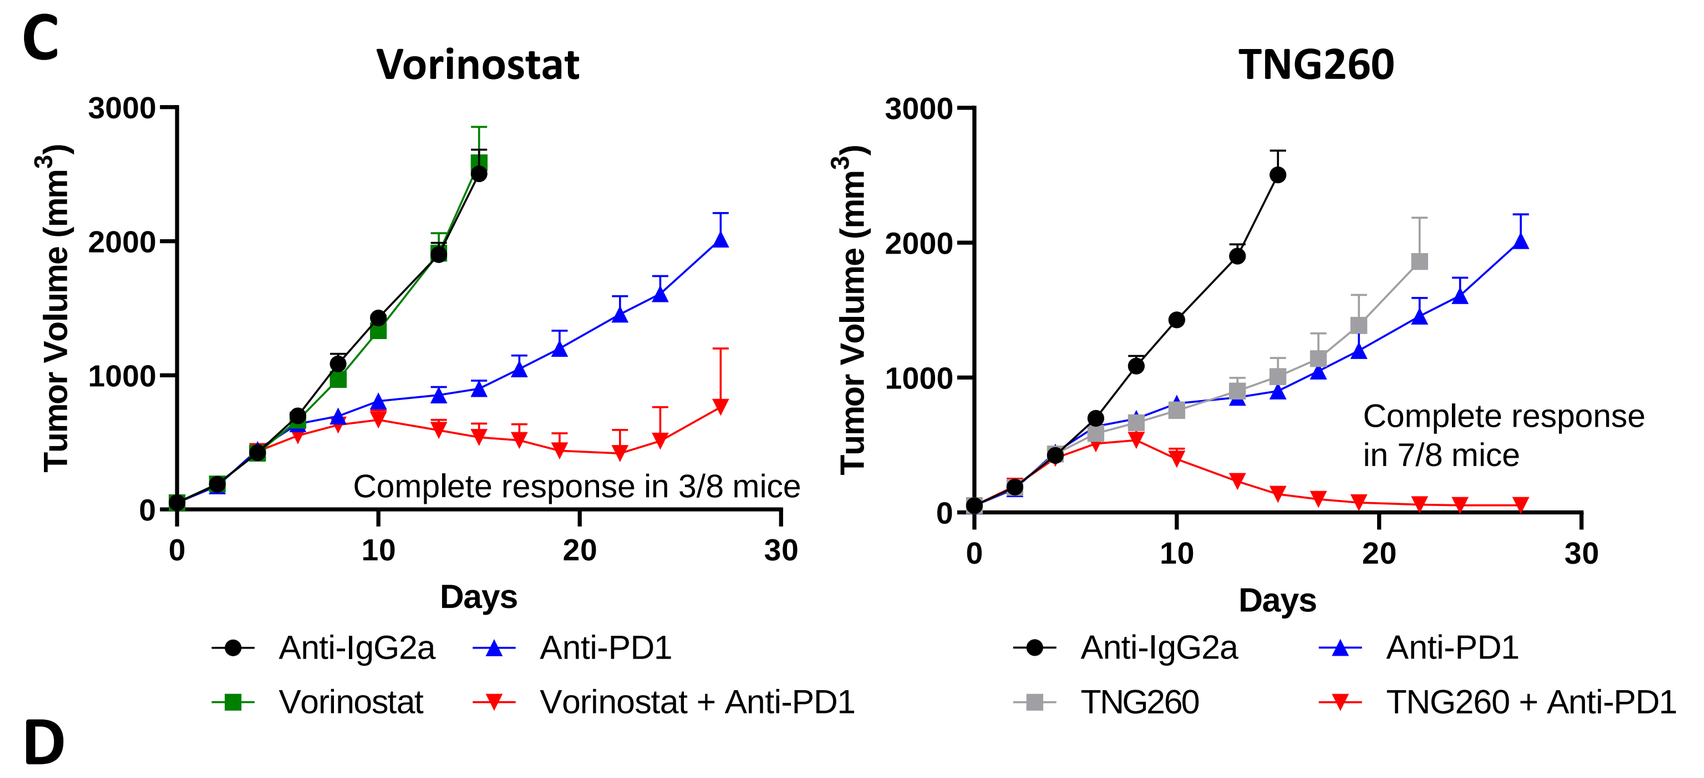
C
Vorinostat
TNG260
Tumor Volume (mm³)
Days
Complete response in 3/8 mice
Complete response in 7/8 mice
Anti-IgG2a
Anti-PD1
Vorinostat
Vorinostat + Anti-PD1
TNG260
TNG260 + Anti-PD1

D

Treatment	Cavg, u (uM)	Fold of HDAC1 IC50
Vorinostat (pan)	0.132*	0.3
TNG260 (CoREST)	0.192	3.5

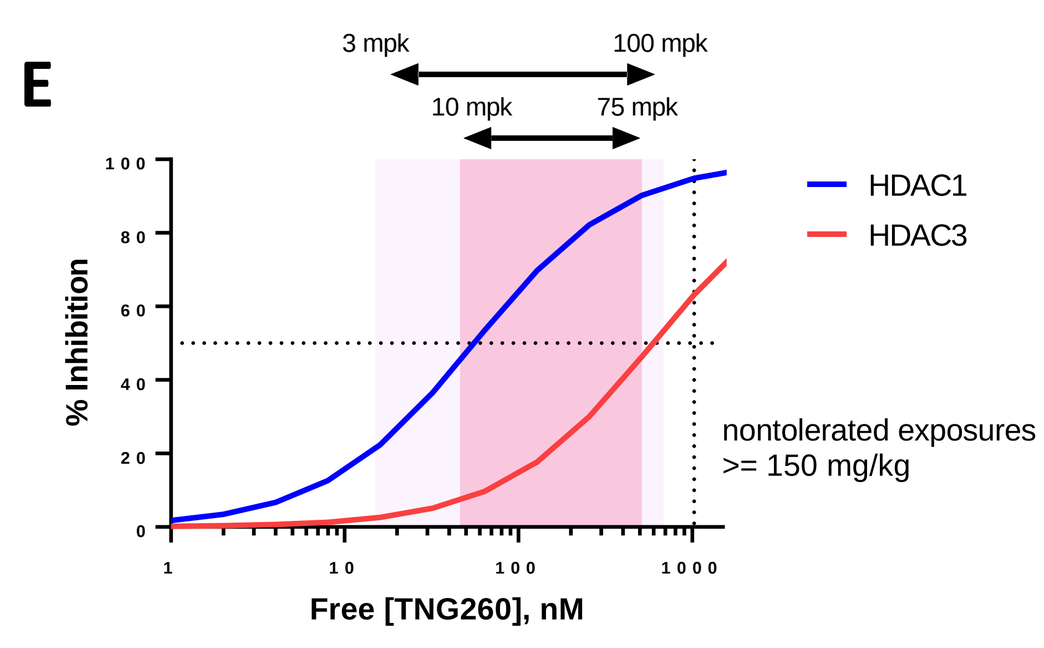
E
3 mpk
100 mpk
10 mpk
75 mpk
% Inhibition
Free [TNG260], nM
HDAC1
HDAC3
nontolerated exposures >= 150 mg/kg

Figure 8. TNG260 does not reduce myeloid and erythroid viability at predicted therapeutic exposures. (A) A colony forming unit assay was performed to evaluate the impact of HDAC inhibitors on the viability of erythroid and myeloid cells. Cells were treated with a dose response of each compound for 14 days. Cell colonies were quantified at the end of the experiment and compared to a solvent control. The efficacious dose range of TNG260 is also plotted. (B) IC50s for each compound were calculated from the erythroid and myeloid colony formation unit assay. These IC50s were normalized to the compound's potency against HDAC1 in the cellular NanoBRET assay to allow for head-to-head compound comparison. (C) A clinically relevant dose of vorinostat for mouse was calculated using body surface area conversion. Mice with STK11-deleted MC38 tumors were treated with vorinostat or TNG260 alone or in combination with anti-PD1. Tumor volumes were monitored over the course of treatment. (D) Table showing Caverage of vorinostat or TNG260 at doses used in part (C) and the coverage of the HDAC1 IC50 at that dose. (E) Plot comparing TNG260 concentrations to HDAC1 or HDAC3 inhibition in vivo. Shaded boxes indicate tolerated and efficacious dose ranges of TNG260.

SUMMARY

- HDAC1 was identified in an unbiased screen as a target that reversed anti-PD1 resistance in STK11-mutant cells
- Existing HDAC inhibitors target the essential gene HDAC3 in addition to HDAC1/2
- TNG260 is a CoREST-selective HDAC1/2 inhibitor
- TNG260 re-sensitizes STK11-deleted tumors to anti-PD1 through increasing T cell activity

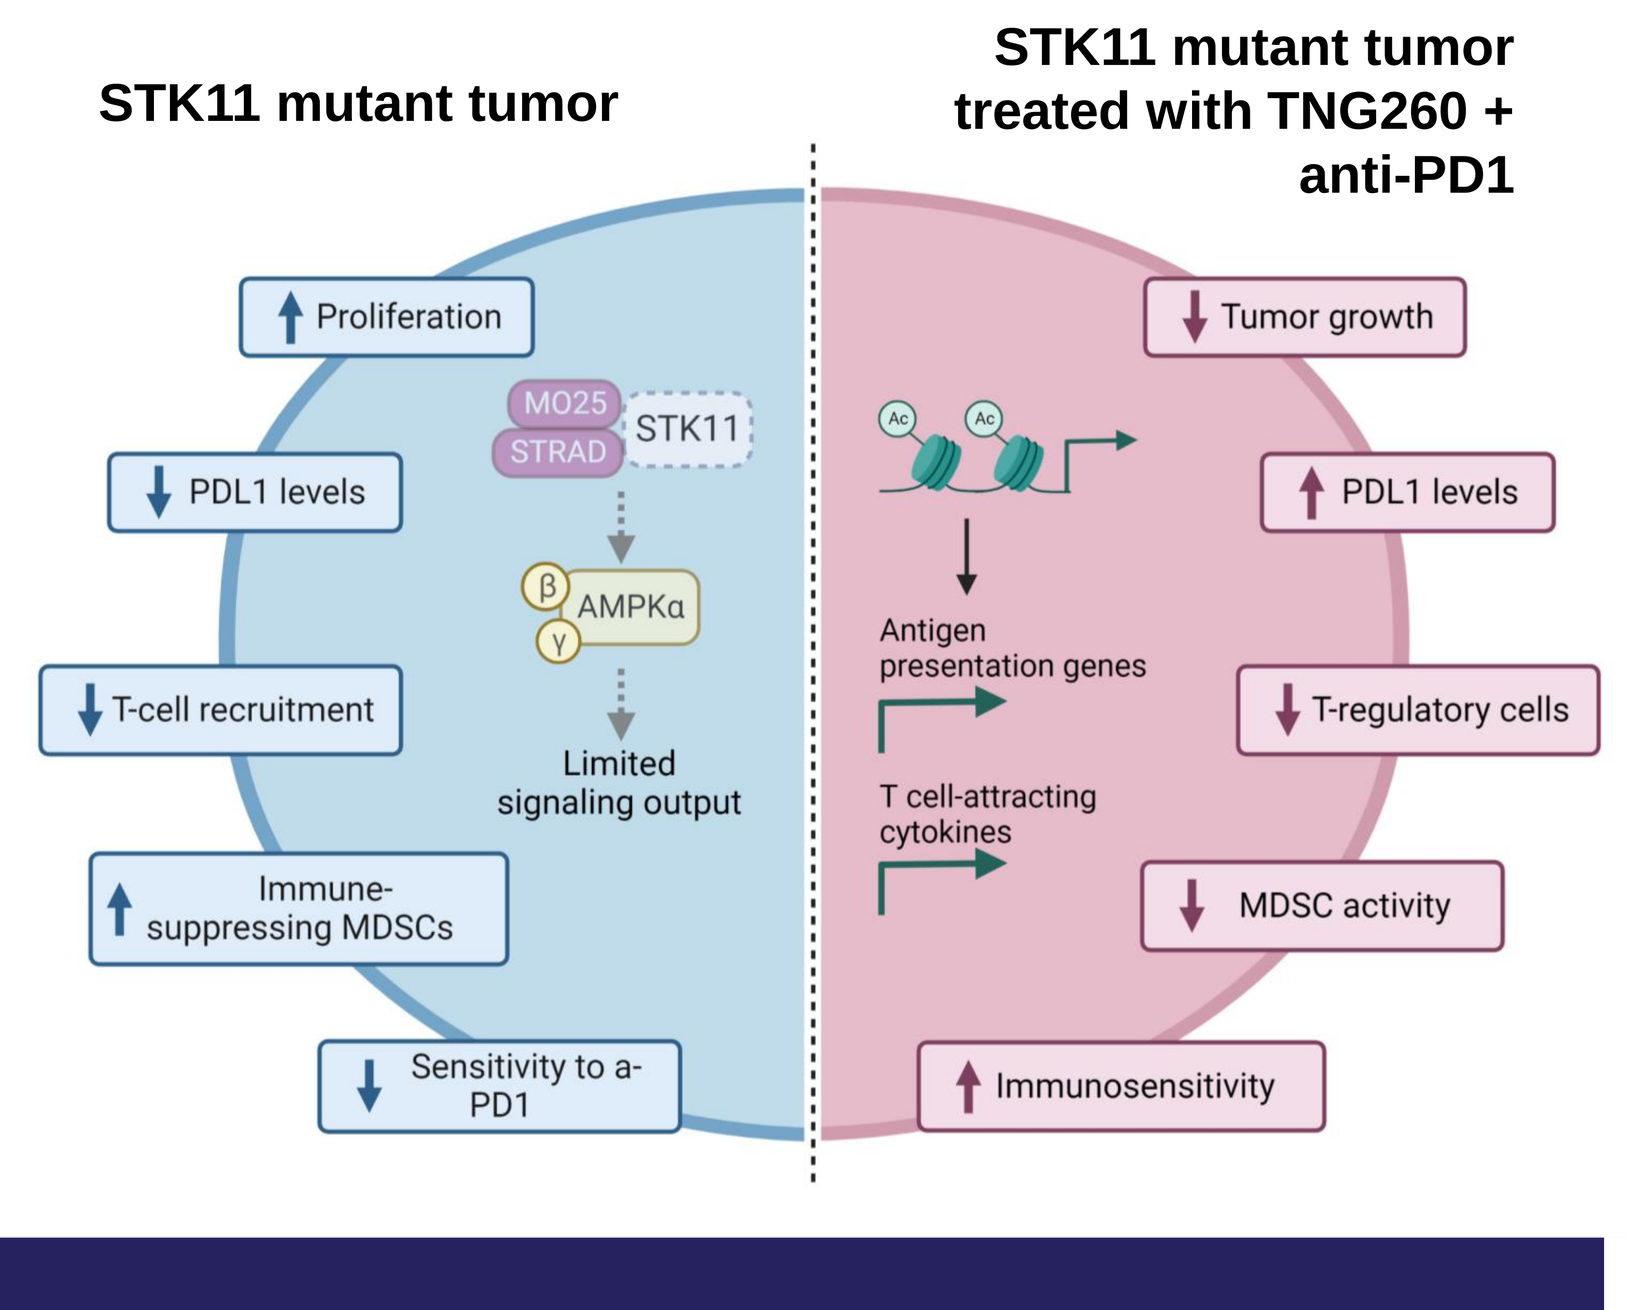
STK11 mutant tumor
STK11 mutant tumor treated with TNG260 + anti-PD1
Proliferation
PDL1 levels
T-cell recruitment
Immune-suppressing MDSCs
Sensitivity to a-PD1
MO25
STRAD
STK11
AMPKα
Limited signaling output
Tumor growth
PDL1 levels
Antigen presentation genes
T cell-attracting cytokines
T-regulatory cells
MDSC activity
Immunosensitivity

ACKNOWLEDGMENTS

The authors gratefully acknowledge the generous contributions from:

- Tango scientists: Preksha Shahagadkar, Shangtao Liu, Binzhang Shen
- The scientific teams at ChemPartner, Crown Biosciences, HDBiosciences, OmicScouts, STEMCELL, and Wuxi AppTec