Leveraging CRISPR-Cas9 screening platform for discovery of novel tumor intrinsic phagocytosis modulators

Lei Ji[1], Grace Trombley[1], Ye Wang[1] , Alyssa Agarwal[1], Wenrong Zhou[2], Serge Gueroussov[1], Henry Weith[1], Sophia Paxton[1], Ashley Choi[1], Tenzing Khendu[1], Samuel Meier[1], Shangtao Liu[1], Binzhang Shen[1], Teng Teng[1], Yi Yu[1], Xuewen Pan[1], Alan Huang[1], Chengyin Min[1,3]

[1]Department of Immunology, Tango Therapeutics, Boston, MA, USA; [2]WuXi AppTec Group, Shanghai, China; [3]Corresponding Author

SITC 2022

1449

BACKGROUND

Targeting “don’t eat me” signaling has achieved profound responses in clinal trials for hematologic malignancies, highlighting the potential therapeutic value of macrophage-mediated phagocytosis for cancer treatment. In recent years, CRISPR-Cas9 based *in vitro* co-culture screens have significantly expanded our knowledge of the key tumor-intrinsic mechanisms mediating cytotoxic lymphocyte killing. However, the interactions between tumor cells and macrophages remain elusive. To illuminate the tumor-intrinsic regulator of macrophage phagocytosis, we developed both positive and negative screening strategies, and deployed multiple primary bone marrow-derived macrophages (BMDM) or macrophage cell line-based co-culture screens.

METHODS

DLD1 and MDA-MB-231 cancer lines were infected with a whole-genome CRISPR-Cas9 library and co-cultured with Raw264.7 or J774A.1 in the presence or absence of the therapeutic antibodies magrolimab or cetuximab. MC38 syngeneic cancer cells were infected with a library of 3000 immune-related genes and co-cultured with primary BMDM. For negative selection, the macrophages were selectively killed off post co-culture and the tumor cells were collected for next generation sequencing (NGS) and statistical analysis. For positive selection, macrophages and tumor cells were fully dissociated from the plate post co-culture, and the mixture of cells was subjected to a CD11b positive selection column. Macrophages and tumor cells were enriched separately for downstream analysis.

RESULTS

For the negative screen, bioinformatic analysis revealed that CD47 knockout in tumor cells strongly sensitized macrophage-mediated clearance of target cells both from naïve and cetuximab-treated conditions. Furthermore, CD47 or EGFR knockout conveyed growth advantage in co-culture upon treatment with magrolimab or cetuximab, respectively. For the positive screen, we developed a method to sequence bar-coded DNA from tumor cells engulfed by macrophages. Consistently, CD47 sgRNAs were significantly enriched inside macrophages in the naïve condition and were depleted upon treatment with magrolimab. Together, these findings demonstrated the robustness of our phagocytosis-based CRISPR screening platform. In additional to validating the role of CD47 in this system, we discovered and validated a group of novel tumor intrinsic regulators of macrophage-mediated phagocytosis.

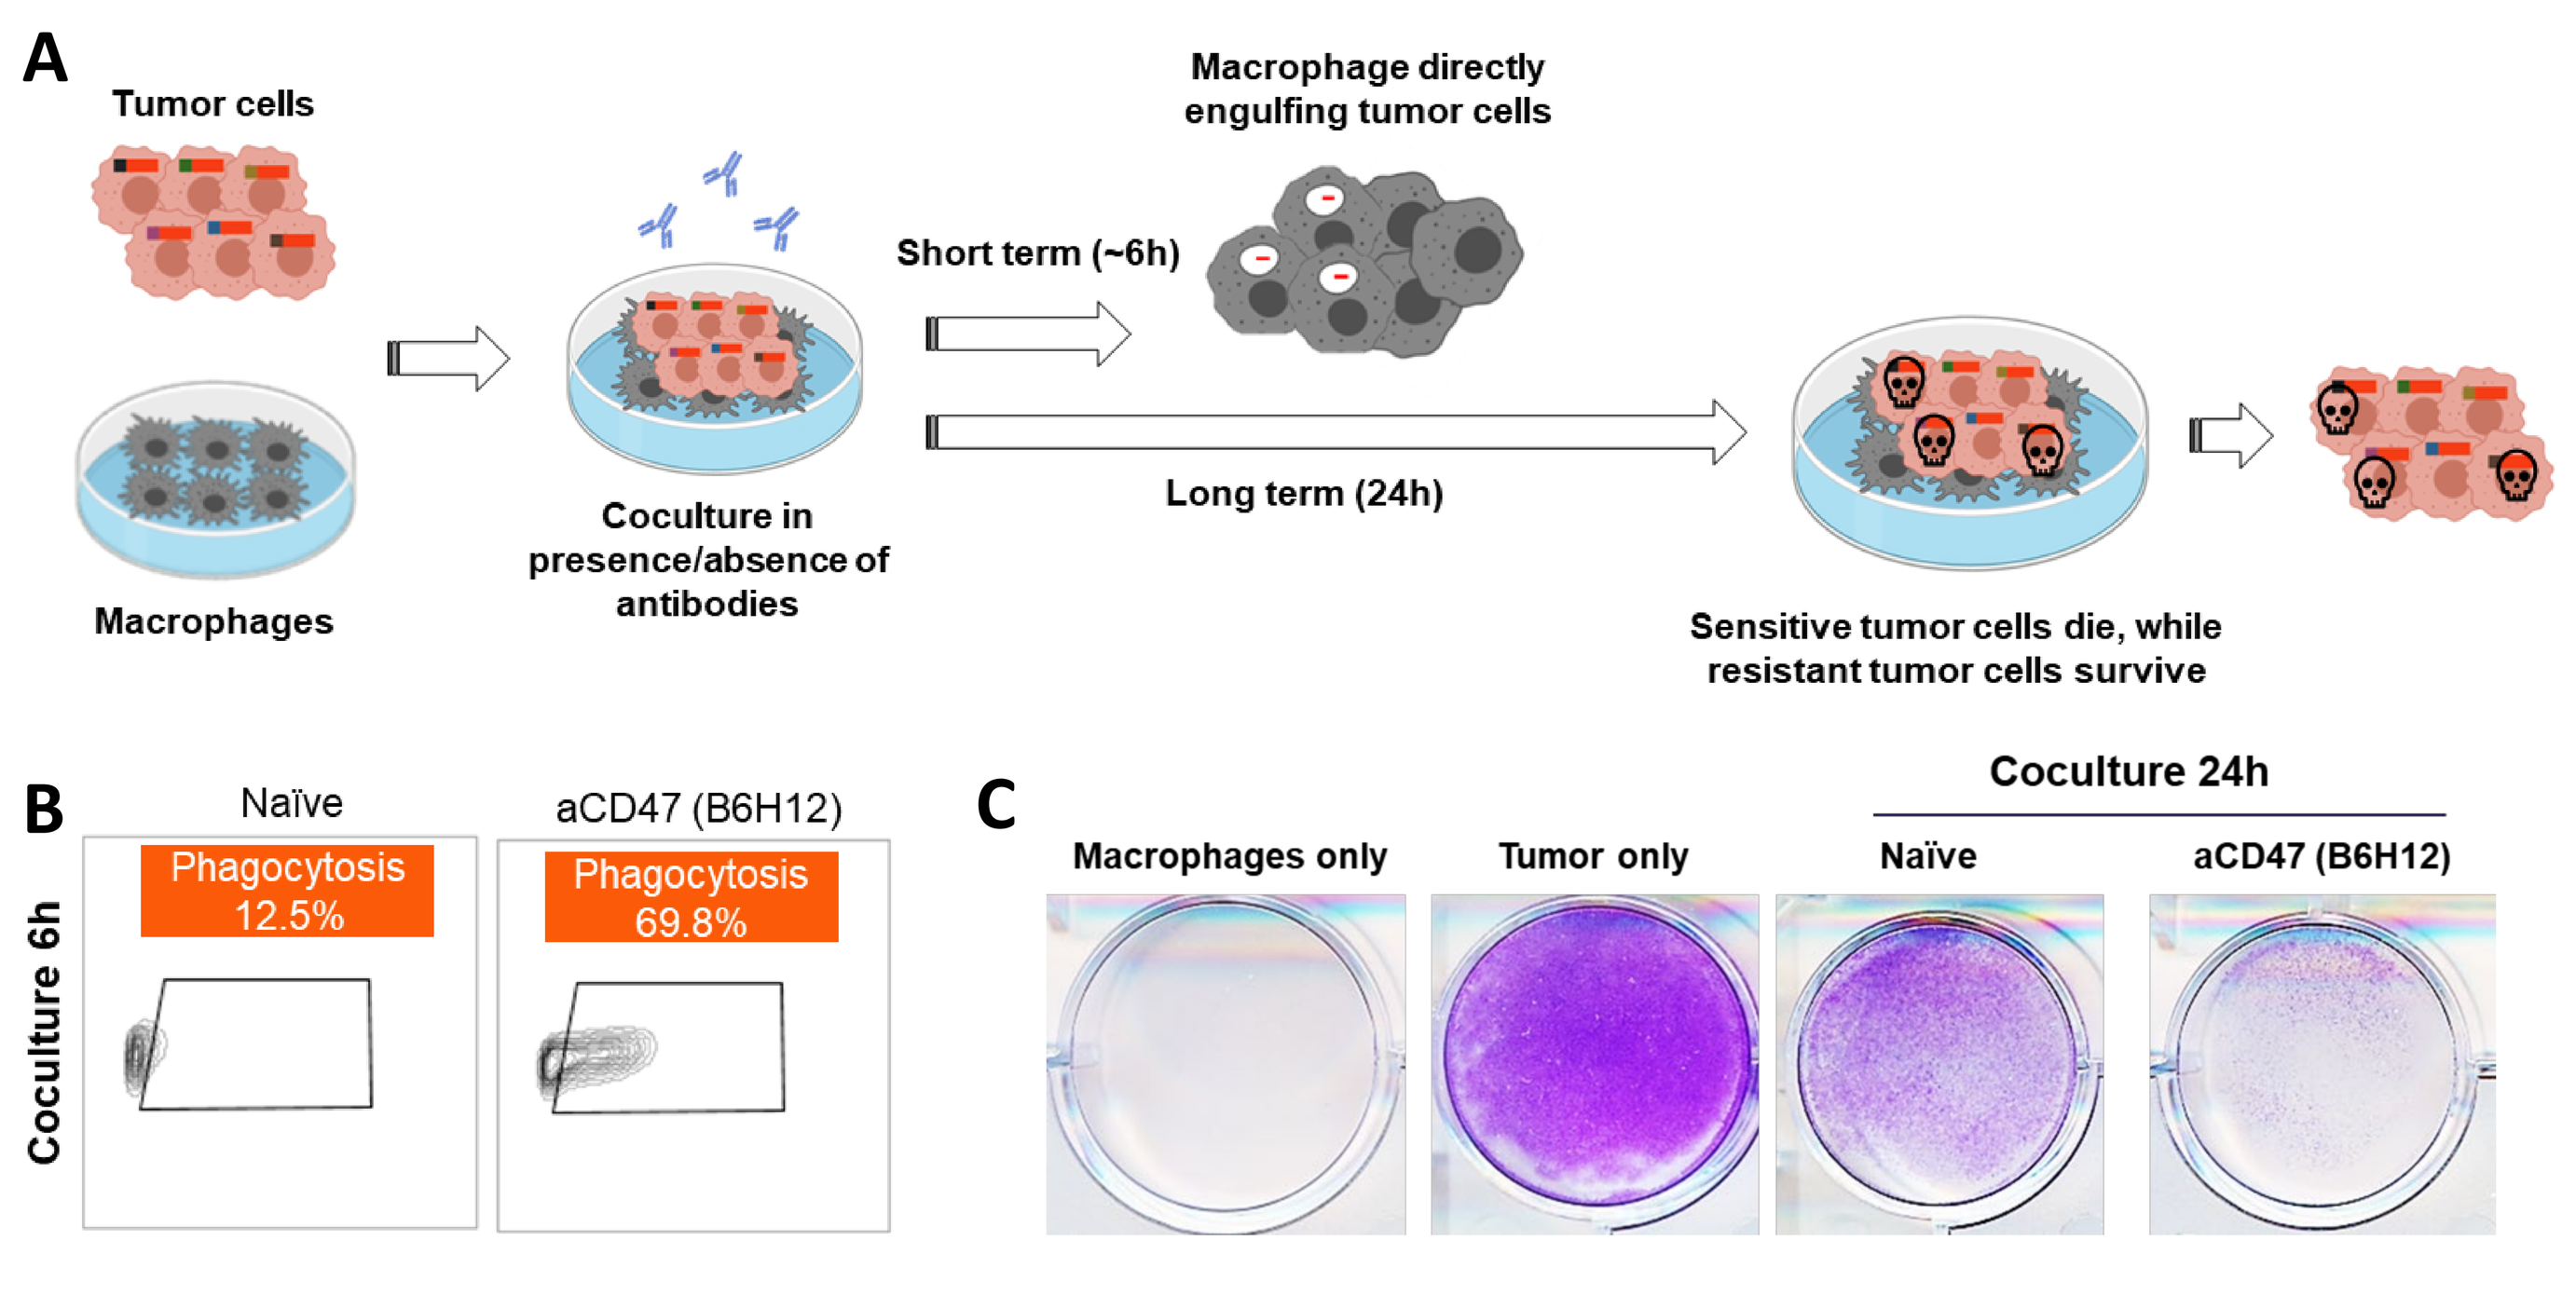
A
Tumor cells
Macrophages
Coculture in presence/absence of antibodies
Short term (~6h)
Macrophage directly engulfing tumor cells
Long term (24h)
Sensitive tumor cells die, while resistant tumor cells survive
B
Naïve
aCD47 (B6H12)
Phagocytosis 12.5%
Phagocytosis 69.8%
Coculture 6h
C
Coculture 24h
Macrophages only
Tumor only
Naïve
aCD47 (B6H12)

Figure 1: Development of *in vitro* tumor/macrophage co-culture platform. (A) Schematic diagram showing *in vitro* tumor/macrophage coculture and its two different stages; (B) Flow cytometry detecting tumor cell-engulfing macrophages after 6h of co-culture; (C) Crystal violet staining of remaining tumor cells after 24h of co-culture.

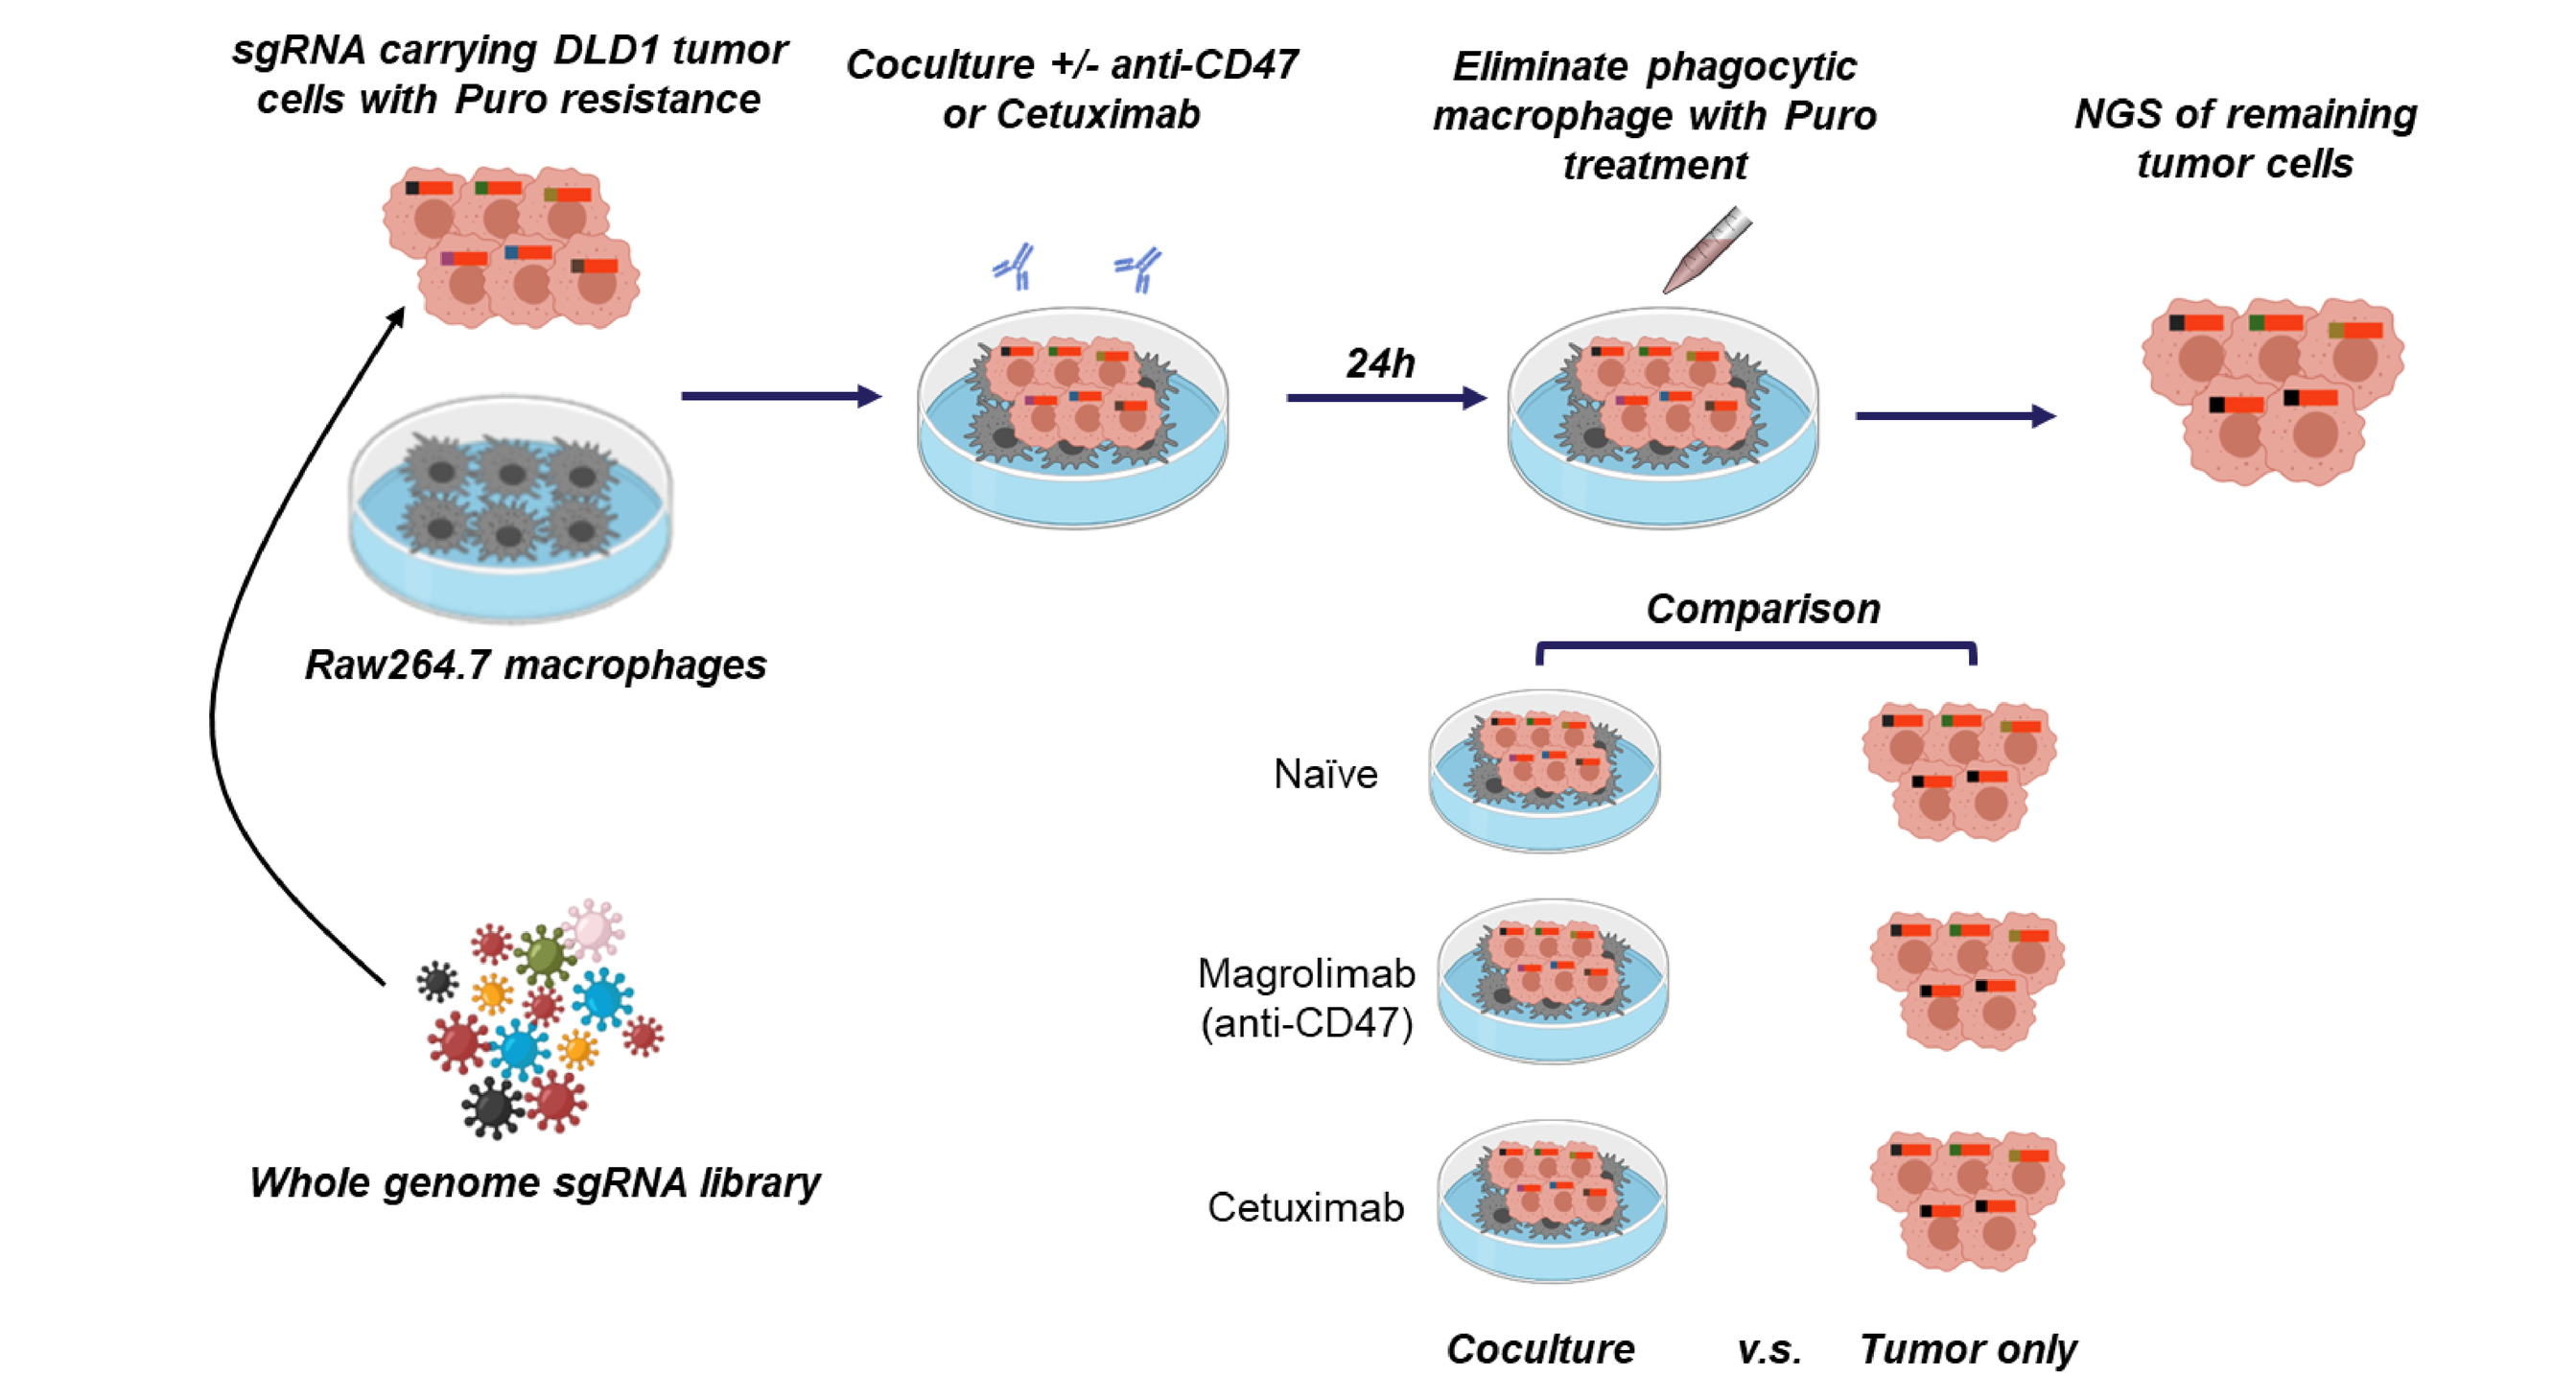
sgRNA carrying DLD1 tumor cells with Puro resistance
Coculture +/- anti-CD47 or Cetuximab
Eliminate phagocytic macrophage with Puro treatment
NGS of remaining tumor cells
24h
Raw264.7 macrophages
Whole genome sgRNA library
Comparison
Naïve
Magrolimab (anti-CD47)
Cetuximab
Coculture
v.s.
Tumor only

Figure 2: Negative selection screen strategy for *in vitro* DLD1/Raw264.7 co-culture. Comparison analysis of NGS data was made between remaining tumor cells from coculture and tumor only across 3 treatment conditions.

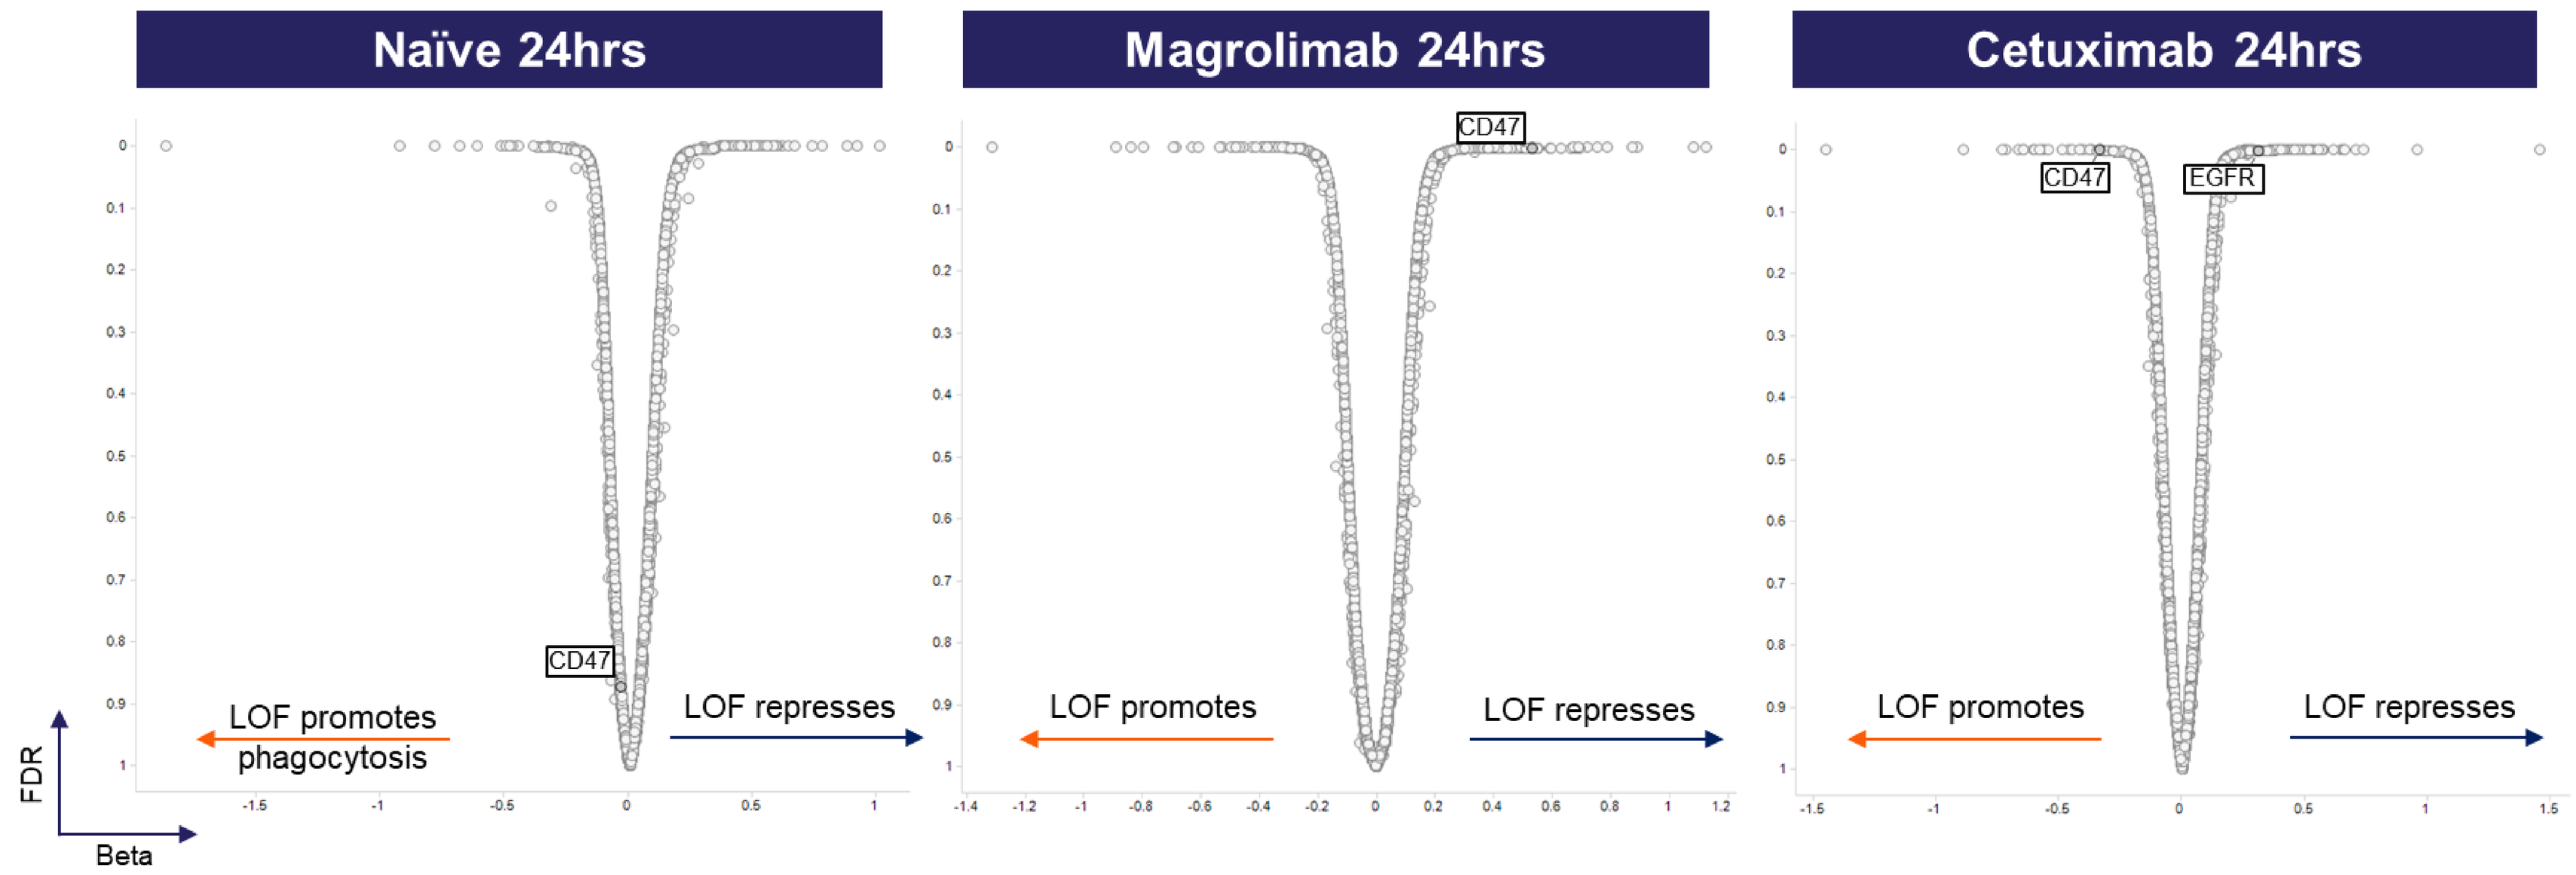
Naïve 24hrs
Magrolimab 24hrs
Cetuximab 24hrs
CD47
EGFR
LOF promotes phagocytosis
LOF promotes
LOF represses
FDR
Beta

Figure 3: Genome-wide CRISPR negative screen identifies tumor intrinsic phagocytosis regulators in DLD1/Raw264.7 co-culture. Hits on the left side indicate depletion of these genes promotes phagocytosis; Hits on the right side indicate depletion of these genes repress phagocytosis.

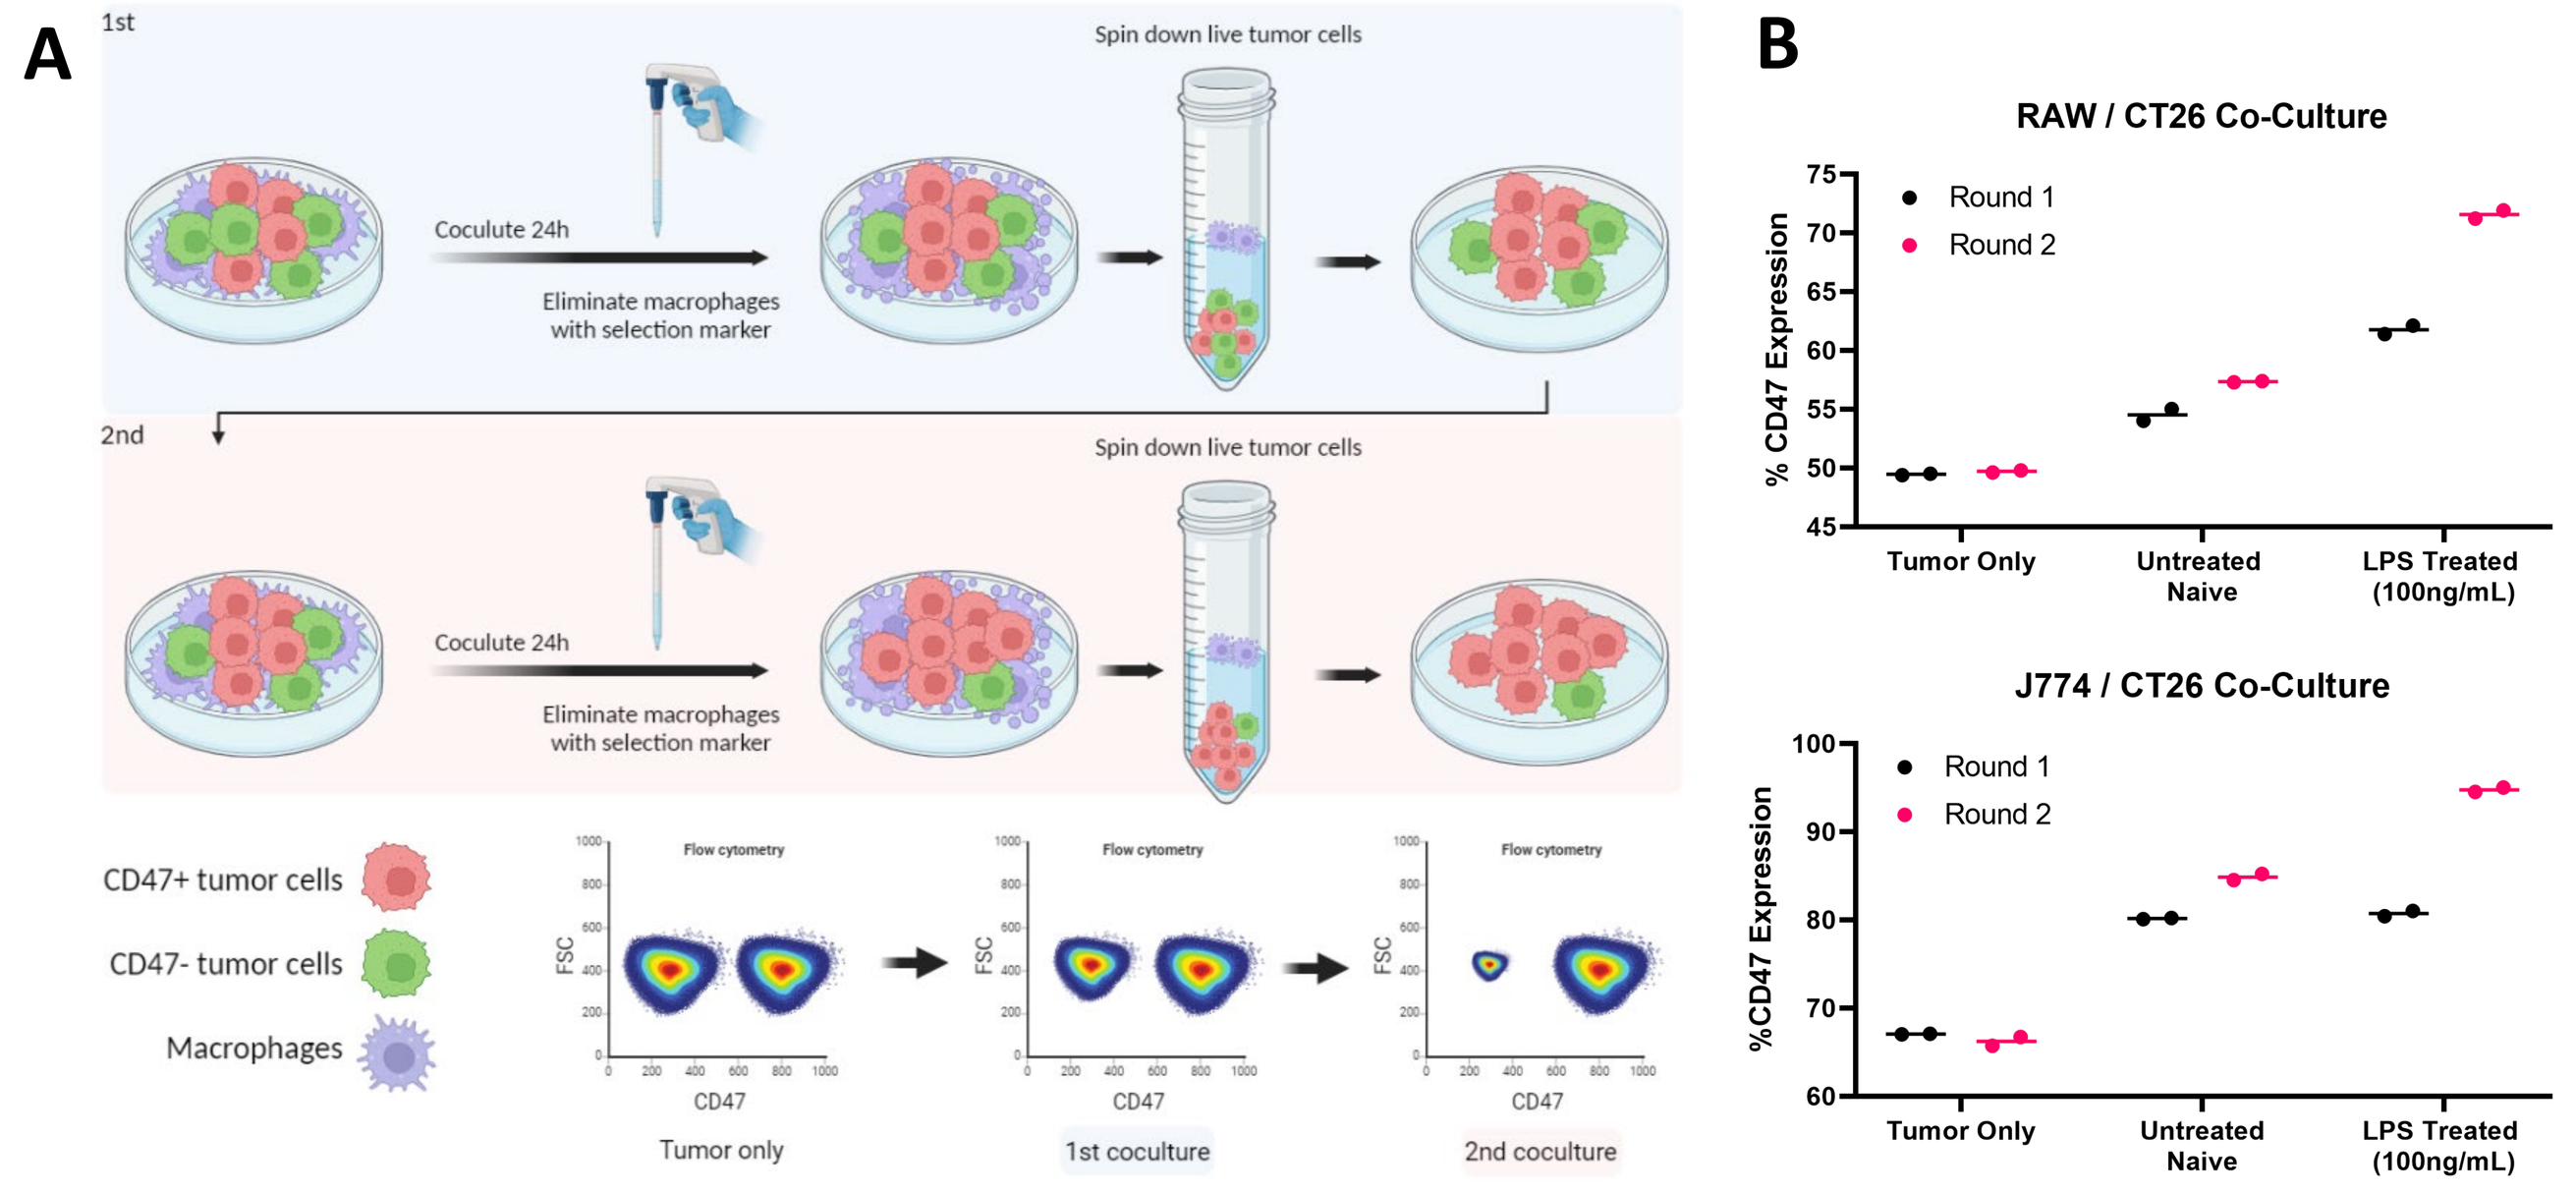
A
1st
2nd
Coculte 24h
Eliminate macrophages with selection marker
Spin down live tumor cells
CD47+ tumor cells
CD47- tumor cells
Macrophages
Flow cytometry
Tumor only
1st coculture
2nd coculture
B
RAW / CT26 Co-Culture
J774 / CT26 Co-Culture
Round 1
Round 2
% CD47 Expression
Tumor Only
Untreated Naive
LPS Treated (100ng/mL)

Figure 4: Development of *in vitro* tumor/macrophage two-round competition co-culture platform. (A) Schematic diagram of 2-round co-culture; (B) CD47+ tumor cells are further enriched after 2-round co-culture.

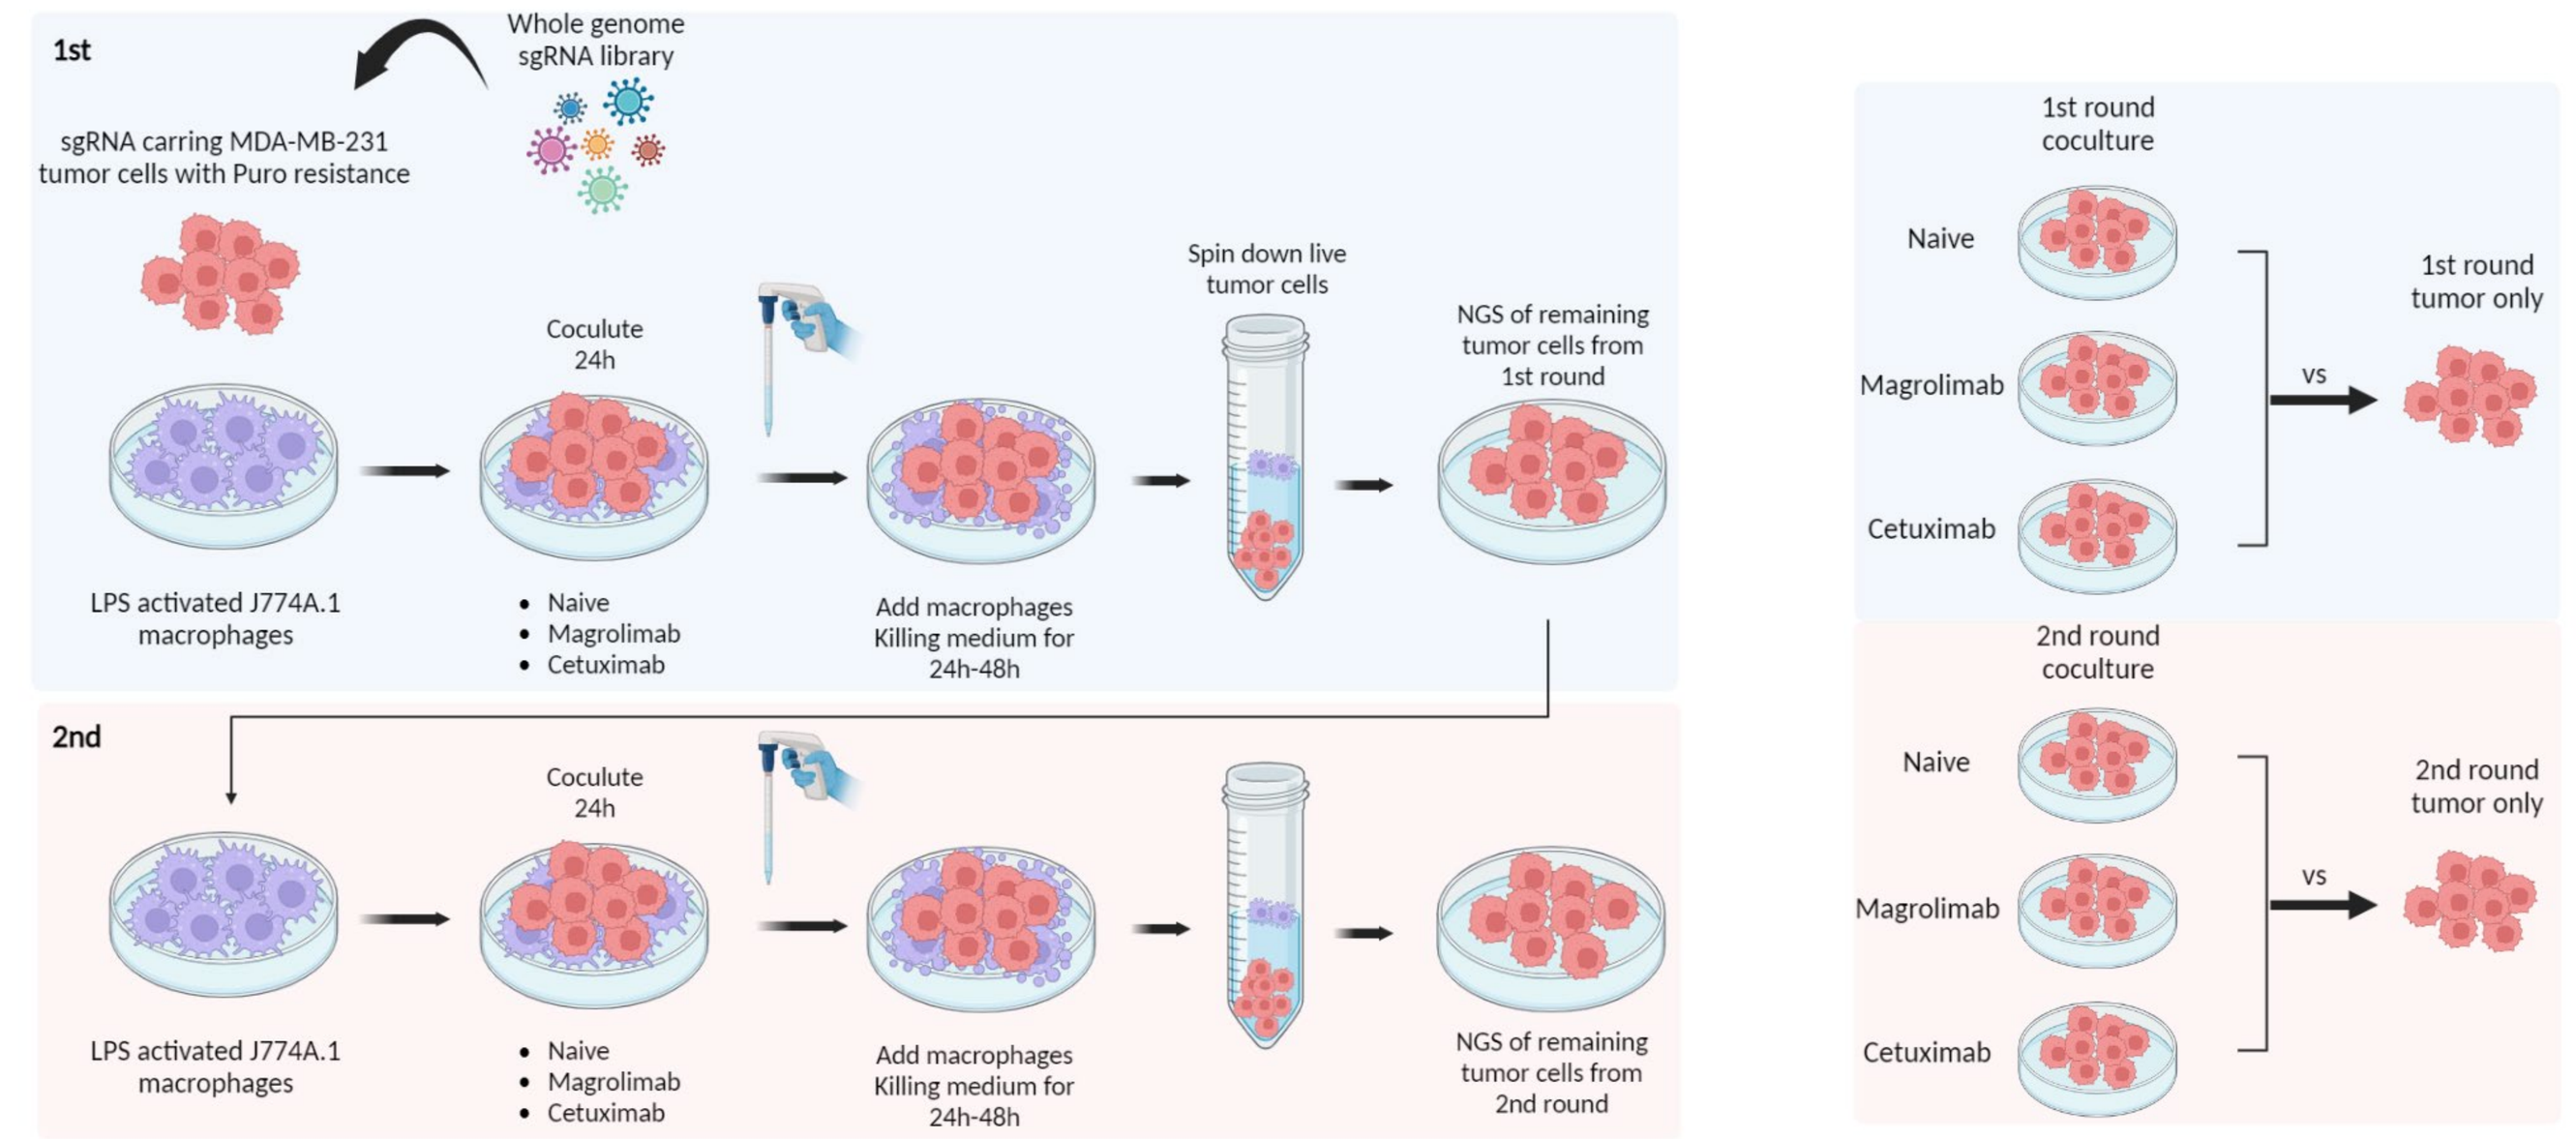
1st
2nd
sgRNA carring MDA-MB-231 tumor cells with Puro resistance
Whole genome sgRNA library
LPS activated J774A.1 macrophages
Coculte 24h
Naive
Magrolimab
Cetuximab
Add macrophages Killing medium for 24h-48h
Spin down live tumor cells
NGS of remaining tumor cells from 1st round
NGS of remaining tumor cells from 2nd round
1st round coculture
2nd round coculture
1st round tumor only
2nd round tumor only
vs

Figure 5: Negative selection screen strategy for *in vitro* MDA-MB-231 two-round co-culture with LPS-activated J774A.1

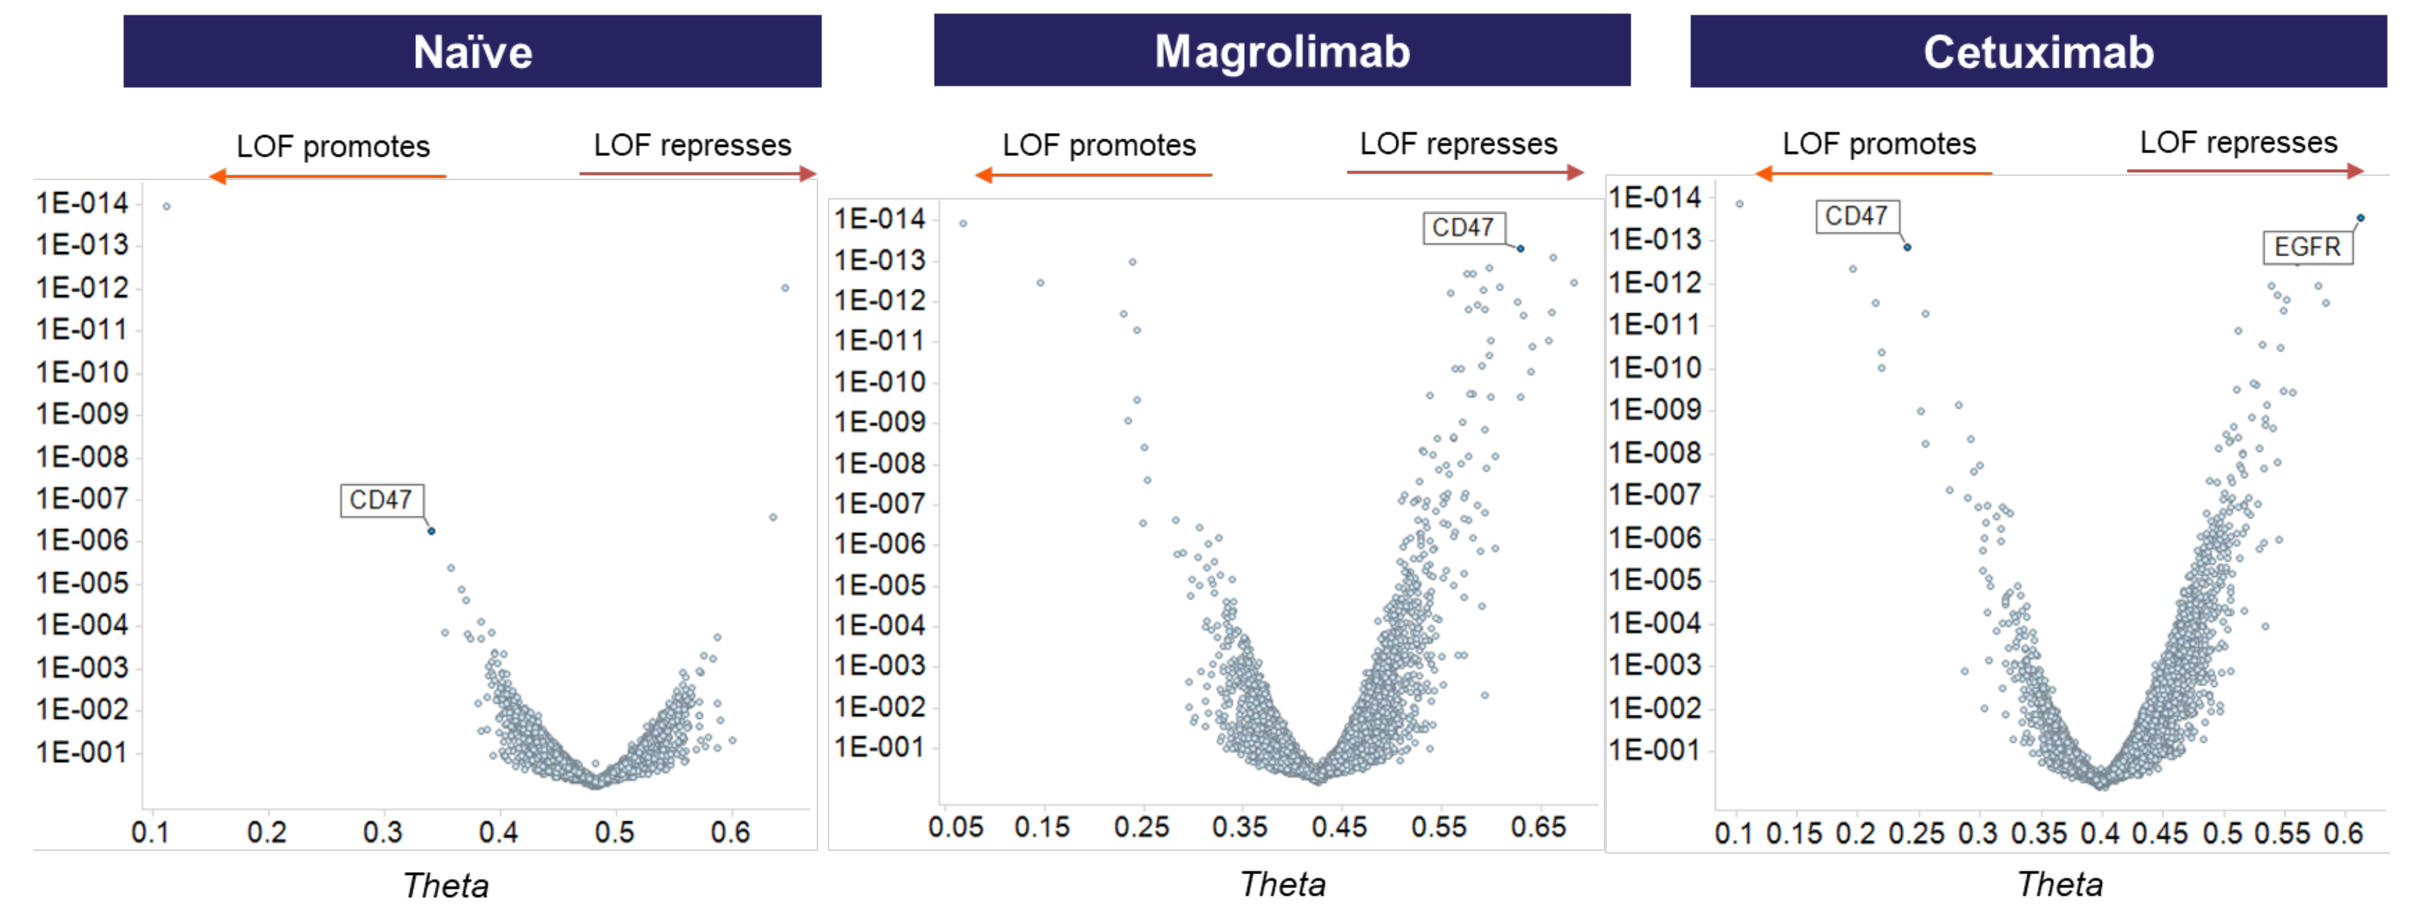
Naïve
Magrolimab
Cetuximab
LOF promotes
LOF represses
CD47
EGFR
Theta

Figure 6: Genome-wide CRISPR negative screen identifies tumor intrinsic phagocytosis regulators in MDA-MB-231 two-round co-culture with LPS-activated J774A.1

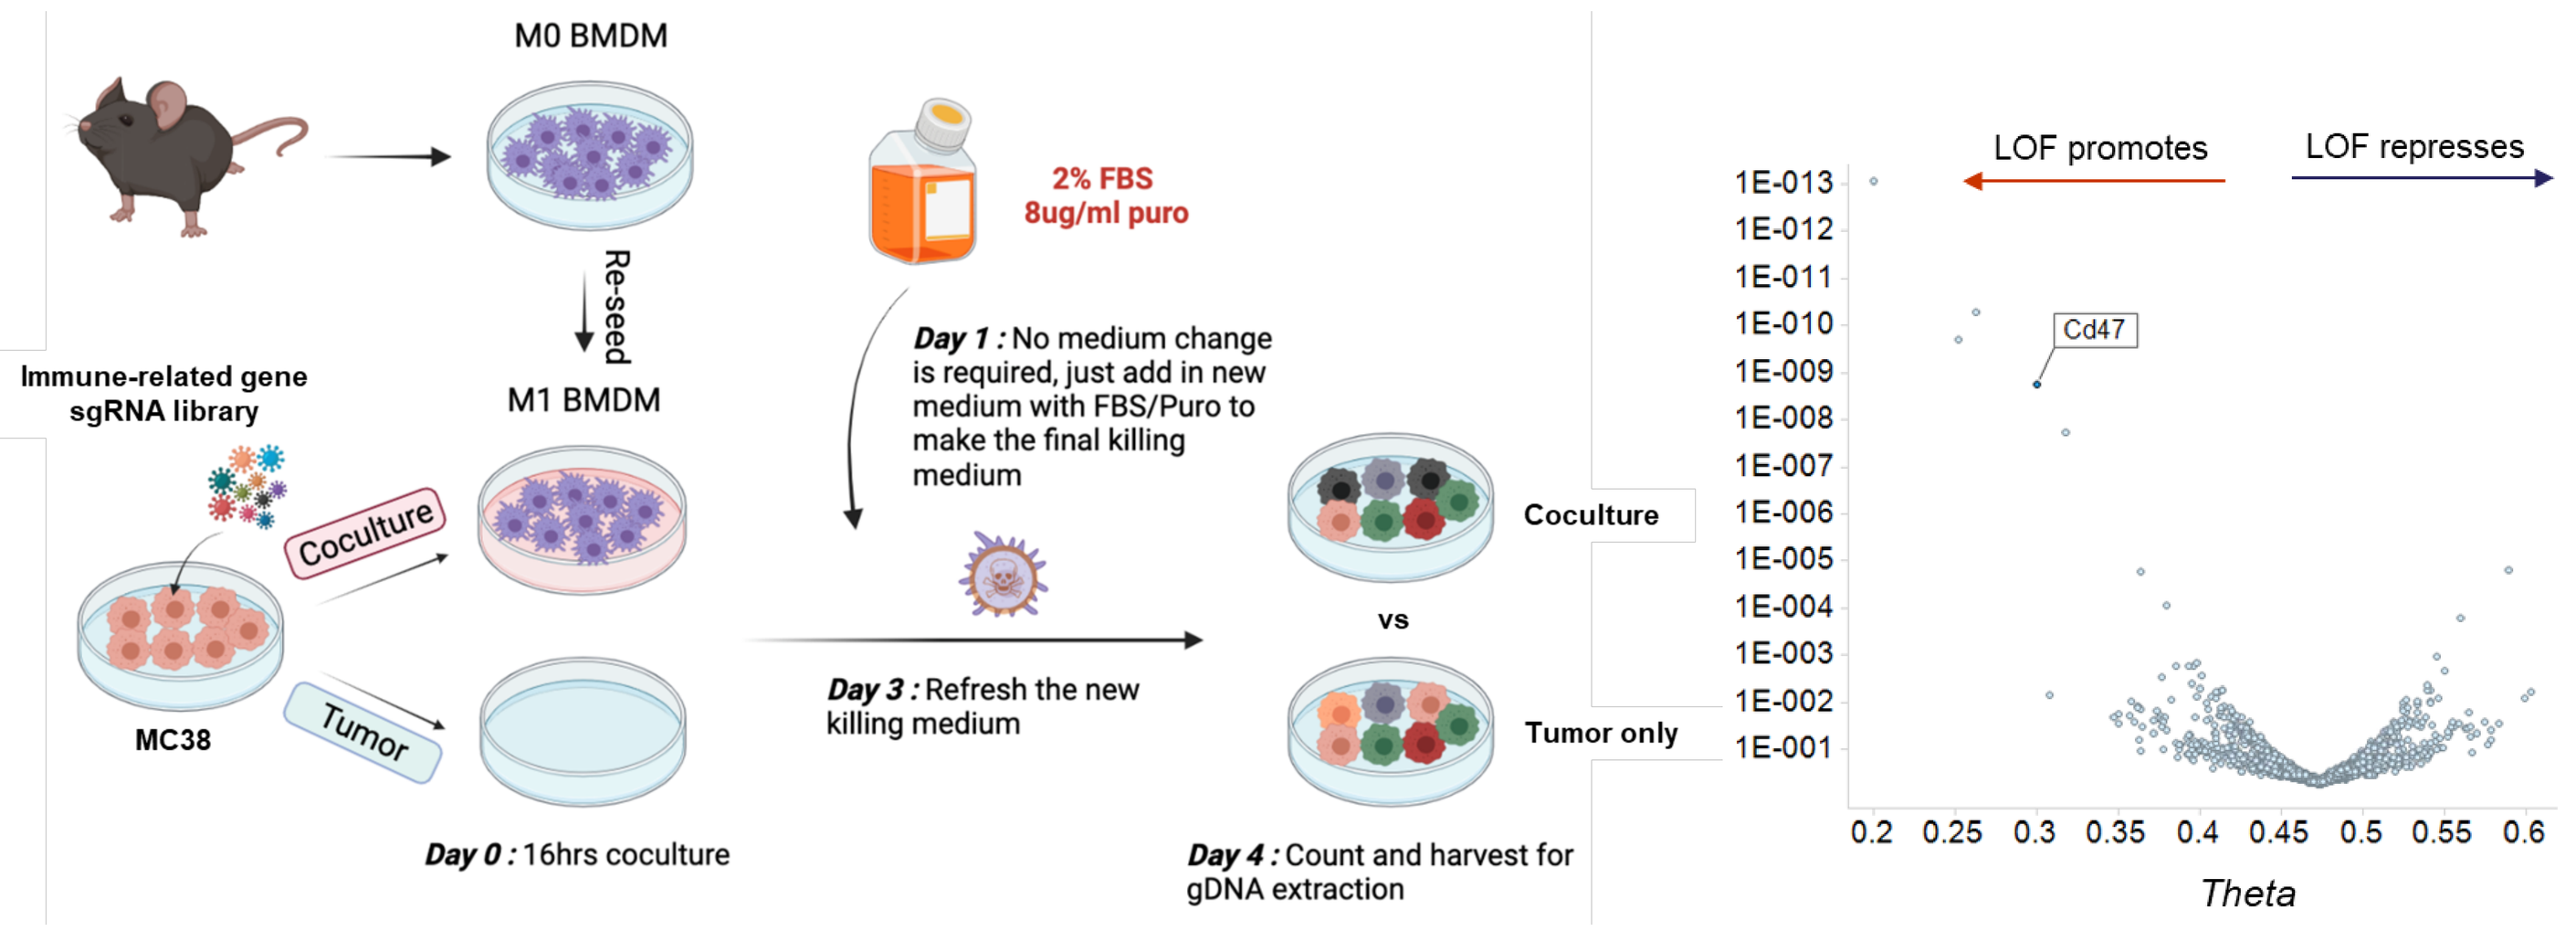
M0 BMDM
Re-seed
M1 BMDM
Immune-related gene sgRNA library
MC38
Coculture
Tumor
2% FBS 8ug/ml puro
Day 1 : No medium change is required, just add in new medium with FBS/Puro to make the final killing medium
Day 3 : Refresh the new killing medium
Day 0 : 16hrs coculture
Day 4 : Count and harvest for gDNA extraction
Coculture
vs
Tumor only
LOF promotes
LOF represses
Cd47
Theta

Figure7: Negative selection screen strategy for *in vitro* MC38/Primary BMDM co-culture and tumor intrinsic phagocytosis regulators identified

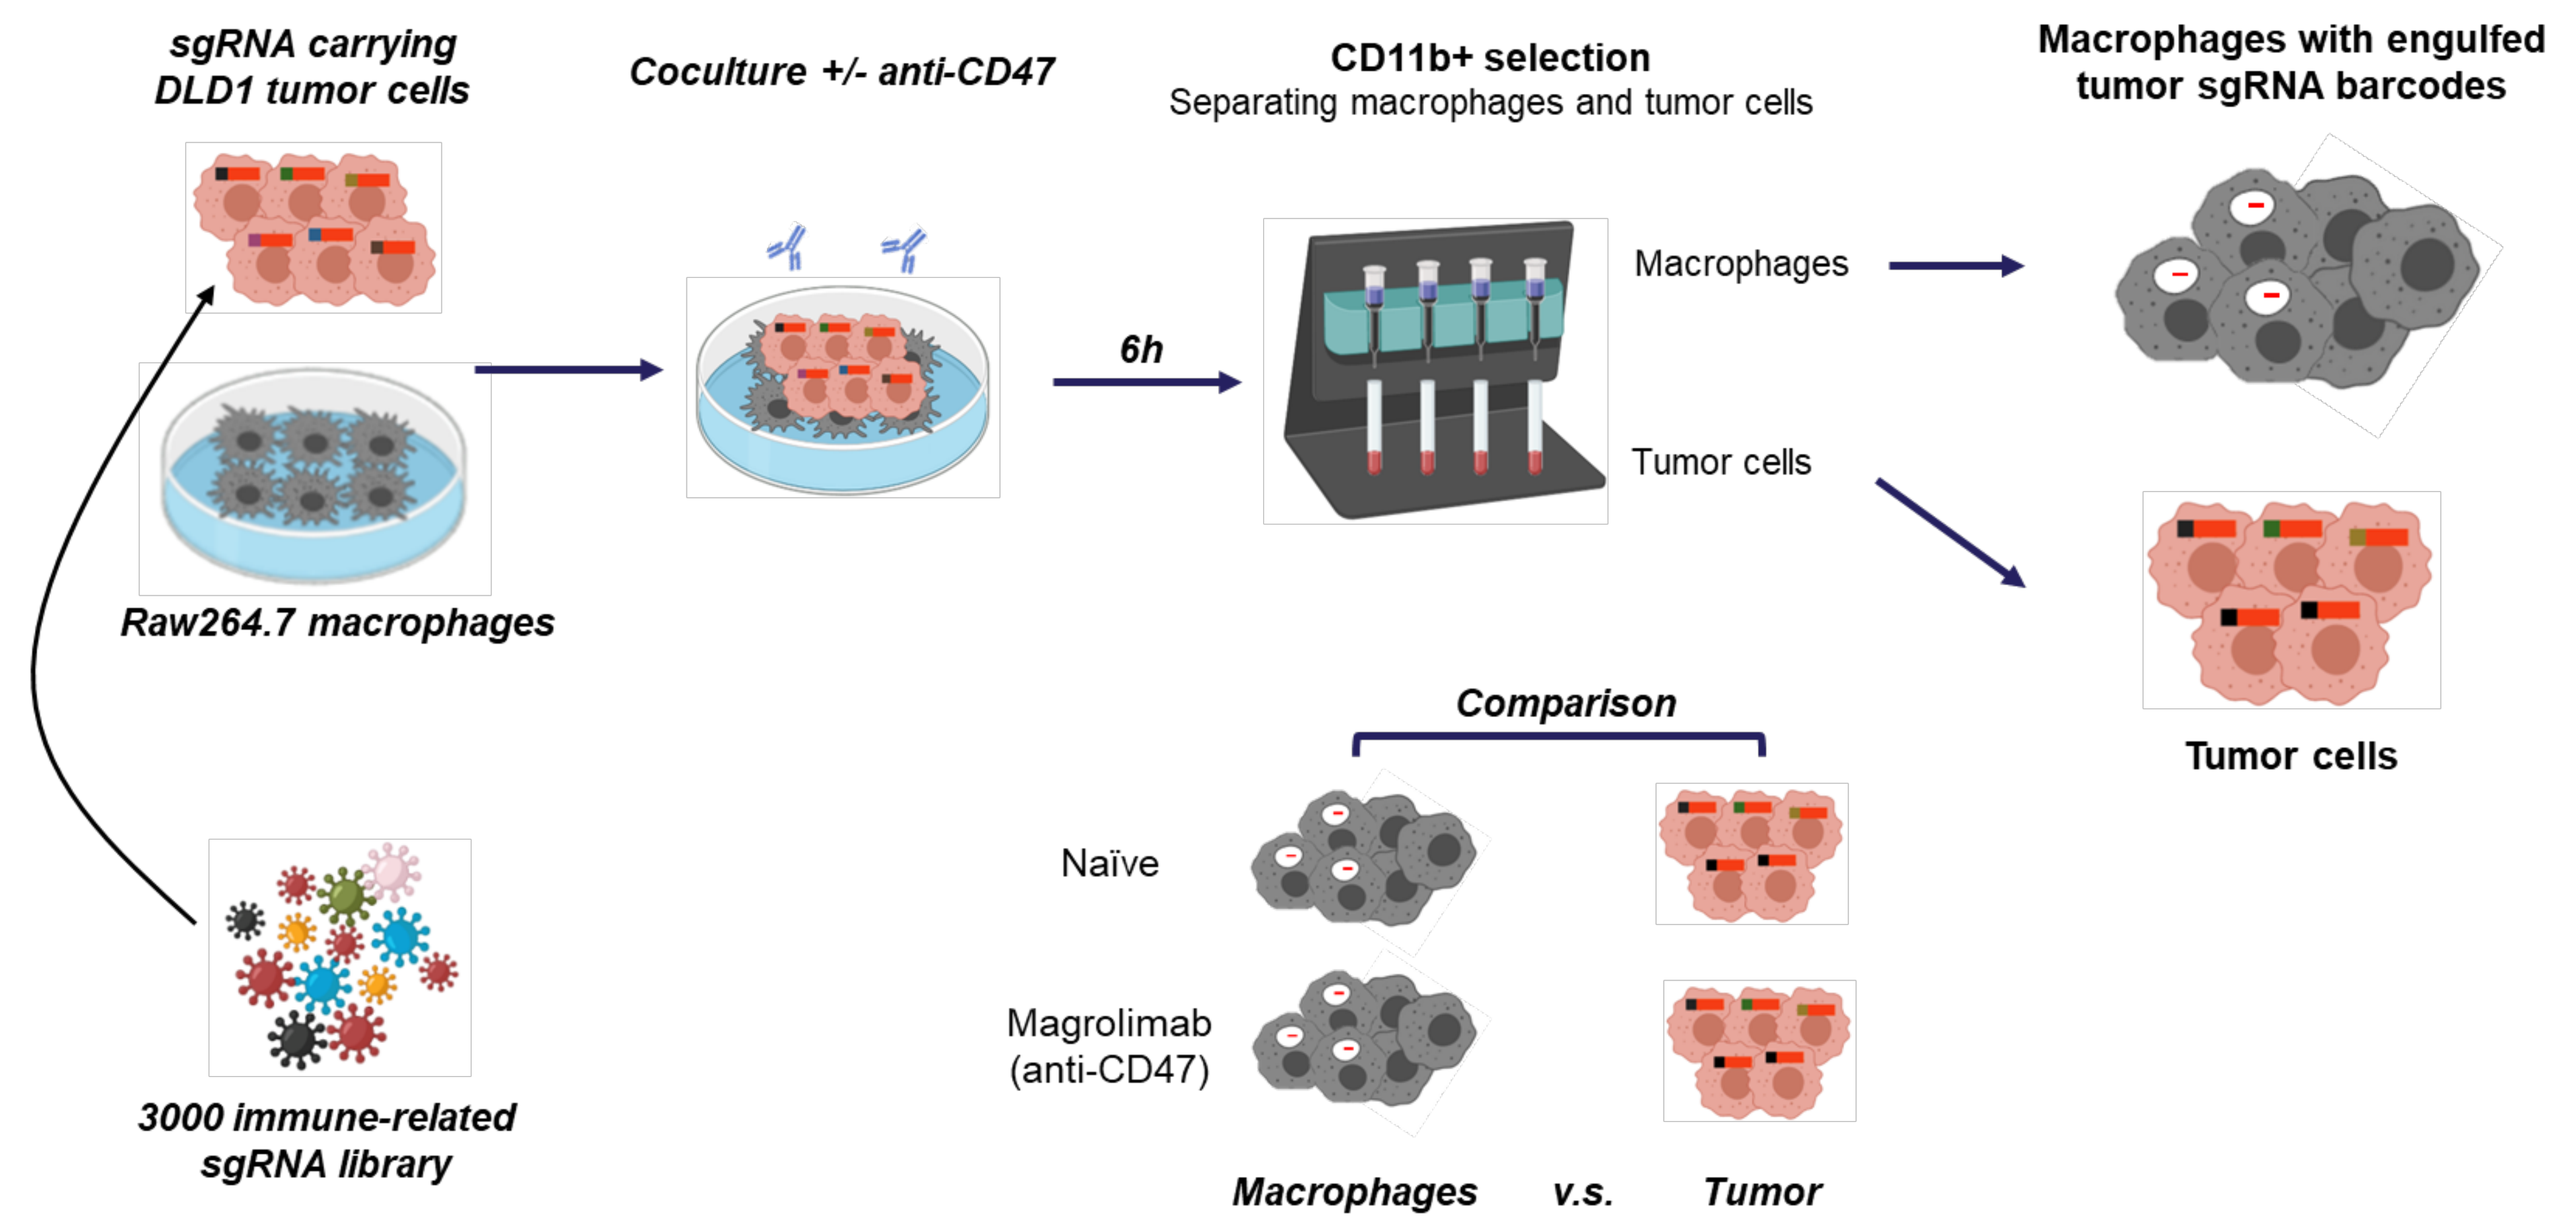
sgRNA carrying DLD1 tumor cells
Coculture +/- anti-CD47
CD11b+ selection
Separating macrophages and tumor cells
Macrophages with engulfed tumor sgRNA barcodes
6h
Macrophages
Tumor cells
Raw264.7 macrophages
3000 immune-related sgRNA library
Comparison
Naïve
Magrolimab (anti-CD47)
Macrophages
v.s.
Tumor

Figure 8: Positive selection screen strategy for *in vitro* DLD1/Raw264.7 co-culture. Comparison analysis of NGS data was made between macrophages and tumor cells from co-culture.

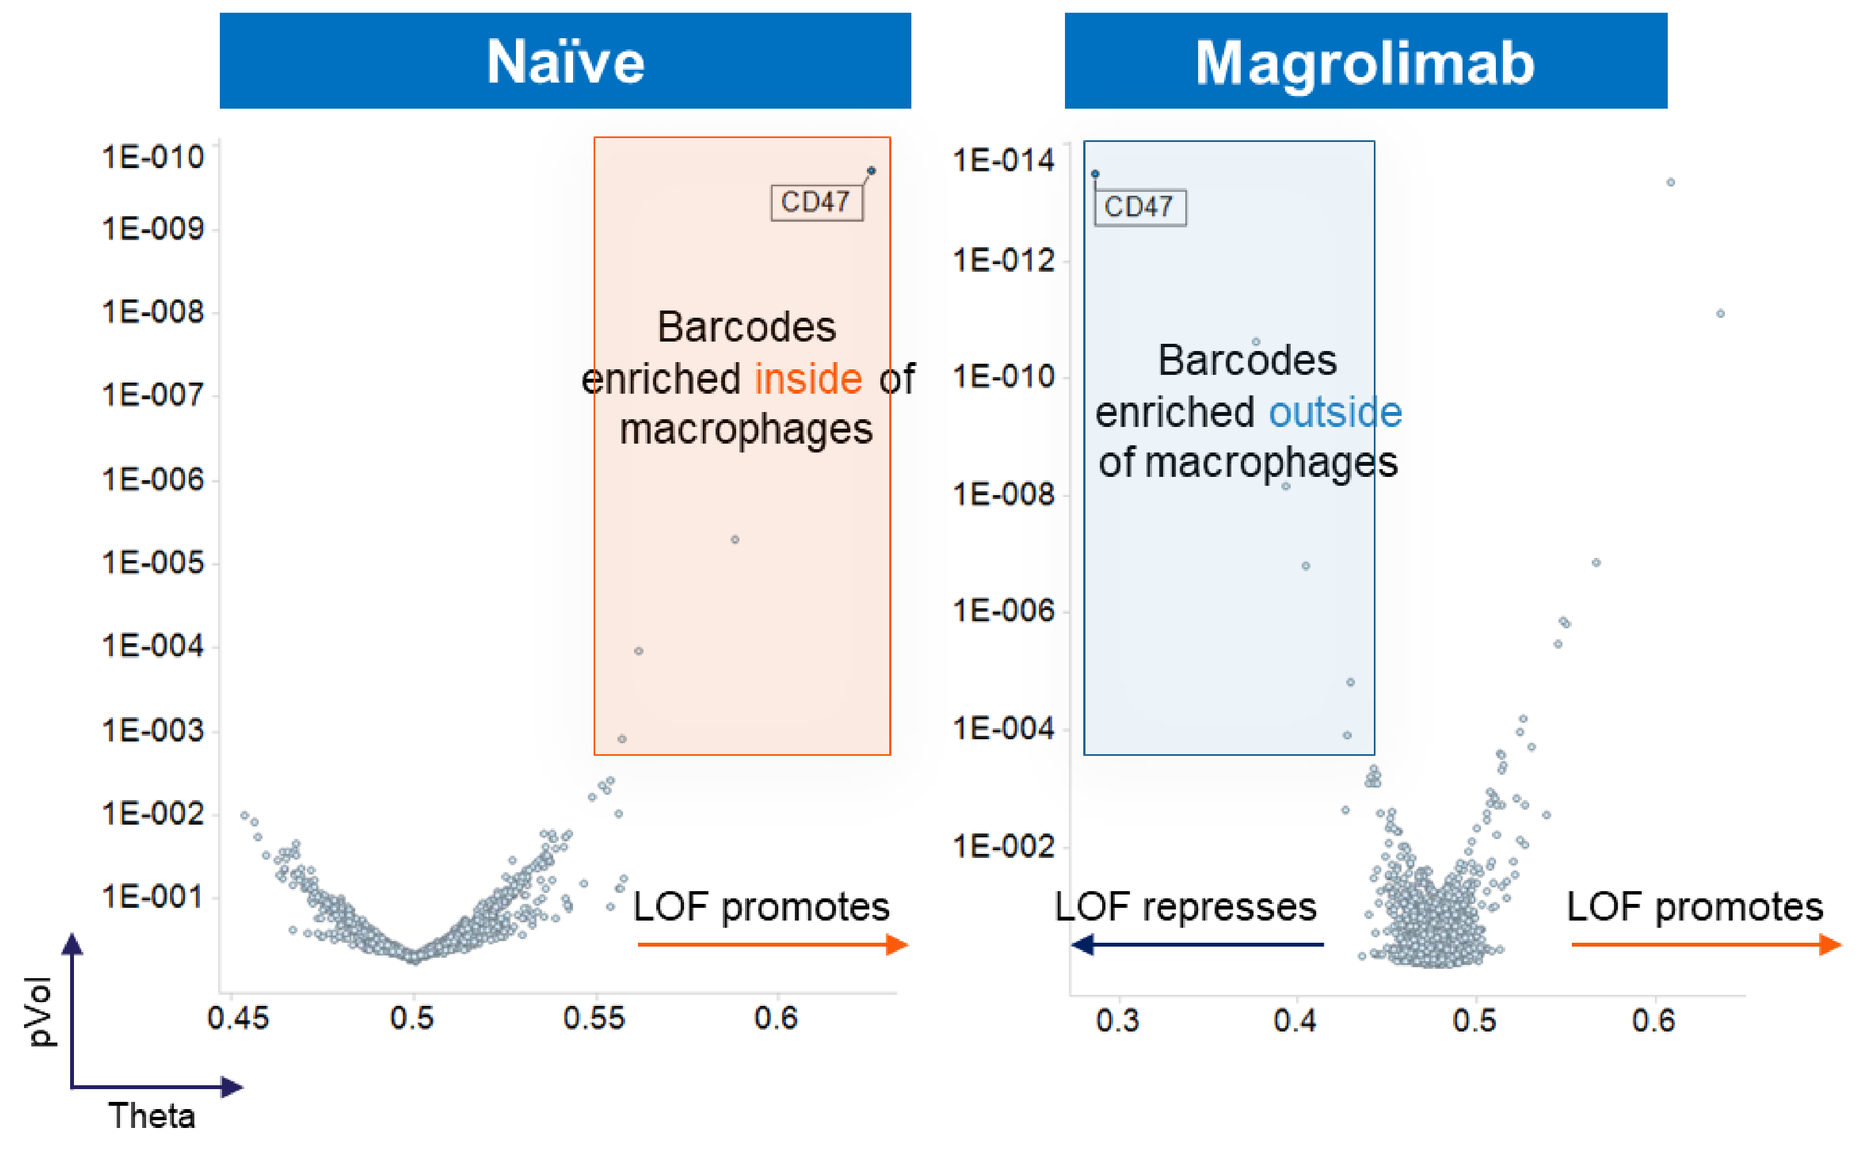
Naïve
Magrolimab
CD47
Barcodes enriched inside of macrophages
Barcodes enriched outside of macrophages
LOF promotes
LOF represses
pVol
Theta

Figure 9: CRISPR positive screen identifies tumor intrinsic phagocytosis regulators in DLD1/Raw264.7 co-culture

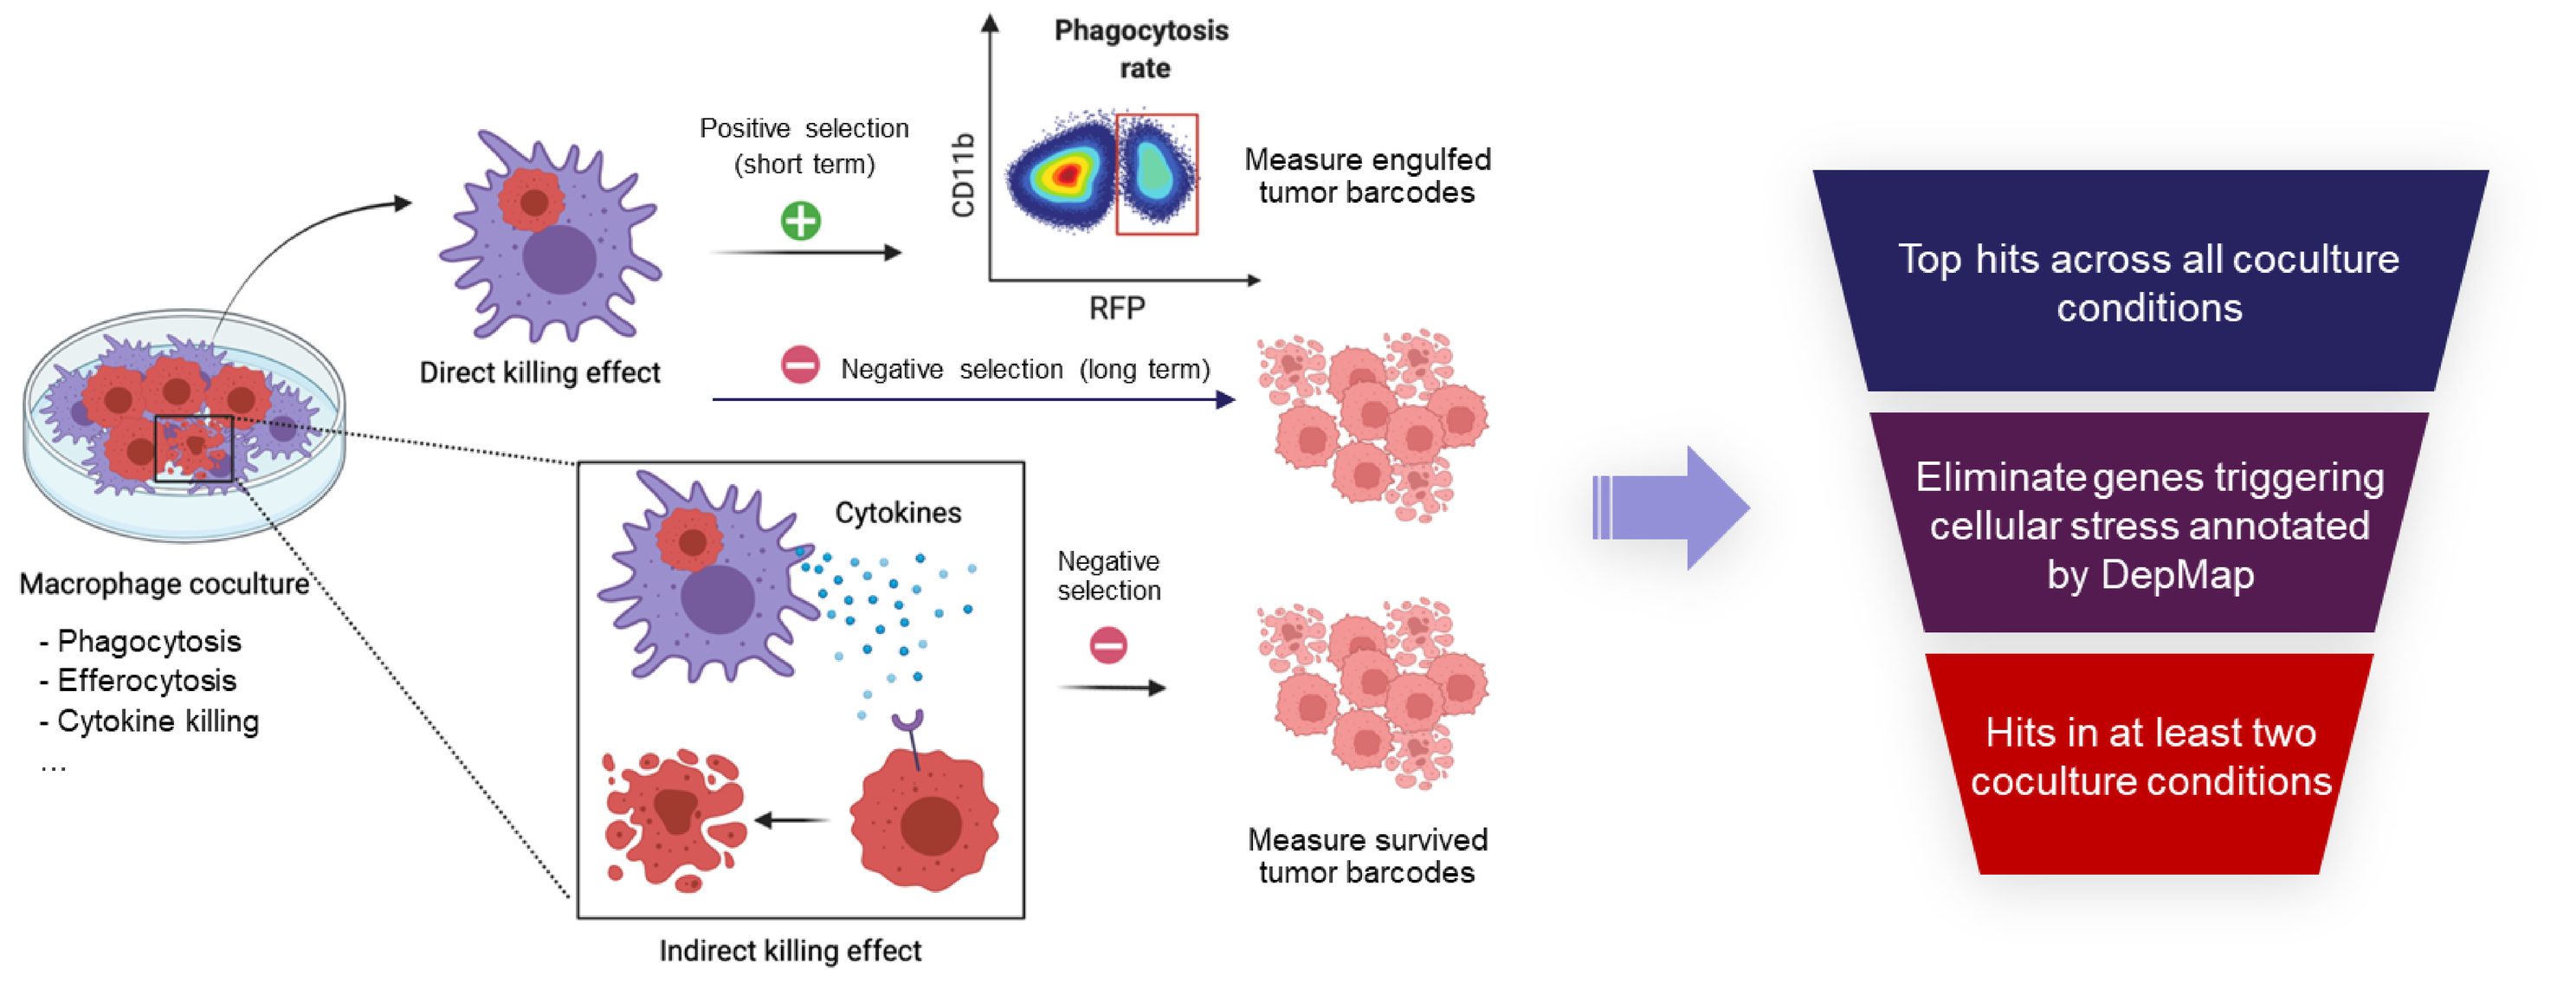
Macrophage coculture
- Phagocytosis
- Efferocytosis
- Cytokine killing
...
Direct killing effect
Positive selection (short term)
Phagocytosis rate
CD11b
RFP
Measure engulfed tumor barcodes
Negative selection (long term)
Cytokines
Negative selection
Indirect killing effect
Measure survived tumor barcodes
Top hits across all coculture conditions
Eliminate genes triggering cellular stress annotated by DepMap
Hits in at least two coculture conditions

Figure 10: Strategy for phagocytosis regulator hits interrogation. Short term positive selection identifies targets directly involved in enhancing macrophage engulfment of tumor cells; Long term negative selection identifies targets involved in tumor cells resistant to phagocytosis and/or macrophage-derived cytokines-mediated killing.

CONCLUSION

We have successfully established and implemented multiple macrophage and tumor cell line co-culture screening platforms, and systematically revealed tumor intrinsic regulators of phagocytosis, uncovering multiple known and novel therapeutic targets regulating vulnerability of tumor cells to macrophage-mediated phagocytosis. Our proprietary high-throughput phagocytosis CRISPR-based screening platform provides an unbiased and rapid solution to study macrophage and tumor cell interactions and discover novel targets for cancer therapy.