Whole genome CRISPR-Cas9 screens in a cancer cell line panel co-cultured with antigen-specific cytotoxic CD8 T cells are a powerful engine for immuno-oncology drug target discovery

TANGO therapeutics

Serge Gueroussov, Disha Subramanya, Jessica Finkler, Bailey Smith, Lei Ji, Ashley Choi, Tenzing Khendu, Samuel Meier, Shangtao Liu, Binzhang Shen, Teng Teng, Yi Yu, Alan Huang, Chengyin Min

ABSTRACT

Background:
Cytotoxic T lymphocytes (CTLs) are a key driver of the anti-tumor immune response. Understanding the molecular mechanisms mediating this process can reveal therapeutic targets whose pharmacological inhibition can increase responses to immunotherapy. In recent years, forward genetic screens with CRISPR-Cas9 have been successfully applied to study the interaction between tumor cells and CTLs in vitro *(1)*. Here, we extend this approach to a broad panel of human cancer cell lines and define methods for prioritization and validation of immuno-oncology therapeutic targets.

Methods:
A panel of seven HLA-A*02 cancer cell lines were prioritized to span multiple cancer lineages. Tumor cells were infected with a whole-genome CRISPR-Cas9 library and co-cultured with primary human CD8+ T cells expressing the NYESO1 HLA-A*02-restricted T cell receptor. Next-generation sequencing and statistical analysis were used to define the top genes that affected tumor cell killing by CTLs. We validated the top hits from the screen in vitro and developed an adoptive cell transfer model to validate the sensitizing effects in vivo.

Results:
Comparison of screen hits uncovered both known and novel pathways that sensitize tumor cell lines to CTL killing, including surface checkpoint molecules, epigenetic regulators, genes that control cytokine response, autophagy, post-transcriptional regulation, and cell surface glycosylation. We prioritized genes for validation based on effect size, druggability, and TCGA correlation between target expression and an immune-deficient tumor microenvironment. We showcase our approach using the known immune regulator, PTPN2 *(2)*. PTPN2 knockout in tumor cells sensitized them to CTL-mediated killing in co-culture assay in vitro and adoptive cell transfer (ACT) model in vivo.

Conclusions:
Our cancer cell and T cell co-culture CRISPR screening platform revealed multiple known as well as novel genes to comprehensively characterize the mechanisms regulating tumor cell killing by CTLs, providing a rich resource of therapeutic targets to advance into drug discovery.

SCREENING STRATEGY AND T CELL ENGINEERING

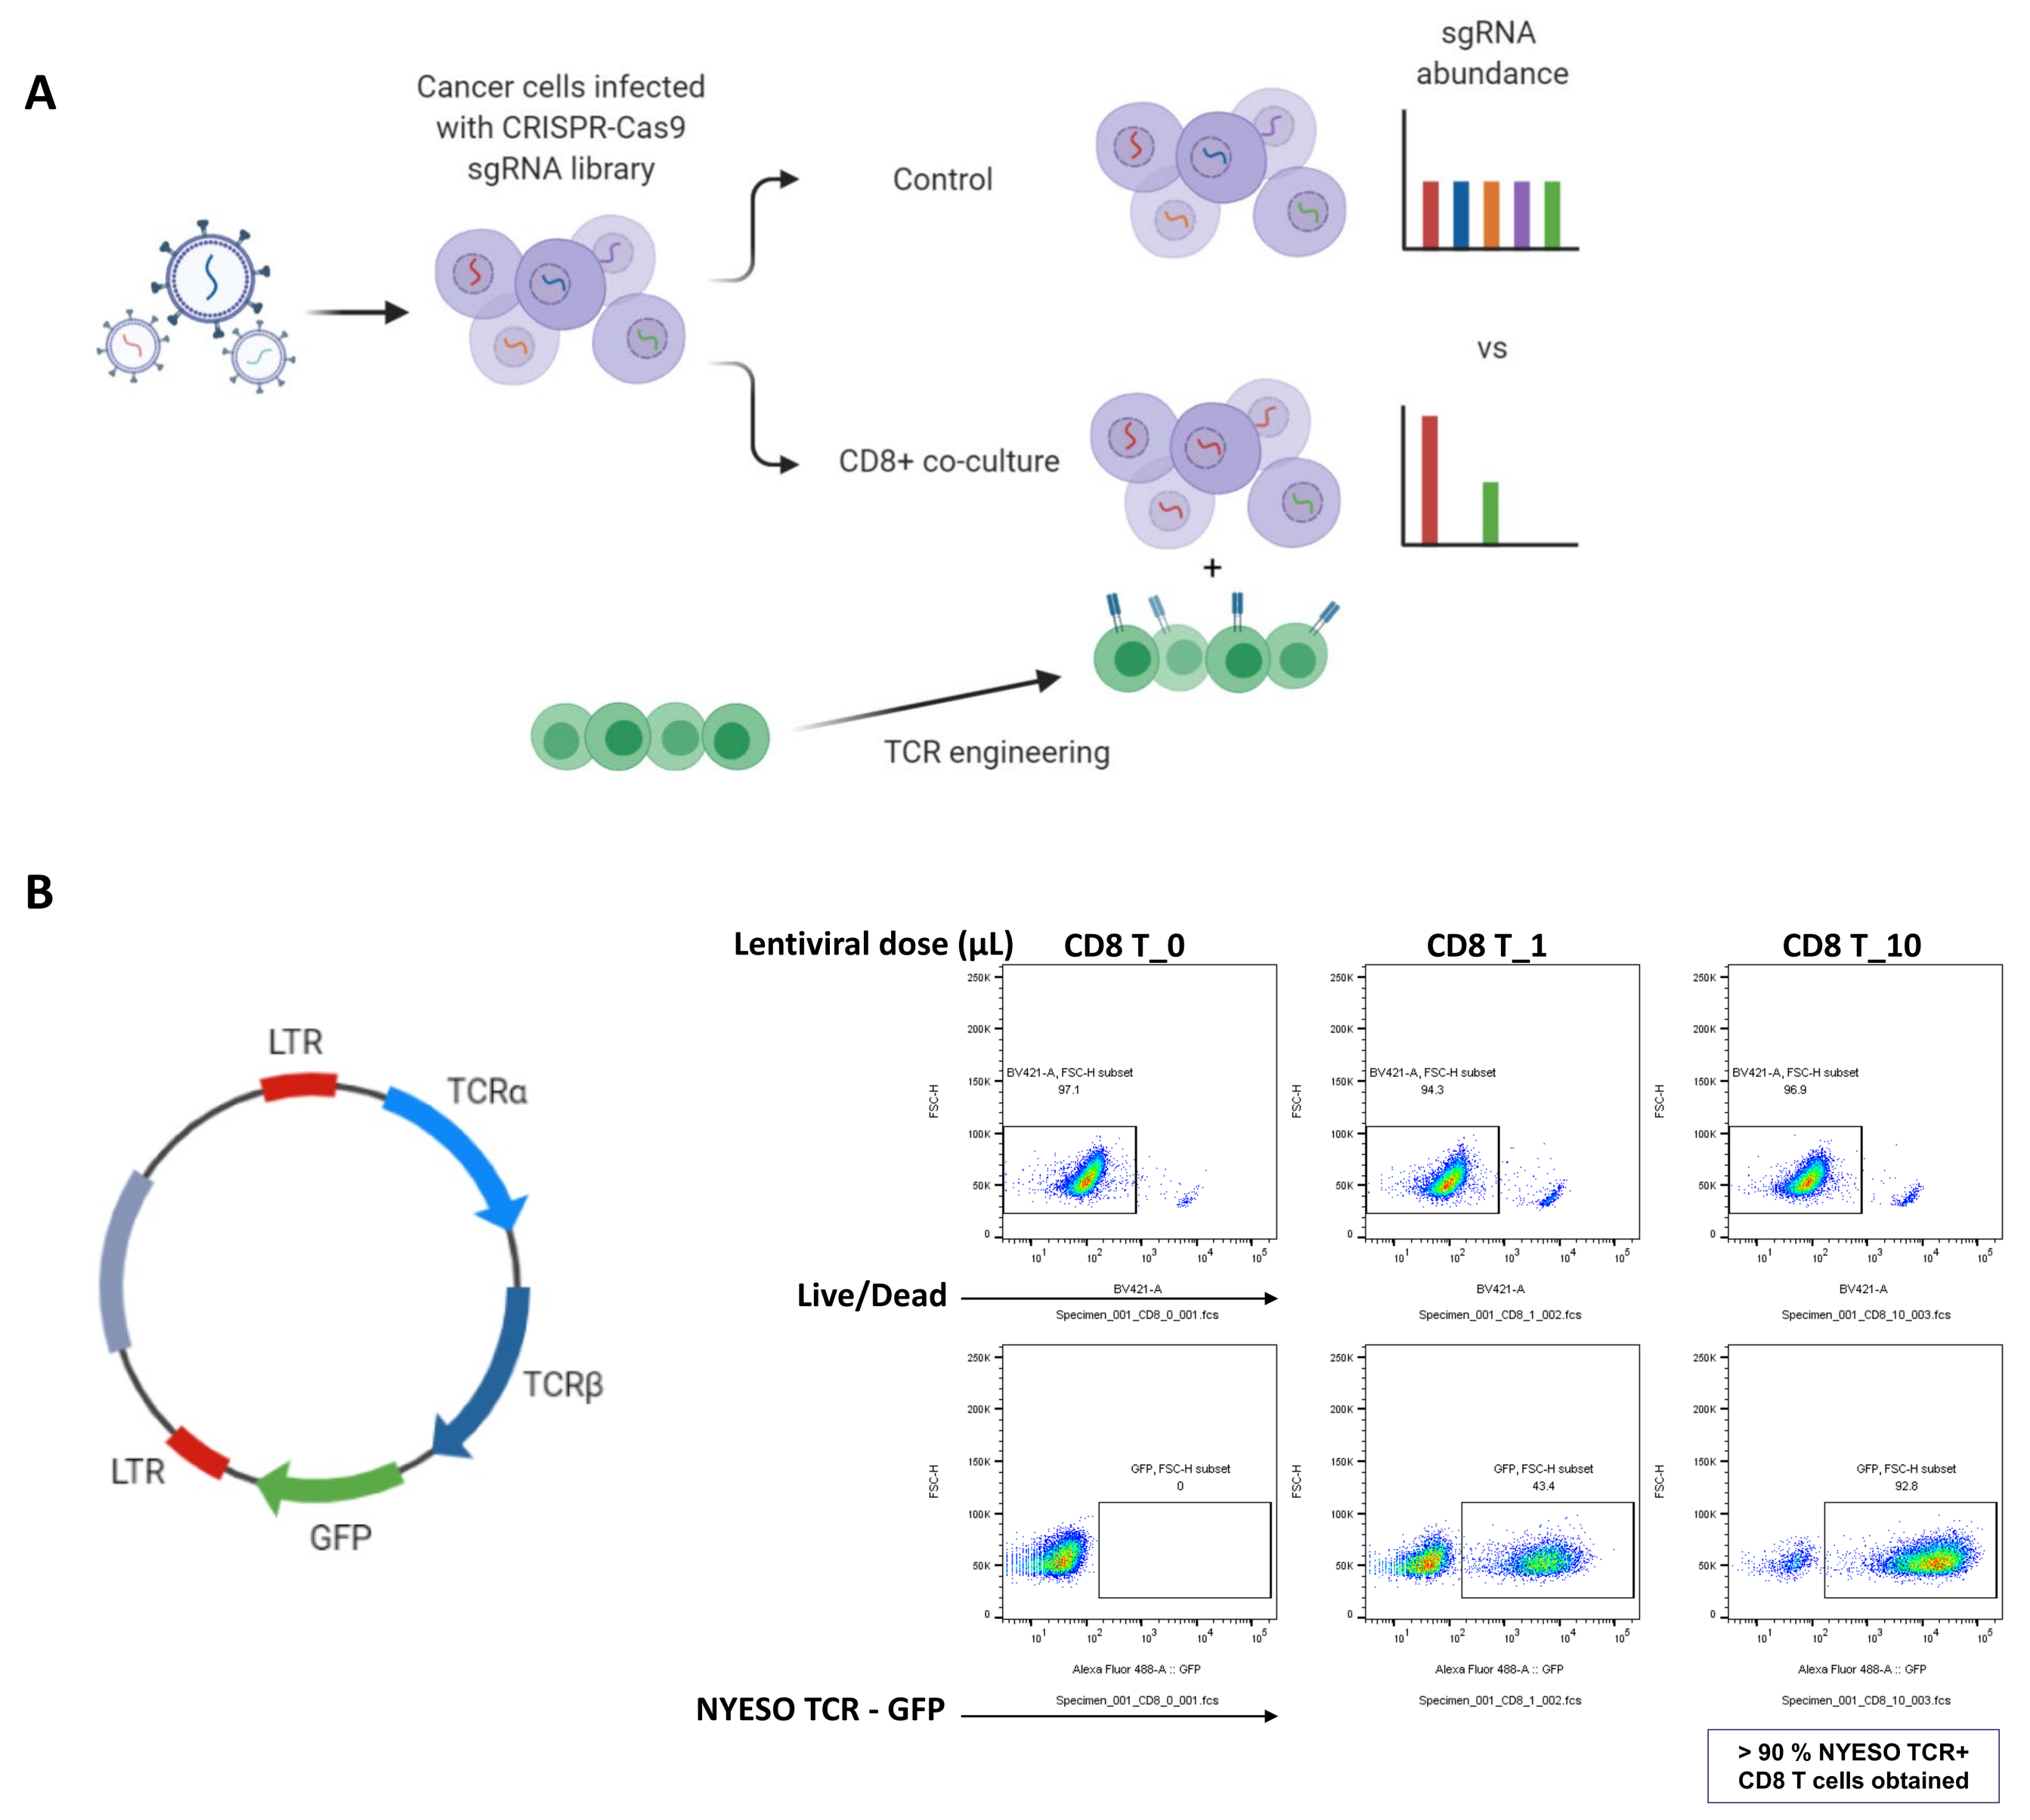
A
Cancer cells infected with CRISPR-Cas9 sgRNA library
Control
CD8+ co-culture
sgRNA abundance
vs
TCR engineering
B
LTR
TCRα
TCRβ
GFP
LTR
Lentiviral dose (µL)
CD8 T_0
CD8 T_1
CD8 T_10
Live/Dead
NYESO TCR - GFP
> 90 % NYESO TCR+ CD8 T cells obtained

Figure 1: T cell co-culture platform. (A) Schematic diagram of screening strategy (B) Vector schematic diagram and confirmation of transgene expression in NYESO TCR+ CD8 T cells isolated from healthy human donors.

CANCER CELL LINE SELECTION

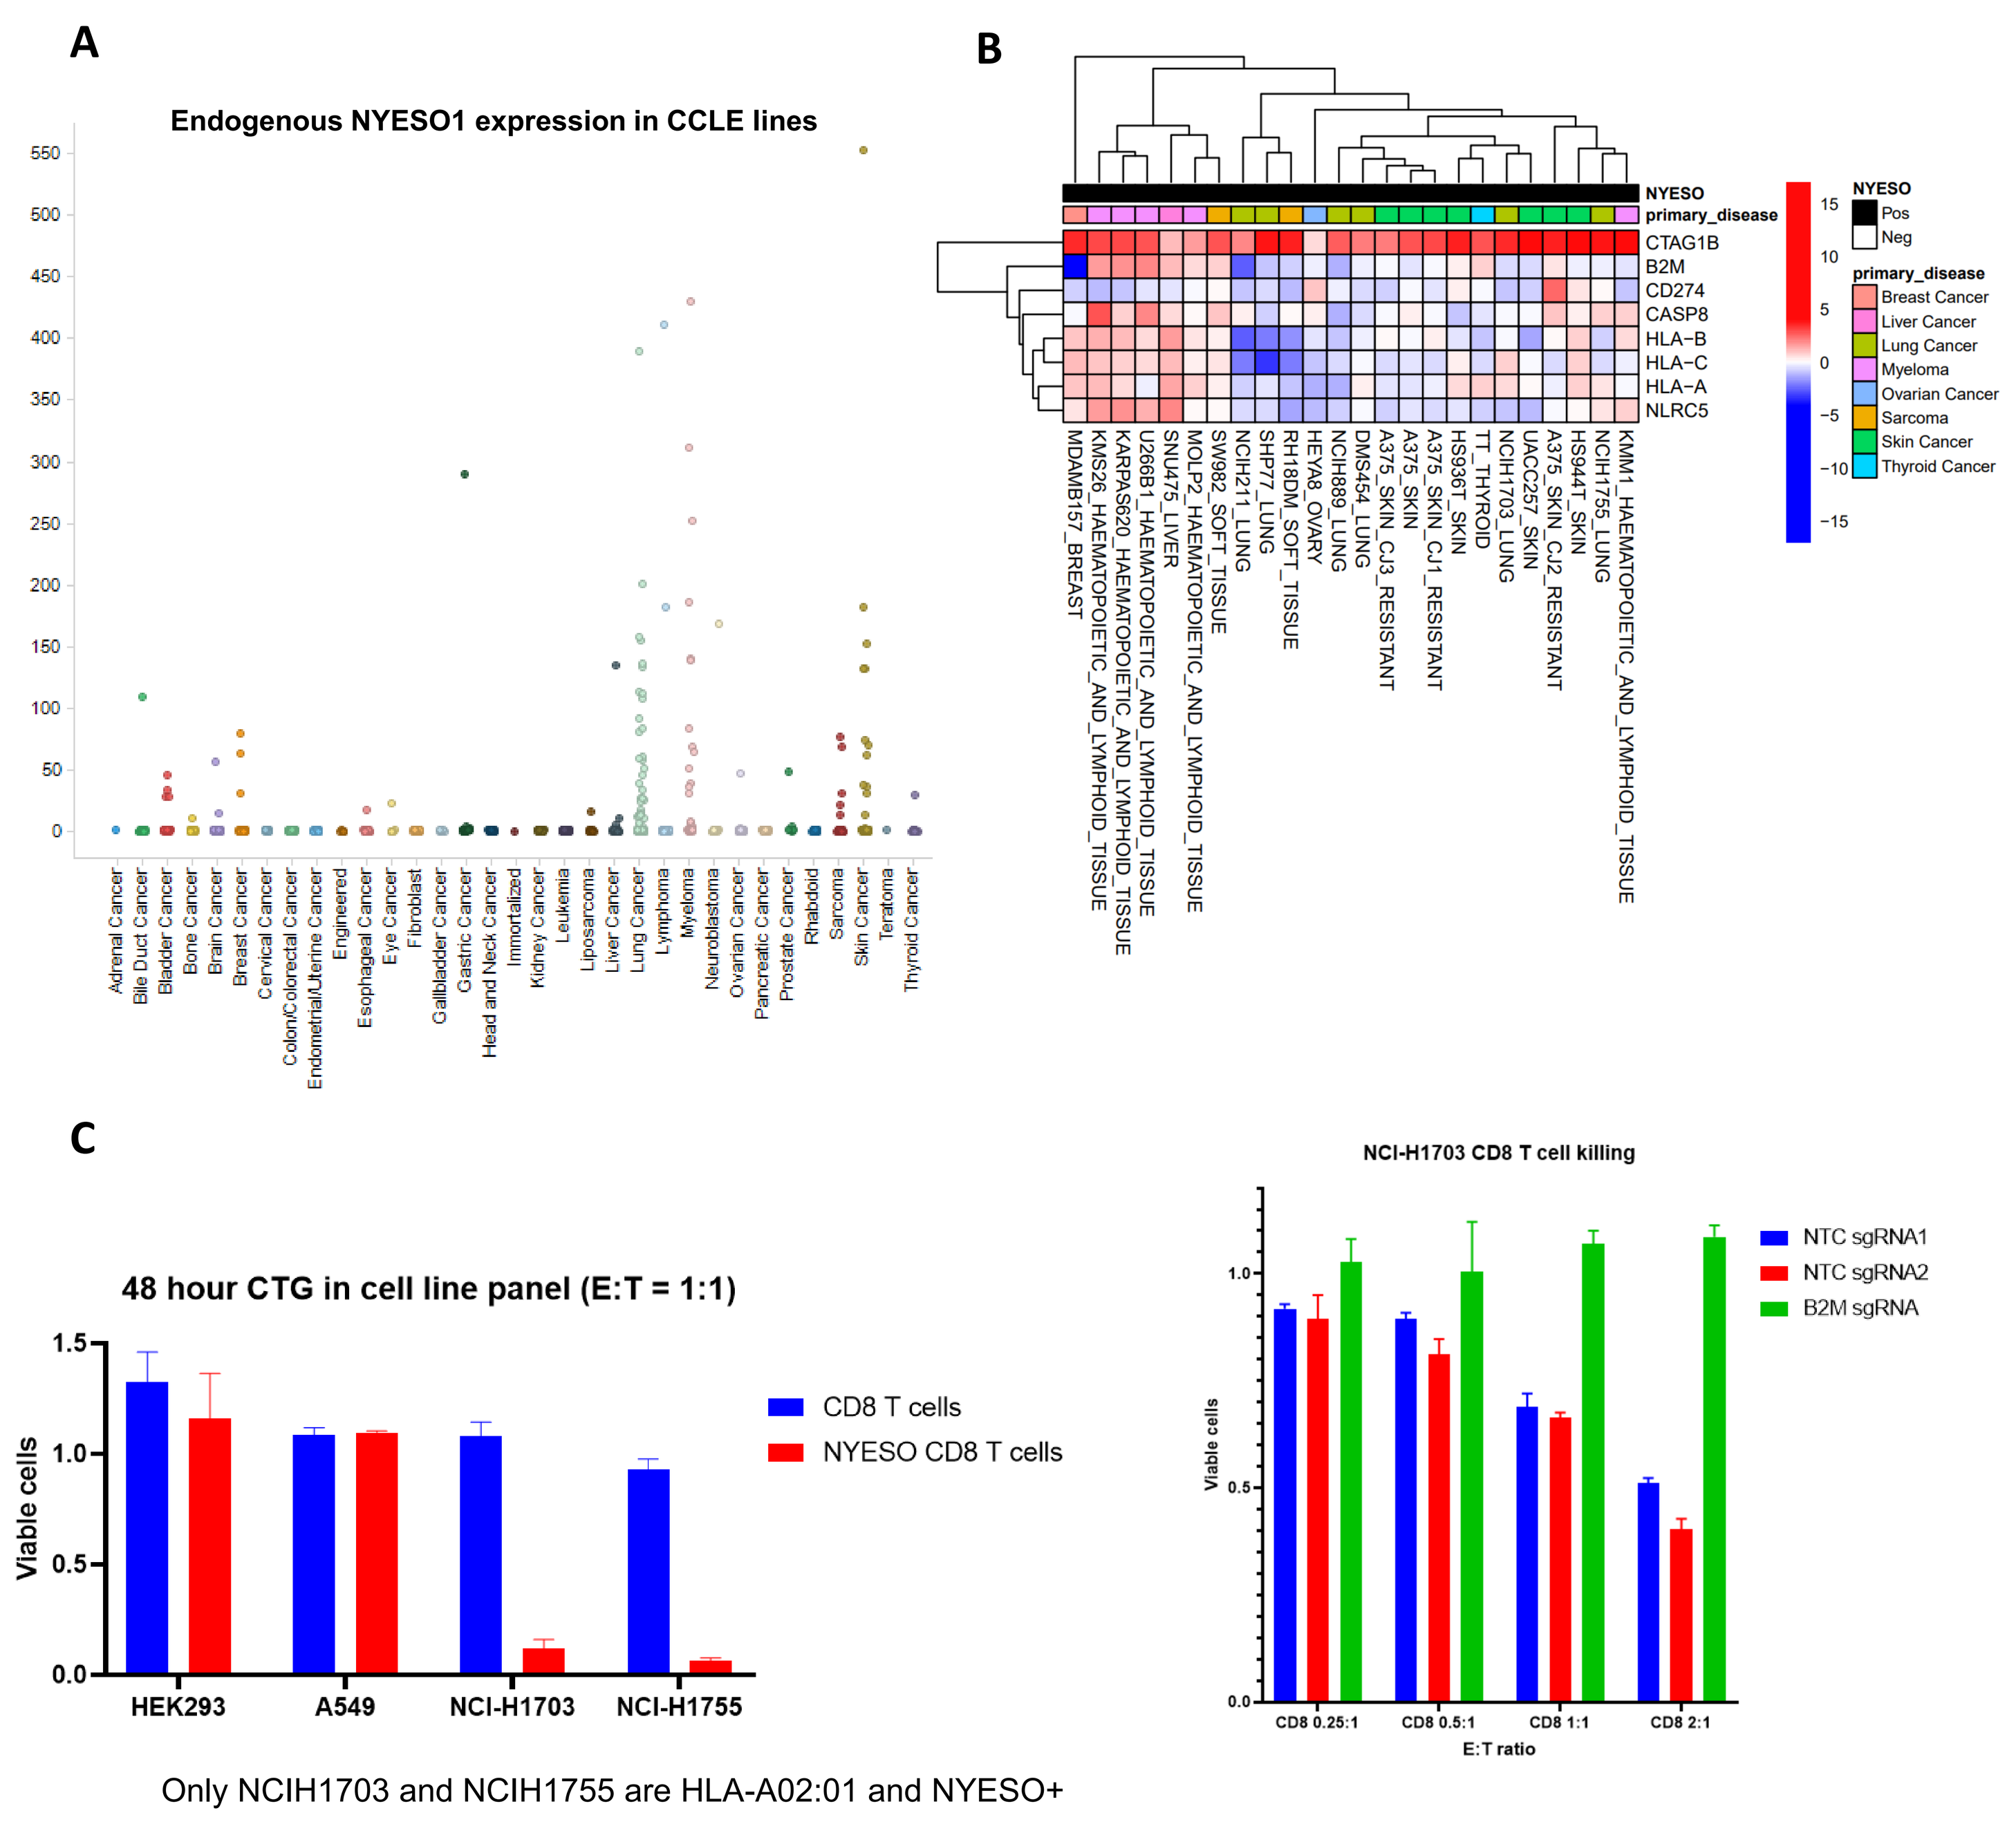
A
Endogenous NYESO1 expression in CCLE lines
TPM
B
NYESO
primary_disease
CTAG1B
B2M
CD274
CASP8
HLA-B
HLA-C
HLA-A
NLRC5
C
48 hour CTG in cell line panel (E:T = 1:1)
Viable cells
CD8 T cells
NYESO CD8 T cells
HEK293
A549
NCI-H1703
NCI-H1755
Only NCIH1703 and NCIH1755 are HLA-A02:01 and NYESO+
NCI-H1703 CD8 T cell killing
NTC sgRNA1
NTC sgRNA2
B2M sgRNA
E:T ratio

Figure 2: Cancer cell line selection for co-culture screens. (A) Lineage restriction of endogenous NYESO1 (CTAG1A/B) expression in CCLE cell lines. (B) MHC-I expression in HLA-A02:01, NYESO positive cell lines. Cell lines with correct haplotype but no endogenous NYESO expression were also identified and engineered with lentiviral-delivered NYESO transgene. (C) In vitro co-culture validation of antigen-specific killing of select cell lines.

RESULTS

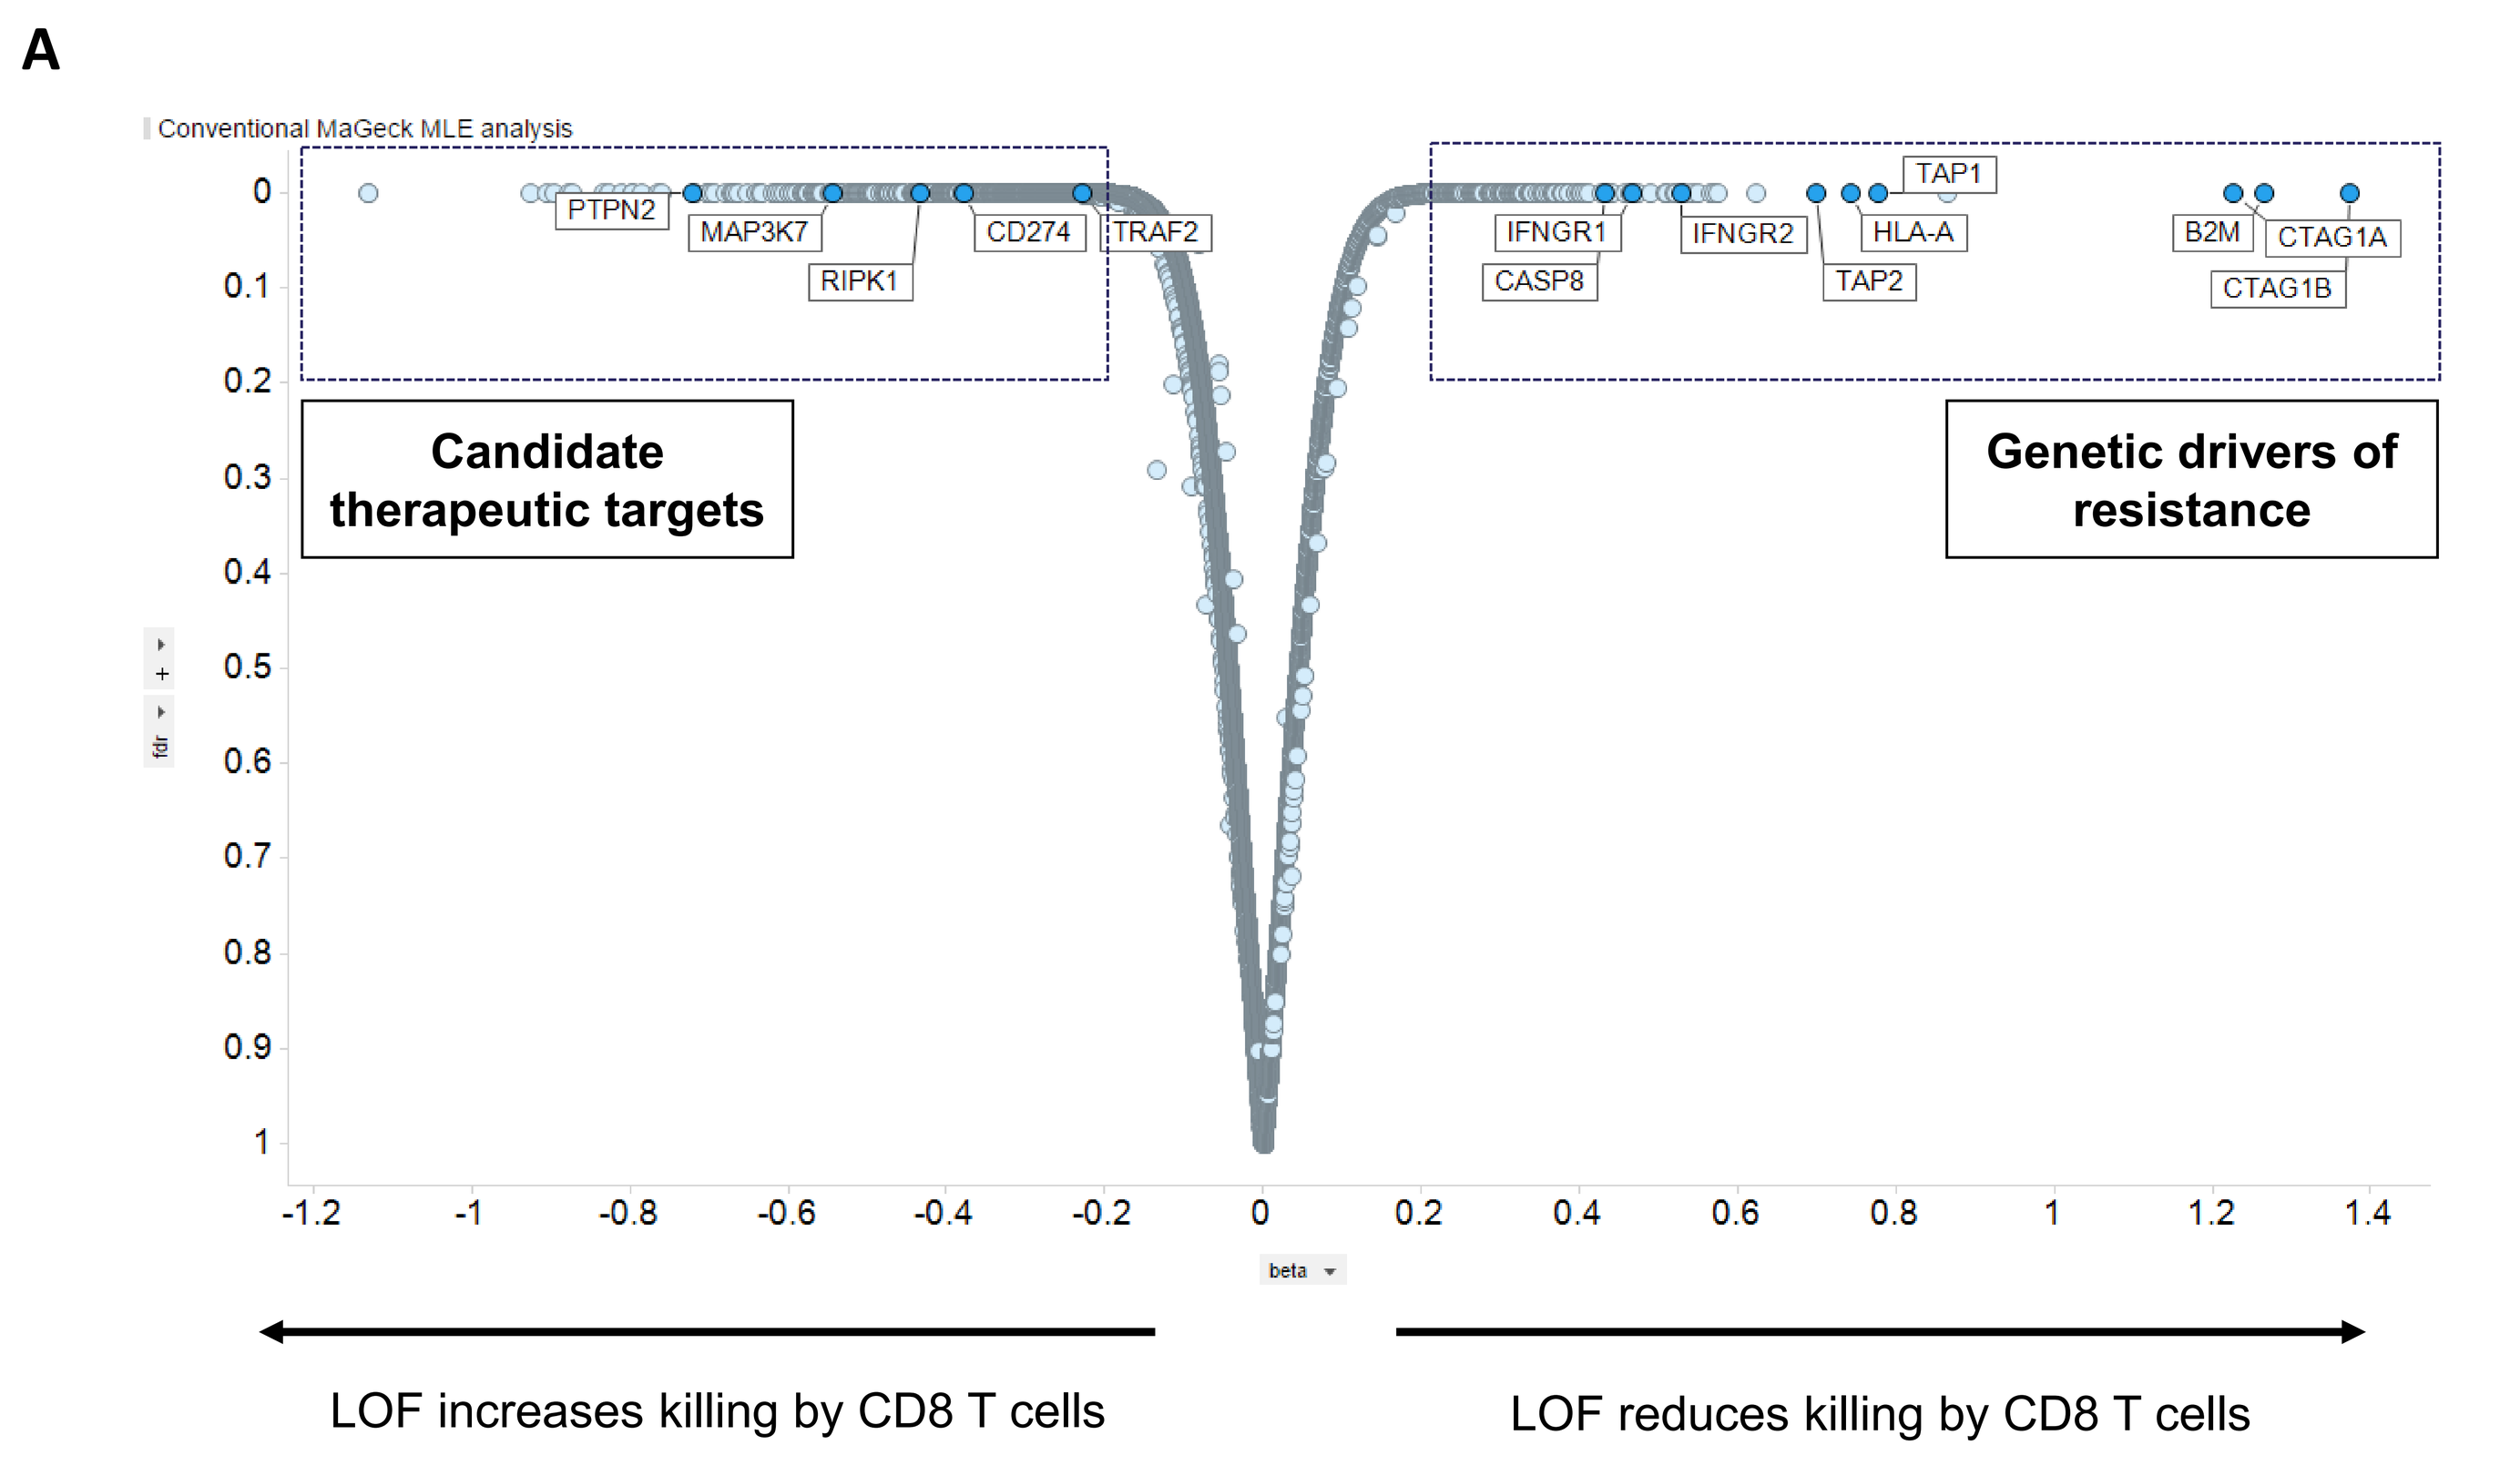
A
Conventional MaGeck MLE analysis
PTPN2
MAP3K7
RIPK1
CD274
TRAF2
IFNGR1
CASP8
IFNGR2
TAP1
HLA-A
TAP2
B2M
CTAG1A
CTAG1B
Candidate therapeutic targets
Genetic drivers of resistance
LOF increases killing by CD8 T cells
LOF reduces killing by CD8 T cells

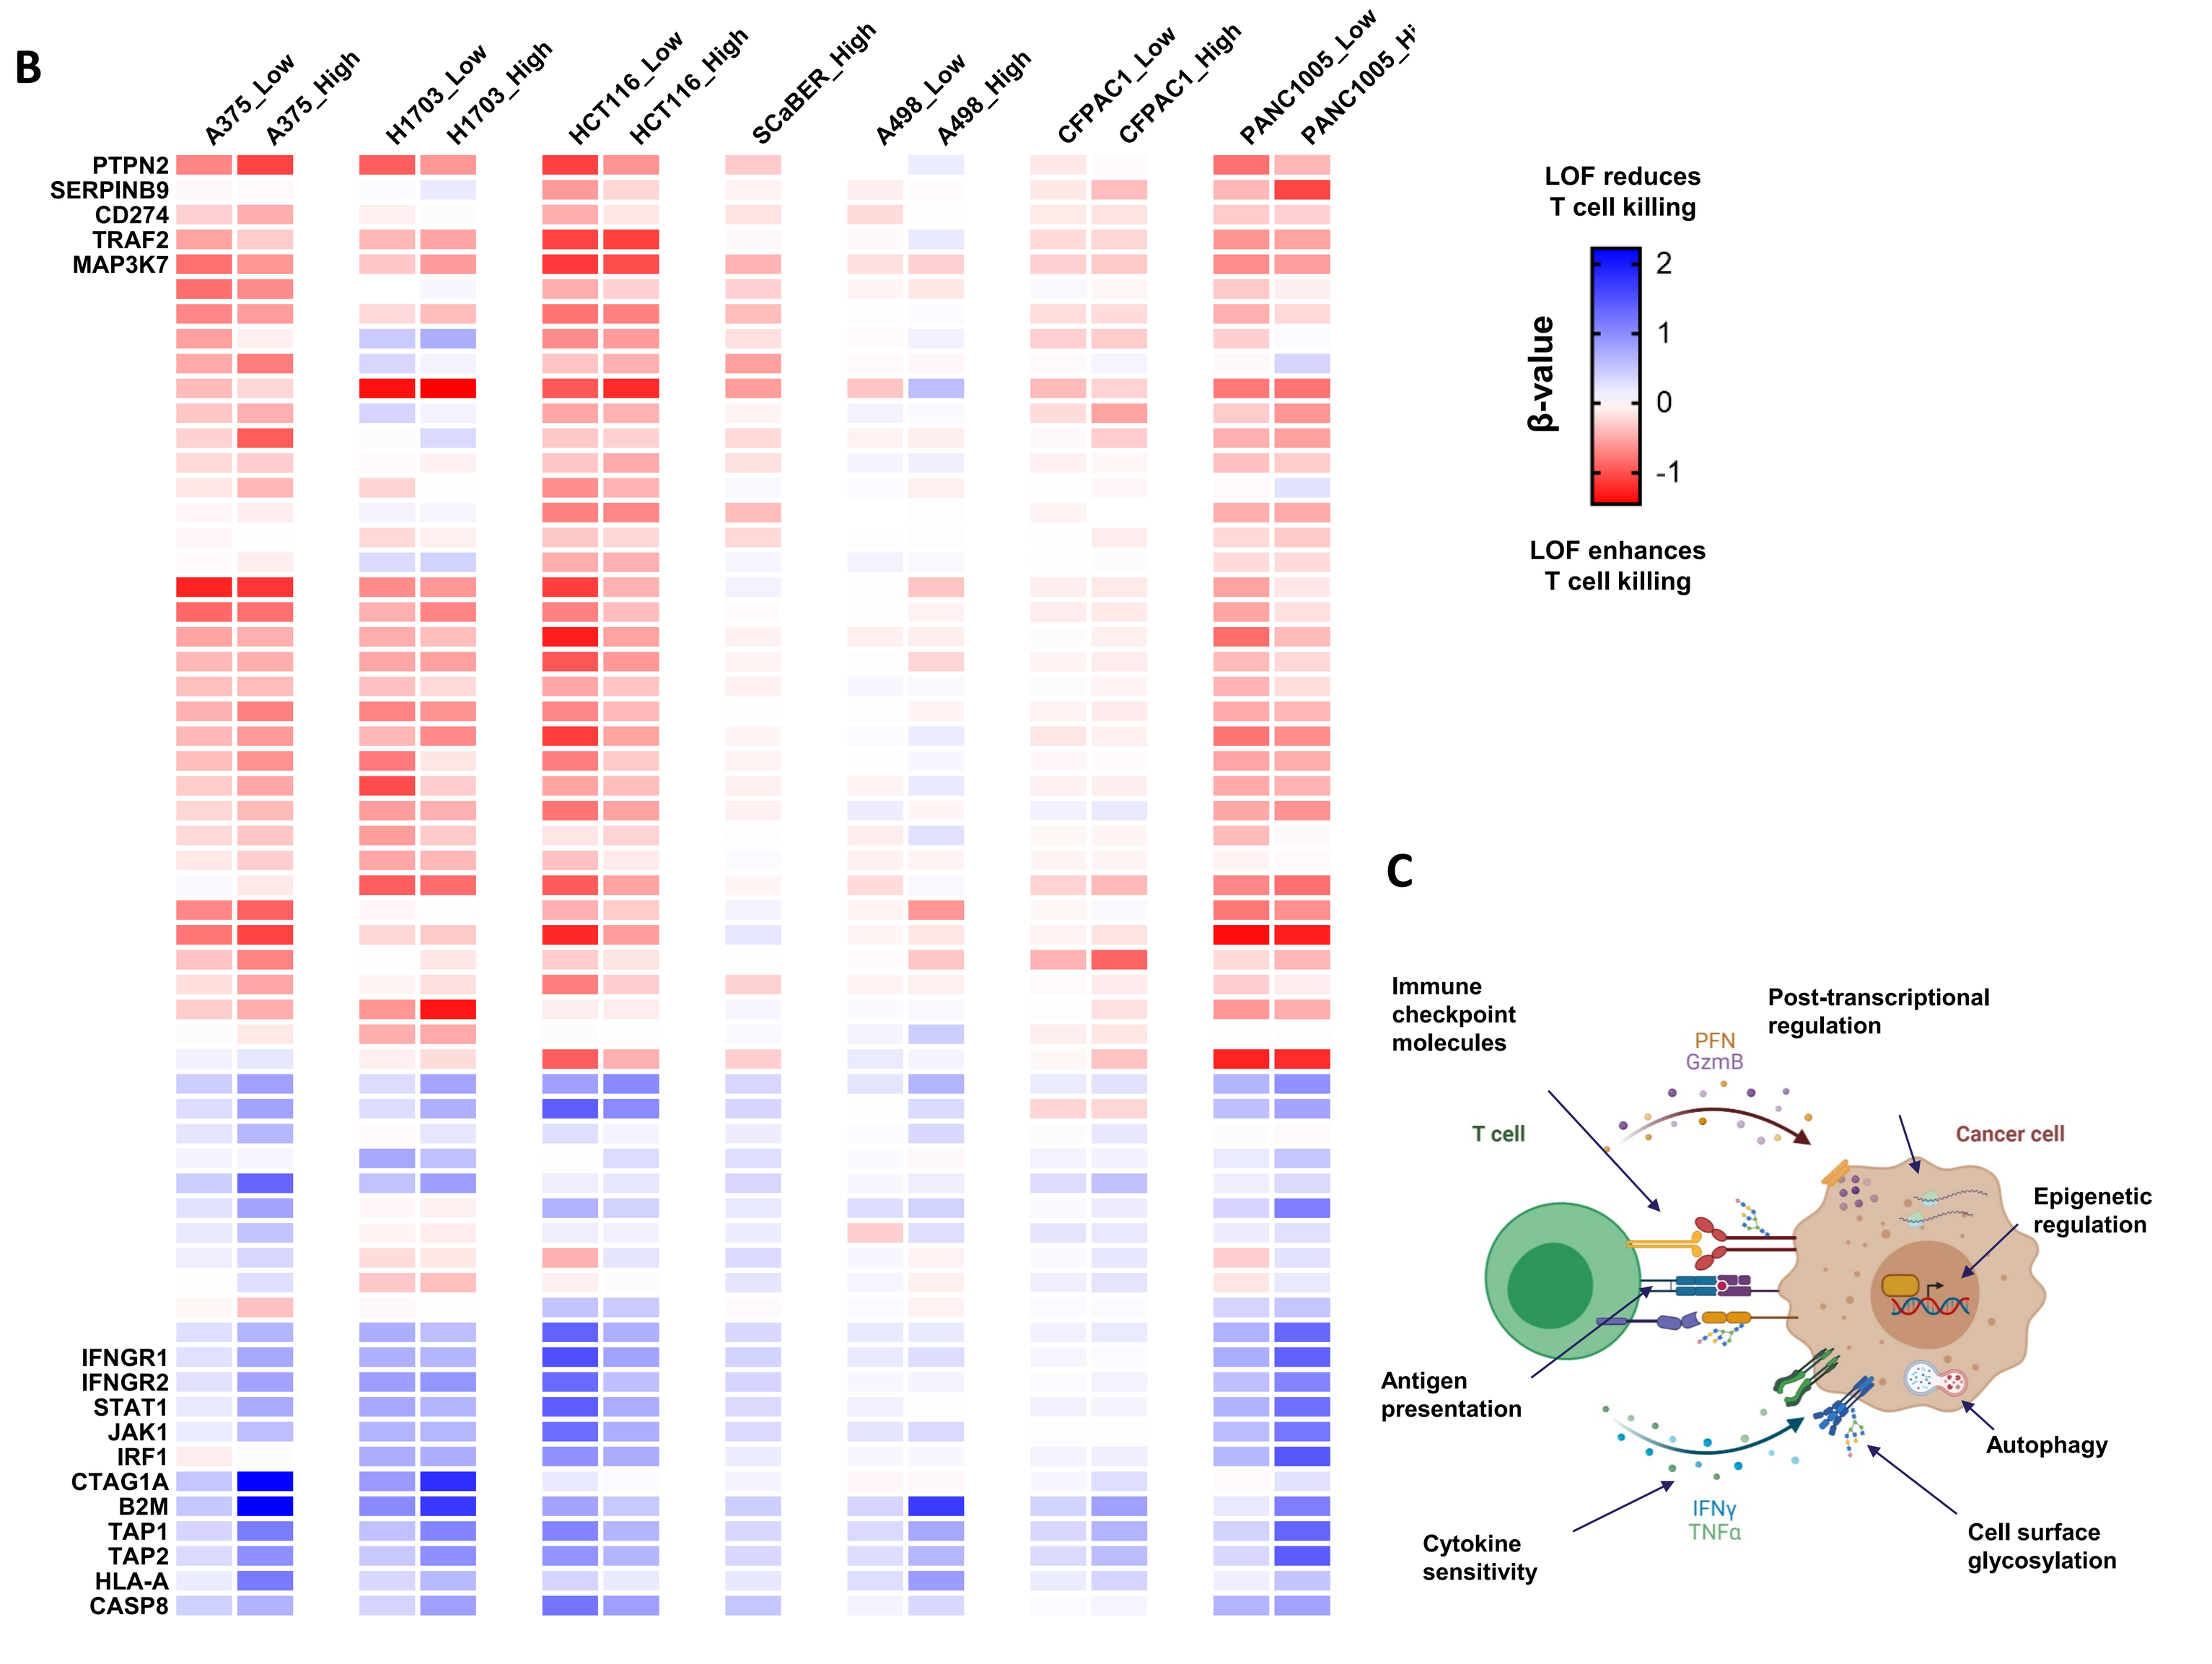
B
A375_Low
A375_High
H1703_Low
H1703_High
HCT116_Low
HCT116_High
SCaBER_High
A498_Low
A498_High
CFPAC1_Low
CFPAC1_High
PANC1005_Low
PTPN2
SERPINB9
CD274
TRAF2
MAP3K7
IFNGR1
IFNGR2
STAT1
JAK1
IRF1
CTAG1A
B2M
TAP1
TAP2
HLA-A
CASP8
LOF reduces T cell killing
β-value
LOF enhances T cell killing
C
Immune checkpoint molecules
Post-transcriptional regulation
PFN
GzmB
T cell
Cancer cell
Epigenetic regulation
Antigen presentation
Autophagy
Cytokine sensitivity
IFNγ
TNFα
Cell surface glycosylation

Figure 3: Summary of screen results from human cell line panel. (A) Example of a genome-wide screen in A375 cell line highlighting select genes that upon knockout, lead to sensitization or resistance to T cell killing. (B) Heatmap summarizing the top hits from profiling multiple human cancer cell lines. (C) Summary of key pathways controlling tumor killing by CD8 T cells.

IN VITRO TARGET VALIDATION

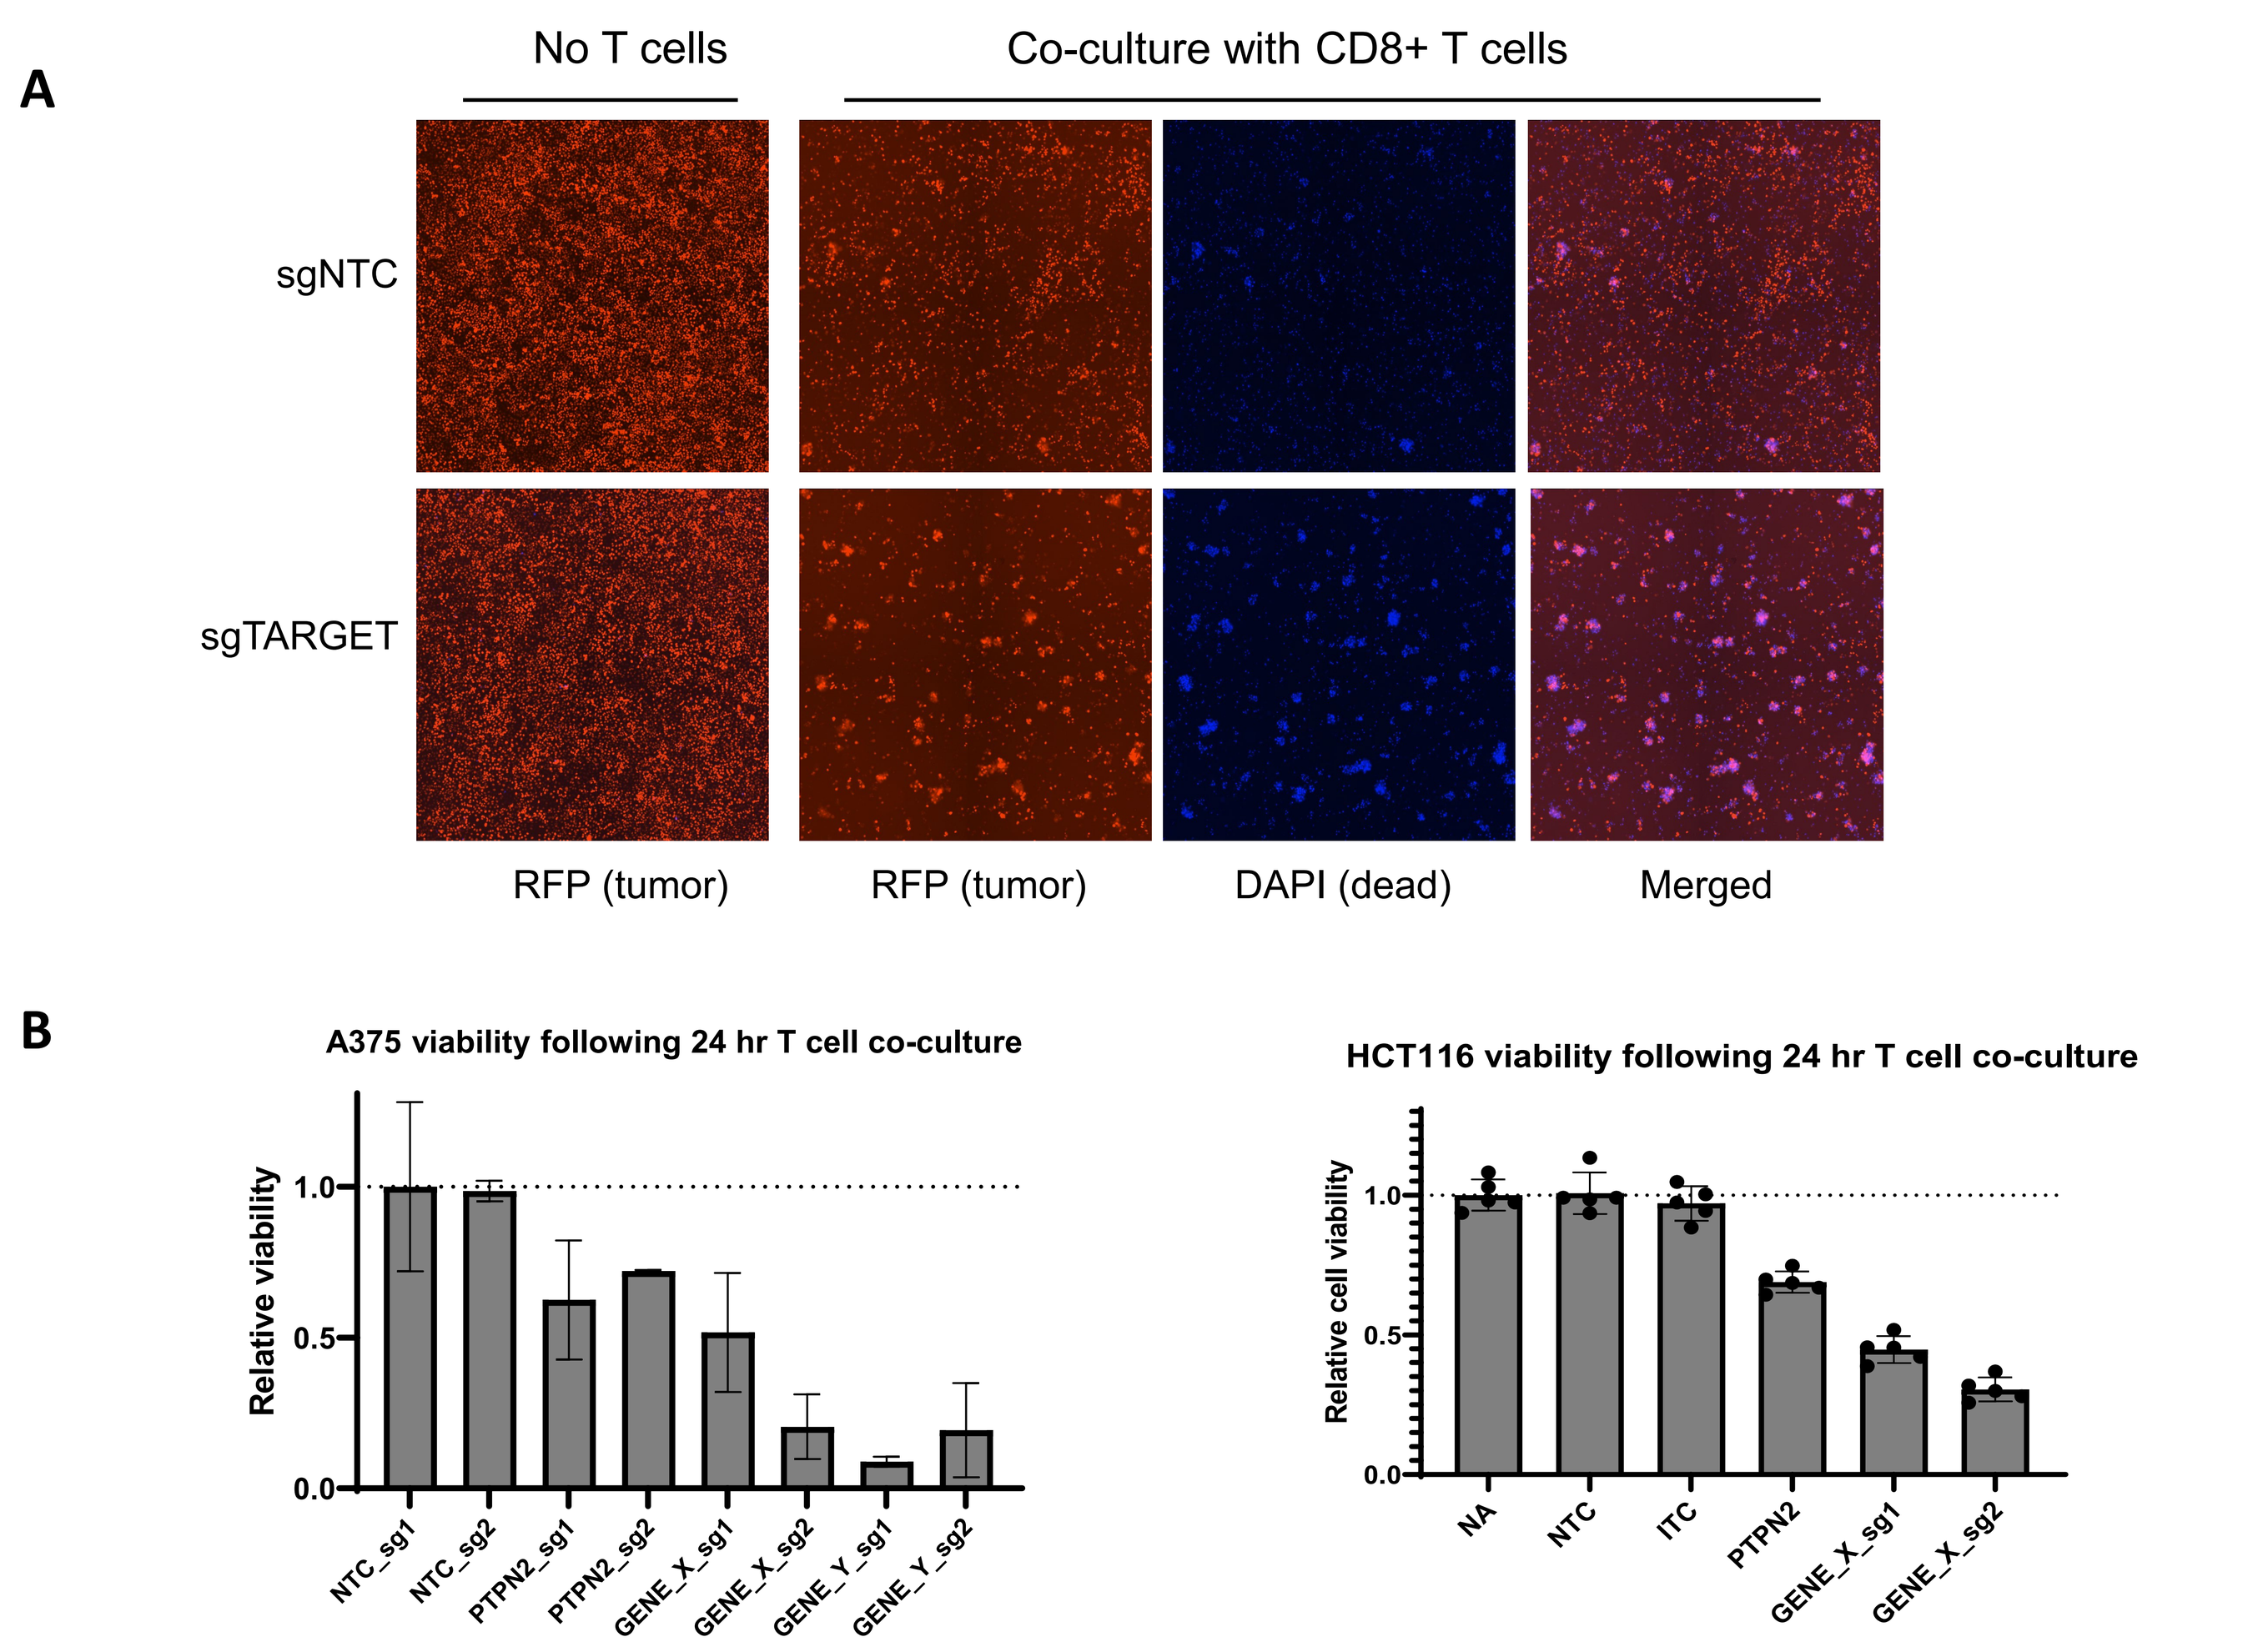
A
No T cells
Co-culture with CD8+ T cells
sgNTC
sgTARGET
RFP (tumor)
RFP (tumor)
DAPI (dead)
Merged
B
A375 viability following 24 hr T cell co-culture
Relative viability
HCT116 viability following 24 hr T cell co-culture
Relative cell viability

Figure 4: Microscopy assay for in vitro validation of co-culture hits. (A) Celigo based assay showing typical example of the observed phenotype for knockout of a sensitizing gene. (B) Genetic validation of PTPN2 and additional sensitizers using Celigo-based assay.

IN VIVO TARGET VALIDATION USING ACT MODEL

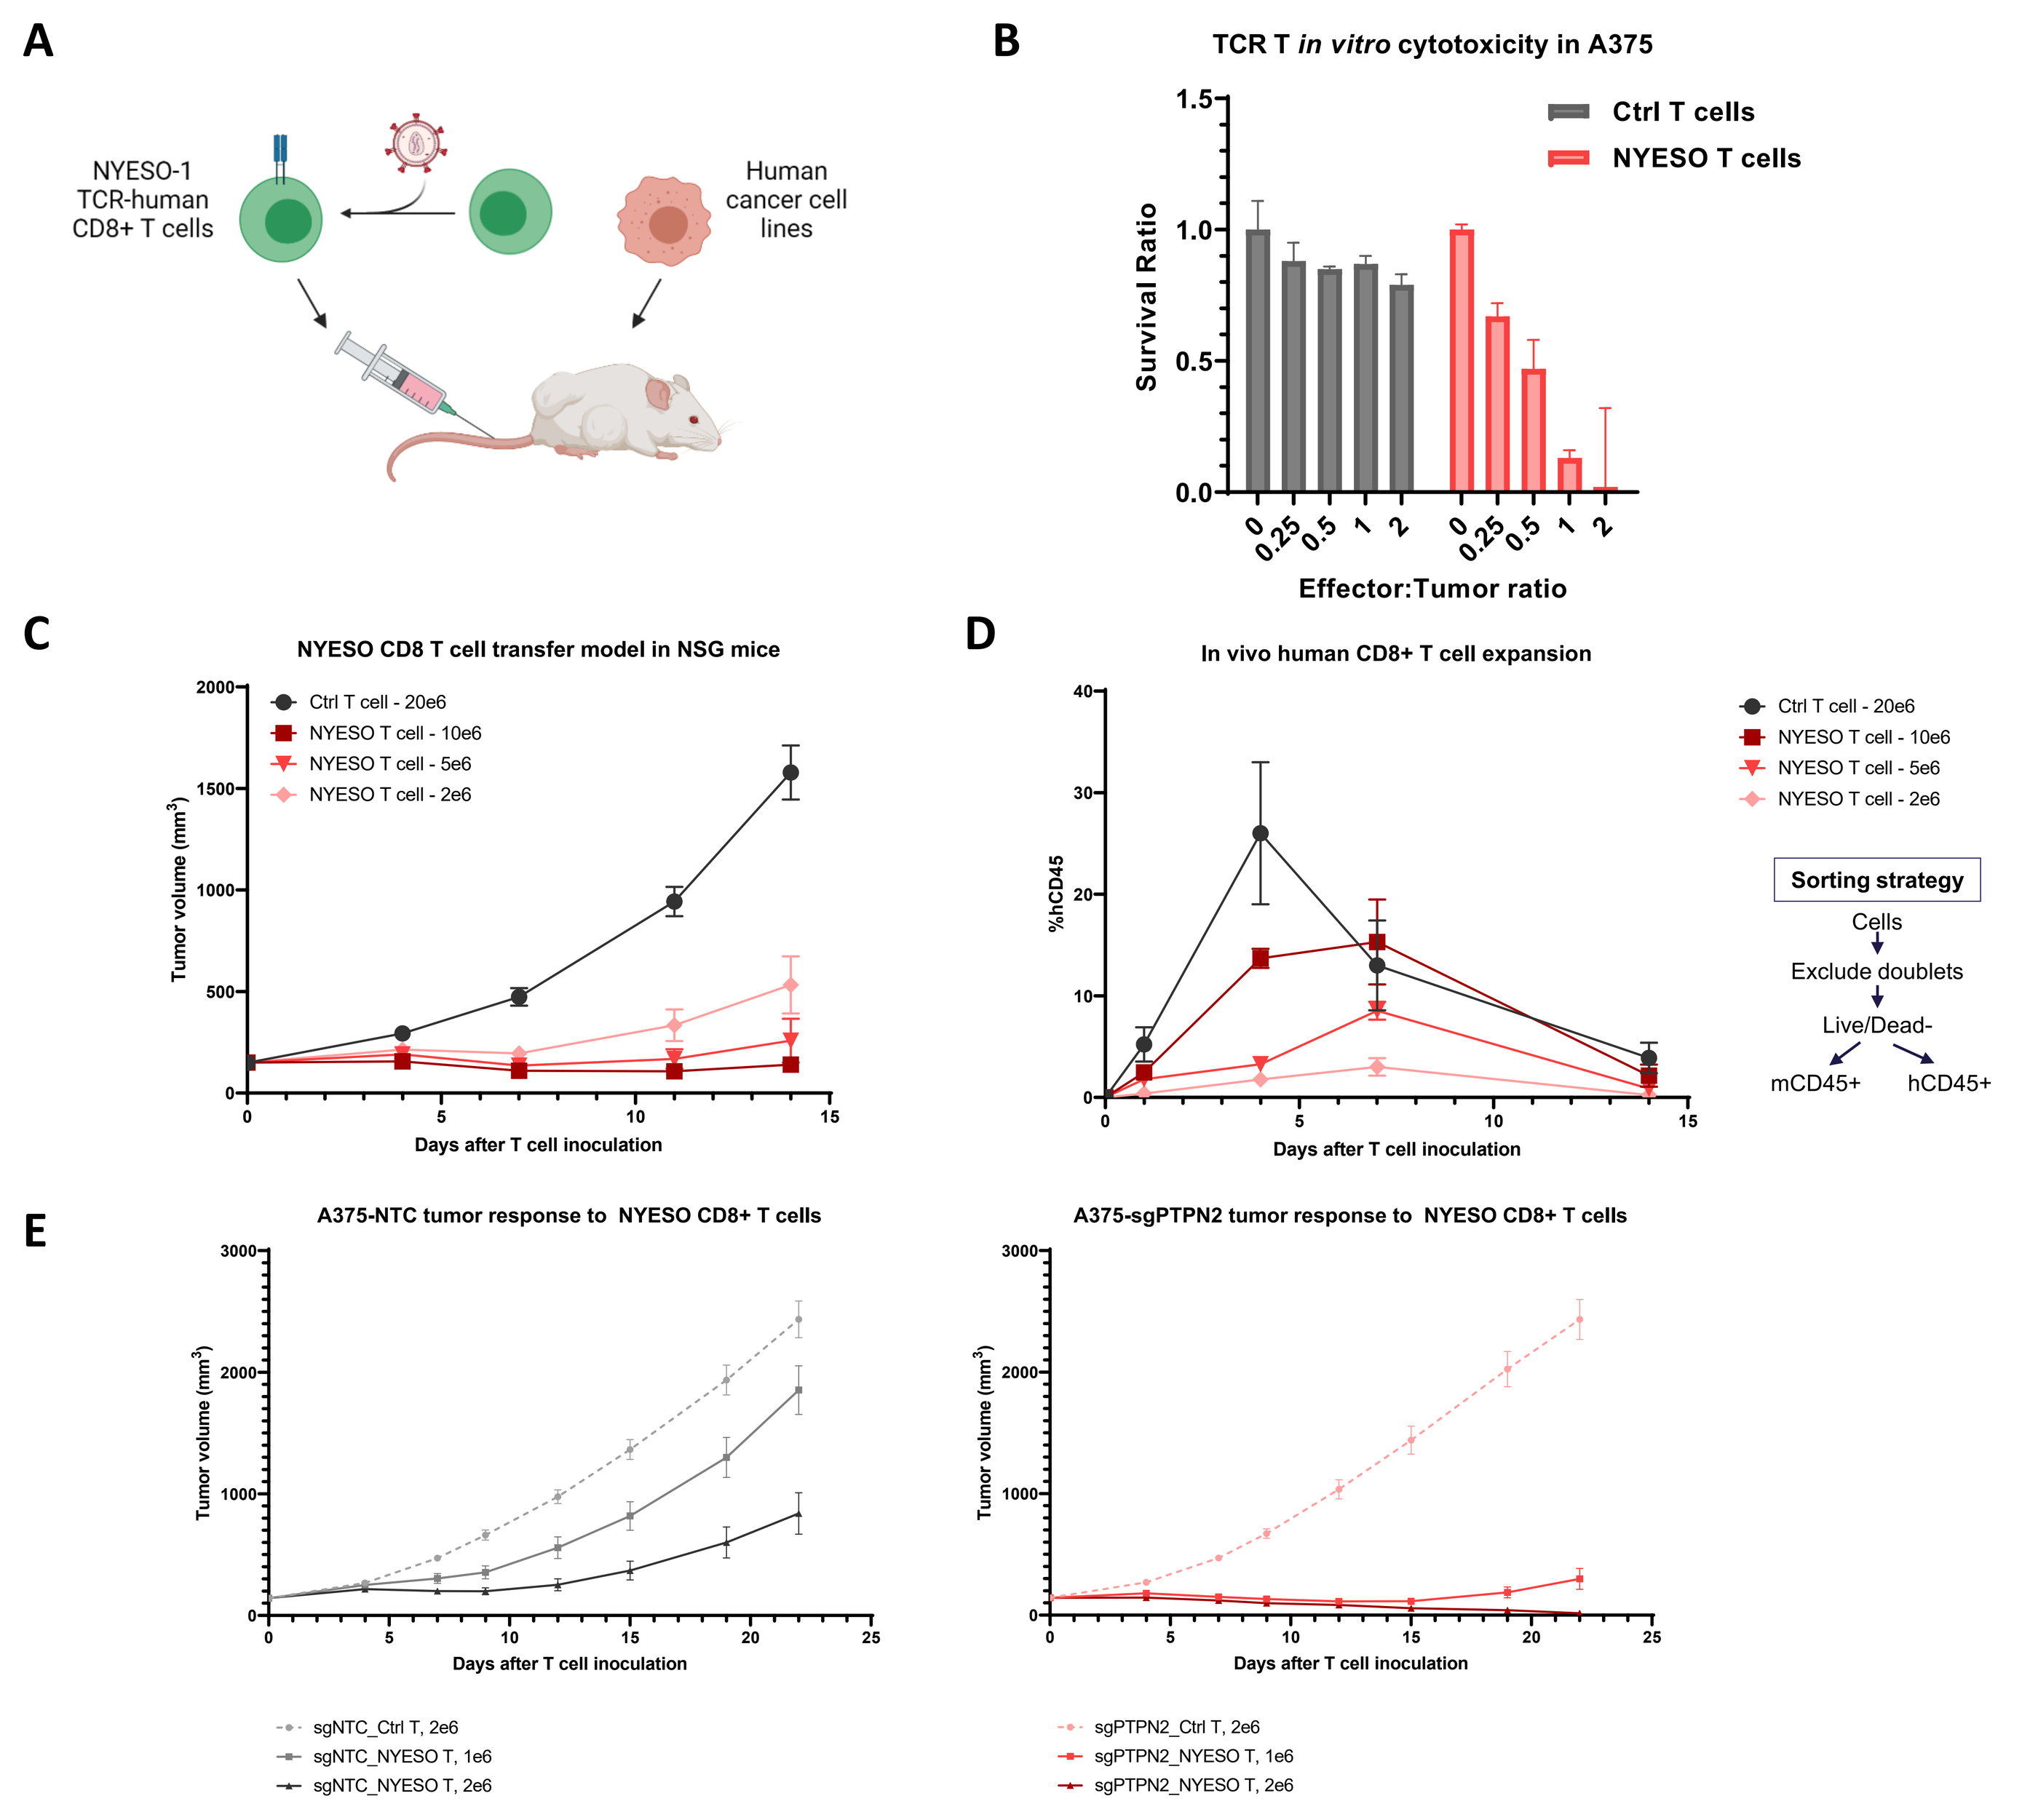
A
NYESO-1 TCR-human CD8+ T cells
Human cancer cell lines
B
TCR T in vitro cytotoxicity in A375
Survival Ratio
Ctrl T cells
NYESO T cells
Effector:Tumor ratio
C
NYESO CD8 T cell transfer model in NSG mice
Days after T cell inoculation
D
In vivo human CD8+ T cell expansion
%hCD45
Sorting strategy
Cells
Exclude doublets
Live/Dead-
mCD45+
hCD45+
E
A375-NTC tumor response to NYESO CD8+ T cells
A375-sgPTPN2 tumor response to NYESO CD8+ T cells

Figure 5: Human A375 cell line xenograft model with adoptive transfer of NYESO TCR+ CD8 T cells for target validation. (A) Schematic diagram of adoptive cell transfer model. (B) In vitro confirmation of antigen-specific killing of A375 melanoma cells by NYESO TCR+ CD8 T cells. (C) In vivo dose finding experiment showing dose-dependent tumor growth inhibition following infusion of NYESO TCR+ CD8 T cells. (D) Quantification of CD8+ T cell expansion from panel (C). (E) Measuring the effect of PTPN2 knockout by CRISPR/Cas9 RNP electroporation prior to cell inoculation compared to a non-target control sgRNA.

CONCLUSION/SUMMARY

- An in-vitro screening platform was established for screening human cancer genes that mediate sensitization and resistance to antigen-specific CD8 T cell killing
- Cell lines were selected based on intact MHC-I expression and HLA-A02:01 haplotype.
- Screen was performed in 7 cancer cell lines, highlighting key pathways involved in regulating killing of human cancer lines by antigen-specific CD8 T cells.
- Candidate genes were validated using in vitro and in vivo systems, showcased using the known sensitizer PTPN2

REFERENCES

1. Patel, S. J. *et al.* Identification of essential genes for cancer immunotherapy. *Nature* **548**, 537–542 (2017).
2. Manguso, R. T. *et al.* In vivo CRISPR screening identifies Ptpn2 as a cancer immunotherapy target. *Nature* **547**, 413–418 (2017).

ACKNOWLEDGEMENTS

The authors gratefully acknowledge the generous contributions from the scientific teams at ChemPartner and WuXi AppTec.