POLB knockout is synthetic lethal with PARP inhibition leading to complete and durable responses in BRCA-mutant tumor xenografts

Madhavi Bandi, Katherine Lazarides, Justin Engel, Tianshu Feng, Michele Meseonznik, Ashley Choi, Yi Yu, Shangtao Liu, Samuel Meier, Hongxiang Zhang, Binzhang Shen, Robert Tjin, Doug Whittington, Brian McMillan, Brian Doyon, Mu-Sen Liu, Xuewen Pan, Erik Wilker, Alan Huang, Jannik N. Andersen, William Mallender

Abstract #10

ABSTRACT

Despite the clinical benefit of PARP1/2 inhibitors (PARPi), which are FDA-approved for the treatment of certain BRCA-mutant cancers, many patients achieve incomplete disease control and develop progressive disease. Motivated by this clinical need, we utilized our CRISPR target discovery screening platform to identify novel targets that synergize with PARP inhibitor treatment. By conducting parallel screens in both BRCA-mutant and wildtype cells, we identified DNA polymerase beta (POLB) as a novel target that - when combined with PARPi - selectively kills BRCA-mutant lines while sparing normal cells. POLB knockout and cDNA rescue experiments using both BRCA1 and BRCA2-mutant isogenic cell lines further demonstrated that the catalytic activity of POLB is required for synthetic lethality with PARPi. Most strikingly, POLB knockout combined with sub-therapeutic doses of PARPi, led to profound tumor regression and prevented in vivo tumor regrowth, even after cessation of drug treatment. Mechanistically, POLB knockout is associated with increased single and double strand DNA breaks, accumulation of poly-ADP-ribose polymers, cell cycle arrest, and apoptosis. Together, these results suggest that POLB inhibitors in combination with PARPi have the potential to drive deep and durable responses providing a novel therapeutic option for cancer patients with BRCA1/2-mutations.

POLB was identified as a synthetic lethal target in anchor screens with Olaparib

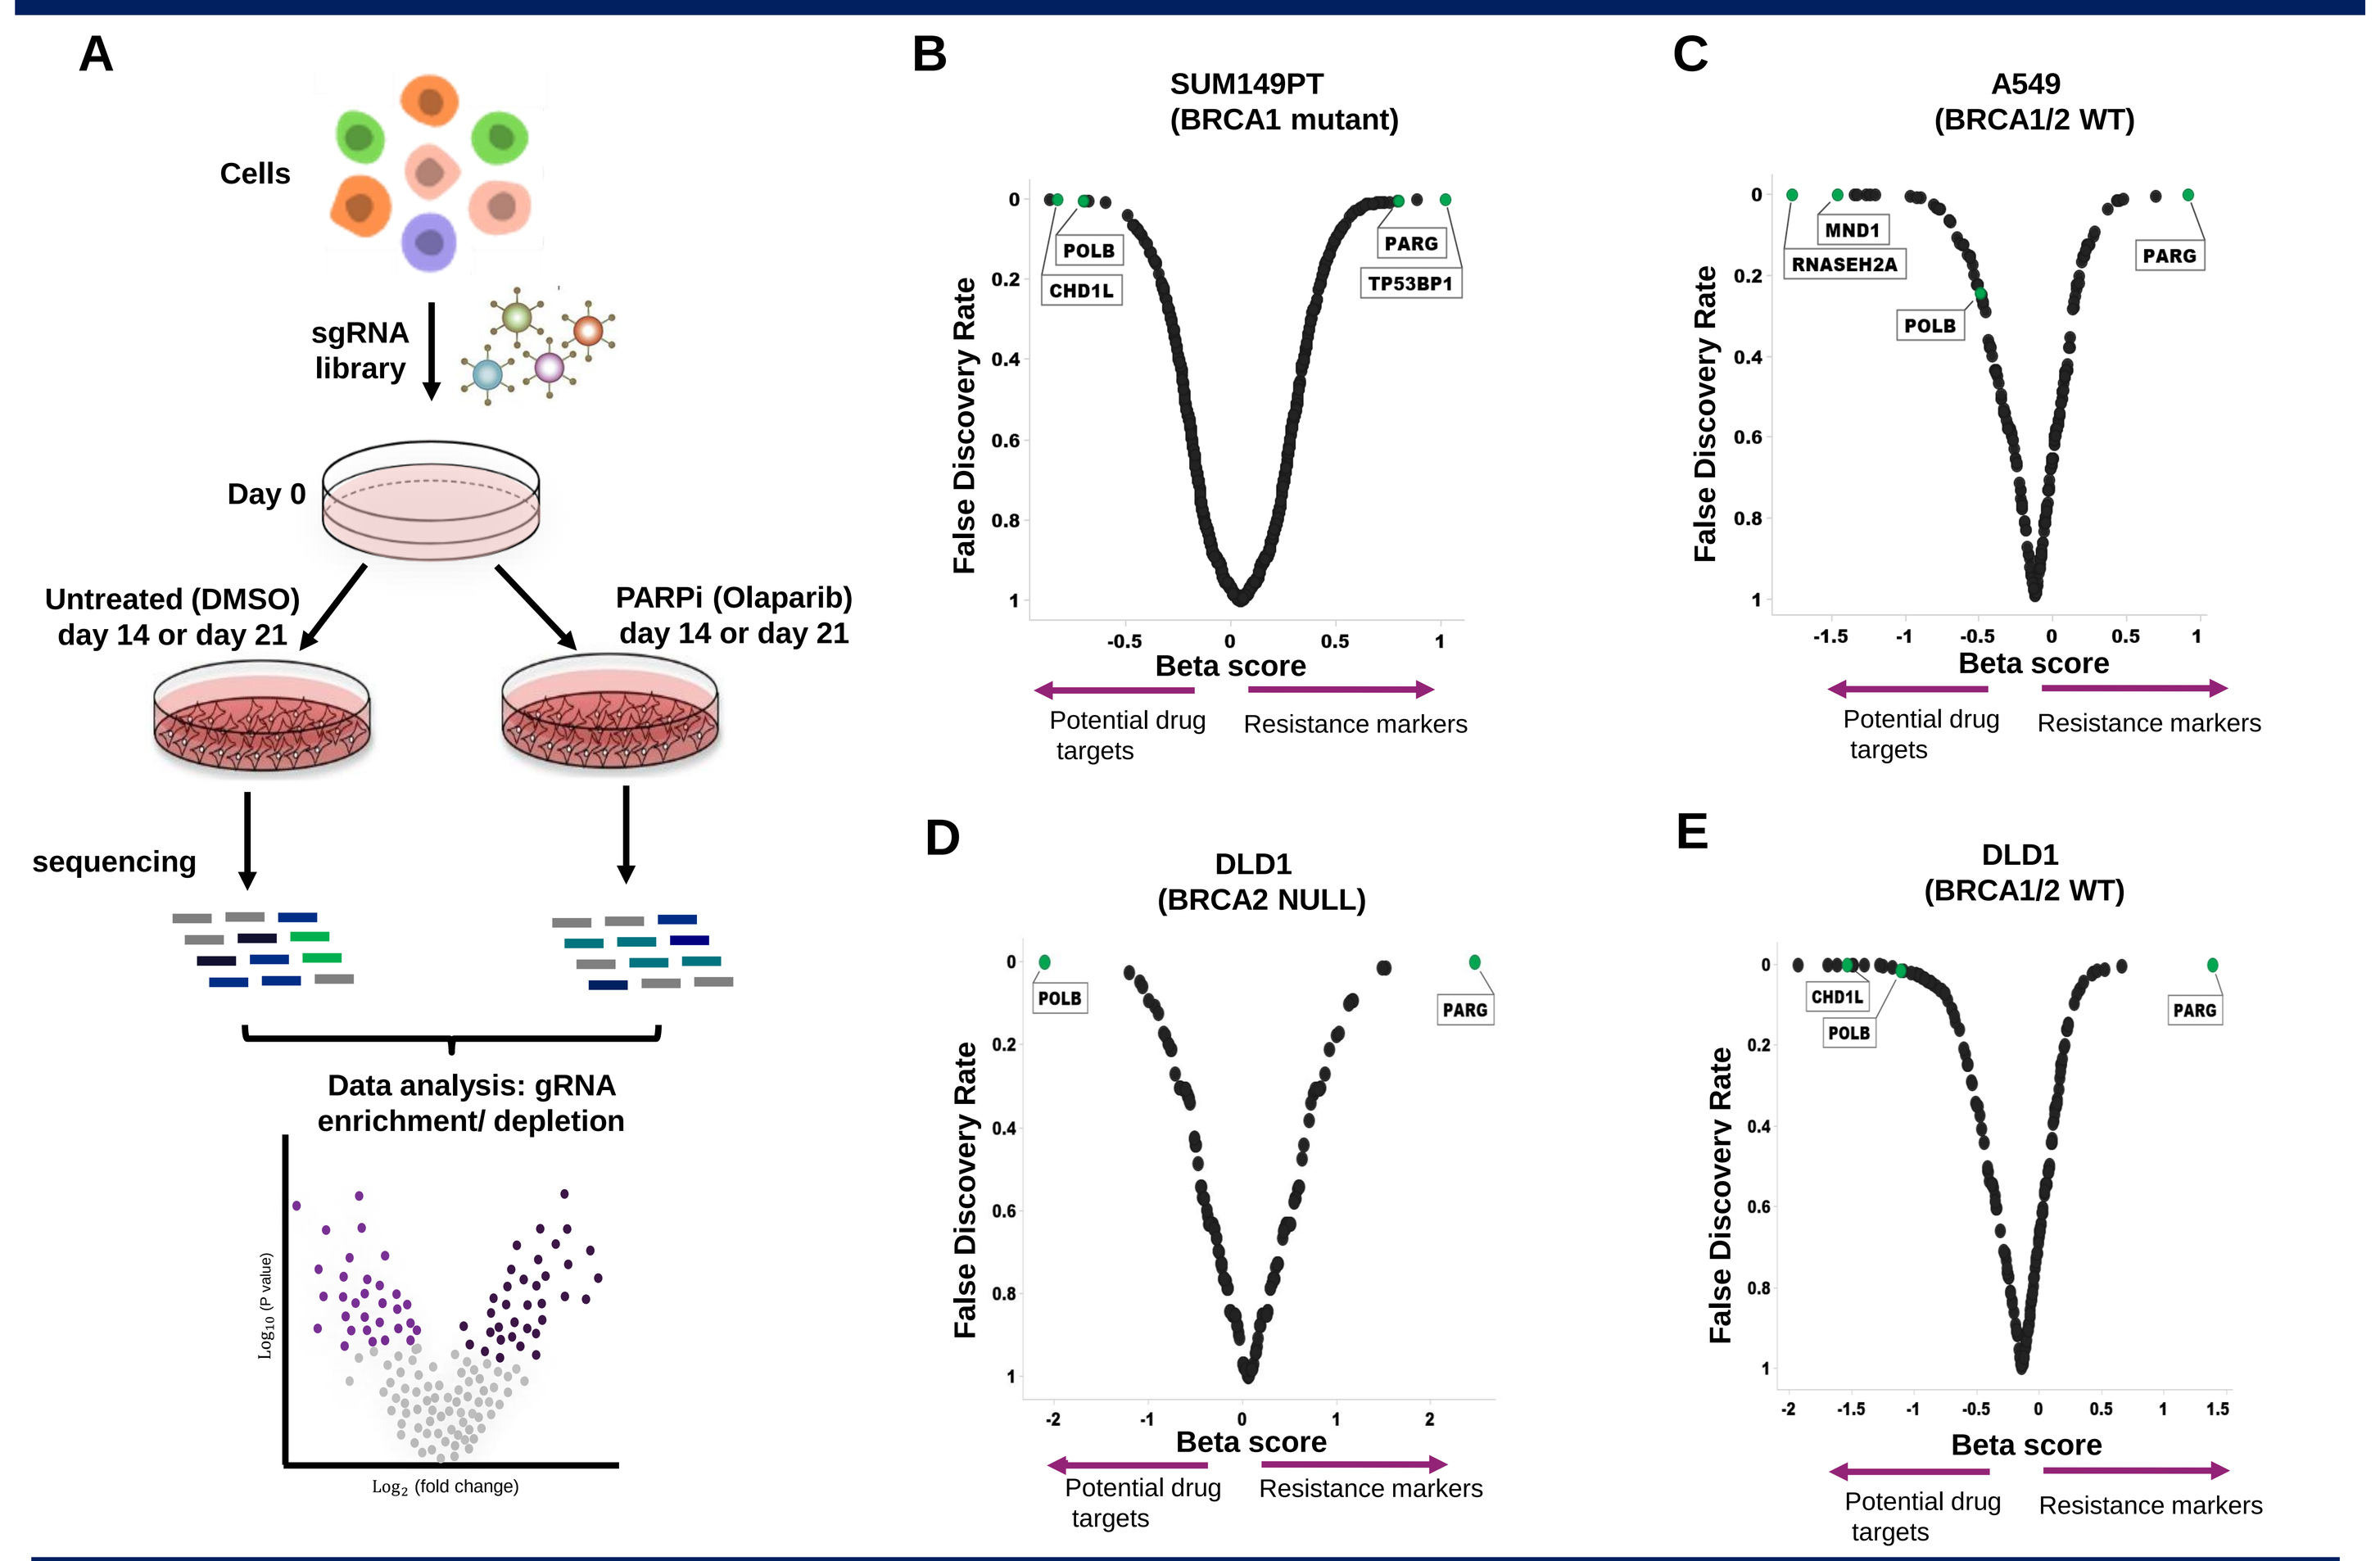
A
Cells
sgRNA library
Day 0
Untreated (DMSO) day 14 or day 21
PARPi (Olaparib) day 14 or day 21
sequencing
Data analysis: gRNA enrichment/ depletion
B
SUM149PT (BRCA1 mutant)
C
A549 (BRCA1/2 WT)
D
DLD1 (BRCA2 NULL)
E
DLD1 (BRCA1/2 WT)
False Discovery Rate
Beta score
Potential drug targets
Resistance markers
POLB
CHD1L
PARG
TP53BP1
MND1
RNASEH2A

Figure 1: CRISPR-based drug anchor screens in the presence or absence of Olaparib. (A) Schematic of CRISPR drug anchor screens performed with +/- Olaparib in 2 BRCA1 mutant cell lines and DLD1 isogenic cell lines. sgRNA enrichment/depletion analysis represented in volcano plots. (B) MDA-MB-436. (C) A549. (D) DLD1 BRCA2 null (-/-). (E) DLD1 BRCA1/2 WT (+/+).

POLB sgRNAs drop out with PARP inhibitor treatment in a time, dose, and BRCA-dependent manner

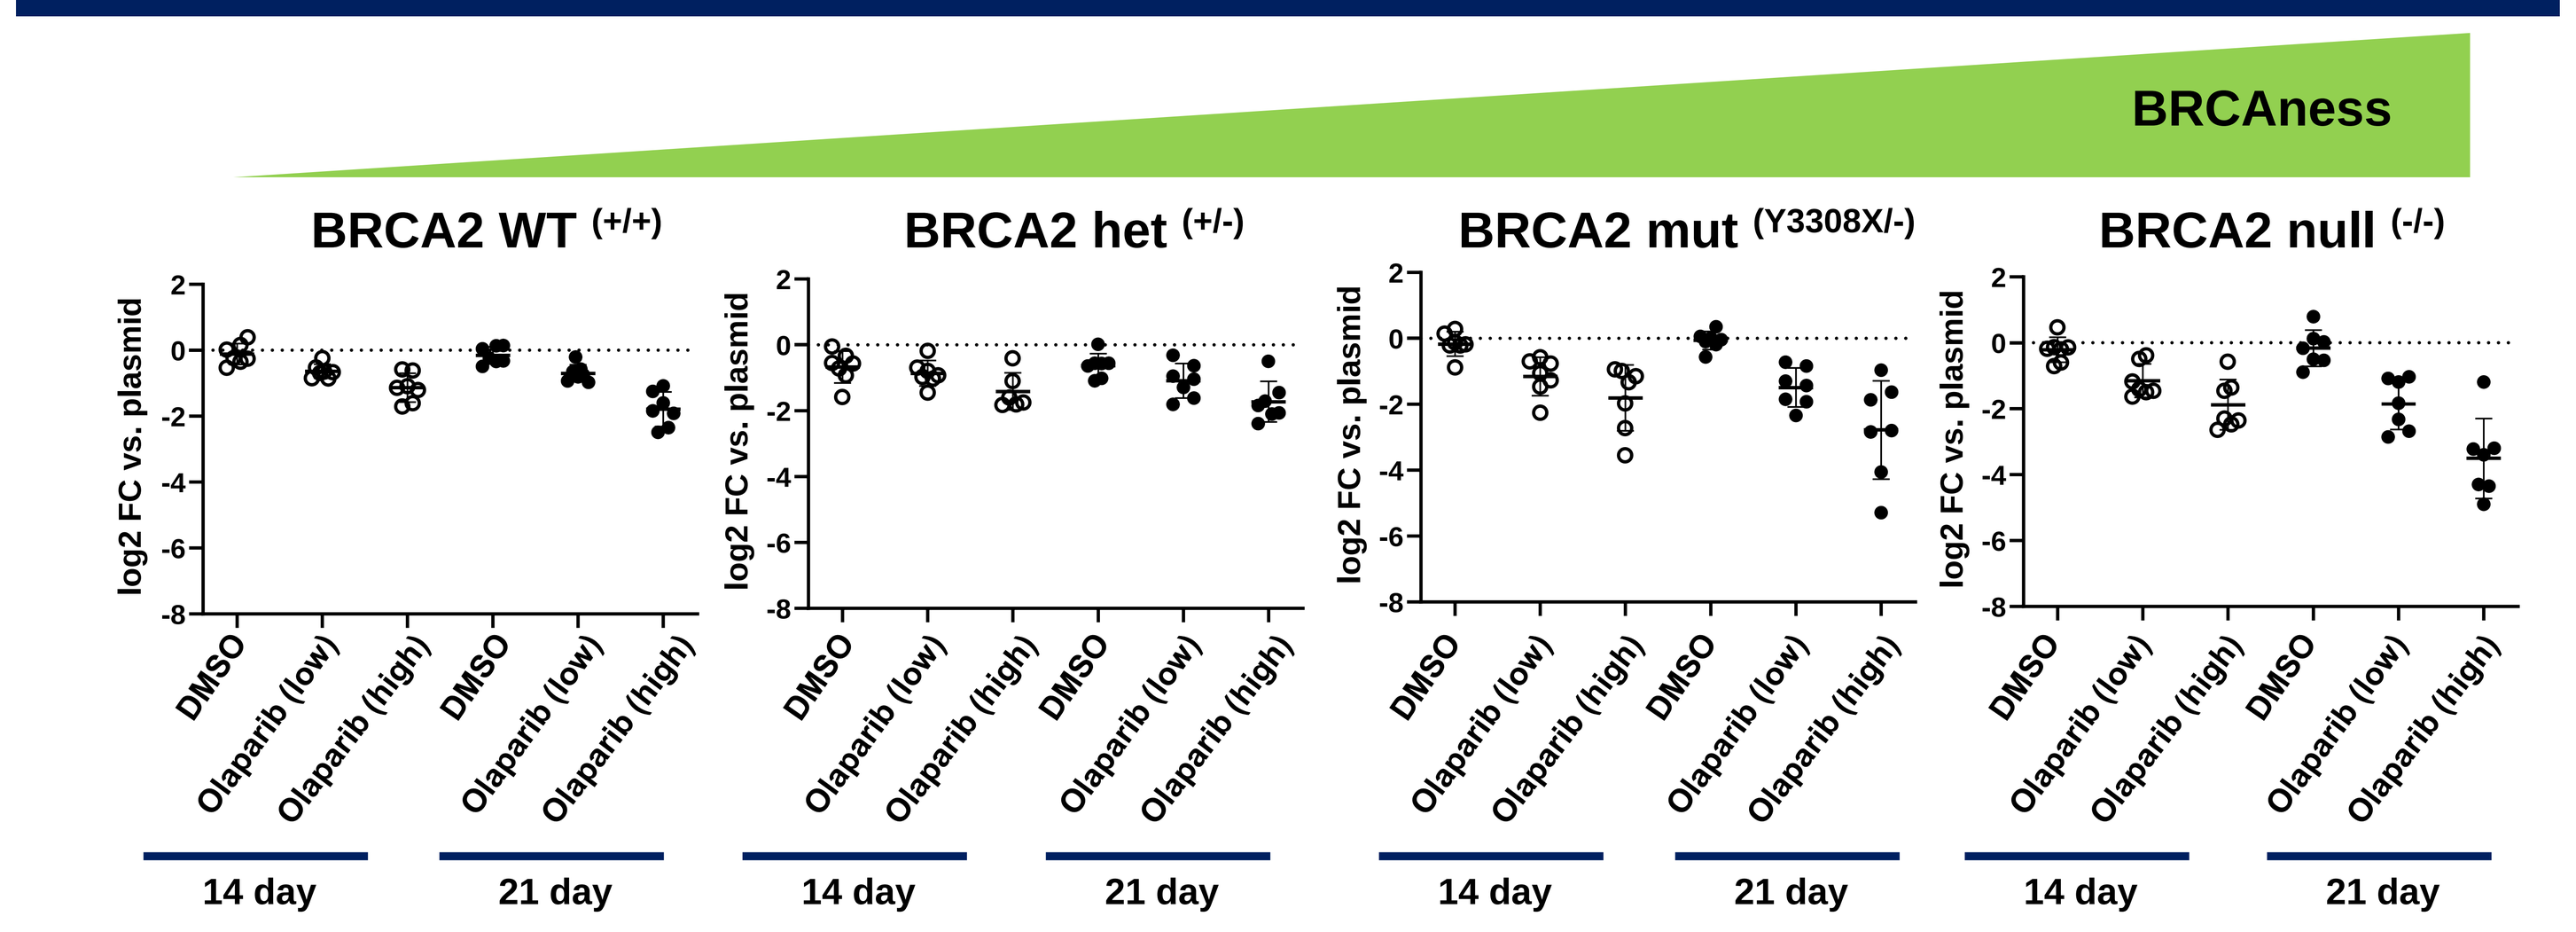
BRCAness
BRCA2 WT (+/+)
BRCA2 het (+/-)
BRCA2 mut (Y3308X/-)
BRCA2 null (-/-)
log2 FC vs. plasmid
DMSO
Olaparib (low)
Olaparib (high)
14 day
21 day

Figure 2: Dropout validation screen of seven independent POLB sgRNAs in DLD1 isogenic cell lines with PARP inhibitor treatment: Fold change (FC) in POLB sgRNAs compared to DMSO control group.

POLB knockout enhances PARP inhibitor response in BRCA1 mutant cells

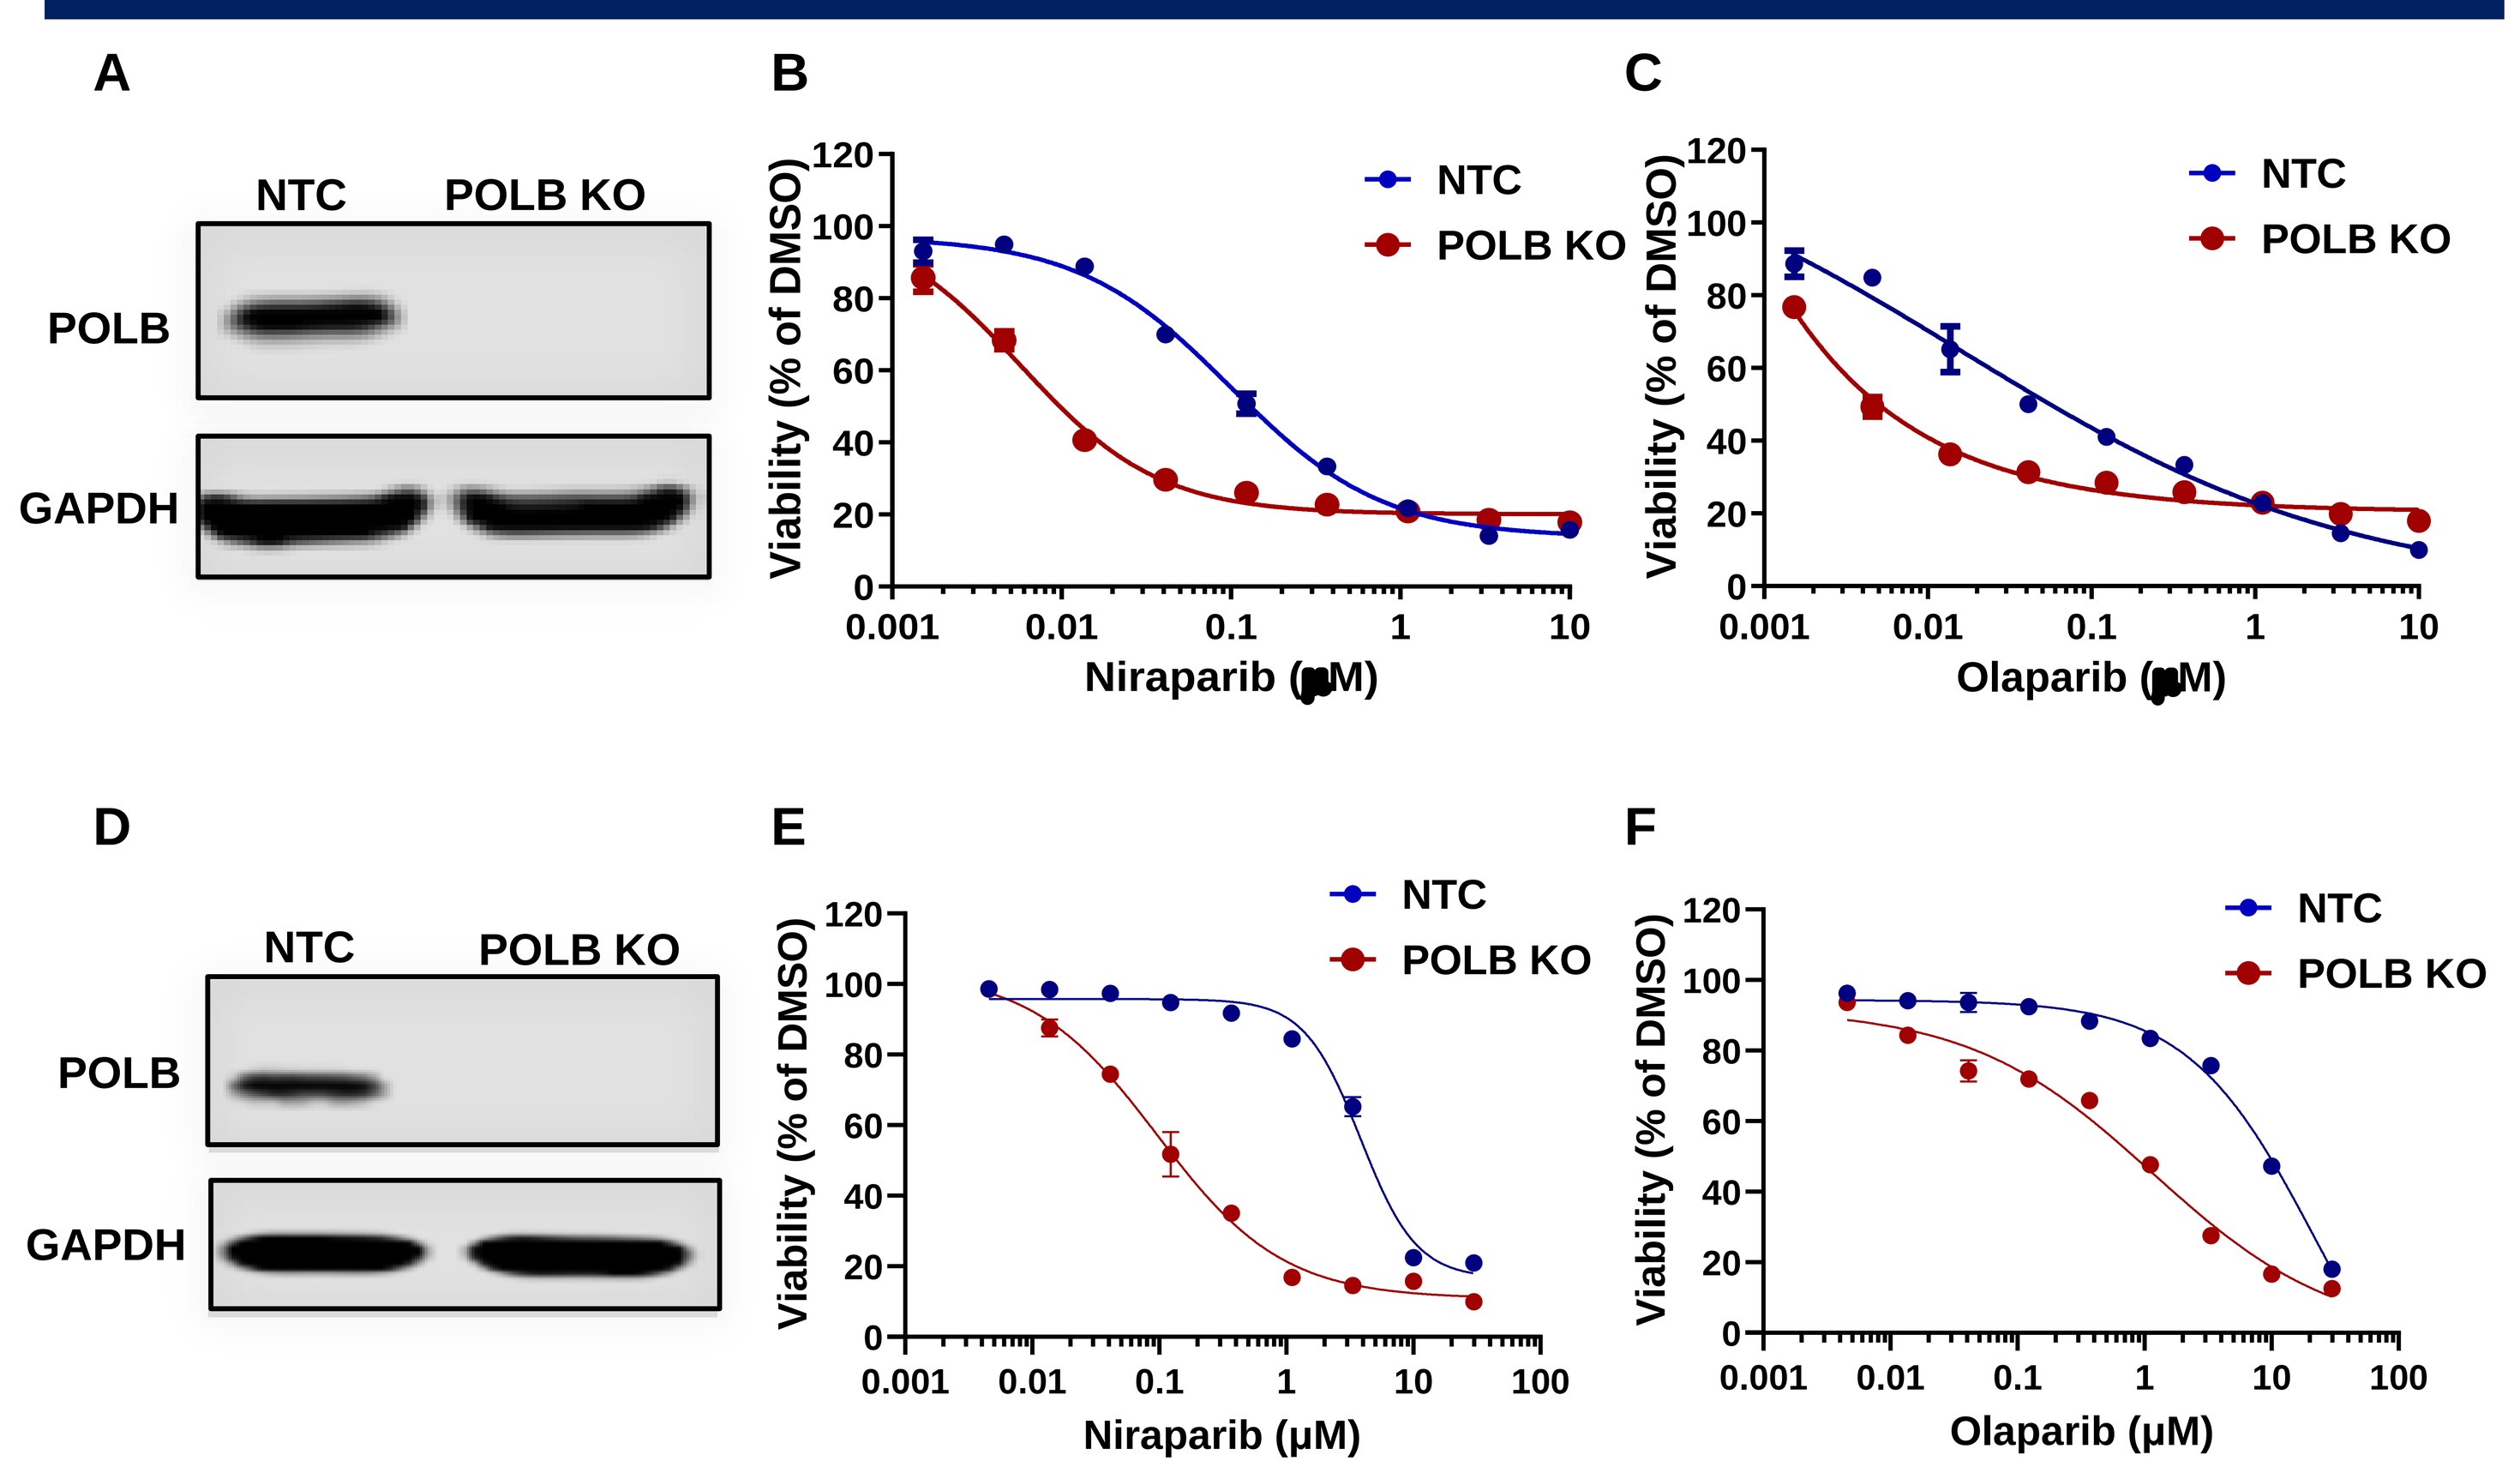
A
NTC
POLB KO
POLB
GAPDH
B
C
D
E
F
Viability (% of DMSO)
Niraparib (μM)
Olaparib (μM)

Figure 3: POLB knockout enhances PARP inhibitor response in BRCA1 mutant cell lines. POLB knockout and non-targeting control (NTC) cell lines were generated in BRCA1 mutant MDAMB436 cells (top panel) and SUM149PT cells (bottom panel). (A, D) Immunoblotting confirming POLB knockout. (B, E) 7-day CTG viability assay with Niraparib. (C, F) 7-day CTG viability assay with Olaparib.

POLB knockout enhances PARP inhibitor-induced growth inhibition in BRCA2 mutant cells

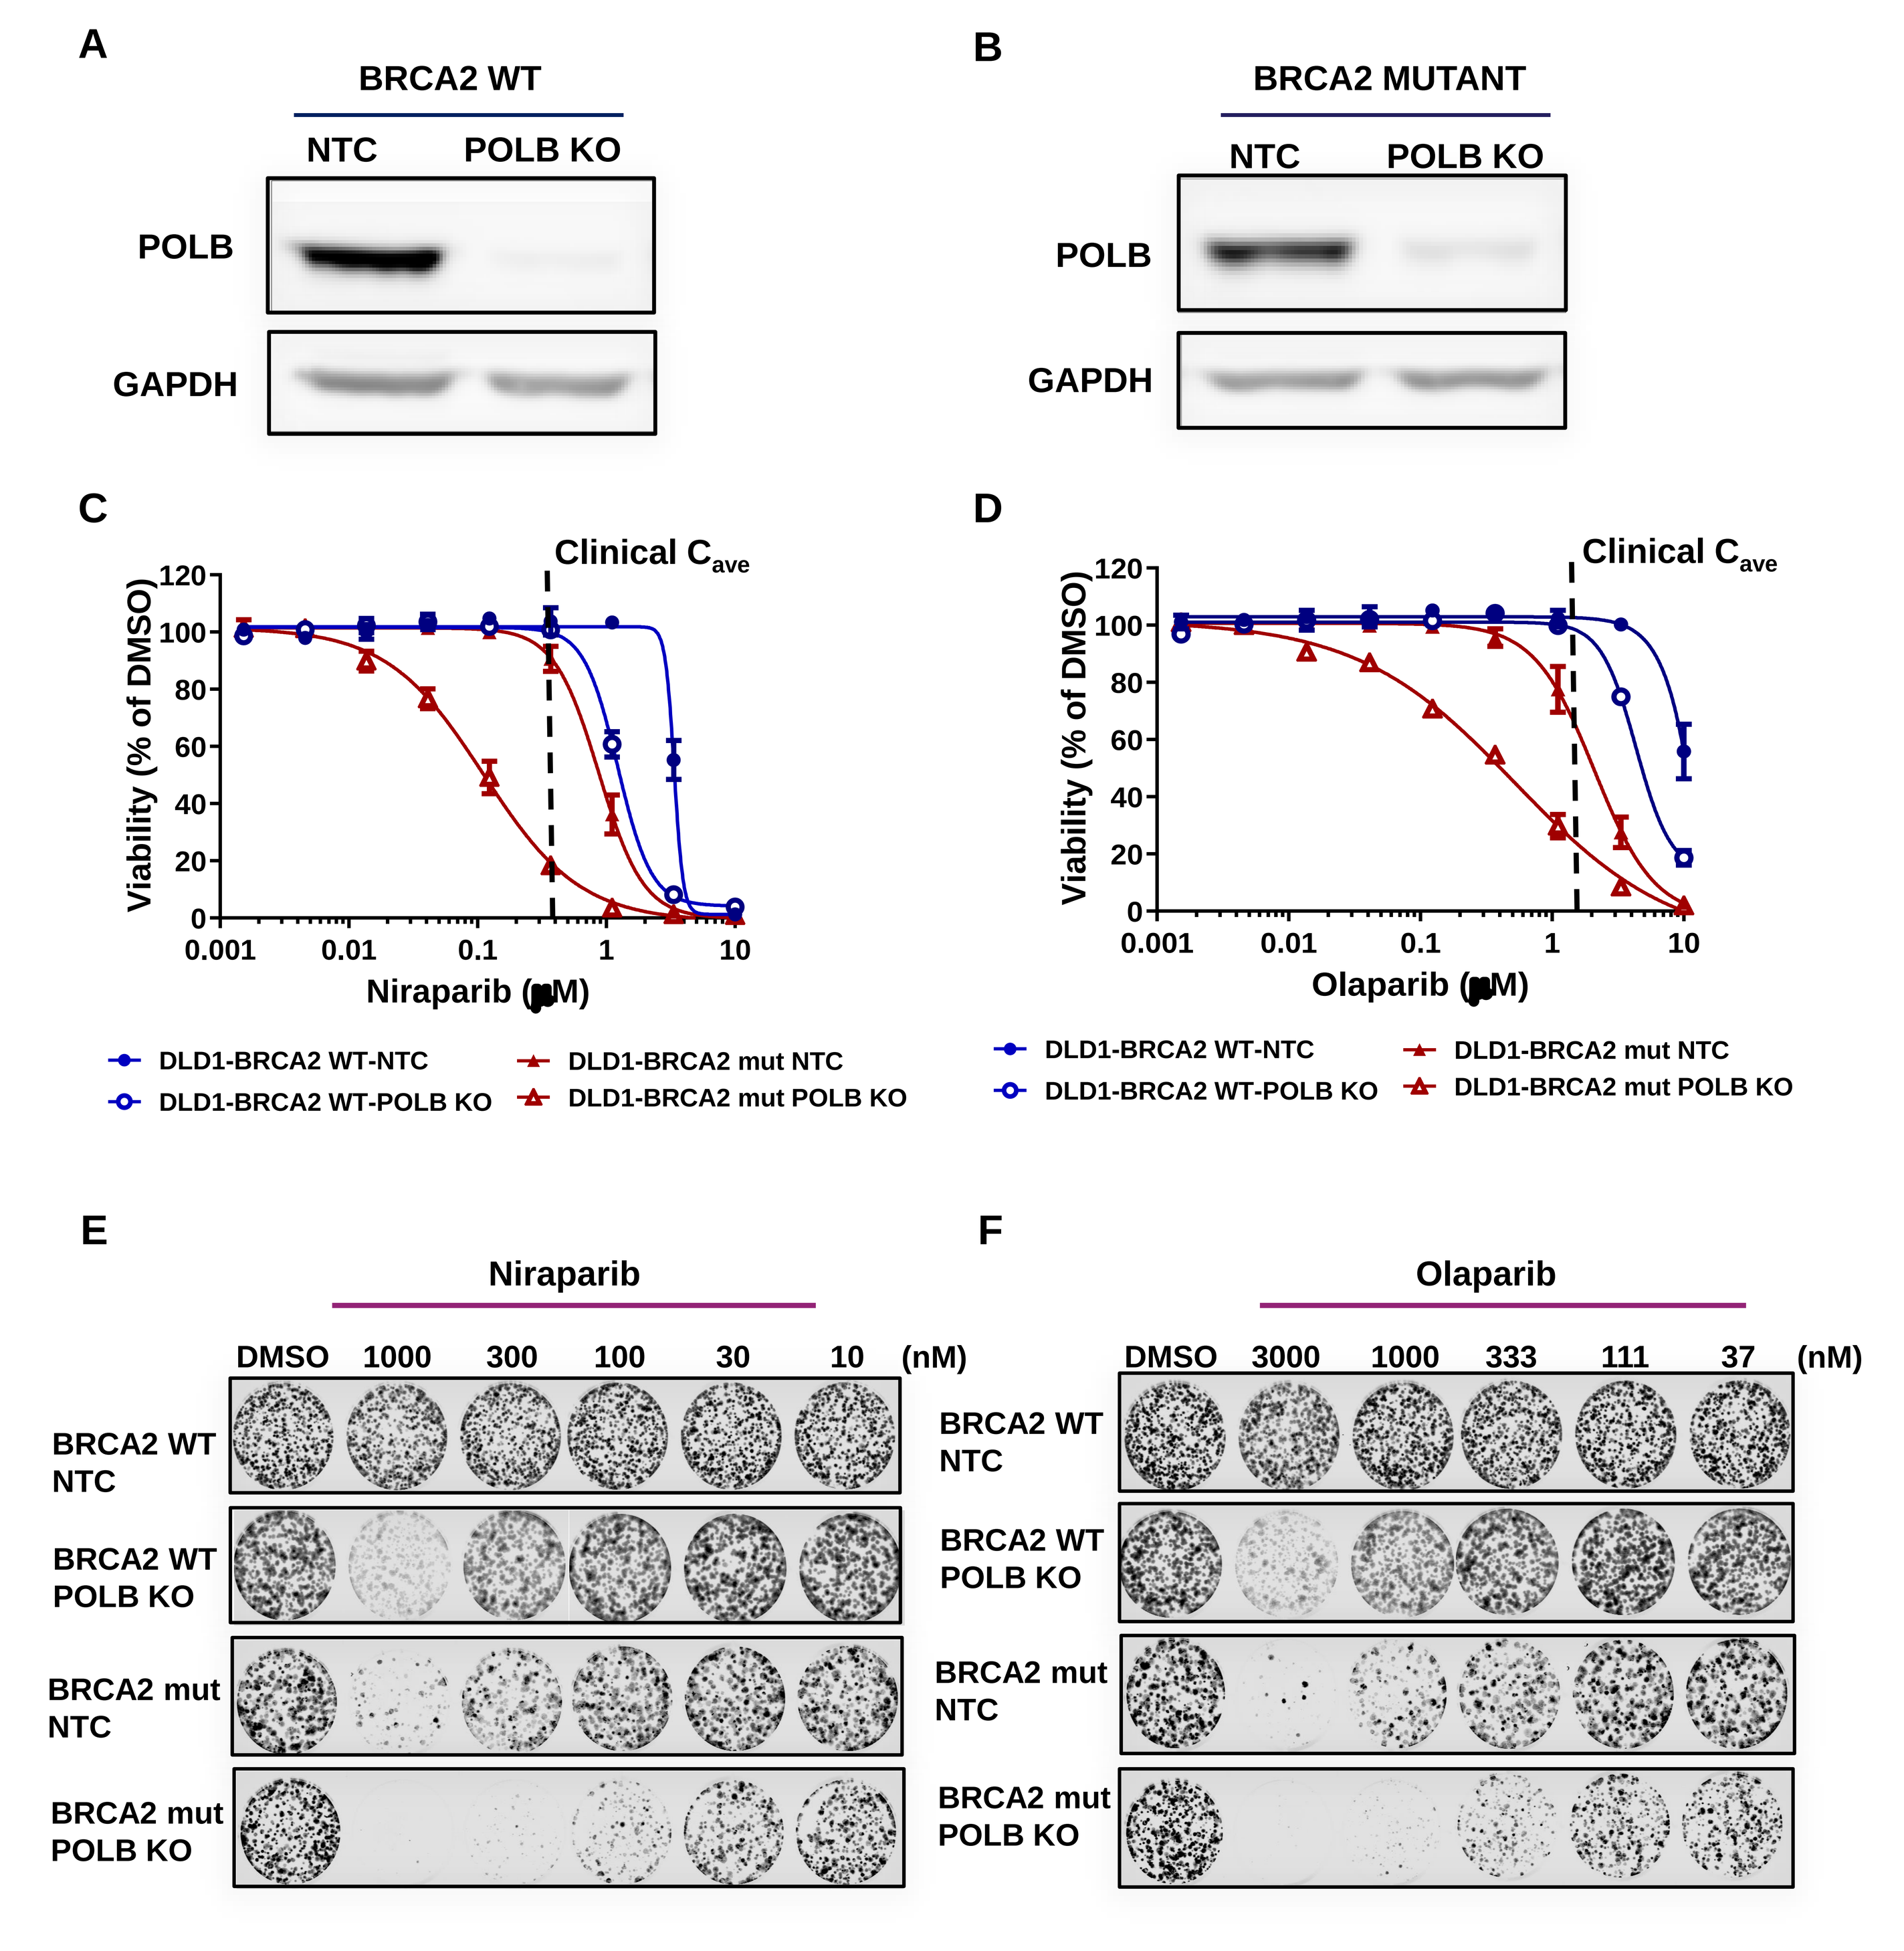
A
BRCA2 WT
B
BRCA2 MUTANT
NTC
POLB KO
POLB
GAPDH
C
D
Clinical C_{ave}
Viability (% of DMSO)
Niraparib (μM)
Olaparib (μM)
DLD1-BRCA2 WT-NTC
DLD1-BRCA2 WT-POLB KO
DLD1-BRCA2 mut NTC
DLD1-BRCA2 mut POLB KO
E
Niraparib
DMSO 1000 300 100 30 10 (nM)
F
Olaparib
DMSO 3000 1000 333 111 37 (nM)
BRCA2 WT NTC
BRCA2 WT POLB KO
BRCA2 mut NTC
BRCA2 mut POLB KO

Figure 4: POLB knockout enhances PARPi-induced growth inhibition in BRCA2 mutant cells. POLB knockout and non-targeting sgRNA control cells were generated using DLD1 isogenic cell lines. (A, B) Western blot confirming POLB knockout. (C) 7-day viability assay with Niraparib. (D) 7-day viability assay with Olaparib. (E) 14-day colony formation assays with Niraparib. (F) 14-day colony formation assays with Olaparib.

Inhibition of polymerase and lyase activities of POLB are required for synthetic lethal activity with PARP inhibitor

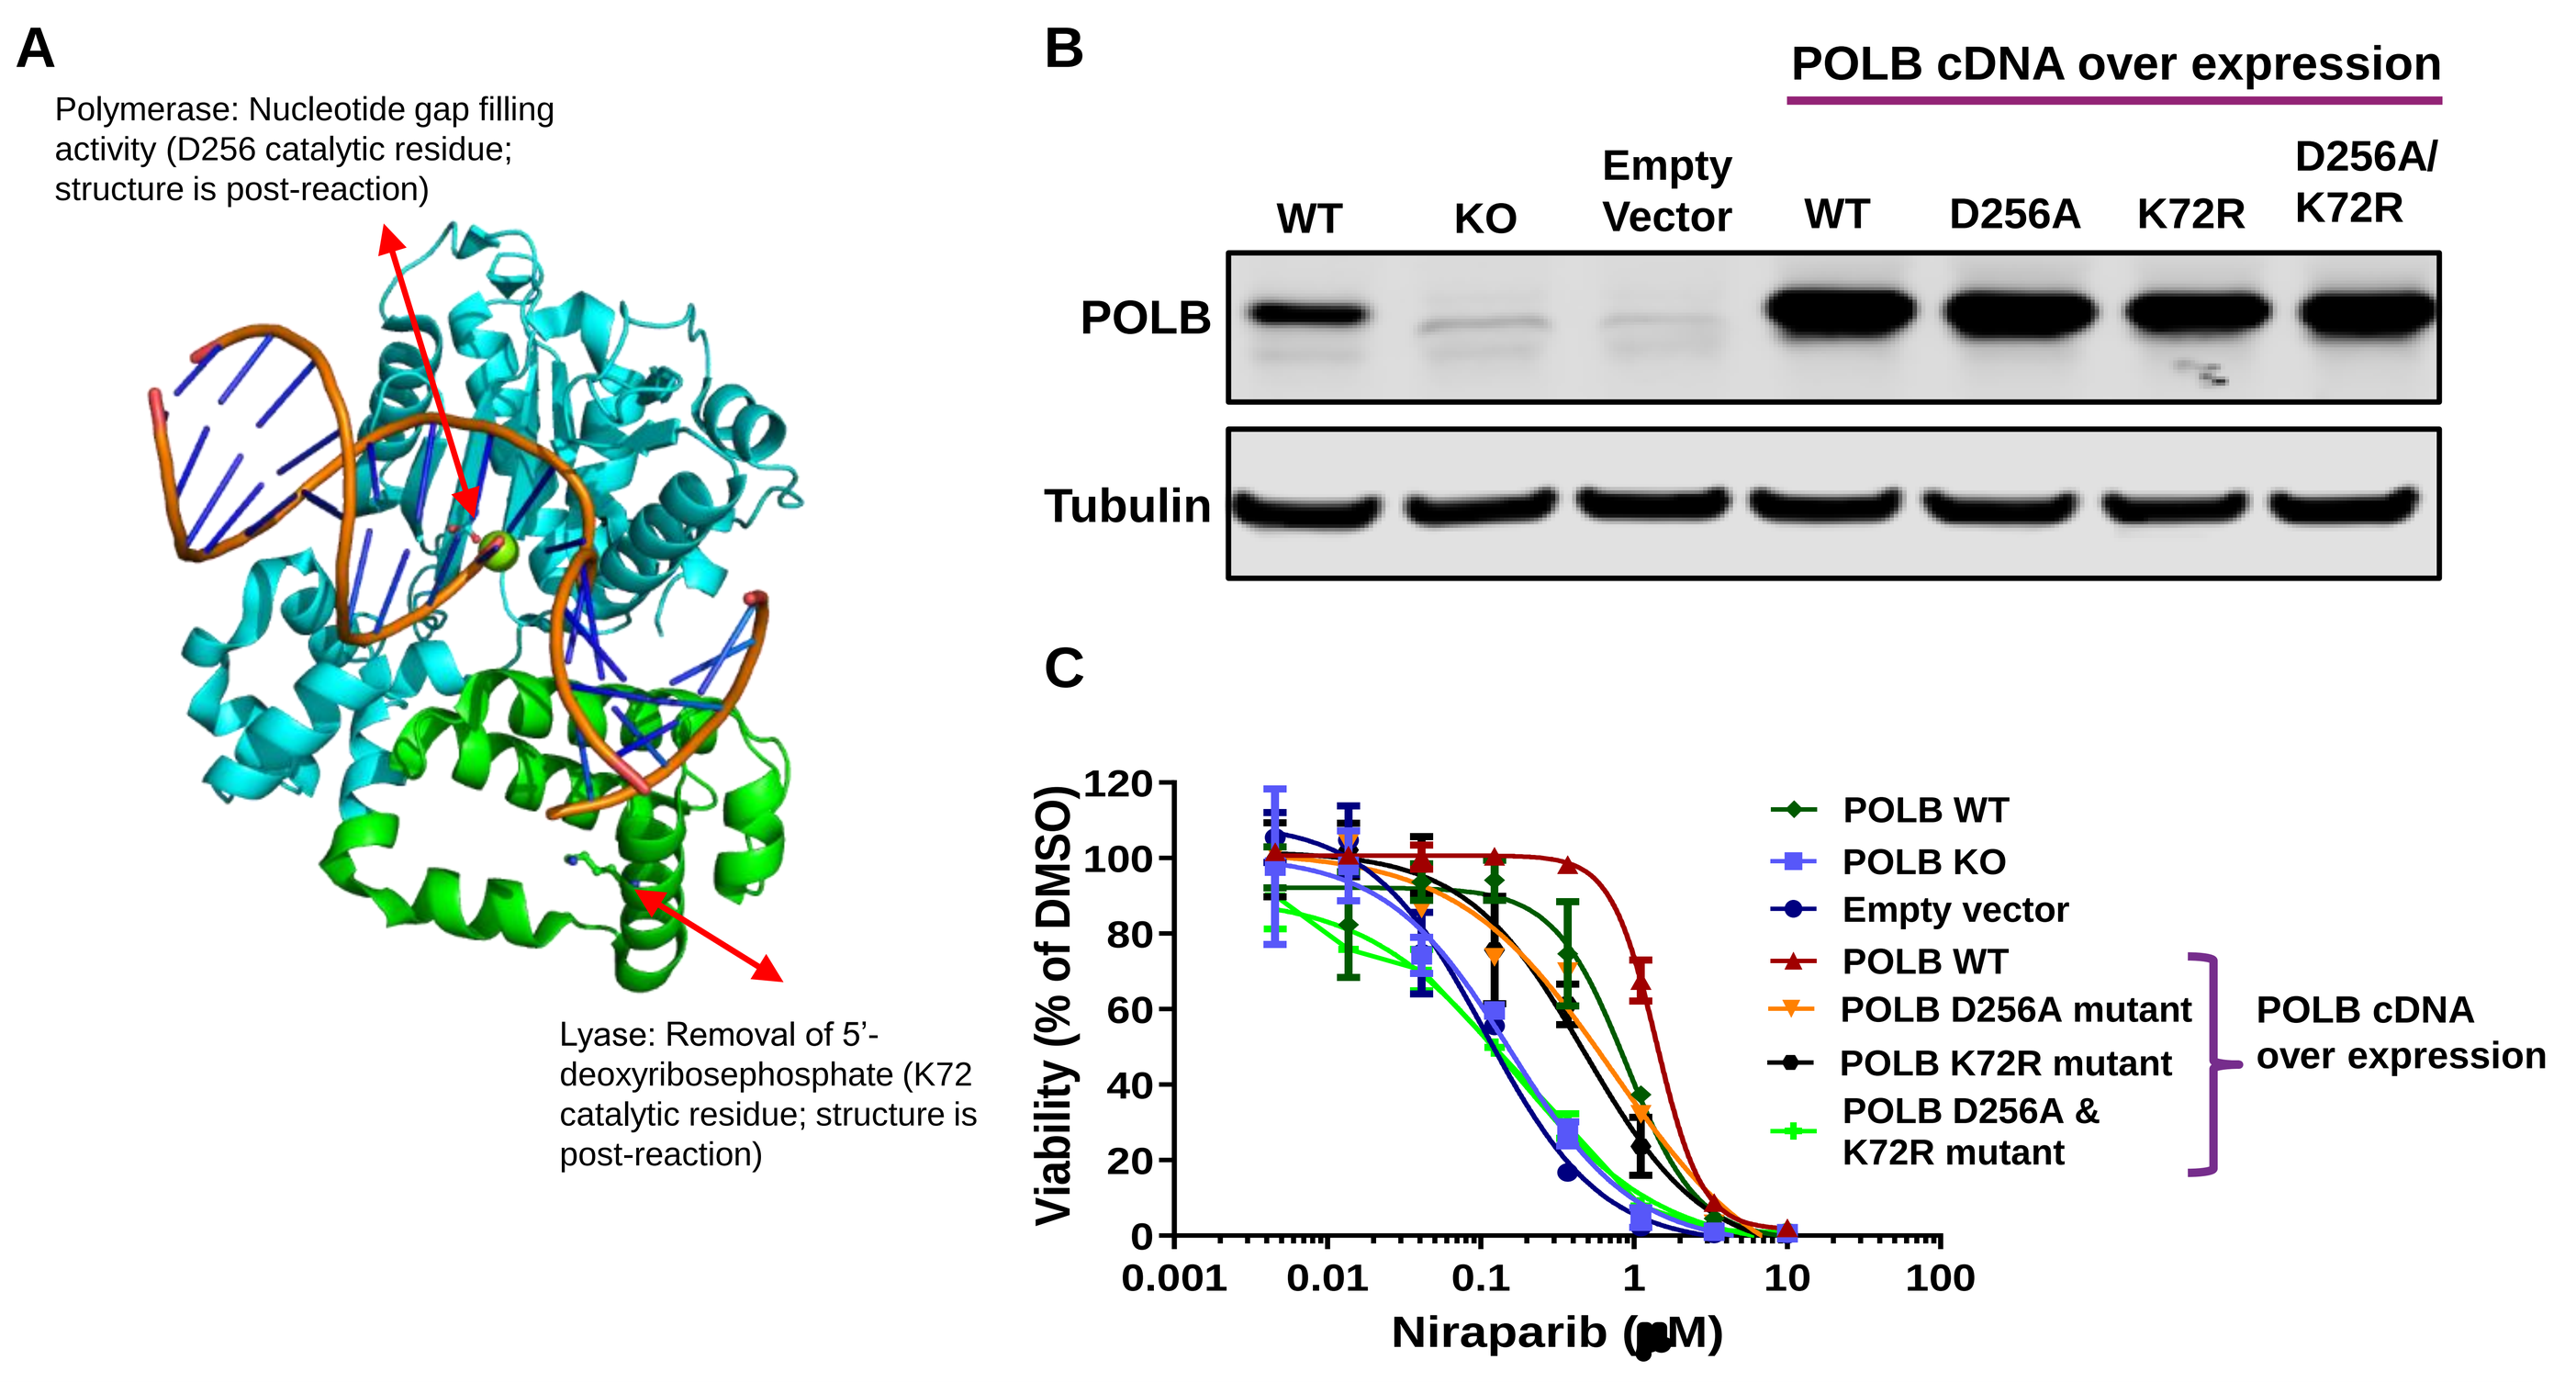
A
Polymerase: Nucleotide gap filling activity (D256 catalytic residue; structure is post-reaction)
Lyase: Removal of 5'-deoxyribosephosphate (K72 catalytic residue; structure is post-reaction)
B
POLB cDNA over expression
WT
KO
Empty Vector
WT
D256A
K72R
D256A/ K72R
POLB
Tubulin
C
Viability (% of DMSO)
Niraparib (μM)
POLB WT
POLB KO
Empty vector
POLB WT
POLB D256A mutant
POLB K72R mutant
POLB D256A & K72R mutant
POLB cDNA over expression

Figure 5: Dual loss of Polymerase and Lyase activities of POLB are required for synthetic lethal activity with PARPi: (A) Crystal structure of POLB showing catalytic residues of polymerase and lyase activities (Ref 1). (B) Immunoblotting analysis confirming POLB WT cDNA or mutant cDNA overexpression (C) 7-day viability assay with Niraparib.

Single-strand breaks accumulate in the absence of POLB with PARP inhibitor treatment

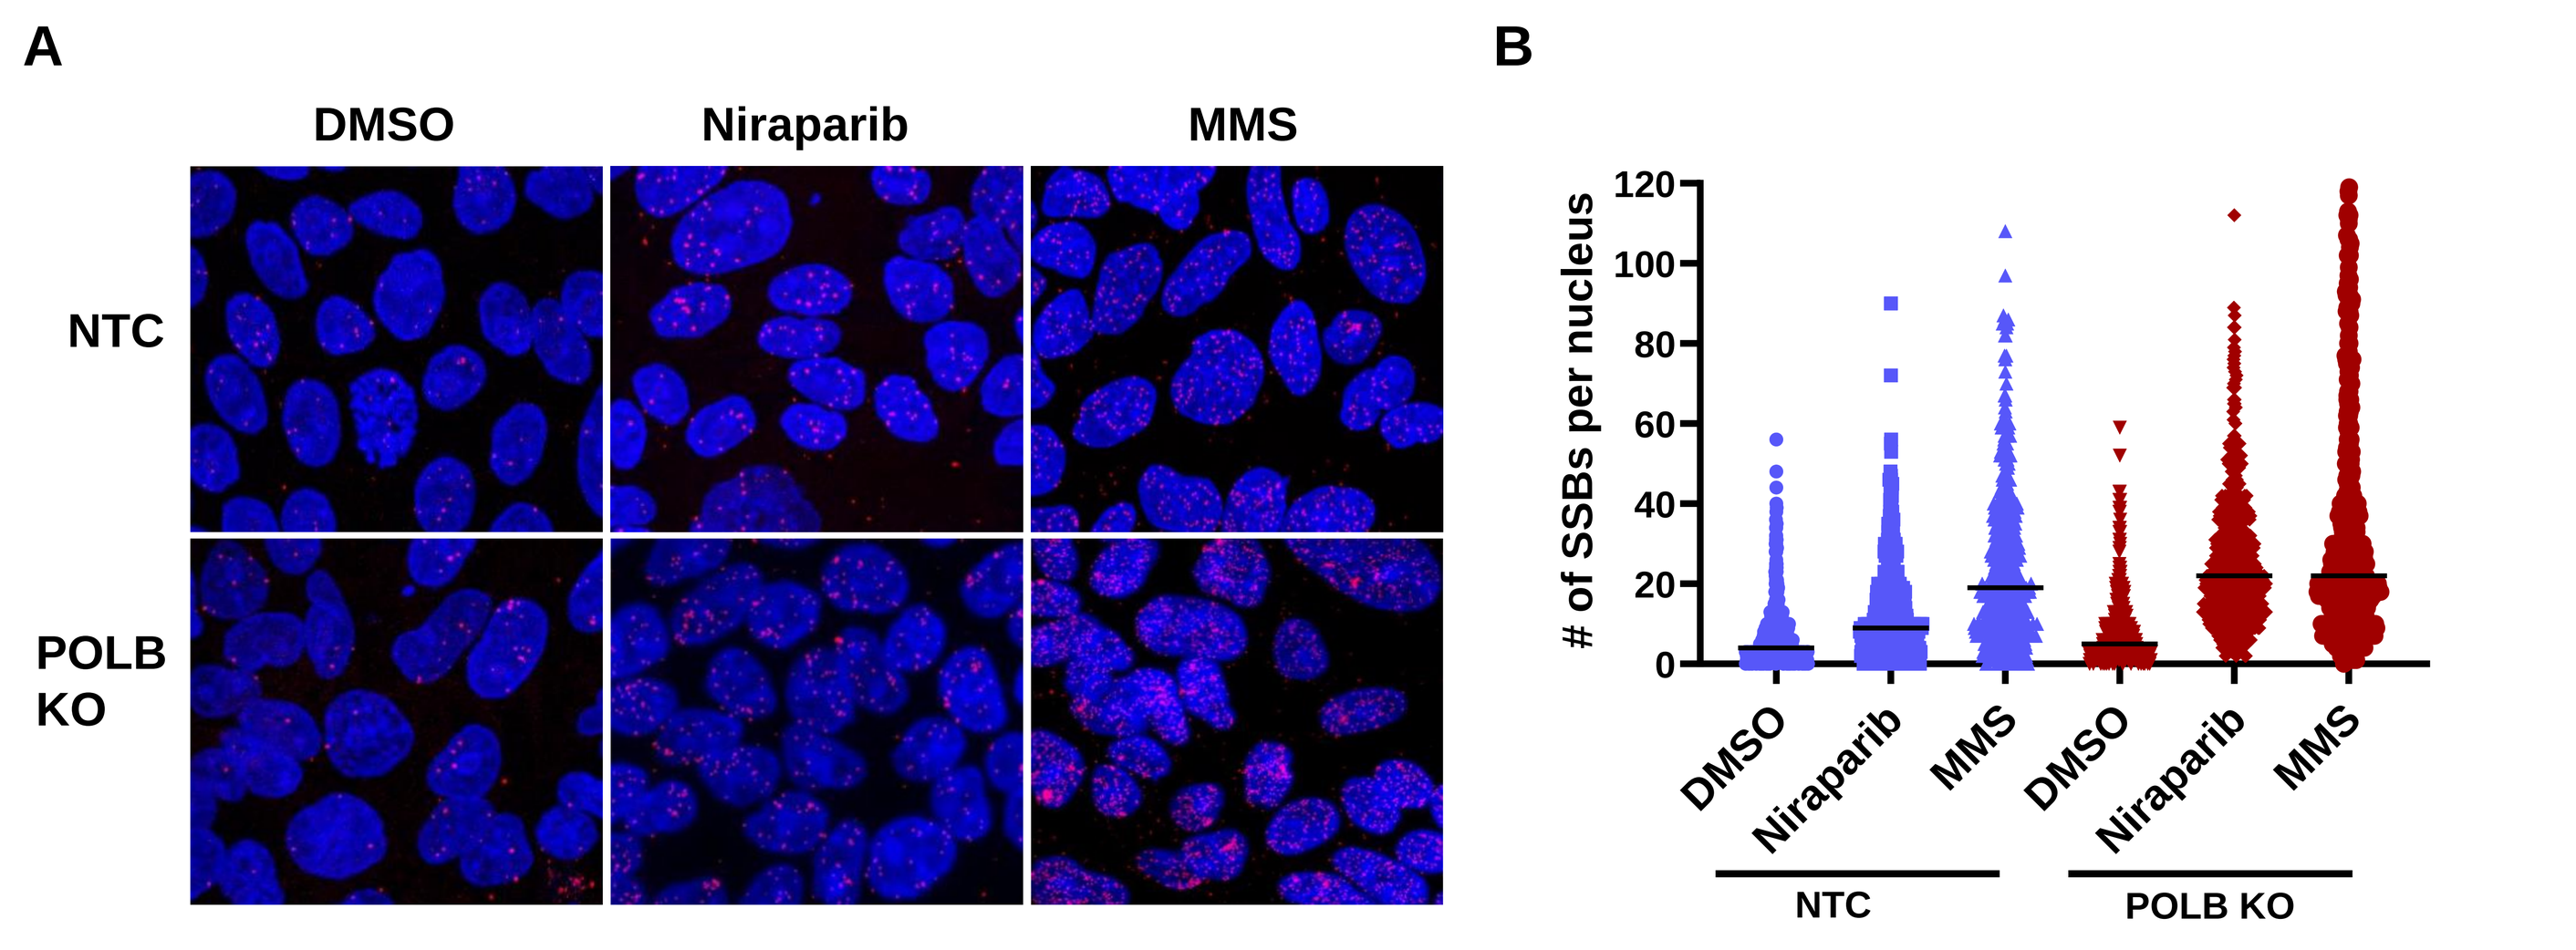
A
DMSO
Niraparib
MMS
NTC
POLB KO
B
of SSBs per nucleus
DMSO
Niraparib
MMS
NTC
POLB KO

Figure 6: Quantification of single-strand breaks in response to POLB knockout (A) sSTRIDE analysis to assess single-strand breaks (Blue, nuclear stain; Pink, single strand breaks) in DLD1 BRCA2 mutant cells. sSTRIDE technology is described in Ref 2. (B) Graph representing the quantification of single-strand breaks in more than 1000 cells per sample.

Knockout of POLB in BRCA1 mutant tumors enhances response to PARP inhibitor treatment

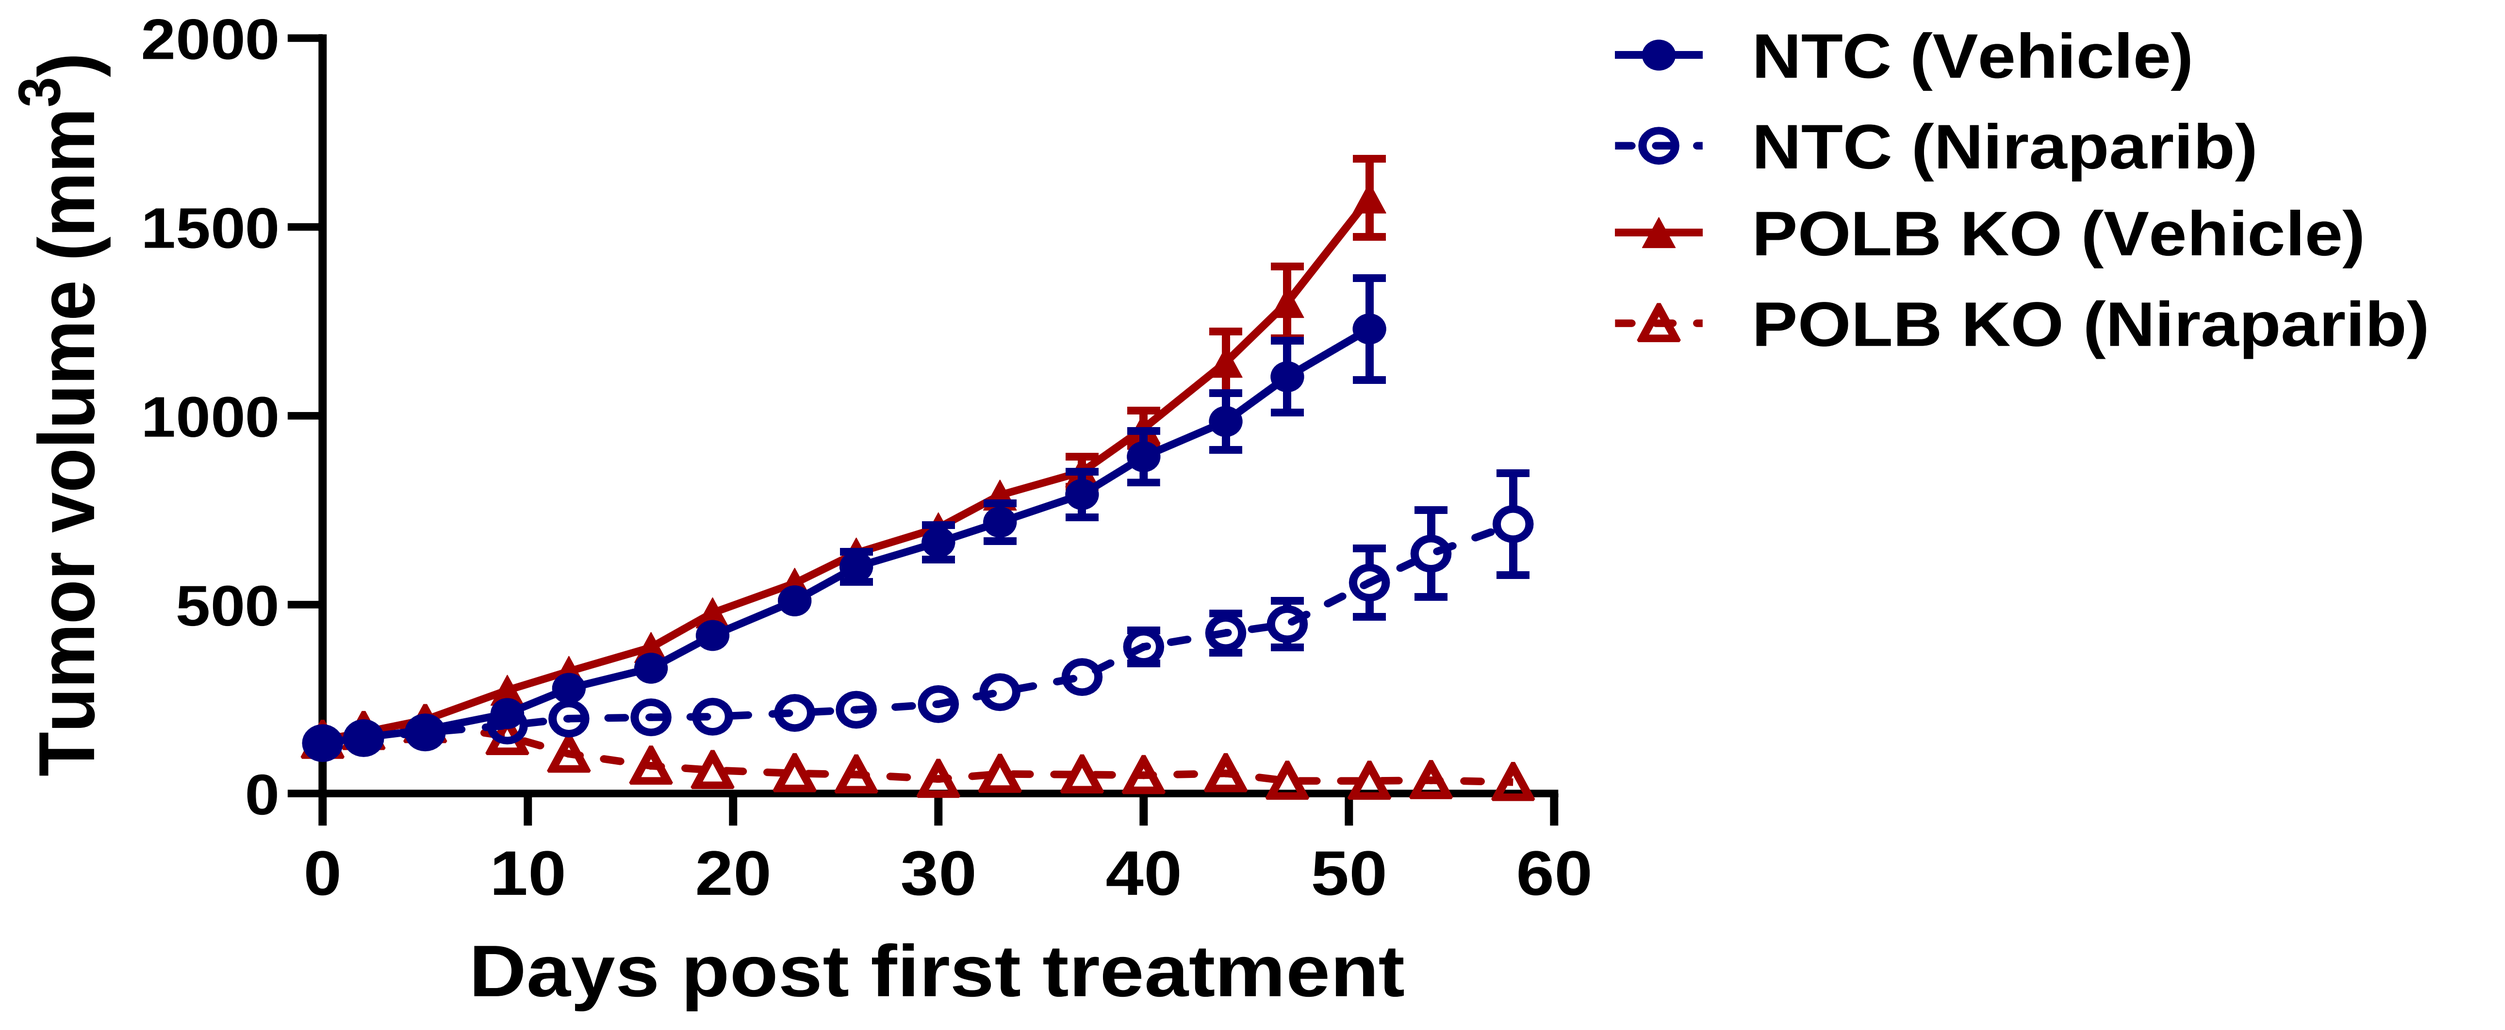
Tumor volume (mm^3)
Days post first treatment
NTC (Vehicle)
NTC (Niraparib)
POLB KO (Vehicle)
POLB KO (Niraparib)

Figure 7: Loss of POLB in BRCA1 mutant cells enhances PARP inhibitor-induced tumor growth inhibition. Xenograft tumors generated from Non-targeting control and POLB knockout MDA-MB-436 cells treated with either vehicle or 15mpk Niraparib for 28 days, PO, QD dosing.

Complete and durable tumor growth inhibition with POLB KO + PARP inhibitor treatment

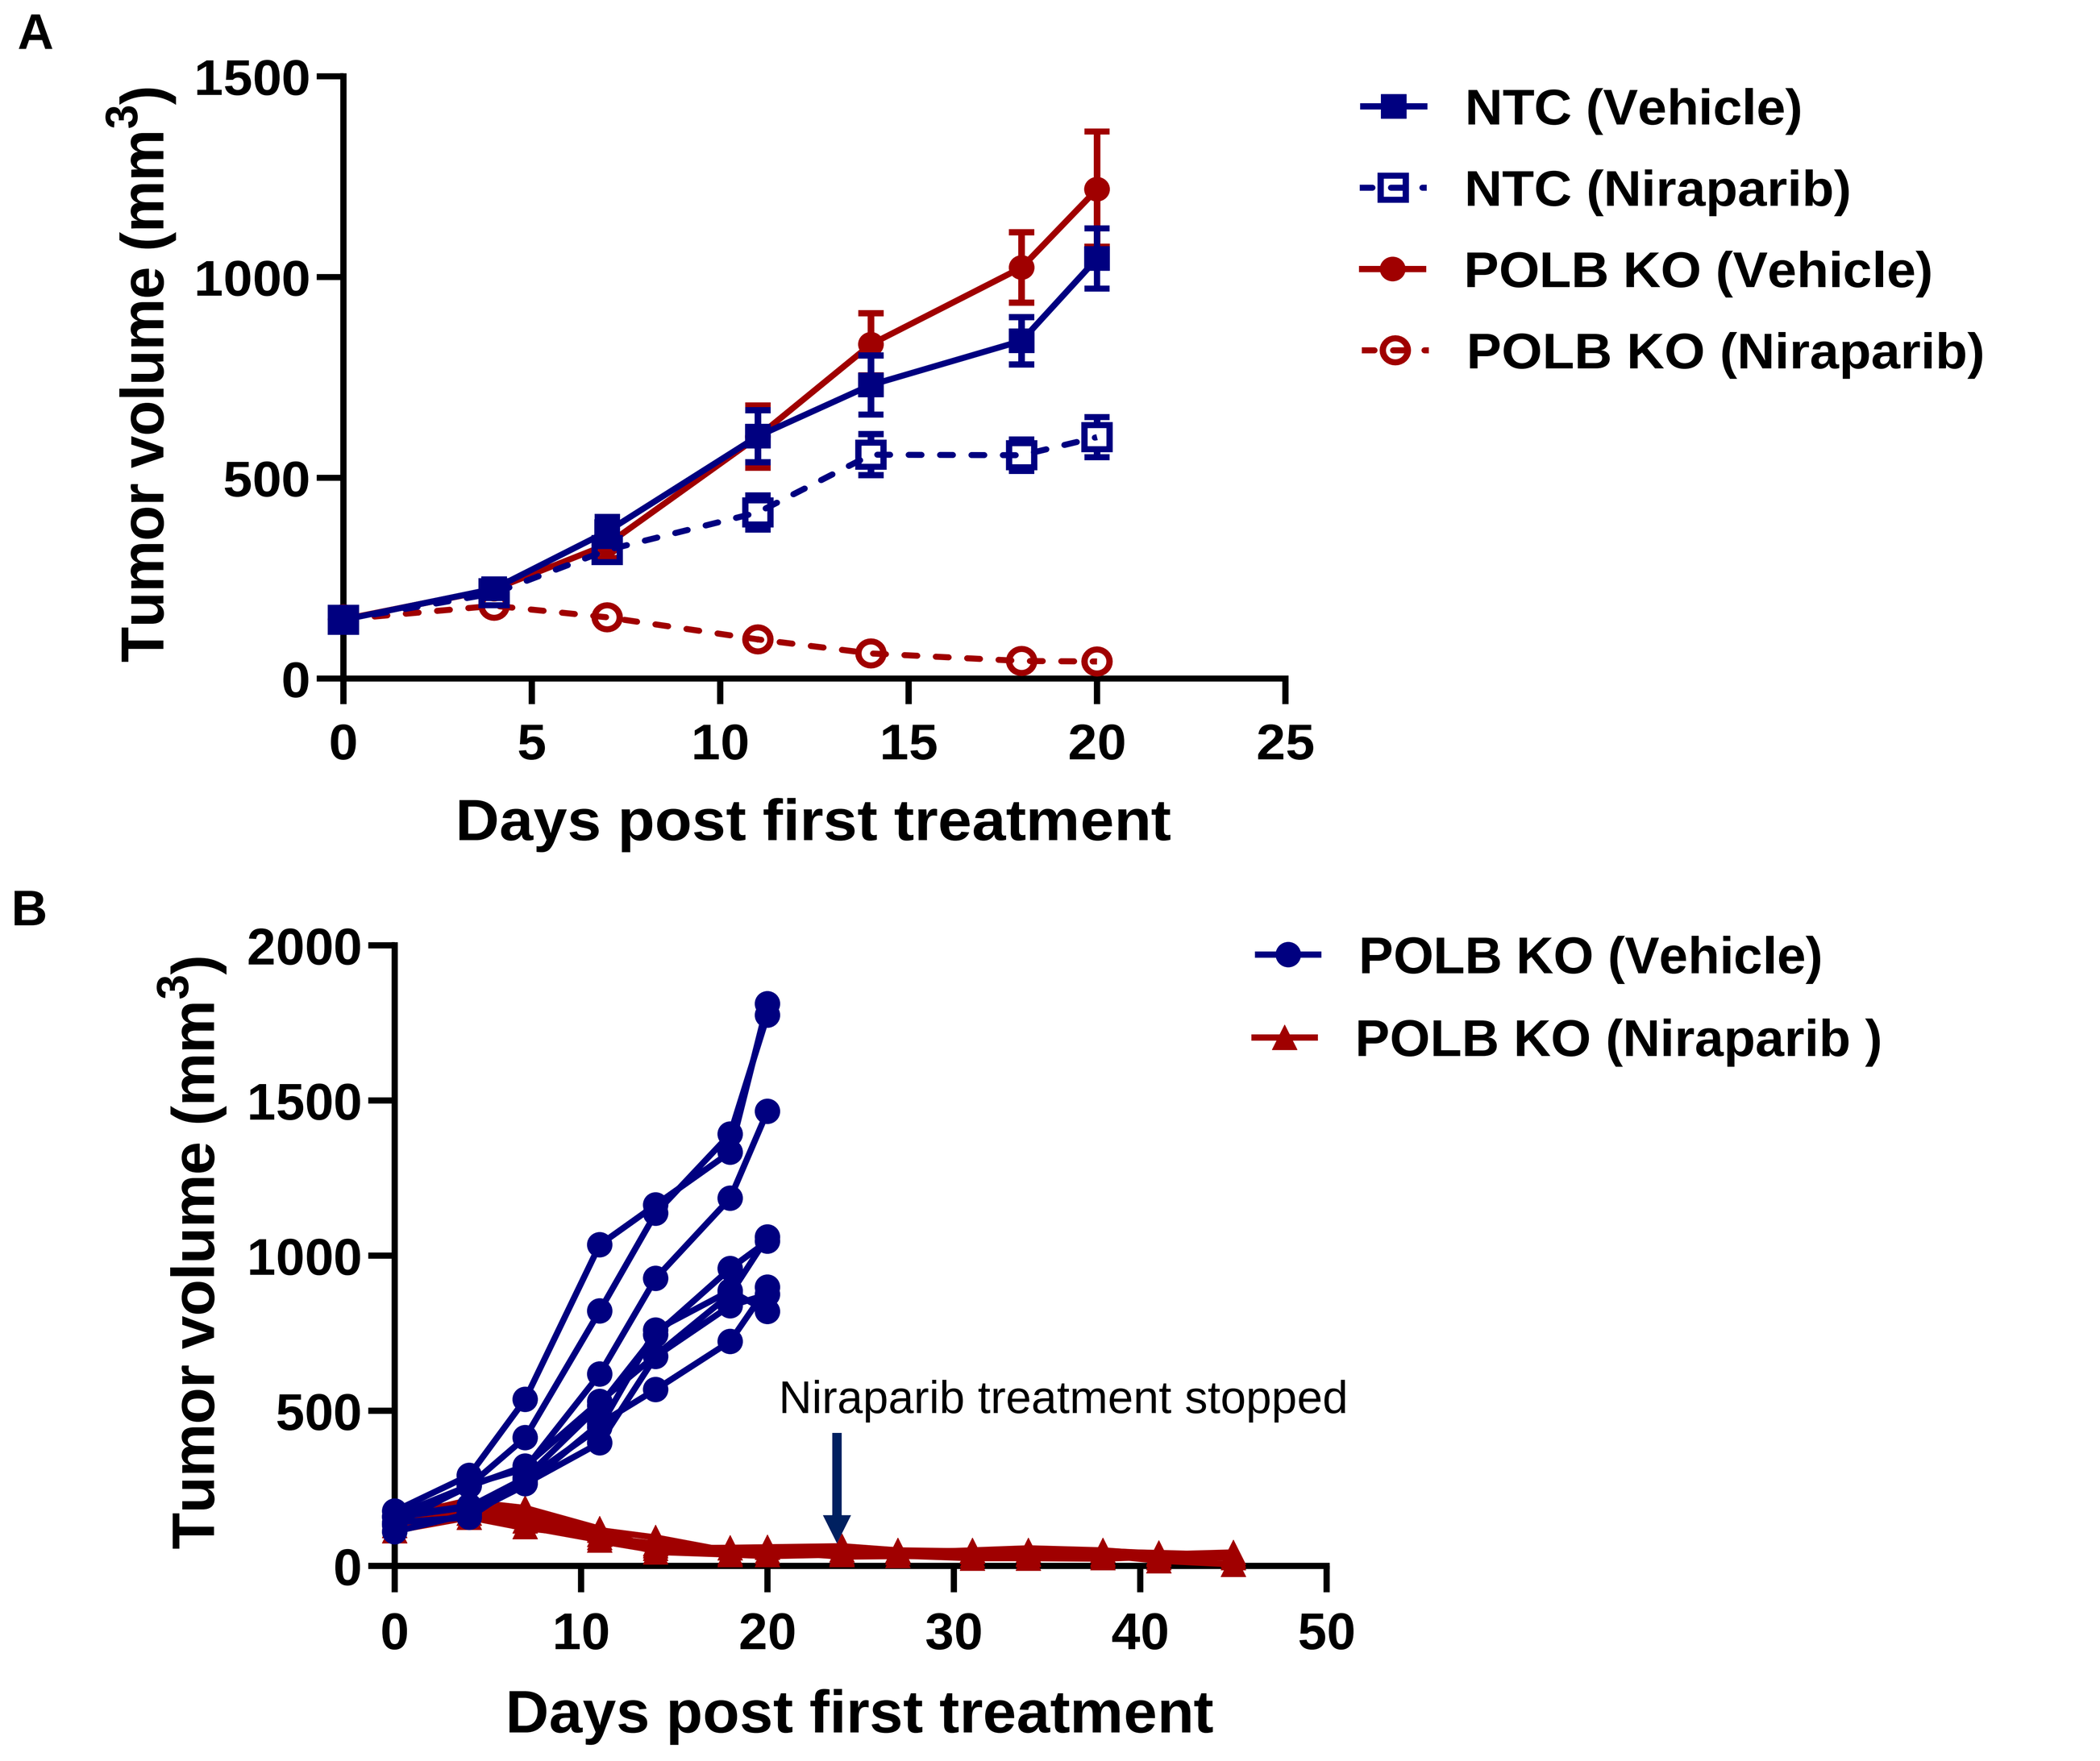
A
Tumor volume (mm^3)
Days post first treatment
NTC (Vehicle)
NTC (Niraparib)
POLB KO (Vehicle)
POLB KO (Niraparib)
B
POLB KO (Vehicle)
POLB KO (Niraparib)
Niraparib treatment stopped

Figure 8: Loss of POLB leads to complete and durable response with PARP inhibitor treatment. (A) Xenograft tumors generated from DLD1 BRCA2 null POLB knockout cells treated with either vehicle or Niraparib at 30mpk, PO, QD dosing. (B) Tumors generated from DLD1 BRCA2 null POLB knockout cells from individual animals either treated with vehicle or niraparib post cessation of treatment at 28 days.

SUMMARY

- **Olaparib drug anchor screens identify POLB as a synthetic lethal target in BRCA1/2 mutant cancers**
- **Genetic validation confirms strong synergy with PARP inhibitor in vitro and in vivo at clinically relevant doses, including complete and durable response after cessation of niraparib treatment**
- **Inhibition of polymerase and lyase activity of POLB is required for the synthetic lethal interaction with PARP inhibitors based on cDNA rescue studies**
- **Knockout of POLB with PARP inhibitor treatment induces single-strand breaks, leading to double-strand breaks, and enhanced cell cycle arrest and apoptosis supporting a synthetic lethal model**

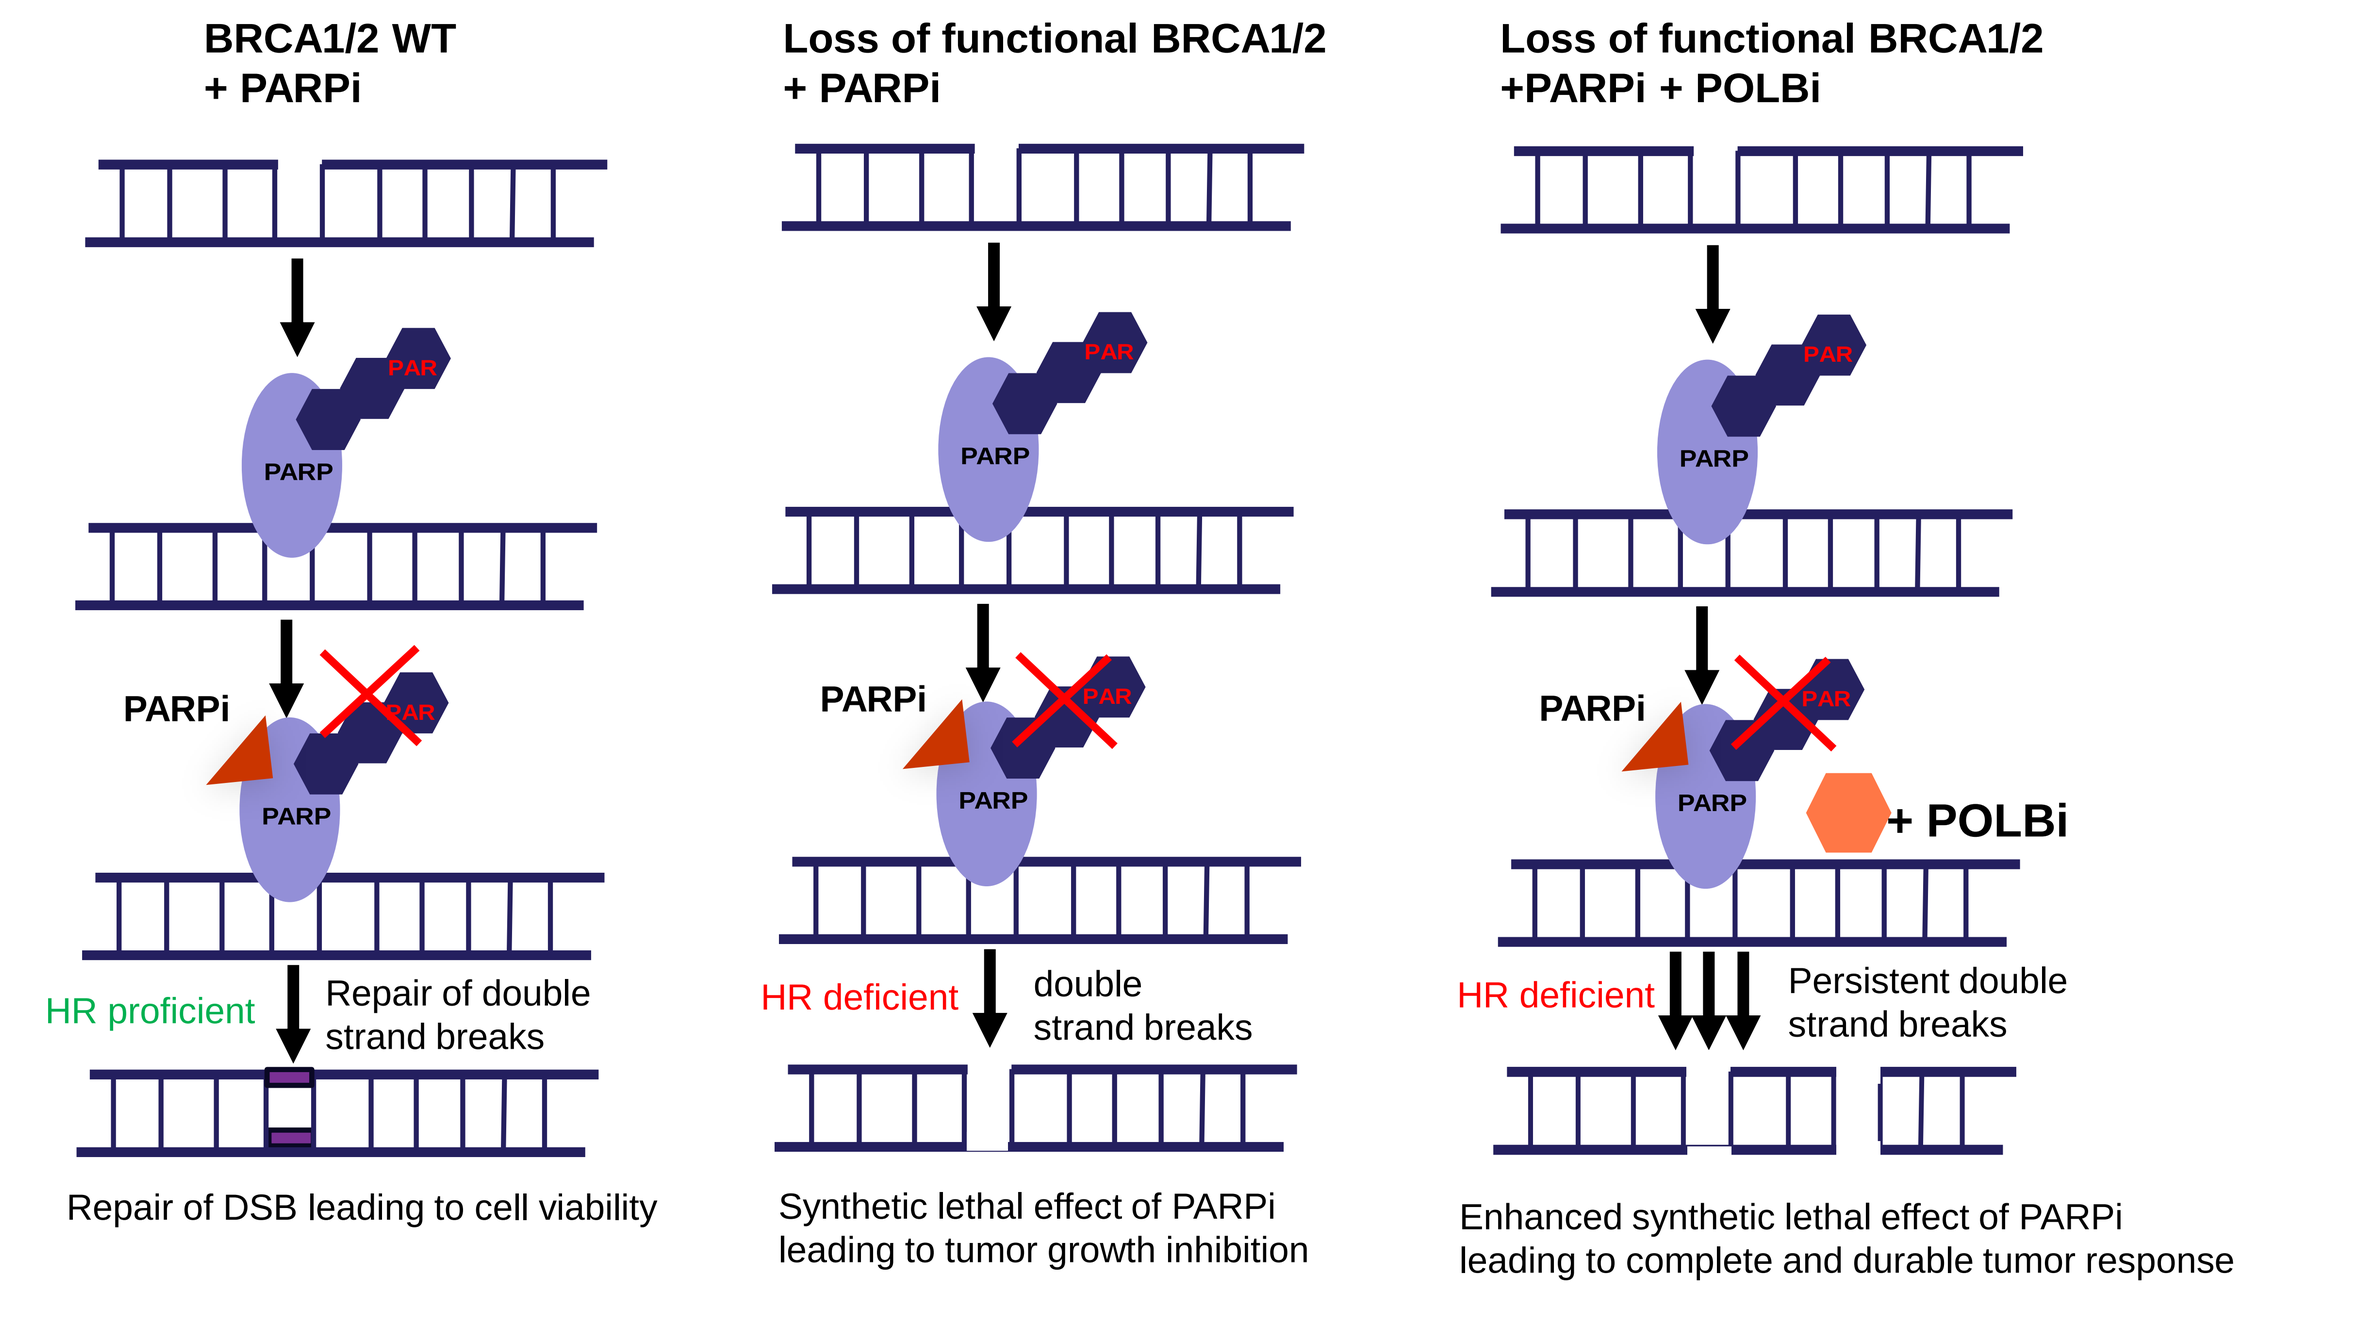
BRCA1/2 WT + PARPi
Loss of functional BRCA1/2 + PARPi
Loss of functional BRCA1/2 +PARPi + POLBi
PARP
PAR
PARPi
+ POLBi
HR proficient
Repair of double strand breaks
HR deficient
double strand breaks
Persistent double strand breaks
Repair of DSB leading to cell viability
Synthetic lethal effect of PARPi leading to tumor growth inhibition
Enhanced synthetic lethal effect of PARPi leading to complete and durable tumor response

ACKNOWLEDGEMENTS

The authors gratefully acknowledge the generous contributions from teams at Biortus, ChemPartner, WuXi AppTec, and intoDNA

REFERENCES

1. Batra, V. K. et al. Magnesium-induced assembly of a complete DNA polymerase catalytic complex. Structure. 14, 757-66 (2006).
2. Kordon, M. et al. STRIDE—a fluorescence method for direct, specific in situ detection of individual single- or double-strand DNA breaks in fixed cells. Nucleic Acids Research, Volume 48, Issue 3, 20 February 2020
3. Ryan, C. et al. Complex synthetic lethality in cancer. Nature Genetics, 55, 2039-2048 2023